FP129

Newly Administered Immunosuppressive Therapy (IST) Has No Impact on Long-term Antiproteinuric Effect of Sparsentan (SPAR), a Dual Angiotensin and Endothelin Receptor Antagonist, in Patients With Primary Focal Segmental Glomerulosclerosis (FSGS): Interim Analysis of the Phase 2 DUET Trial

Howard Trachtman,[1] Ivan Rychlik,[2] Robert Haws,[3] Carla Nester,[4] Alessia Fornoni,[5] Radko Komers[6]

[1]NYU Medical Center, New York, NY, USA; [2]3rd Medical School, Charles' University, Prague, Czech Republic; [3]Marshfield Clinic, Marshfield, WI, USA; [4]Stead Family Department of Pediatrics, University of Iowa, IA, USA; [5]University of Miami Health System, Miami, FL, USA; [6]Retrophin, Inc., San Diego, CA, USA

Introduction

- Sparsentan is a first-in-class, orally active, dual-acting, selective antagonist of the angiotensin II Type 1 receptor and the endothelin type A receptor in development for the treatment of primary focal segmental glomerulosclerosis (FSGS)[1]
- In the 8-week, double-blind (DB) period in the phase 2 DUET trial in patients with primary FSGS, sparsentan 200, 400, and 800 mg/d resulted in greater reduction in proteinuria compared with irbesartan 300 mg/d[2]
- All patients completing the DB period entered an open-label extension (OLE) and received sparsentan, regardless of treatment in the DB period
- In an interim analysis (data cutoff: June 9, 2016), sparsentan resulted in further decline in proteinuria over 48 weeks in all groups[3]
- Potential effects of initiation of new immunosuppressive therapy (IST) on proteinuria reduction with sparsentan are of interest

Objective

- To determine the potential impact of newly initiated IST on the effects of sparsentan treatment during the OLE in the DUET study

Methods

Study Design

- DUET (NCT01613118) is a phase 2, randomized, active-controlled study with an 8-week DB treatment period
- Patients were allocated to 200, 400, and 800 mg/d sparsentan cohorts and randomized within these cohorts to receive DB treatment with sparsentan or active control (irbesartan 300 mg/d)
- All patients completing DB treatment continued in an OLE and received only sparsentan for an additional 144 weeks at the same dose as in the original cohort
 - During the OLE, dose adjustments were allowed for safety or efficacy
- Initiation of new IST was not permitted during the 8-week DB period but was allowed during the OLE

