SUN-037

No Impact of Newly Initiated Immunosuppressive Therapy Observed on Long-Term Antiproteinuric Effect of Sparsentan in Focal Segmental Glomerulosclerosis: Interim 84-Week Analysis of the DUET Trial

Vladimír Tesař,[1] Howard Trachtman,[2] Edward Murphy,[3] Beatrice Ferguson,[3] Radko Komers[3]

[1]Charles University, General University Hospital, Prague, Czech Republic; [2]NYU School of Medicine, NYU Langone Health, New York, NY, USA; [3]Retrophin, Inc., San Diego, CA, USA.

Introduction

- Sparsentan, a first-in-class, orally active, dual-acting selective antagonist of the angiotensin II type 1 receptor and the endothelin type A receptor, is in development for the treatment of primary and genetic focal segmental glomerulosclerosis (FSGS)[1,2]
- In the ongoing DUET study in patients with FSGS, sparsentan (over the pooled 200, 400, and 800 mg/d dose range) resulted in greater reduction in proteinuria vs irbesartan 300 mg/d during an 8-week double-blind (DB) period[2]
- Sparsentan continues to be evaluated in an open-label extension (OLE) of the DUET study
- In an interim analysis (data cutoff: November 2017), treatment with sparsentan in the OLE resulted in sustained antiproteinuric effects and modest further declines in proteinuria over a total of 84 weeks[3]
- Potential effects of initiating new immunosuppressive therapy (IST) (which was permitted during the OLE) on reduction of proteinuria and preservation of kidney function in sparsentan-treated patients are of interest

Objective

- This was an interim, 84-week analysis to determine the potential impact of newly initiated IST on the antiproteinuric effects of sparsentan treatment in FSGS patients enrolled in the DUET study OLE

Methods

Study Design

- DUET (NCT01613118) is a phase 2, randomized, DB, active-controlled study
- Key DUET eligibility criteria: patients aged 8-75 years (US) or 18-75 years (EU) with biopsy-proven FSGS, baseline urinary protein-to-creatinine ratio (UP/C) ≥1 g/g, and estimated glomerular filtration rate (eGFR) >30 mL/min/1.73 m^2
- Patients were allocated to 200, 400, and 800 mg/d sparsentan cohorts and randomized within these cohorts to receive DB treatment with sparsentan or active control (irbesartan 300 mg/d)
- Patients who completed the 8-week DB treatment continued in an ongoing OLE (**Figure 1**). In the OLE, patients randomized to DB treatment with sparsentan continued sparsentan (SPAR:SPAR group) and those randomized to irbesartan were switched to sparsentan (IRB:SPAR group)
- Initiation of new IST (eg, steroids, alkylating agents, mycophenolate mofetil, calcineurin inhibitors, rituximab) was not allowed during the DB period but was permitted during the OLE