Figure 1. DUET study design, selected inclusion criteria, and dosing

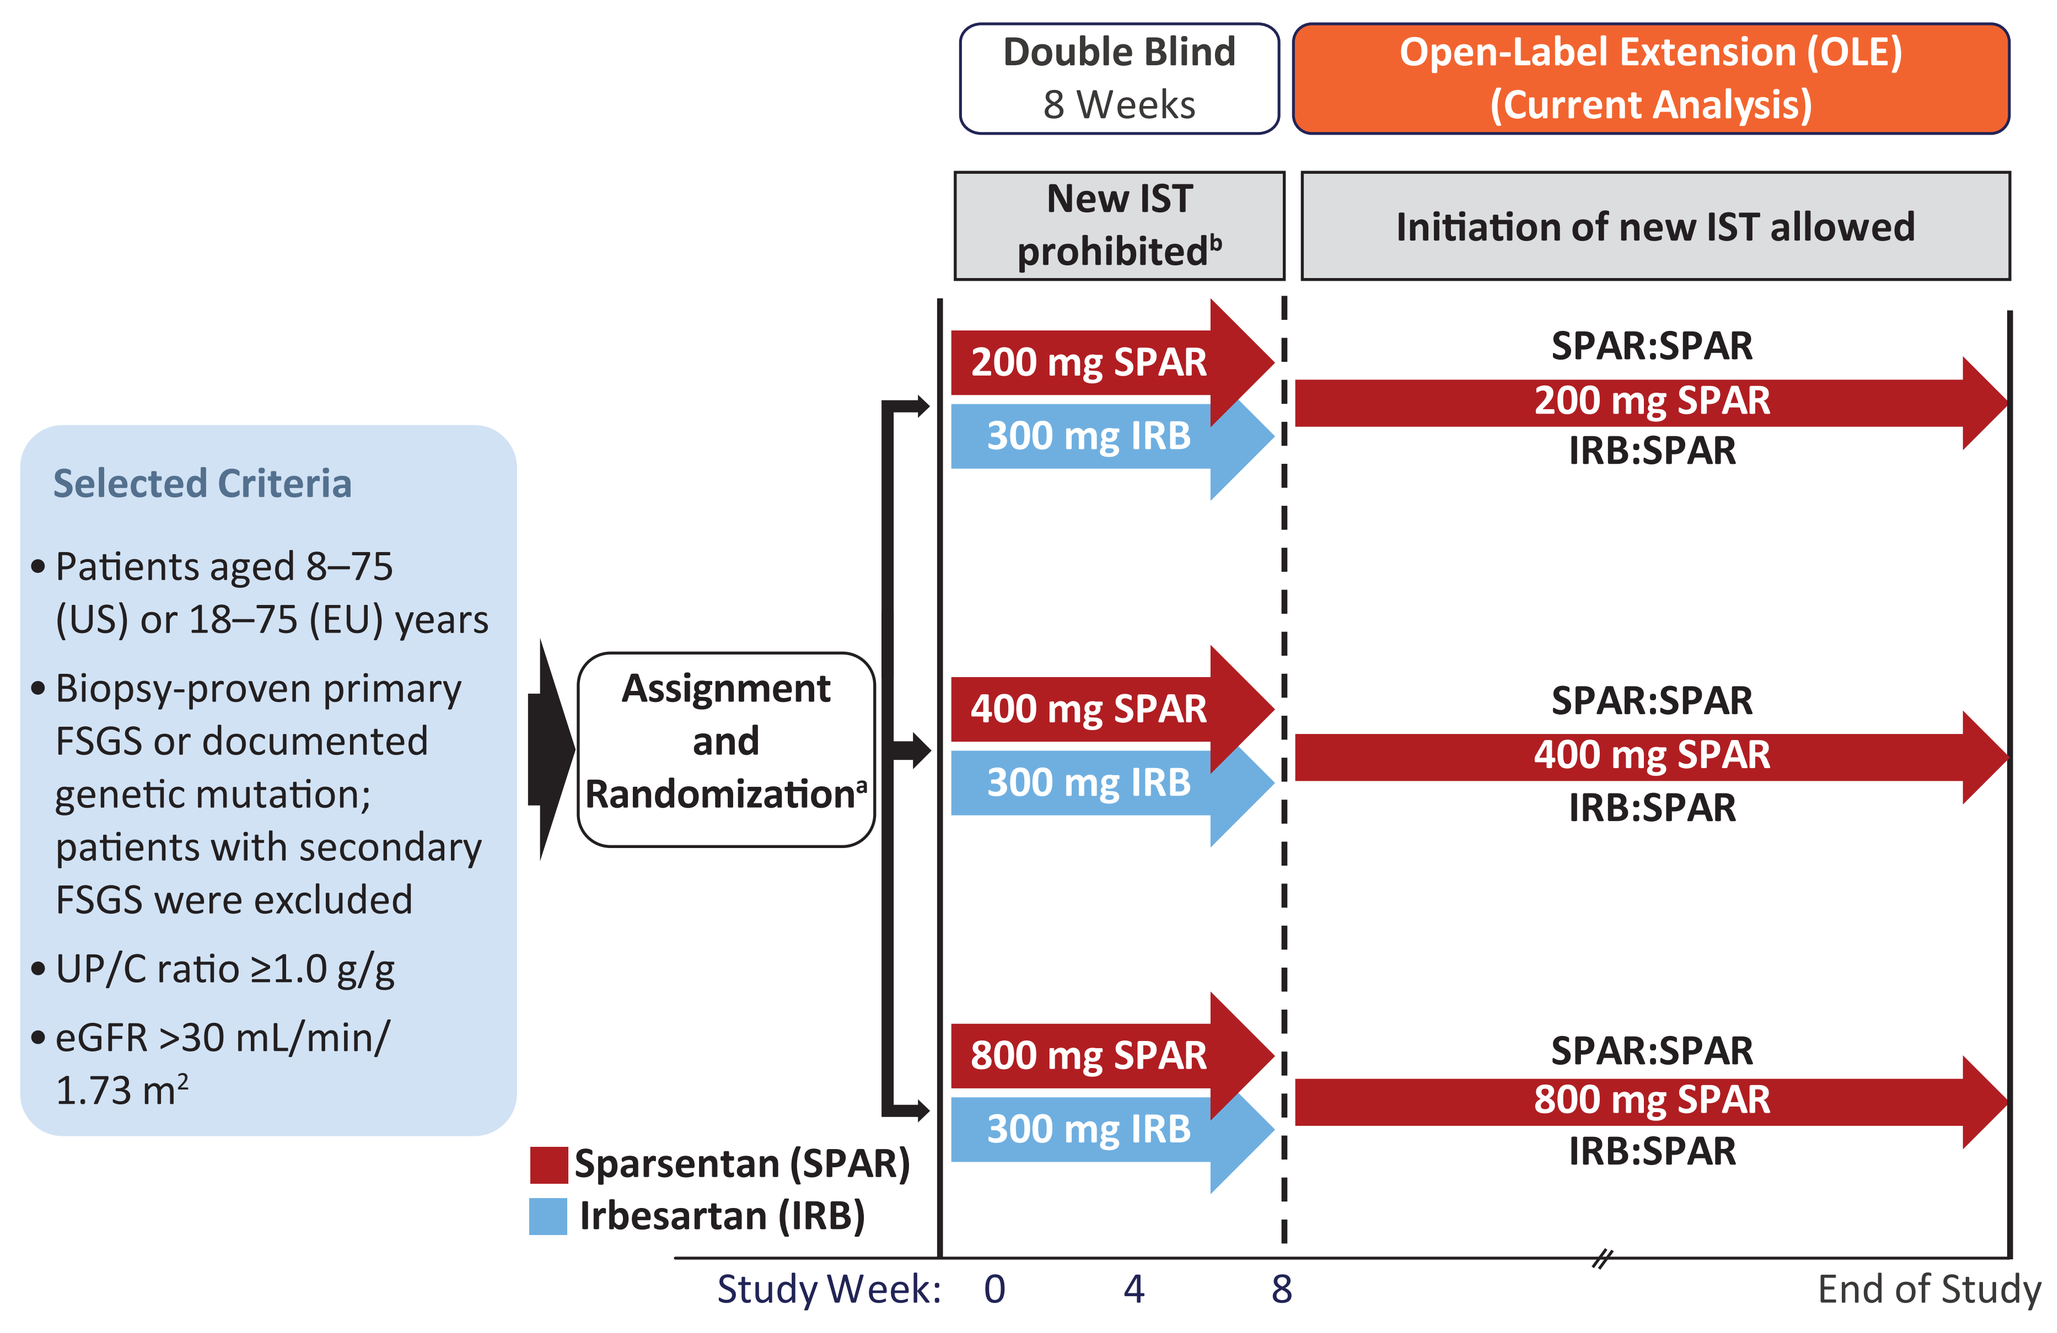

Double Blind
8 Weeks
Open-Label Extension (OLE)
(Current Analysis)
New IST prohibited[b]
Initiation of new IST allowed
Selected Criteria
• Patients aged 8–75 (US) or 18–75 (EU) years
• Biopsy-proven primary FSGS or documented genetic mutation; patients with secondary FSGS were excluded
• UP/C ratio ≥1.0 g/g
• eGFR >30 mL/min/1.73 m²
Assignment and Randomization[a]
200 mg SPAR
300 mg IRB
SPAR:SPAR
200 mg SPAR
IRB:SPAR
400 mg SPAR
300 mg IRB
SPAR:SPAR
400 mg SPAR
IRB:SPAR
800 mg SPAR
300 mg IRB
SPAR:SPAR
800 mg SPAR
IRB:SPAR
Sparsentan (SPAR)
Irbesartan (IRB)
Study Week: 0 4 8
End of Study

Note: Patients were assigned to dose cohort, then randomized to receive sparsentan or irbesartan within the dose cohort. Study drug was administered orally, once daily. Patients weighing ≤50 kg received half the assigned daily dose of sparsentan or irbesartan.
[a]After 2 weeks of RASI washout.
[b]If a patient was taking ISTs (except rituximab or cyclophosphamide), the dose and/or levels must have been stable for 1 month before randomization and the investigator should not have had plans to alter the regimen during the first 8 weeks of treatment, except to stabilize levels.
DB = double-blind; eGFR = estimated glomerular filtration rate; FSGS = focal segmental glomerulosclerosis; IRB:SPAR = patients randomized to irbesartan who then transitioned to sparsentan in the OLE; IST, immunosuppressive therapy; RASI = renin-angiotensin system inhibitor; SPAR:SPAR = patients randomized to sparsentan who also received sparsentan in the OLE; UP/C = urinary protein-to-creatinine ratio.

Assessments

- Urinary protein-to-creatinine ratio (UP/C), estimated glomerular filtration rate (eGFR), and blood pressure (BP) over time (interim analysis through 48 weeks)
- FSGS partial remission endpoint (FPRE),[4] defined as the proportion of patients who achieved UP/C ≤1.5 g/g and a >40% reduction in UP/C, at each time point

Statistical Analysis

- This interim analysis of the OLE included follow-up through 48 weeks
- During the 8-week, DB period, the full analysis set was defined as all randomized patients who received ≥1 dose of study drug and had ≥1 post-baseline efficacy evaluation; the efficacy evaluable set included all patients who received ≥1 dose of study drug and had both baseline and Week 8 UP/C measurements
- During the OLE, the full analysis set included all patients who received open-label sparsentan and had any efficacy assessment in the OLE
- Baseline was defined as Week 0 for patients receiving SPAR:SPAR (ie, patients receiving sparsentan in the DB period and OLE), and Week 8 for patients receiving IRB:SPAR (ie, patients receiving irbesartan in the DB period and transitioning to sparsentan in the OLE)
- Changes from baseline in UP/C, eGFR, and BP were assessed through descriptive statistics
 - Statistical significance for mean change from baseline was obtained via evaluation of 95% confidence intervals (CIs); P<0.05 if the 95% CI excluded zero
- The analysis was repeated after truncating data from patients who received IST during the OLE
 - Only those measurements that followed the initiation of IST were excluded

Results

Figure 2. Patient disposition at interim data cutoff

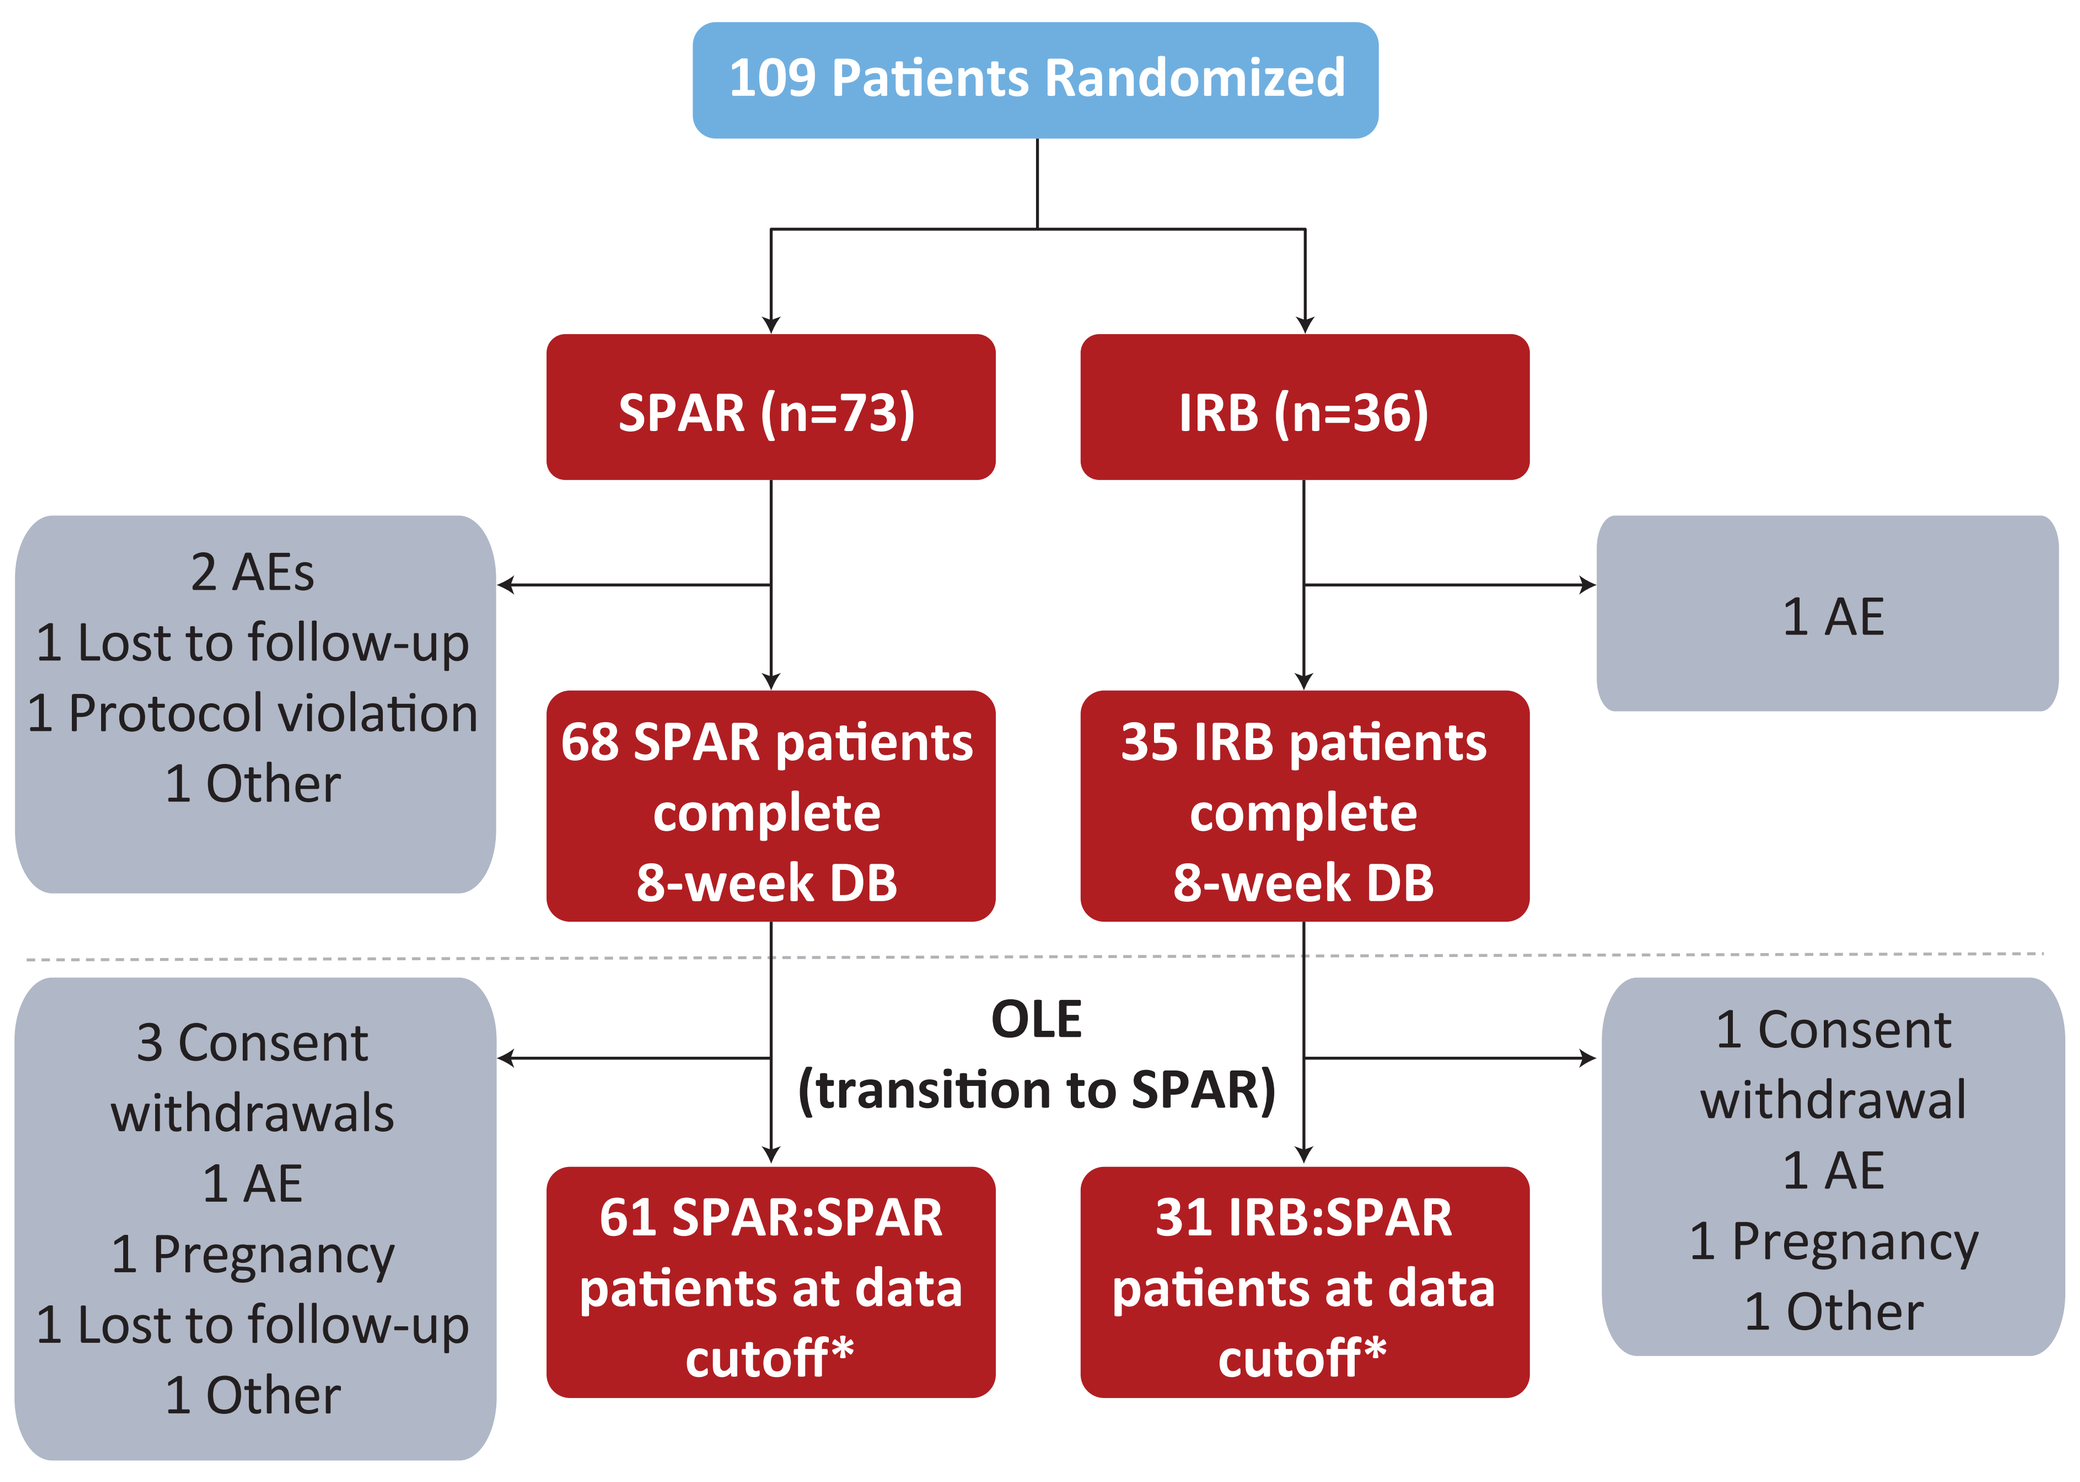

109 Patients Randomized
SPAR (n=73)
IRB (n=36)
2 AEs
1 Lost to follow-up
1 Protocol violation
1 Other
1 AE
68 SPAR patients complete 8-week DB
35 IRB patients complete 8-week DB
OLE
(transition to SPAR)
3 Consent withdrawals
1 AE
1 Pregnancy
1 Lost to follow-up
1 Other
1 Consent withdrawal
1 AE
1 Pregnancy
1 Other
61 SPAR:SPAR patients at data cutoff*
31 IRB:SPAR patients at data cutoff*

*As of June 9, 2016.
AE = adverse event; DB = double-blind; IRB:SPAR = patients randomized to irbesartan who then transitioned to sparsentan in the OLE; OLE = open-label extension; SPAR:SPAR = patients randomized to sparsentan who also received sparsentan in the OLE.

Table 1. Patient demographics and baseline characteristics

Characteristics	Sparsentan, All Doses (n=73)	Irbesartan (n=36)
Age group, n (%)		
Pediatric (aged 8–18 y)	13 (18)	10 (28)
Adult (aged 19–75 y)	60 (82)	26 (72)
Sex, n (%)		
Female	32 (44)	17 (47)
Male	41 (56)	19 (53)
Race, n (%)		
Asian	5 (7)	1 (3)
Black	8 (11)	7 (19)
White	57 (78)	26 (72)
Other	3 (4)	2 (6)
Ethnicity, n (%)		
Hispanic/Latino	14 (19)	6 (17)
Non-Hispanic/Non-Latino	59 (81)	30 (83)
BMI, kg/m², mean (SD)	28.4 (6.1)	28.7 (6.4)
IST at baseline, n (%)	21 (29)	13 (36)
eGFR, mL/min/1.73 m², mean (SD)	74.6 (37.6)	73.1 (41.3)
UP/C ratio, g/g, median (range)	3.61 (0.4–18.7)	3.12 (0.9–10.7)
ACE inhibitor or ARB use before washout, n (%)	59 (81)	32 (89)
Use of ≥1 diuretic/antihypertensive agent, n (%)	40 (55)	20 (56)
Diuretics	26 (36)	9 (25)
Additional antihypertensive treatments	29 (40)	16 (44)

Based on the full analysis set.
ACE = angiotensin-converting enzyme; ARB = angiotensin II receptor blocker; BMI = body mass index; eGFR = estimated glomerular filtration rate; IST = immunosuppressive therapy; SD = standard deviation; UP/C = urinary protein-to-creatinine ratio (24-hour urine).