Figure 1. DUET study design, key inclusion criteria, and dosing

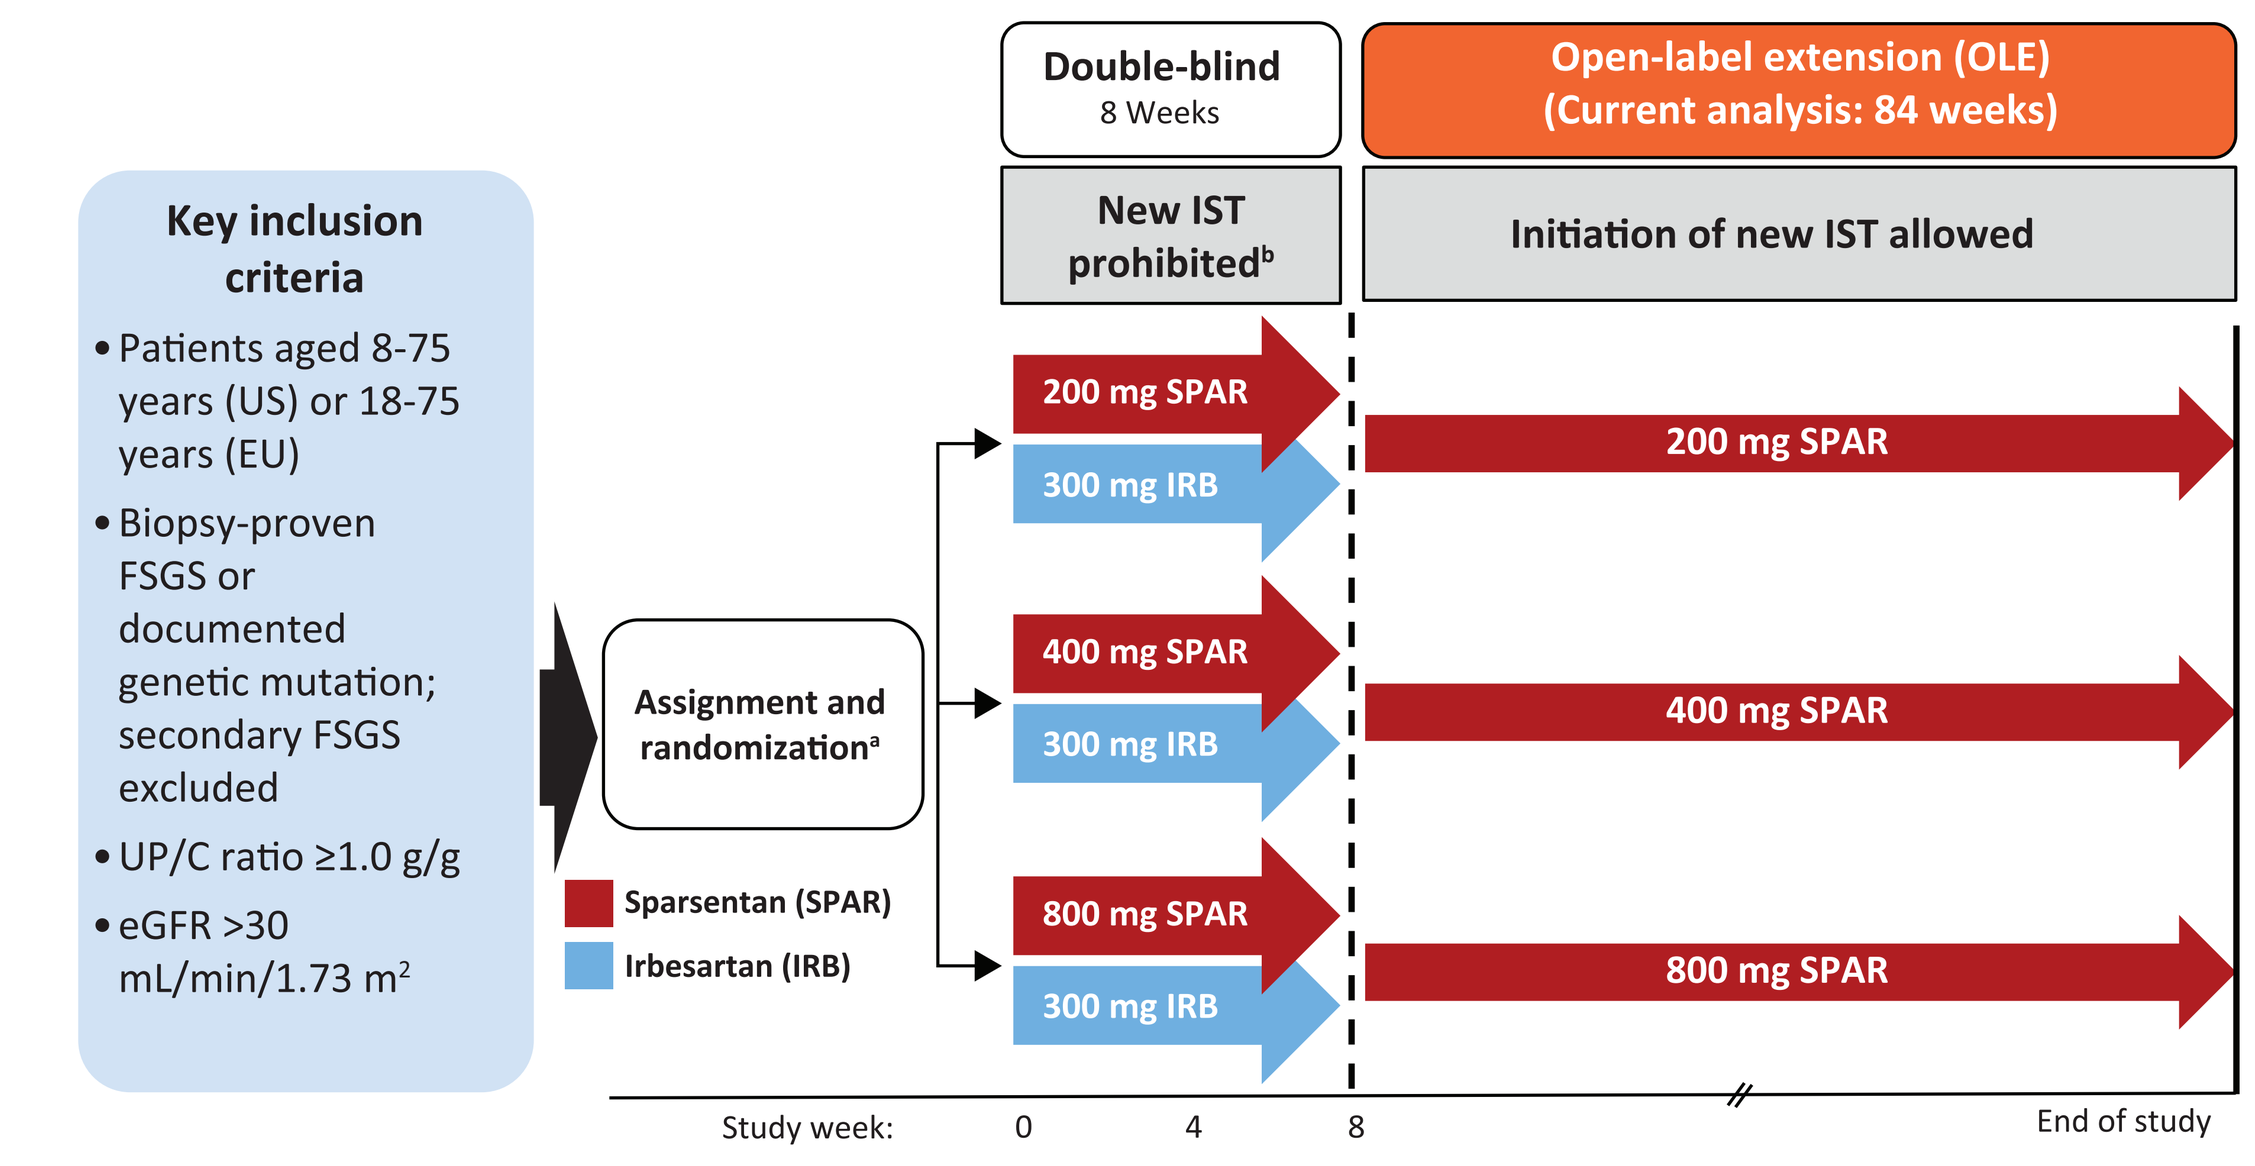
Double-blind
8 Weeks
Open-label extension (OLE)
(Current analysis: 84 weeks)
New IST prohibited[b]
Initiation of new IST allowed
Key inclusion criteria
• Patients aged 8-75 years (US) or 18-75 years (EU)
• Biopsy-proven FSGS or documented genetic mutation; secondary FSGS excluded
• UP/C ratio ≥1.0 g/g
• eGFR >30 mL/min/1.73 m2
Assignment and randomization[a]
Sparsentan (SPAR)
Irbesartan (IRB)
200 mg SPAR
300 mg IRB
400 mg SPAR
800 mg SPAR
Study week: 0 4 8
End of study

Note: Patients were assigned to dose cohort, then randomized to sparsentan or irbesartan within the dose cohort. Study drug was administered orally, once daily. Patients who weighed ≤50 kg received half the assigned daily dose of sparsentan or irbesartan.
[a]After 2 weeks of renin-angiotensin system inhibitor washout. [b]If a patient was taking ISTs (except rituximab or cyclophosphamide), the dose and/or levels must have been stable for 1 month prior to randomization and the investigator should not alter the regimen during the first 8 weeks of treatment, except to stabilize levels.
eGFR, estimated glomerular filtration rate; FSGS, focal segmental glomerulosclerosis; UP/C, urinary protein-to-creatinine ratio.