Table 2. Patients with new IST at or before a given timepoint

Study Period	Study Week	Patients With New IST,[a] n (%) SPAR:SPAR (n=50)[b]	IRB:SPAR (n=33)[b]
DB[c]	0–8	—	—
OLE	16	3 (6)	1 (3)
	24	4 (8)	1 (3)
	36	4 (8)	2 (6)
	48	6 (12)	2 (6)

[a]IST medications that were newly initiated during the OLE were hydrocortisone, methylprednisolone, mycophenolate mofetil, prednisone, and tacrolimus.
[b]Includes all patients who received open-label sparsentan and had any efficacy assessments after receiving open-label sparsentan.
[c]Initiation of new IST was not permitted during the 8-week DB period.
DB = double blind; IRB:SPAR = patients randomized to irbesartan who then transitioned to sparsentan in the OLE; IST = immunosuppressive therapy; OLE = open-label extension; SPAR:SPAR = patients randomized to sparsentan who also received sparsentan in the OLE.

Figure 3. UP/C over 48 weeks

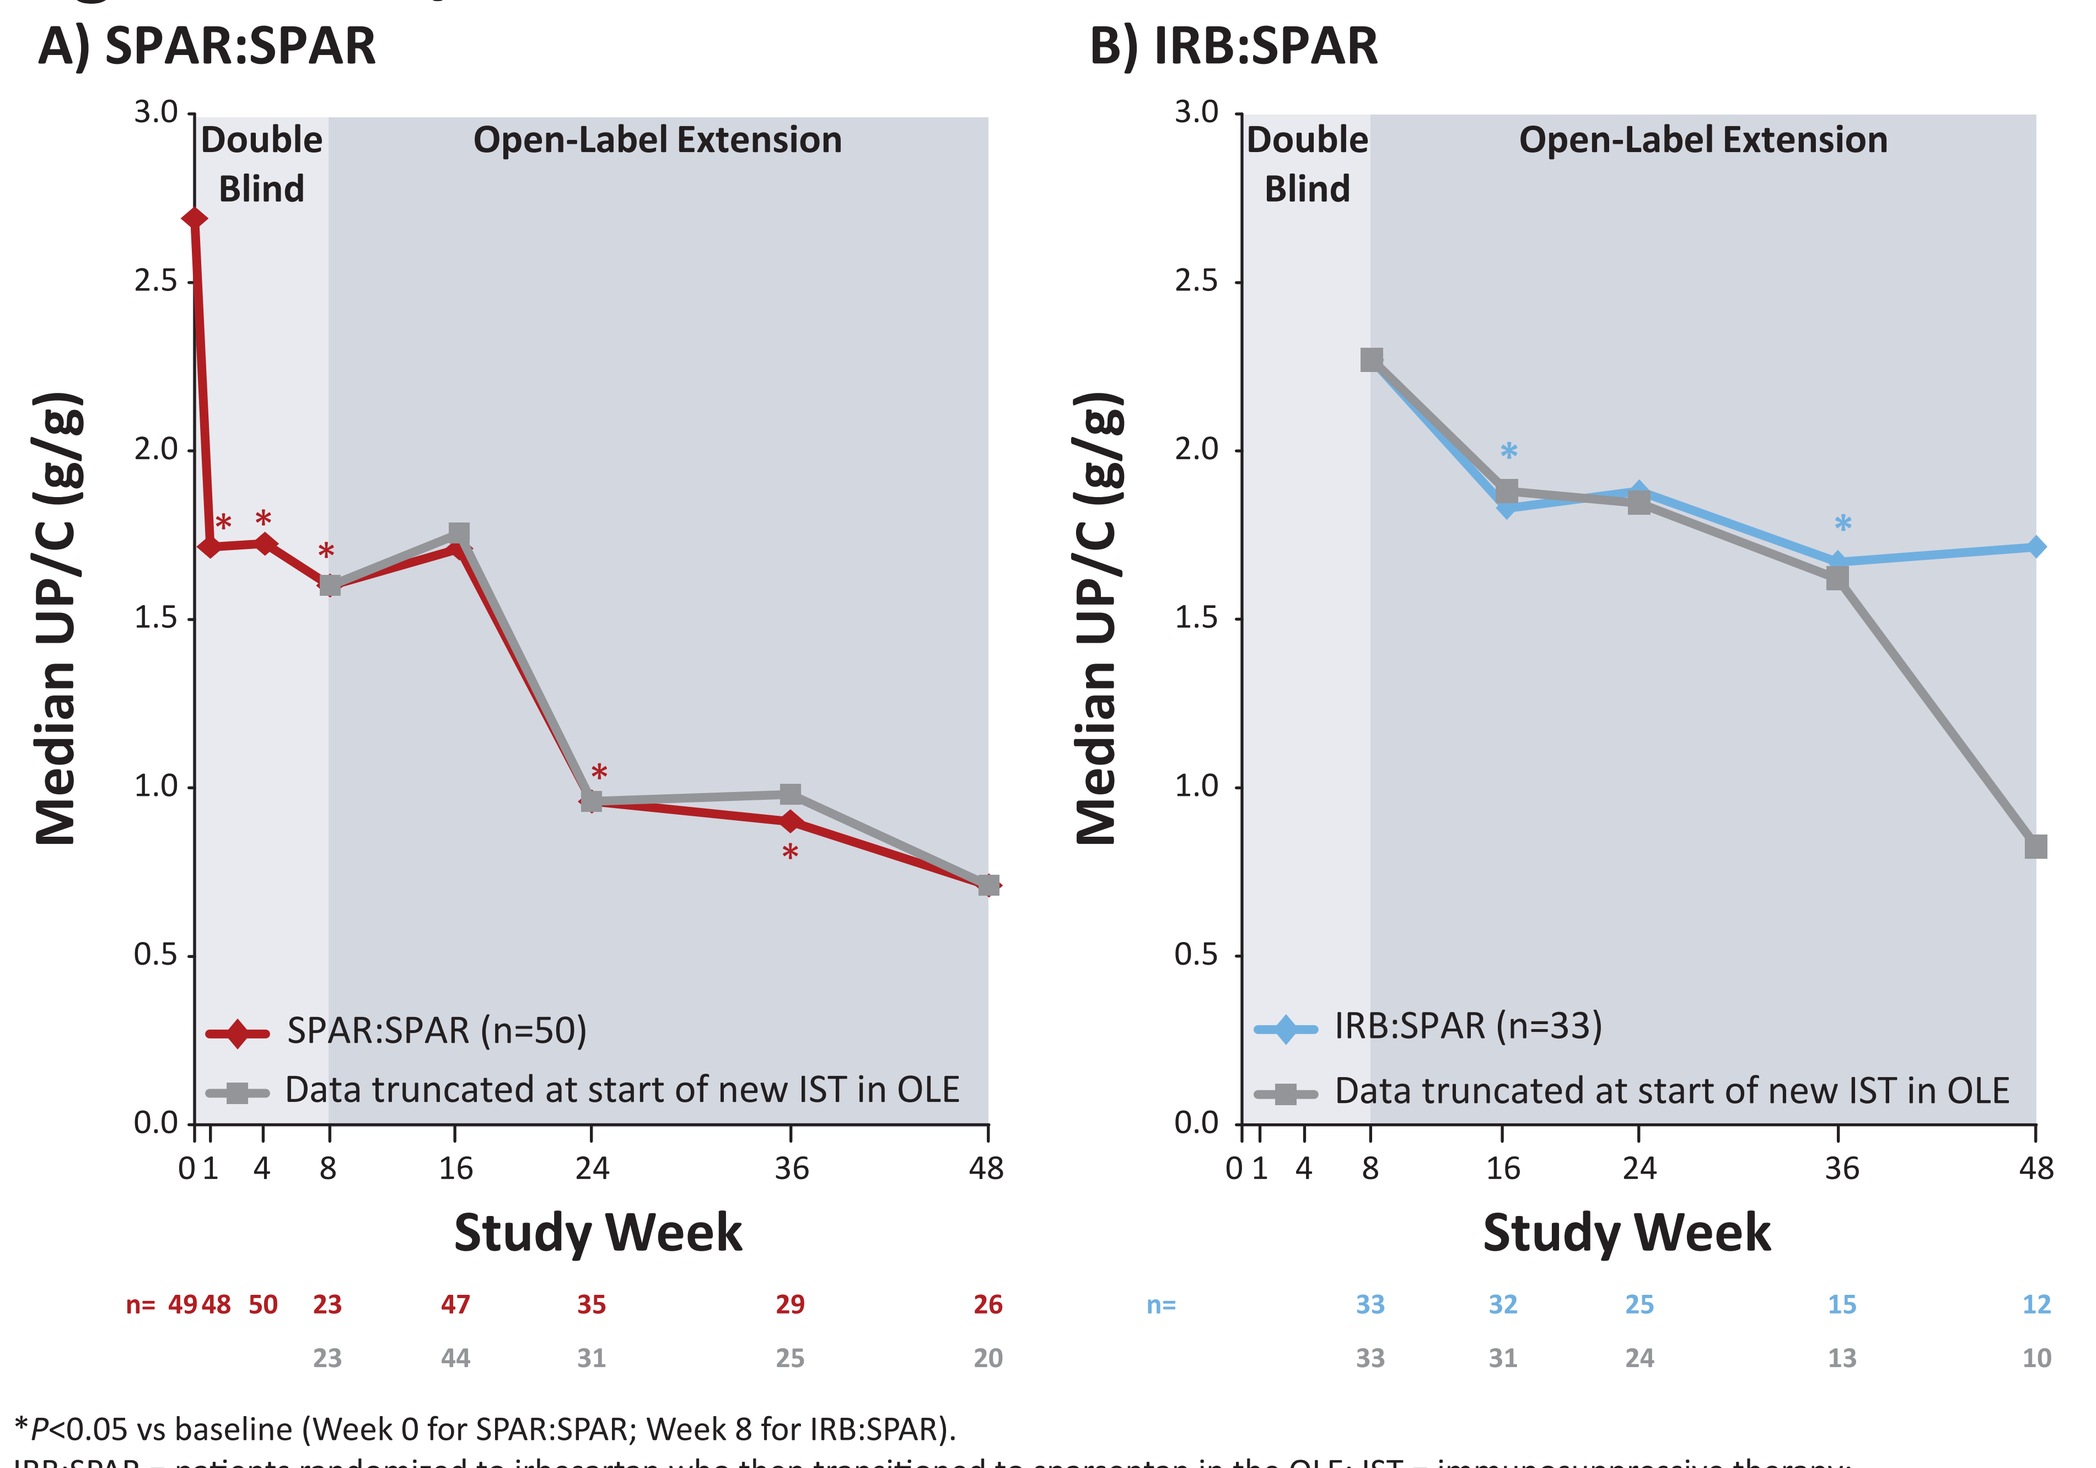

A) SPAR:SPAR
B) IRB:SPAR
Double Blind
Open-Label Extension
Median UP/C (g/g)
SPAR:SPAR (n=50)
IRB:SPAR (n=33)
Data truncated at start of new IST in OLE
Study Week
n= 49 48 50 23 47 35 29 26
23 44 31 25 20
n= 33 32 25 15 12
33 31 24 13 10

*P<0.05 vs baseline (Week 0 for SPAR:SPAR; Week 8 for IRB:SPAR).
IRB:SPAR = patients randomized to irbesartan who then transitioned to sparsentan in the OLE; IST = immunosuppressive therapy; OLE = open-label extension; SPAR:SPAR = patients randomized to sparsentan who also received sparsentan in the OLE; UP/C = urinary protein-to-creatinine ratio (first morning void).

Figure 4. FPRE over 48 weeks

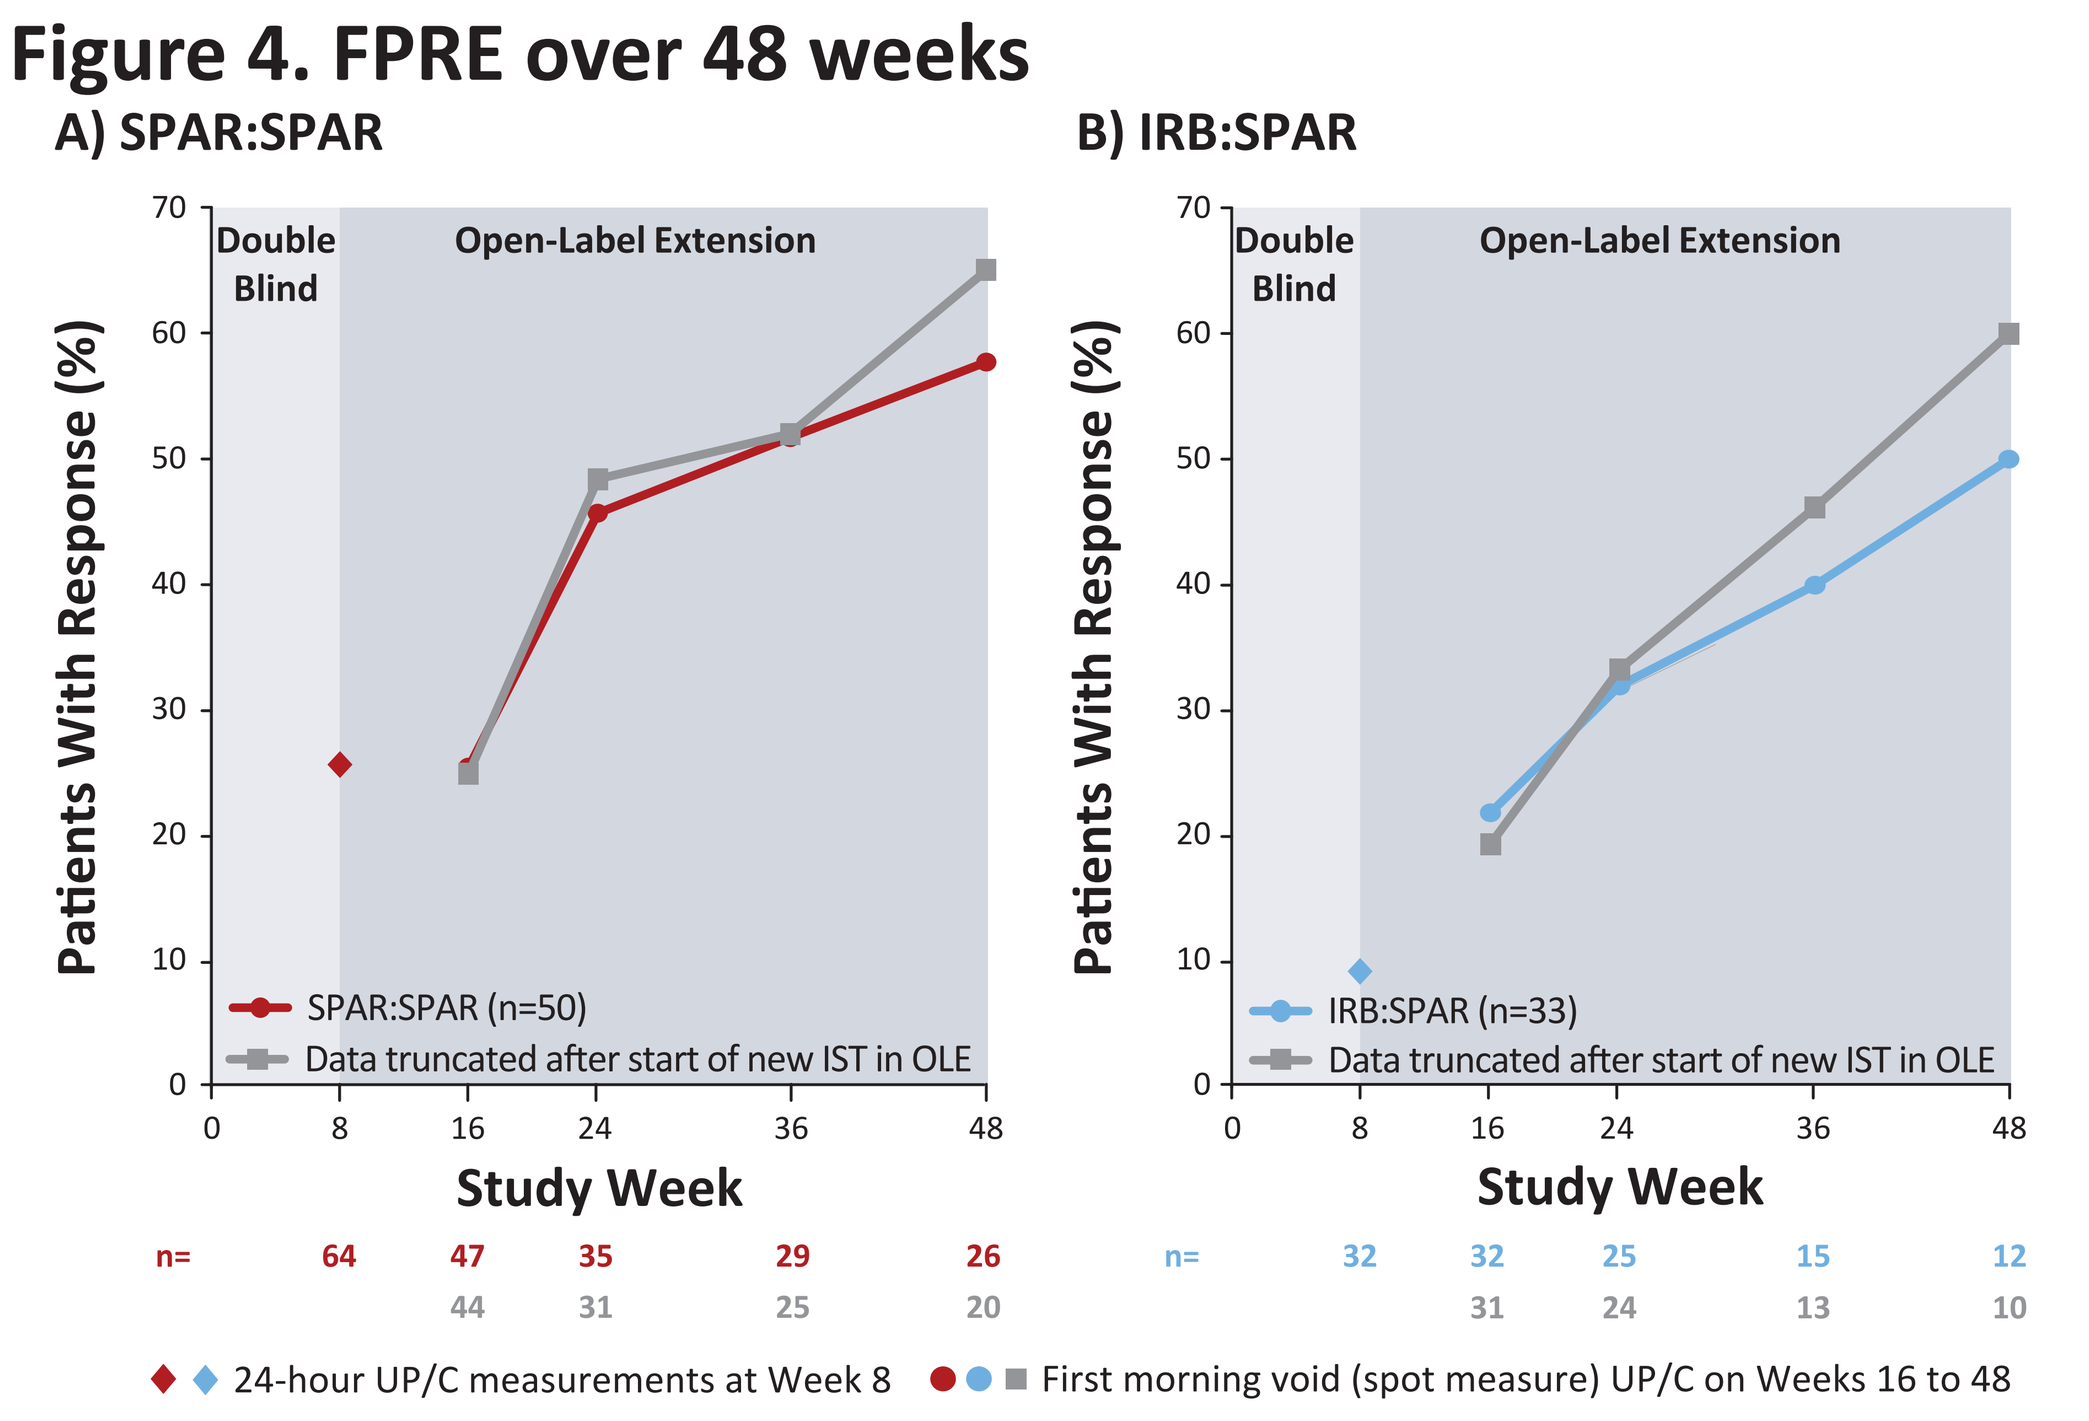

A) SPAR:SPAR
B) IRB:SPAR
Double Blind
Open-Label Extension
Patients With Response (%)
SPAR:SPAR (n=50)
IRB:SPAR (n=33)
Data truncated after start of new IST in OLE
Study Week
n= 64 47 35 29 26
44 31 25 20
n= 32 32 25 15 12
31 24 13 10
24-hour UP/C measurements at Week 8
First morning void (spot measure) UP/C on Weeks 16 to 48

FPRE is defined as UP/C ≤1.5 g/g and >40% reduction in UP/C from baseline. Baseline in the double-blind period defined as Week 0; baseline for the open-label period defined as last observation before start of open-label sparsentan treatment (ie, Week 8). Data for Week 8 is based on the efficacy evaluable set. Data for Weeks 16–48 are based on the full analysis set.
FPRE = FSGS partial remission endpoint; IRB:SPAR = patients randomized to irbesartan who then transitioned to sparsentan in the OLE; IST = immunosuppressive therapy; OLE = open-label extension; SPAR:SPAR = patients randomized to sparsentan who also received sparsentan in the OLE; UP/C = urinary protein-to-creatinine ratio.