Study Measures

- UP/C, FSGS partial remission endpoint (FPRE; UP/C ≤1.5 g/g and >40% UP/C decrease from baseline),[4] eGFR, and blood pressure were measured every 12 weeks to Week 84 (interim analysis)
 - 73% of SPAR:SPAR and 74% of IRB:SPAR patients had UP/C >1.5 g/g at baseline. FPRE analysis did not exclude patients with baseline UP/C ≤1.5 g/g. In those patients, only the >40% reduction in UP/C from baseline criterion was applied.
- Treatment-emergent adverse events (TEAEs) were examined from first dose of sparsentan through Week 84

Data Analysis

- This interim analysis was conducted after 84 weeks of follow-up (data cutoff: November 2017)
- Data from all patients and from the remaining population after exclusion of data obtained from patients after initiation of new IST in the OLE were analyzed separately
 - Only those measurements that followed initiation of IST were excluded
- The full analysis set during OLE included all patients who received open-label sparsentan and had ≥1 efficacy assessment in OLE
- The safety analysis set during OLE included all patients who had ≥1 safety assessment after receiving open-label sparsentan
- Baseline was defined as Week 0 for patients receiving SPAR:SPAR (ie, patients receiving sparsentan in the DB period and OLE), and Week 8 for patients receiving IRB:SPAR (ie, patients receiving irbesartan in the DB period and transitioning to sparsentan in the OLE)
- Changes from baseline in UP/C, eGFR, and blood pressure were examined using descriptive statistics
 - Statistical significance of *P*<0.05 for mean change from baseline was found if 95% confidence intervals (CIs) excluded zero
 - UP/C analyses used geometric means and CI limits calculated by exponentiation of mean and CI limits from natural log transformed data
 - Change from baseline for each patient was calculated as natural log transformation of ratio of observed value at study week and baseline value. Mean and CI limits of ratio were back-transformed by exponentiation.

Results

Figure 2. Patient disposition at 84-week interim data cutoff[a]

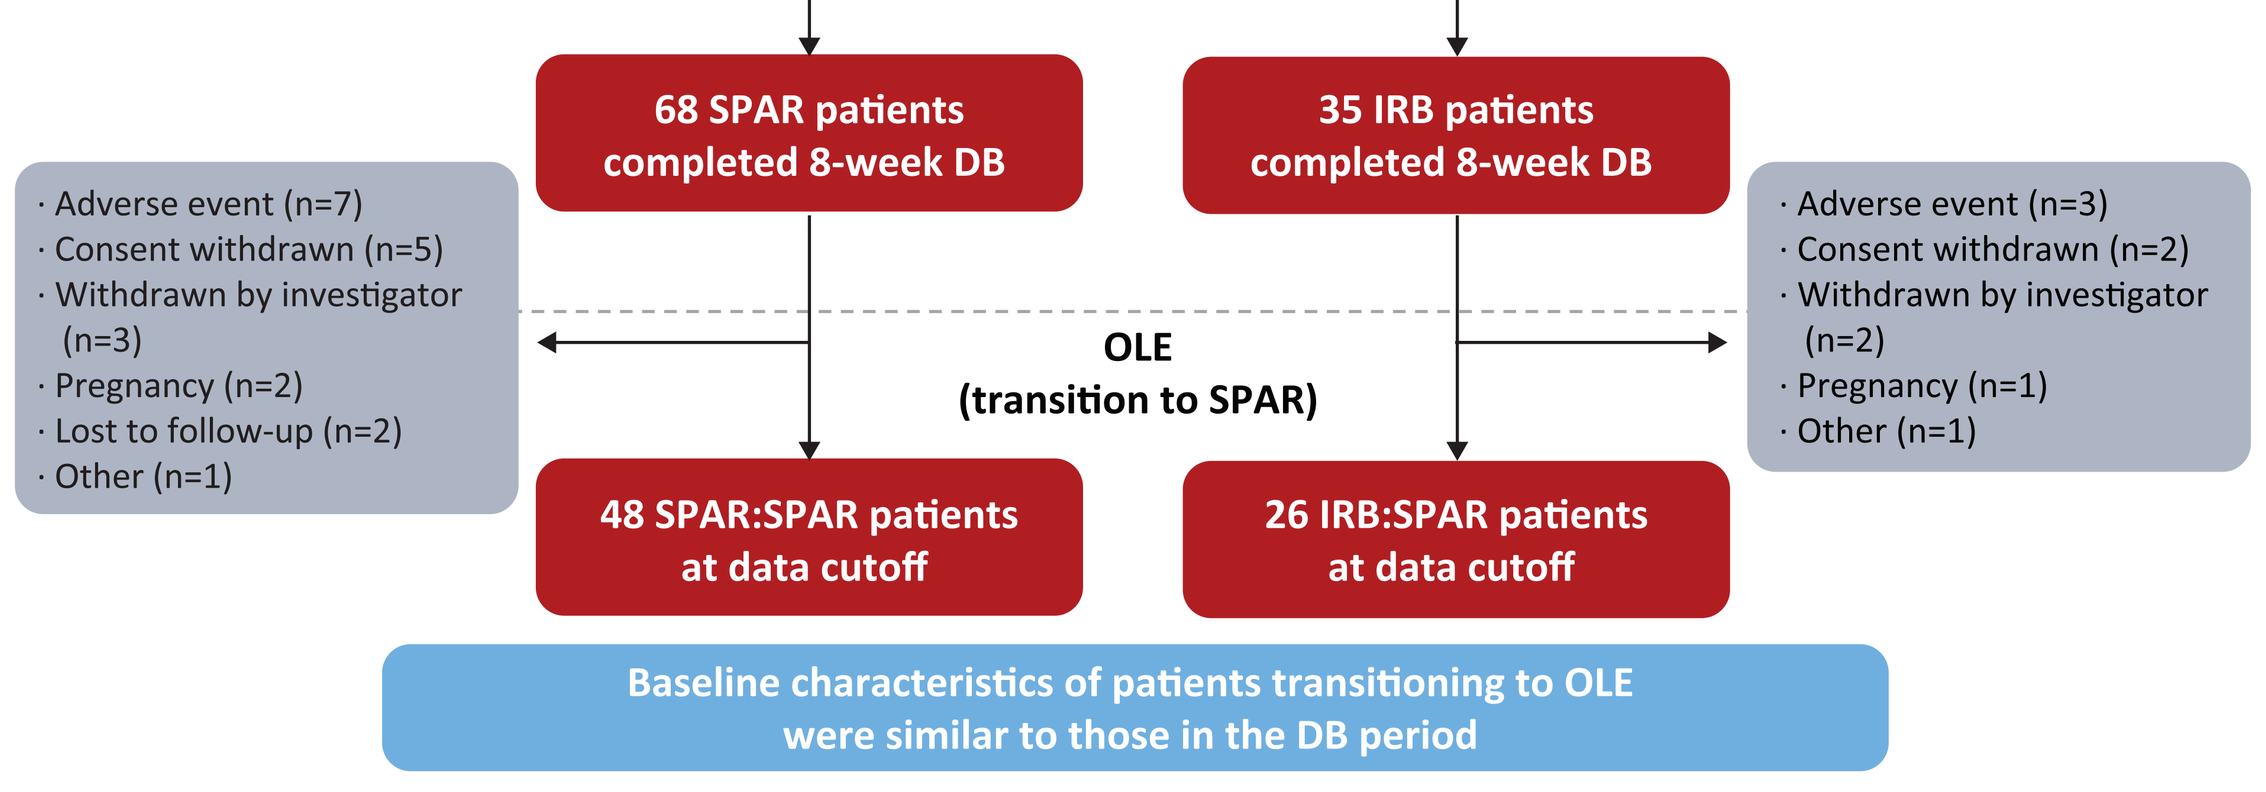
Randomized (N=109)
SPAR (n=73)
IRB (n=36)
68 SPAR patients completed 8-week DB
35 IRB patients completed 8-week DB
· Adverse event (n=7)
· Consent withdrawn (n=5)
· Withdrawn by investigator (n=3)
· Pregnancy (n=2)
· Lost to follow-up (n=2)
· Other (n=1)
OLE
(transition to SPAR)
· Adverse event (n=3)
· Consent withdrawn (n=2)
· Withdrawn by investigator (n=2)
· Pregnancy (n=1)
· Other (n=1)
48 SPAR:SPAR patients at data cutoff
26 IRB:SPAR patients at data cutoff
Baseline characteristics of patients transitioning to OLE were similar to those in the DB period

[a]As of November 2017. One patient in the SPAR:SPAR group was lost to follow-up after completion of the DB period and did not have any assessments in the OLE (n=68 in DB period; n=67 in OLE).
DB, double-blind; IRB, irbesartan; IRB:SPAR, patients who received irbesartan in the DB and sparsentan in the OLE; OLE, open-label extension; SPAR, sparsentan; SPAR:SPAR, patients who received sparsentan in the DB and OLE.

Table 1. Patient demographics and baseline characteristics

Characteristics	Sparsentan, All Doses (n=73)	Irbesartan (n=36)
Age group, n (%)		
Pediatric (8-18 years)	13 (18)	10 (28)
Adult (19-75 years)	60 (82)	26 (72)
Sex, n (%)		
Female	32 (44)	17 (47)
Male	41 (56)	19 (53)
Race, n (%)		
Asian	5 (7)	1 (3)
Black	8 (11)	7 (19)
White	57 (78)	26 (72)
Other	3 (4)	2 (6)
Ethnicity, n (%)		
Hispanic/Latino	14 (19)	6 (17)
Non-Hispanic/Non-Latino	59 (81)	30 (83)
BMI, kg/m^2, mean (SD)	28.4 (6.1)	28.7 (6.4)
IST at baseline, n (%)	21 (29)	13 (36)
eGFR, mL/min/1.73 m^2, median	73.4	65.1
UP/C ratio, g/g, median (range)	3.61 (0.4-18.7)	3.12 (0.9-10.7)
ACE inhibitor or ARB use before washout, n (%)	59 (81)	32 (89)
Use of ≥1 diuretic/antihypertensive agent, n (%)	40 (55)	20 (56)
Diuretics	26 (36)	9 (25)
Additional antihypertensive treatments	29 (40)	16 (44)

Based on the full analysis set in the double-blind period.
ACE, angiotensin-converting enzyme; ARB, angiotensin II receptor blocker; BMI, body mass index; eGFR, estimated glomerular filtration rate; IST, immunosuppressive therapy; SD, standard deviation; UP/C, urinary protein-to-creatinine ratio (24-hour urine).

- At Week 84, a total of 22% (22/102) of patients in the OLE had initiated a new IST
- In the SPAR:SPAR group (n=67), sparsentan resulted in rapid, sustained reduction in UP/C from baseline through 84 weeks in the OLE (**Figure 3A**)
 - Sixteen (24%) SPAR:SPAR patients received newly initiated IST during the OLE, which had no effect on UP/C decline
- In the IRB:SPAR group (n=35), switching to sparsentan resulted in significant reduction in UP/C (Week 16), which was sustained in the OLE through 84 weeks, suggesting additional sparsentan effect following irbesartan cessation (**Figure 3B**)
 - Similar to the SPAR:SPAR group, the IRB:SPAR group showed no effect of newly initiated IST (n=6; 17%) on UP/C decline