Figure 5. eGFR over 48 weeks

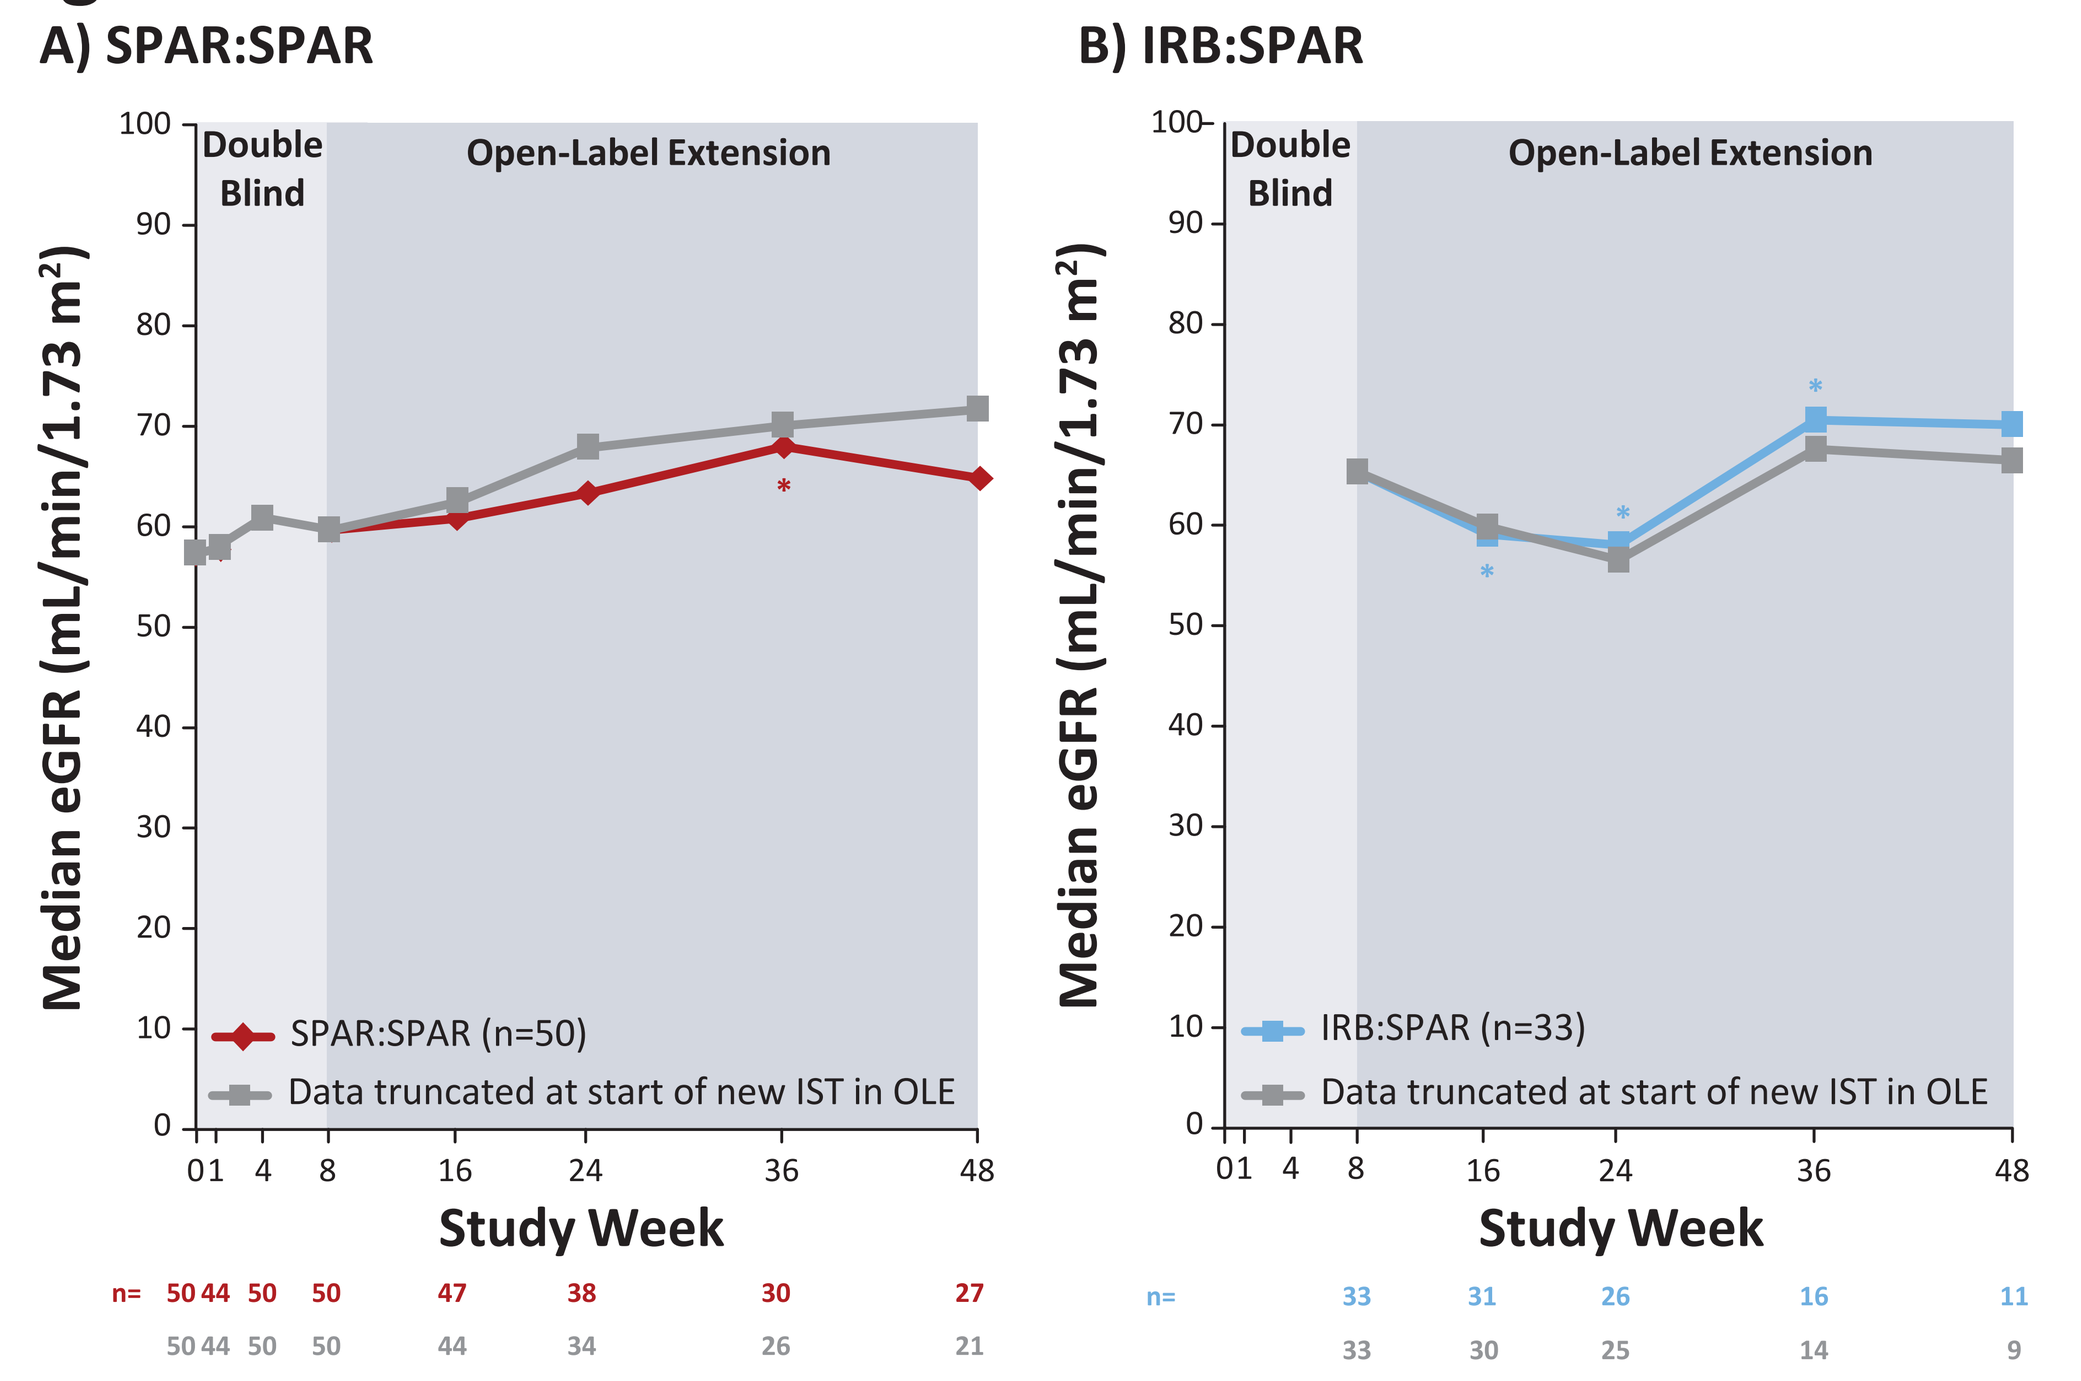

A) SPAR:SPAR
B) IRB:SPAR
Double Blind
Open-Label Extension
Median eGFR (mL/min/1.73 m²)
SPAR:SPAR (n=50)
IRB:SPAR (n=33)
Data truncated at start of new IST in OLE
Study Week
n= 50 44 50 50 47 38 30 27
50 44 50 50 44 34 26 21
n= 33 31 26 16 11
33 30 25 14 9

*P<0.05 vs baseline (Week 0 for SPAR:SPAR and Week 8 for IRB:SPAR).
Based on the full analysis set.
eGFR = estimated glomerular filtration rate; IRB:SPAR = patients randomized to irbesartan who then transitioned to sparsentan in the OLE; IST = immunosuppressive therapy; OLE = open-label extension; SPAR:SPAR = patients randomized to sparsentan who also received sparsentan in the OLE.