Figure 3. UP/C over 84 weeks

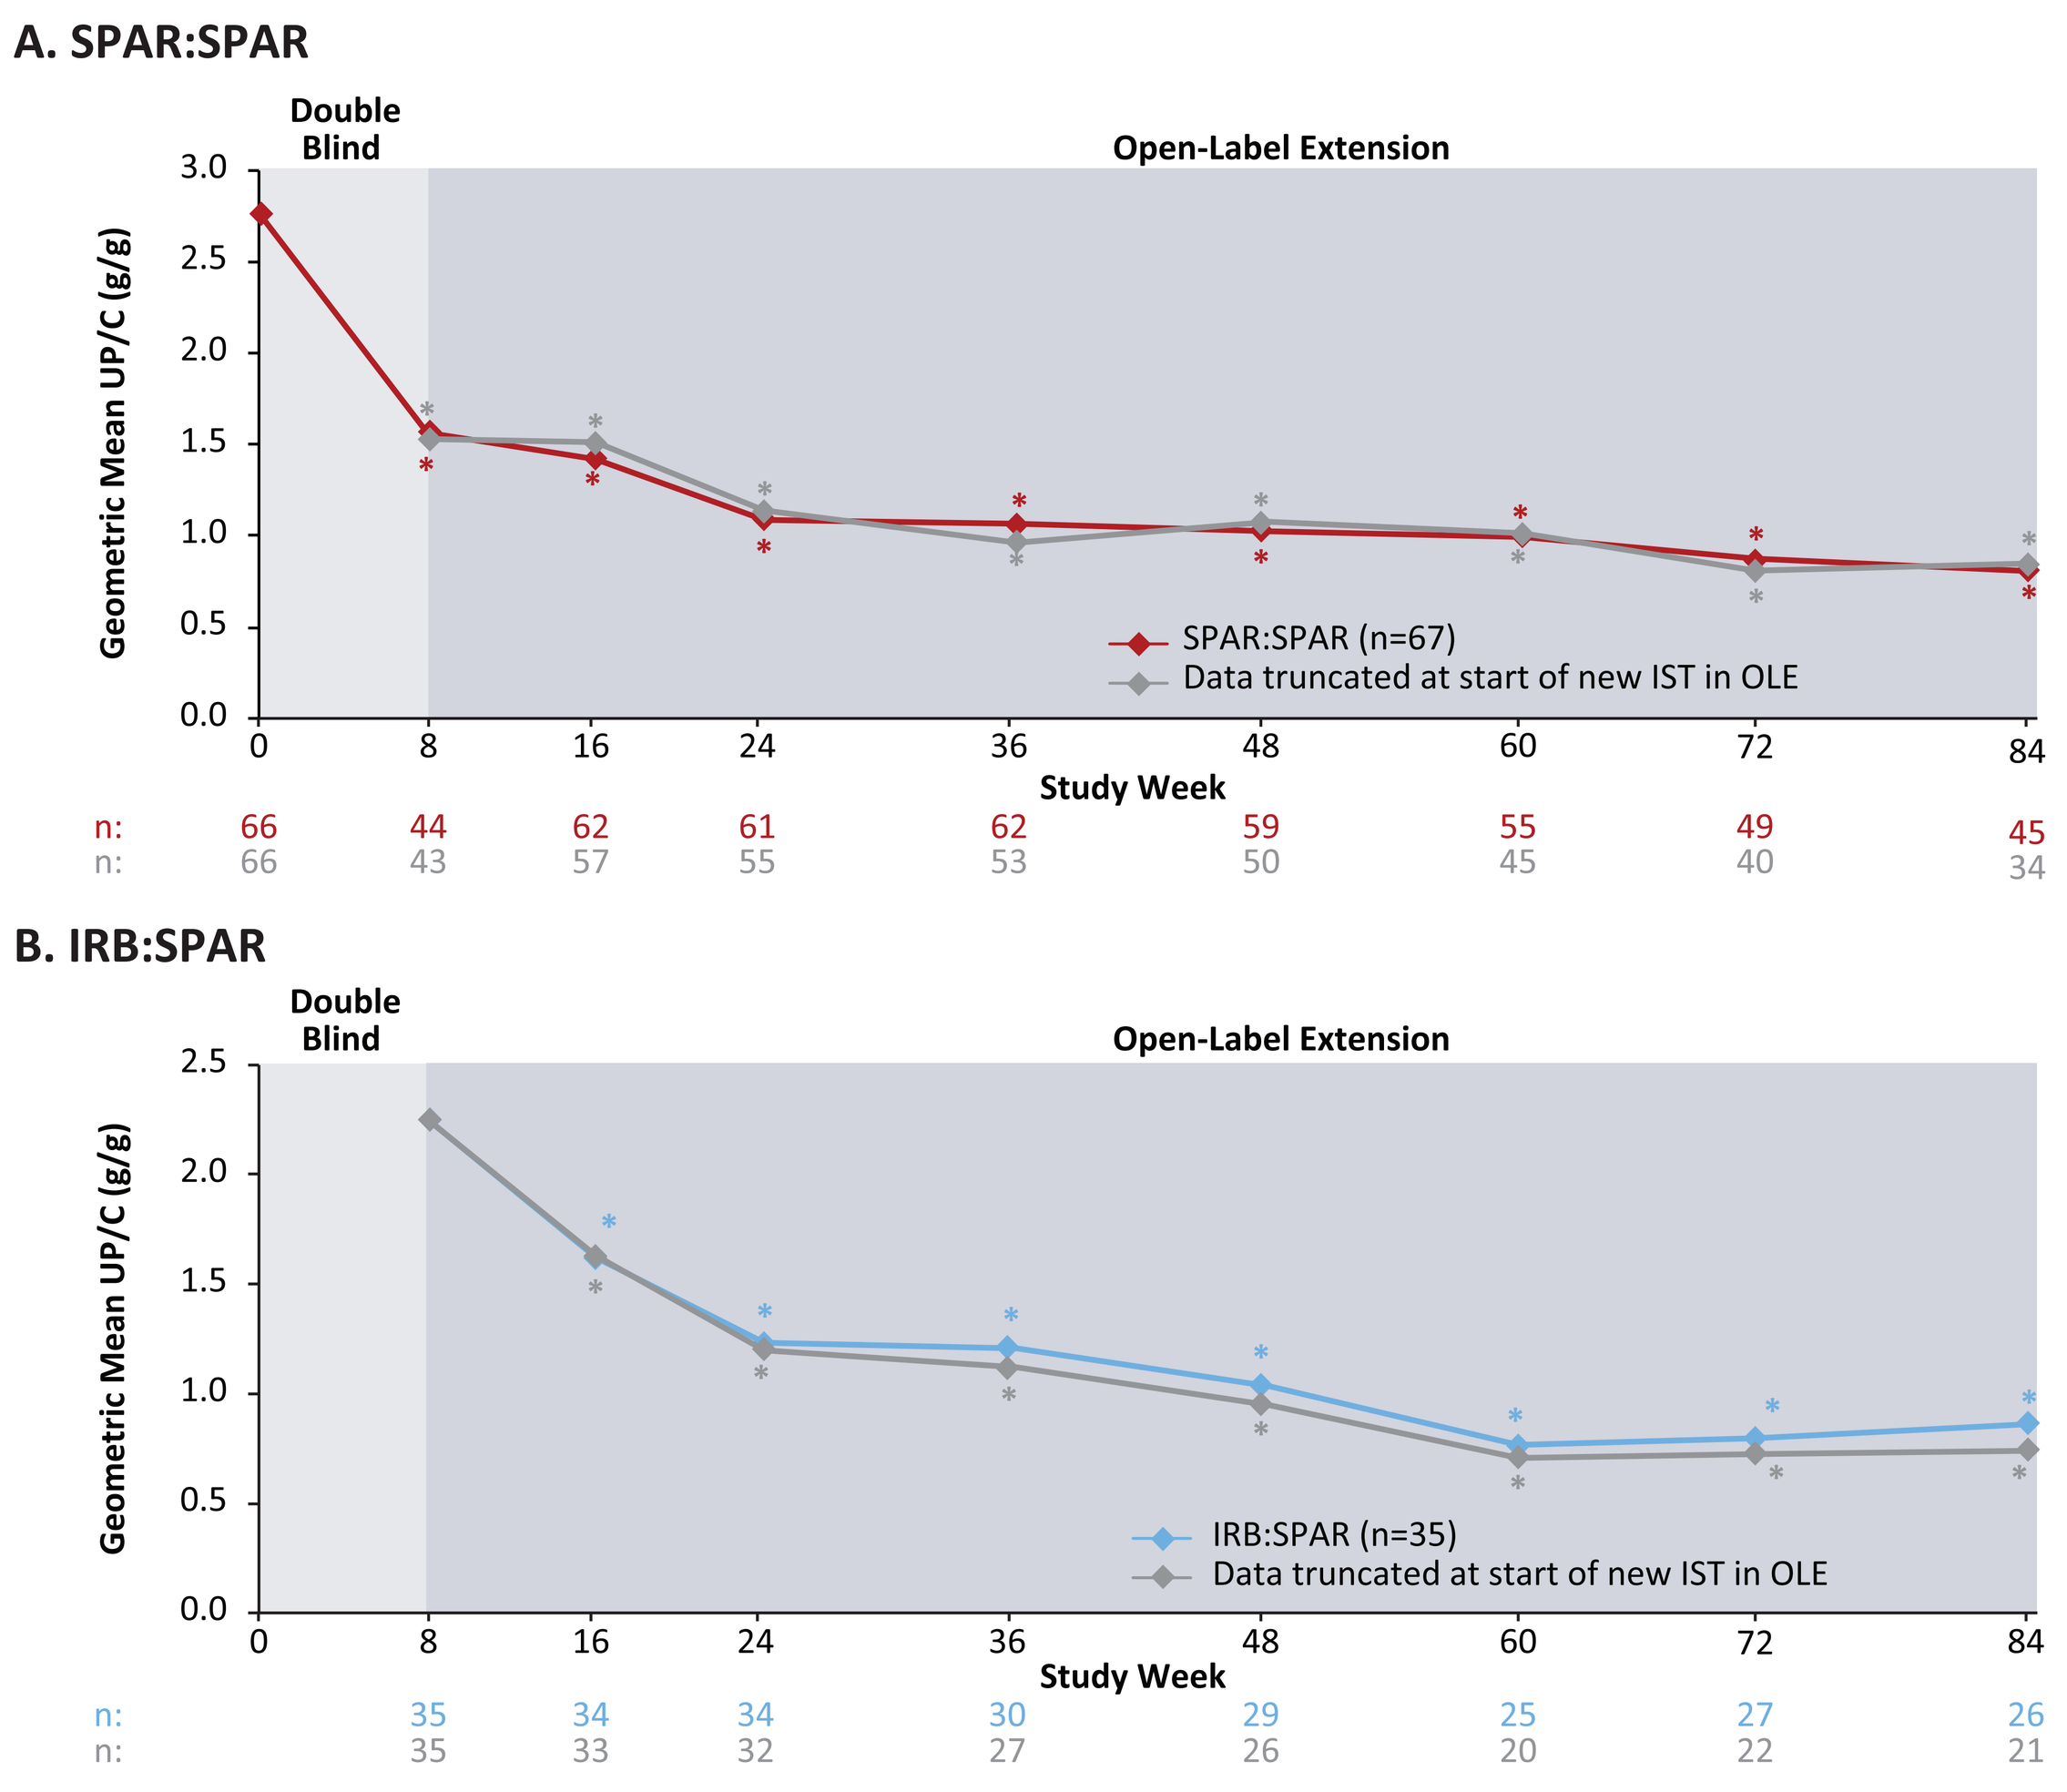
A. SPAR:SPAR
Double Blind
Open-Label Extension
Geometric Mean UP/C (g/g)
SPAR:SPAR (n=67)
Data truncated at start of new IST in OLE
Study Week
n: 66 44 62 61 62 59 55 49 45
n: 66 43 57 55 53 50 45 40 34
B. IRB:SPAR
Double Blind
Open-Label Extension
Geometric Mean UP/C (g/g)
IRB:SPAR (n=35)
Data truncated at start of new IST in OLE
Study Week
n: 35 34 34 30 29 25 27 26
n: 35 33 32 27 26 20 22 21