Figure 6. Blood pressure over 48 weeks

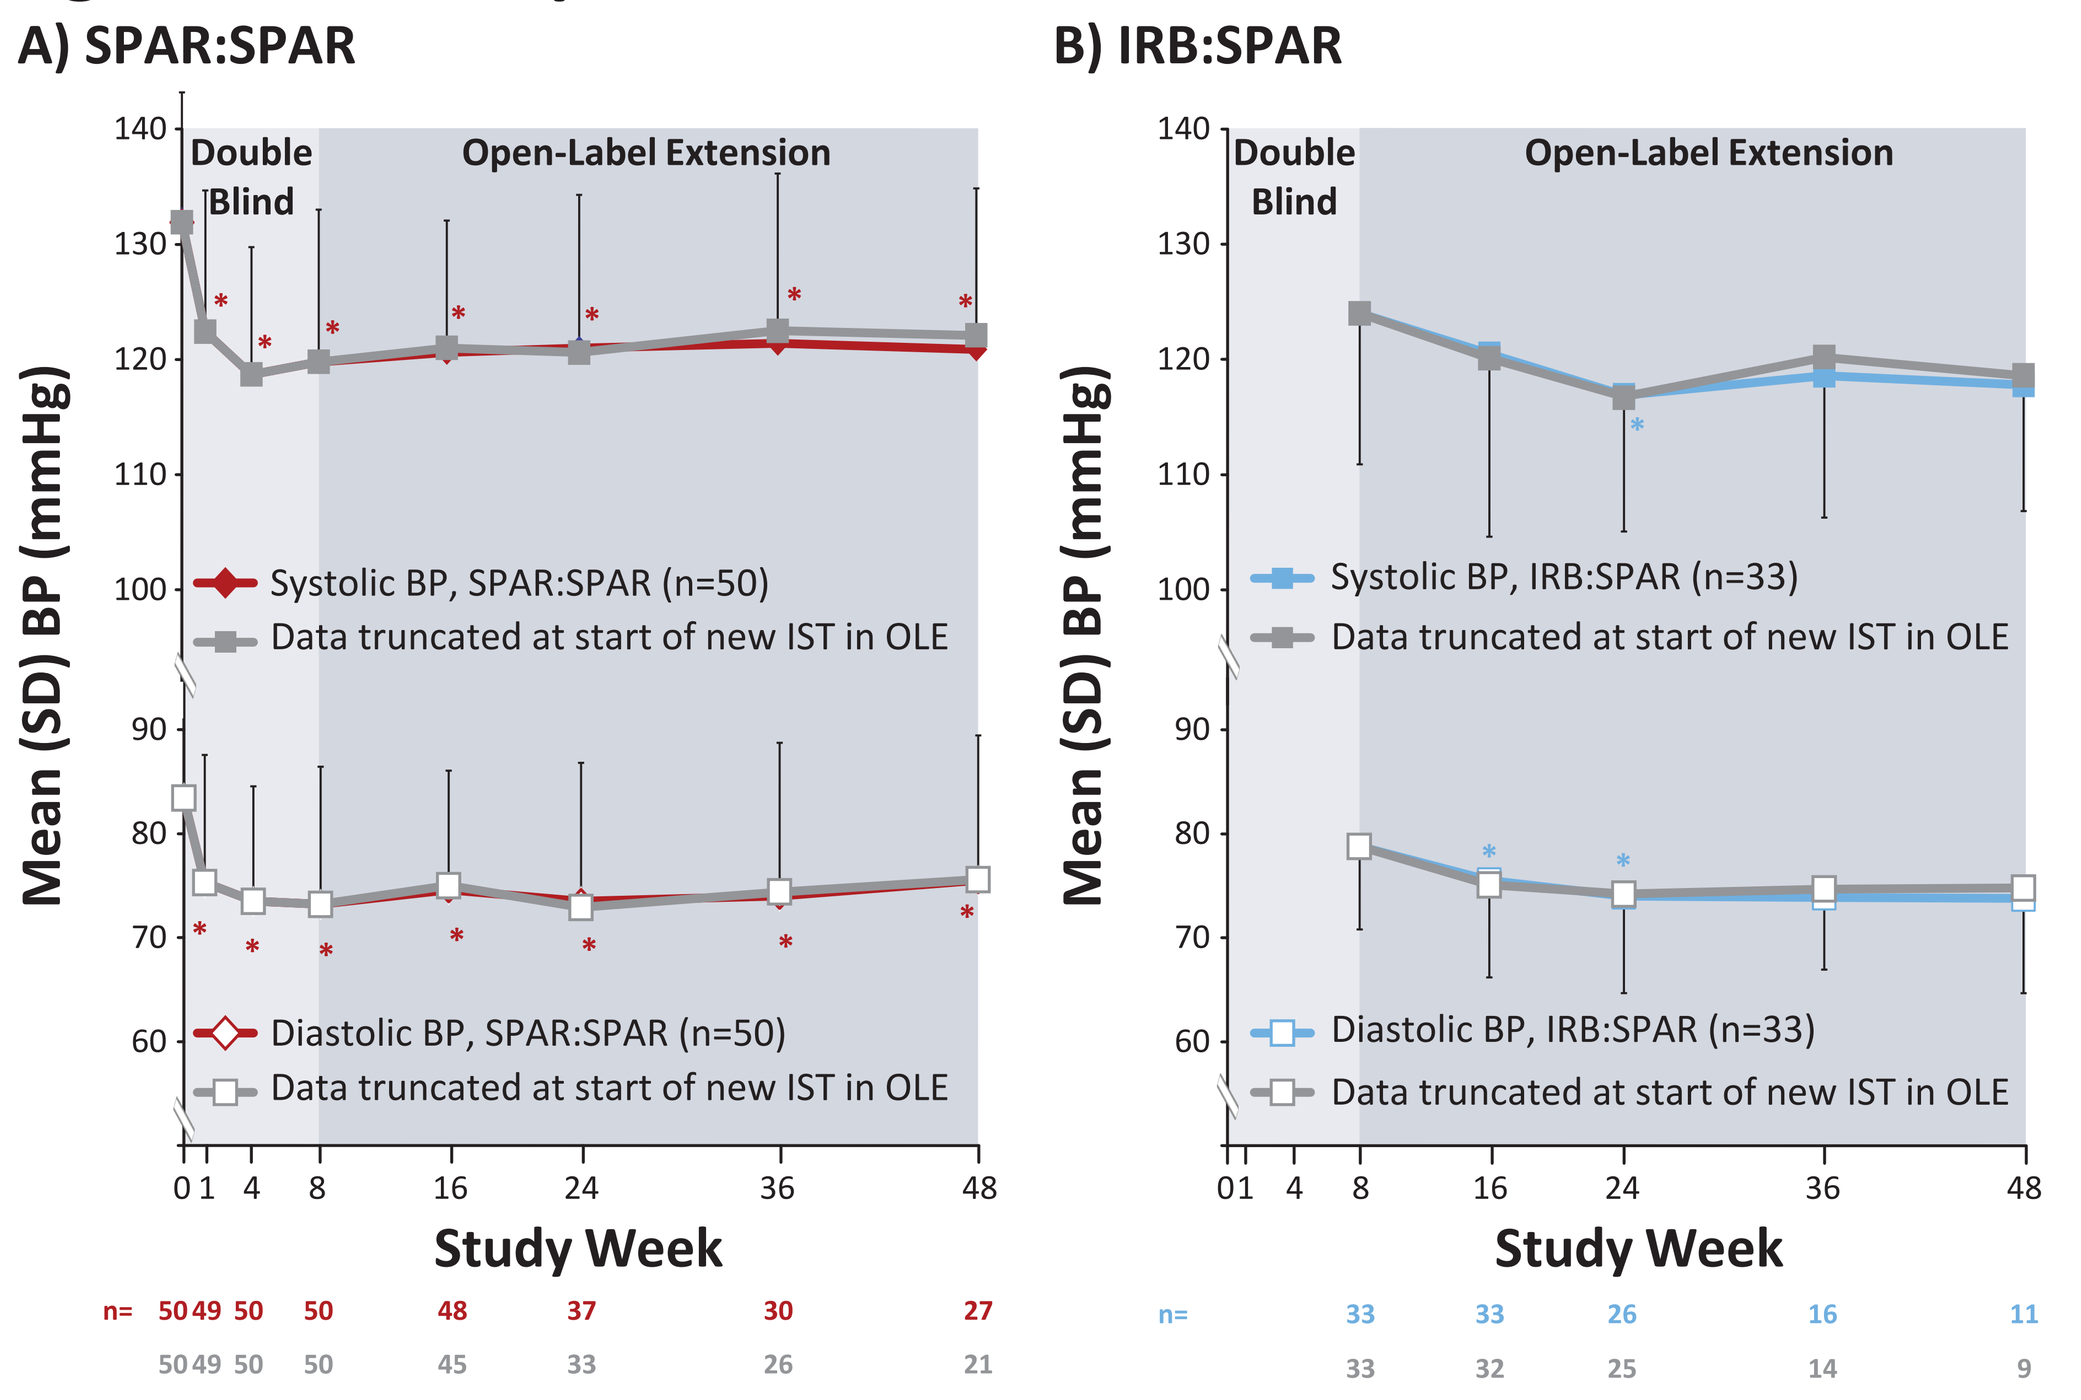

A) SPAR:SPAR
B) IRB:SPAR
Double Blind
Open-Label Extension
Mean (SD) BP (mmHg)
Systolic BP, SPAR:SPAR (n=50)
Systolic BP, IRB:SPAR (n=33)
Diastolic BP, SPAR:SPAR (n=50)
Diastolic BP, IRB:SPAR (n=33)
Data truncated at start of new IST in OLE
Study Week
n= 50 49 50 50 48 37 30 27
50 49 50 50 45 33 26 21
n= 33 33 26 16 11
33 32 25 14 9

*P<0.05 vs baseline (Week 0 for SPAR:SPAR; Week 8 for IRB:SPAR).
Based on the full analysis set. One-sided error bars represent 1 SD.
BP = blood pressure; IRB:SPAR = patients randomized to irbesartan who then transitioned to sparsentan in the OLE; IST = immunosuppressive therapy; OLE = open-label extension; SPAR:SPAR = patients randomized to sparsentan who also received sparsentan in the OLE; SD = standard deviation.

Summary

Original Results

- Patients treated with sparsentan achieved progressive reduction in UP/C over 48 weeks in the OLE of the DUET study
- Transition from irbesartan to sparsentan in the OLE led to further reduction in UP/C
- eGFR remained stable during this follow-up period
- In the SPAR:SPAR group, BP declined initially and remained stable for the remainder of the follow-up period; in the IRB:SPAR group, transition to sparsentan resulted in further reduction in BP

Assessing the Impact of New IST

- Few patients initiated new IST during the OLE
- Truncation of data after initiation of new IST yielded results that were similar to those from the original analysis
- Initiating new IST had no meaningful impact on the course of proteinuria, eGFR, and BP observed under long-term treatment with sparsentan

Conclusions

- Newly initiated IST had no meaningful impact on the effects of sparsentan observed in this interim analysis over 48 weeks of sparsentan treatment (data cutoff: June 9, 2016) in the DUET OLE

References

1. Komers R, Plotkin H. *Am J Physiol Regul Integr Comp Physiol*. 2016;310(10):R877-84.
2. Trachtman H, et al. *J Am Soc Nephrol*. 2016;27(suppl):2B.
3. Trachtman H, et al. *J Am Soc Nephrol*. 2017;28:43-4.
4. Troost JP, et al. *Clin J Am Soc Nephrol*. 2018;13(3):414-21.

Disclosures

HT: Consultancy fees from Kaneka Inc, Otsuka, and ChemoCentryx; consultant to Genzyme and Optherion, Inc. IR, RH, CN: No conflicts of interest to declare. AF: Vice President and CSO, L&F Health LLC. L&F Health LLC and affiliated companies – patent estate covering topics relevant to FSGS; consulting agreements with and/or honoraria from Hoffman-La Roche, Genentech, Mesoblast, Bristol-Myers Squibb, AbbVie, Janssen, Boehringer Ingelheim, AstraZeneca, Pfizer, ChemoCentryx, Dimerix, Mallinckrodt, and Variant Pharmaceuticals, Inc. Variant Pharmaceuticals – licensed worldwide rights to develop and commercialize hydroxypropyl beta cyclodextrin for treatment of kidney disease from L&F Research LLC. RK: Employee of Retrophin, Inc.; may have equity or other financial interest in Retrophin, Inc. Authors had full access to all data and take responsibility for its integrity and the accuracy of all analyses. Editorial support for this poster was provided by Kristen W. Quinn, PhD, of Peloton Advantage, LLC, and was funded by Retrophin, Inc.

Presented at the European Renal Association & European Dialysis and Transplant Association 55th Annual Congress (ERA-EDTA 2018), May 24–27, 2018, Copenhagen, Denmark