*95% CI of the change from baseline (Week 0 for SPAR:SPAR; Week 8 for IRB:SPAR) after log transformation excludes 0. Based on the full analysis set for OLE. UP/C based on first morning void. CI, confidence interval; IRB:SPAR, patients who received irbesartan in the DB and sparsentan in the OLE; IST, immunosuppressive therapy; OLE, open-label extension; SPAR:SPAR, patients who received sparsentan in the DB and OLE; UP/C, urinary protein-to-creatinine ratio.

- The proportion of patients reaching FPRE was similar over the 84 weeks in the OLE, regardless of use of newly initiated IST, in both the SPAR:SPAR (**Figure 4A**) and IRB:SPAR (**Figure 4B**) groups

Figure 4. FPRE over 84 weeks

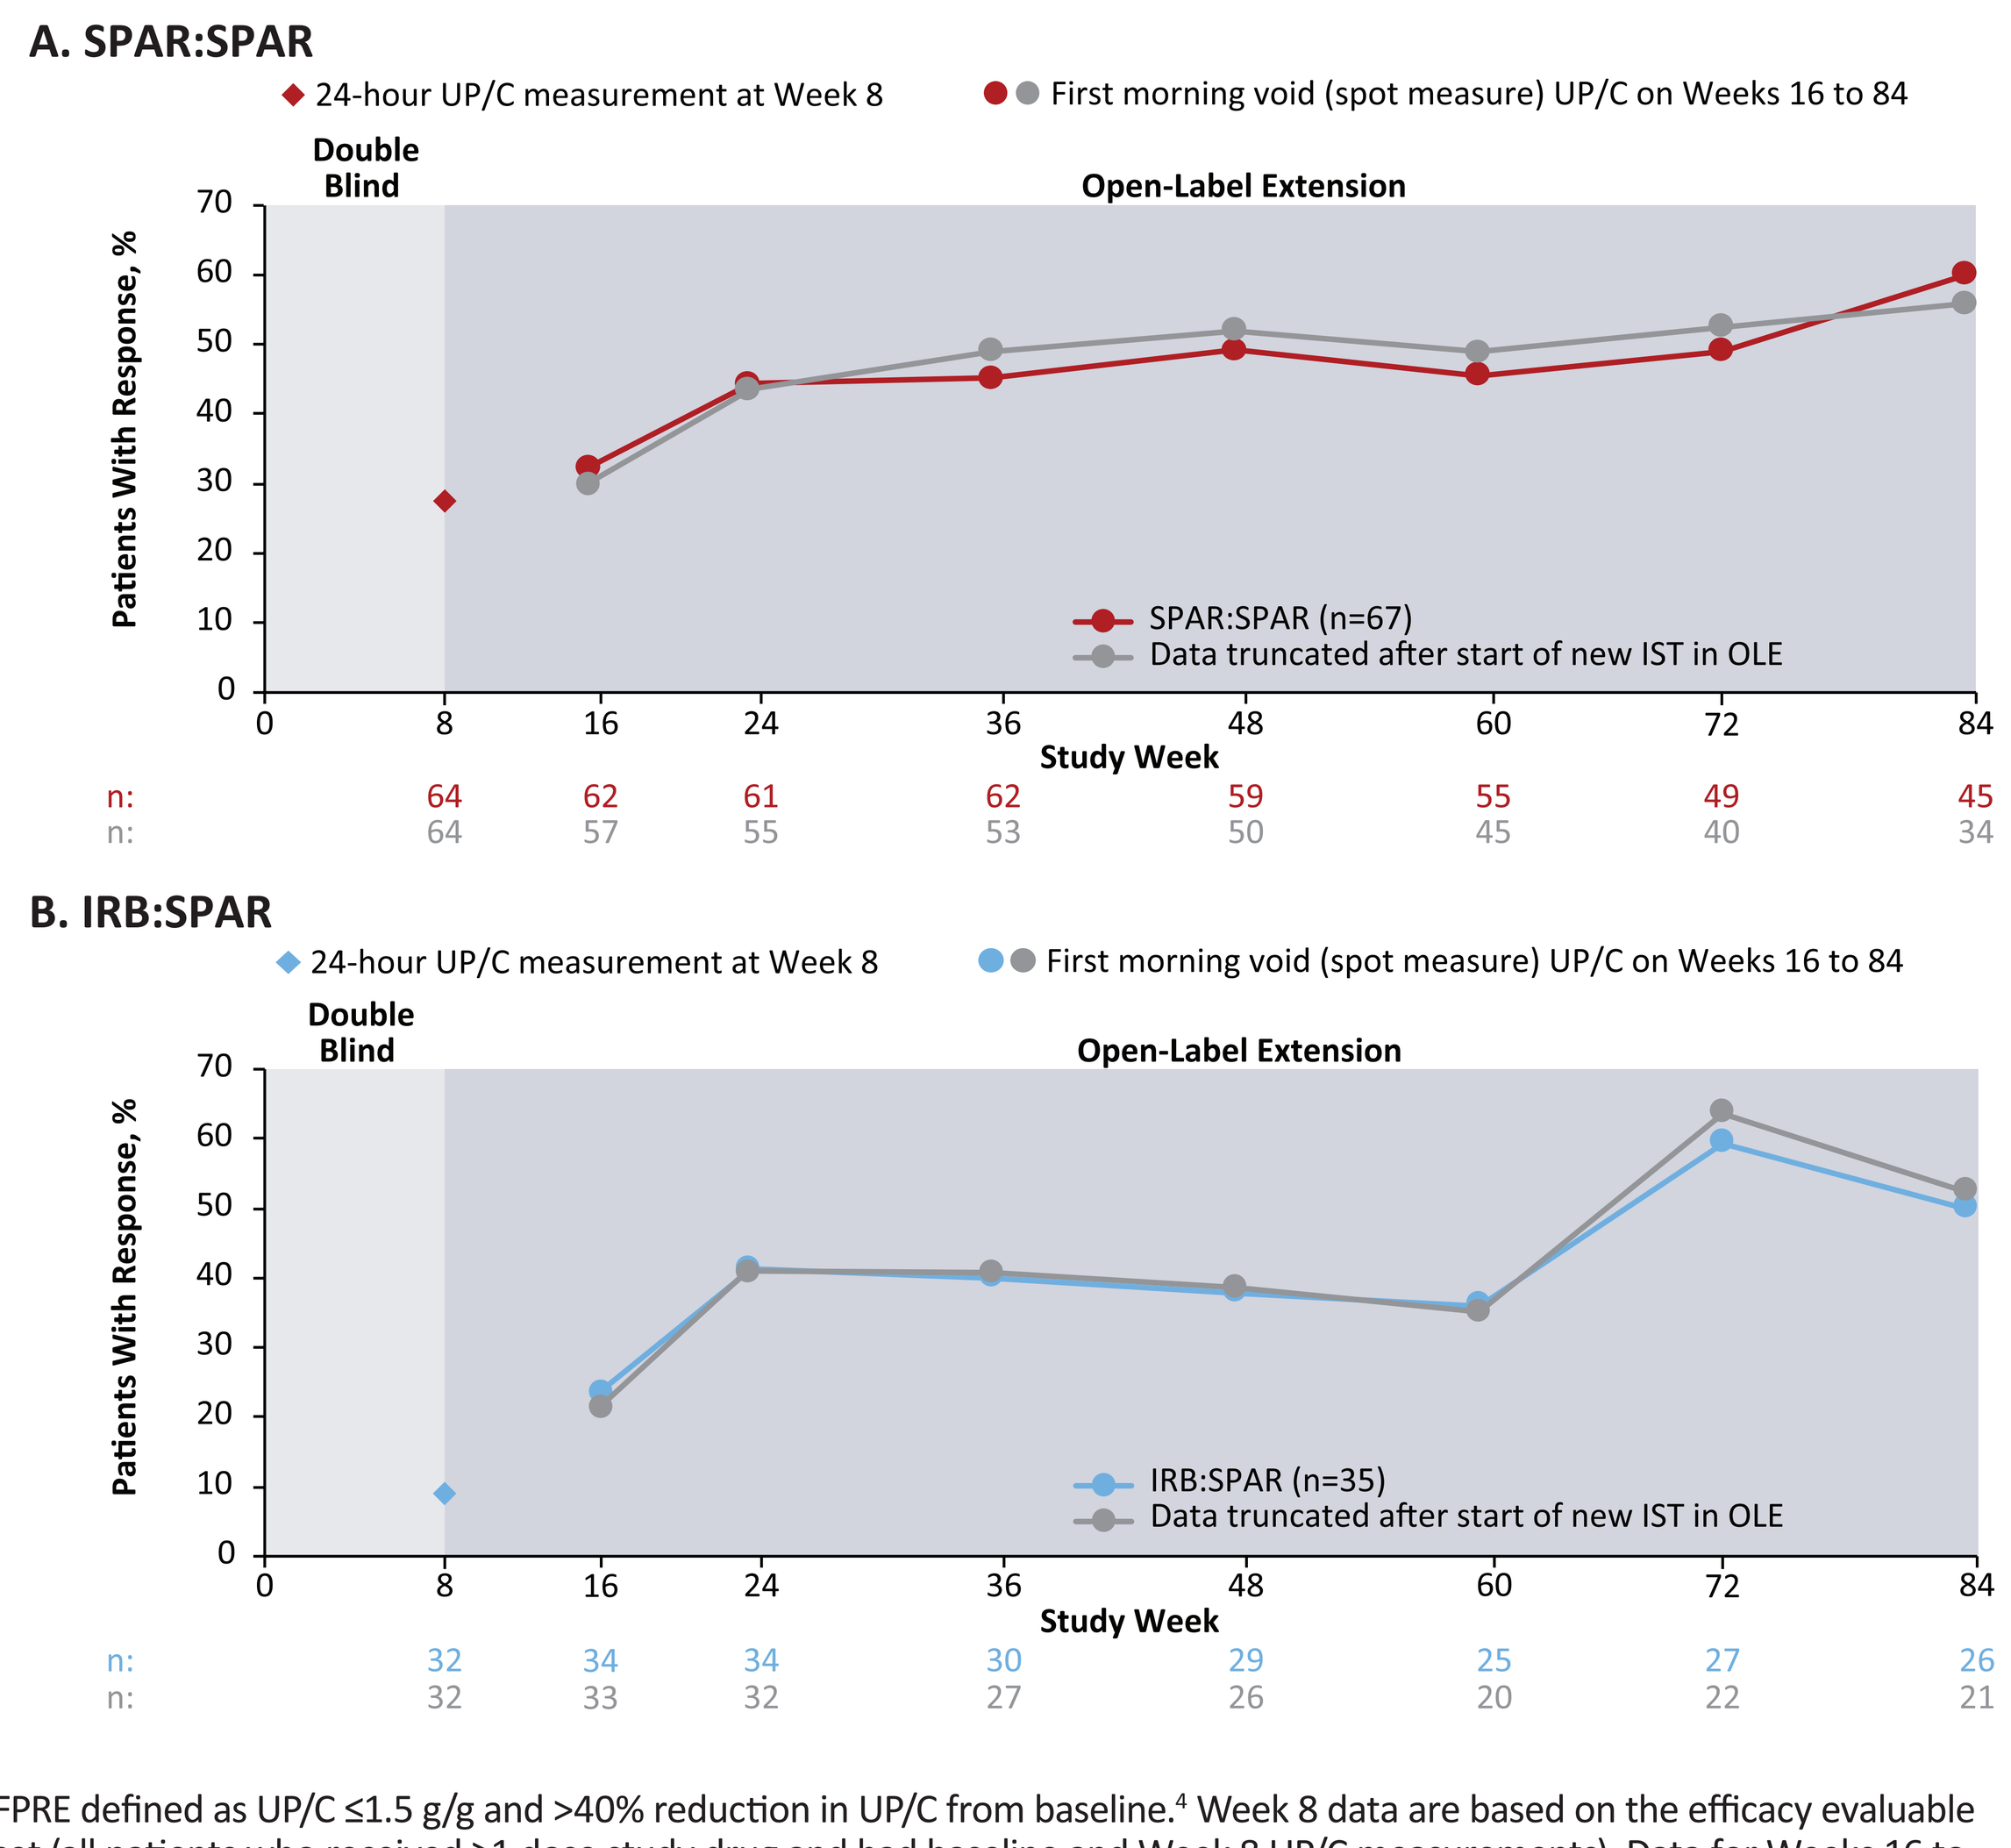
A. SPAR:SPAR
24-hour UP/C measurement at Week 8
First morning void (spot measure) UP/C on Weeks 16 to 84
Double Blind
Open-Label Extension
Patients With Response, %
SPAR:SPAR (n=67)
Data truncated after start of new IST in OLE
Study Week
n: 64 62 61 62 59 55 49 45
n: 64 57 55 53 50 45 40 34
B. IRB:SPAR
24-hour UP/C measurement at Week 8
First morning void (spot measure) UP/C on Weeks 16 to 84
Double Blind
Open-Label Extension
Patients With Response, %
IRB:SPAR (n=35)
Data truncated after start of new IST in OLE
Study Week
n: 32 34 34 30 29 25 27 26
n: 32 33 32 27 26 20 22 21

FPRE defined as UP/C ≤1.5 g/g and >40% reduction in UP/C from baseline.[4] Week 8 data are based on the efficacy evaluable set (all patients who received ≥1 dose study drug and had baseline and Week 8 UP/C measurements). Data for Weeks 16 to 84 are based on the full analysis set for OLE. FPRE, FSGS partial remission endpoint; FSGS, focal segmental glomerulosclerosis; IRB:SPAR, patients who received irbesartan in the DB and sparsentan in the OLE; IST, immunosuppressive therapy; OLE, open-label extension; SPAR:SPAR, patients also received sparsentan in the DB and OLE; UP/C, urinary protein-to-creatinine ratio.

- Newly initiated IST had no effect on the course of eGFR over 84 weeks in the OLE in both the SPAR:SPAR (**Figure 5A**) and IRB:SPAR (**Figure 5B**) groups

Figure 5. eGFR over 84 weeks

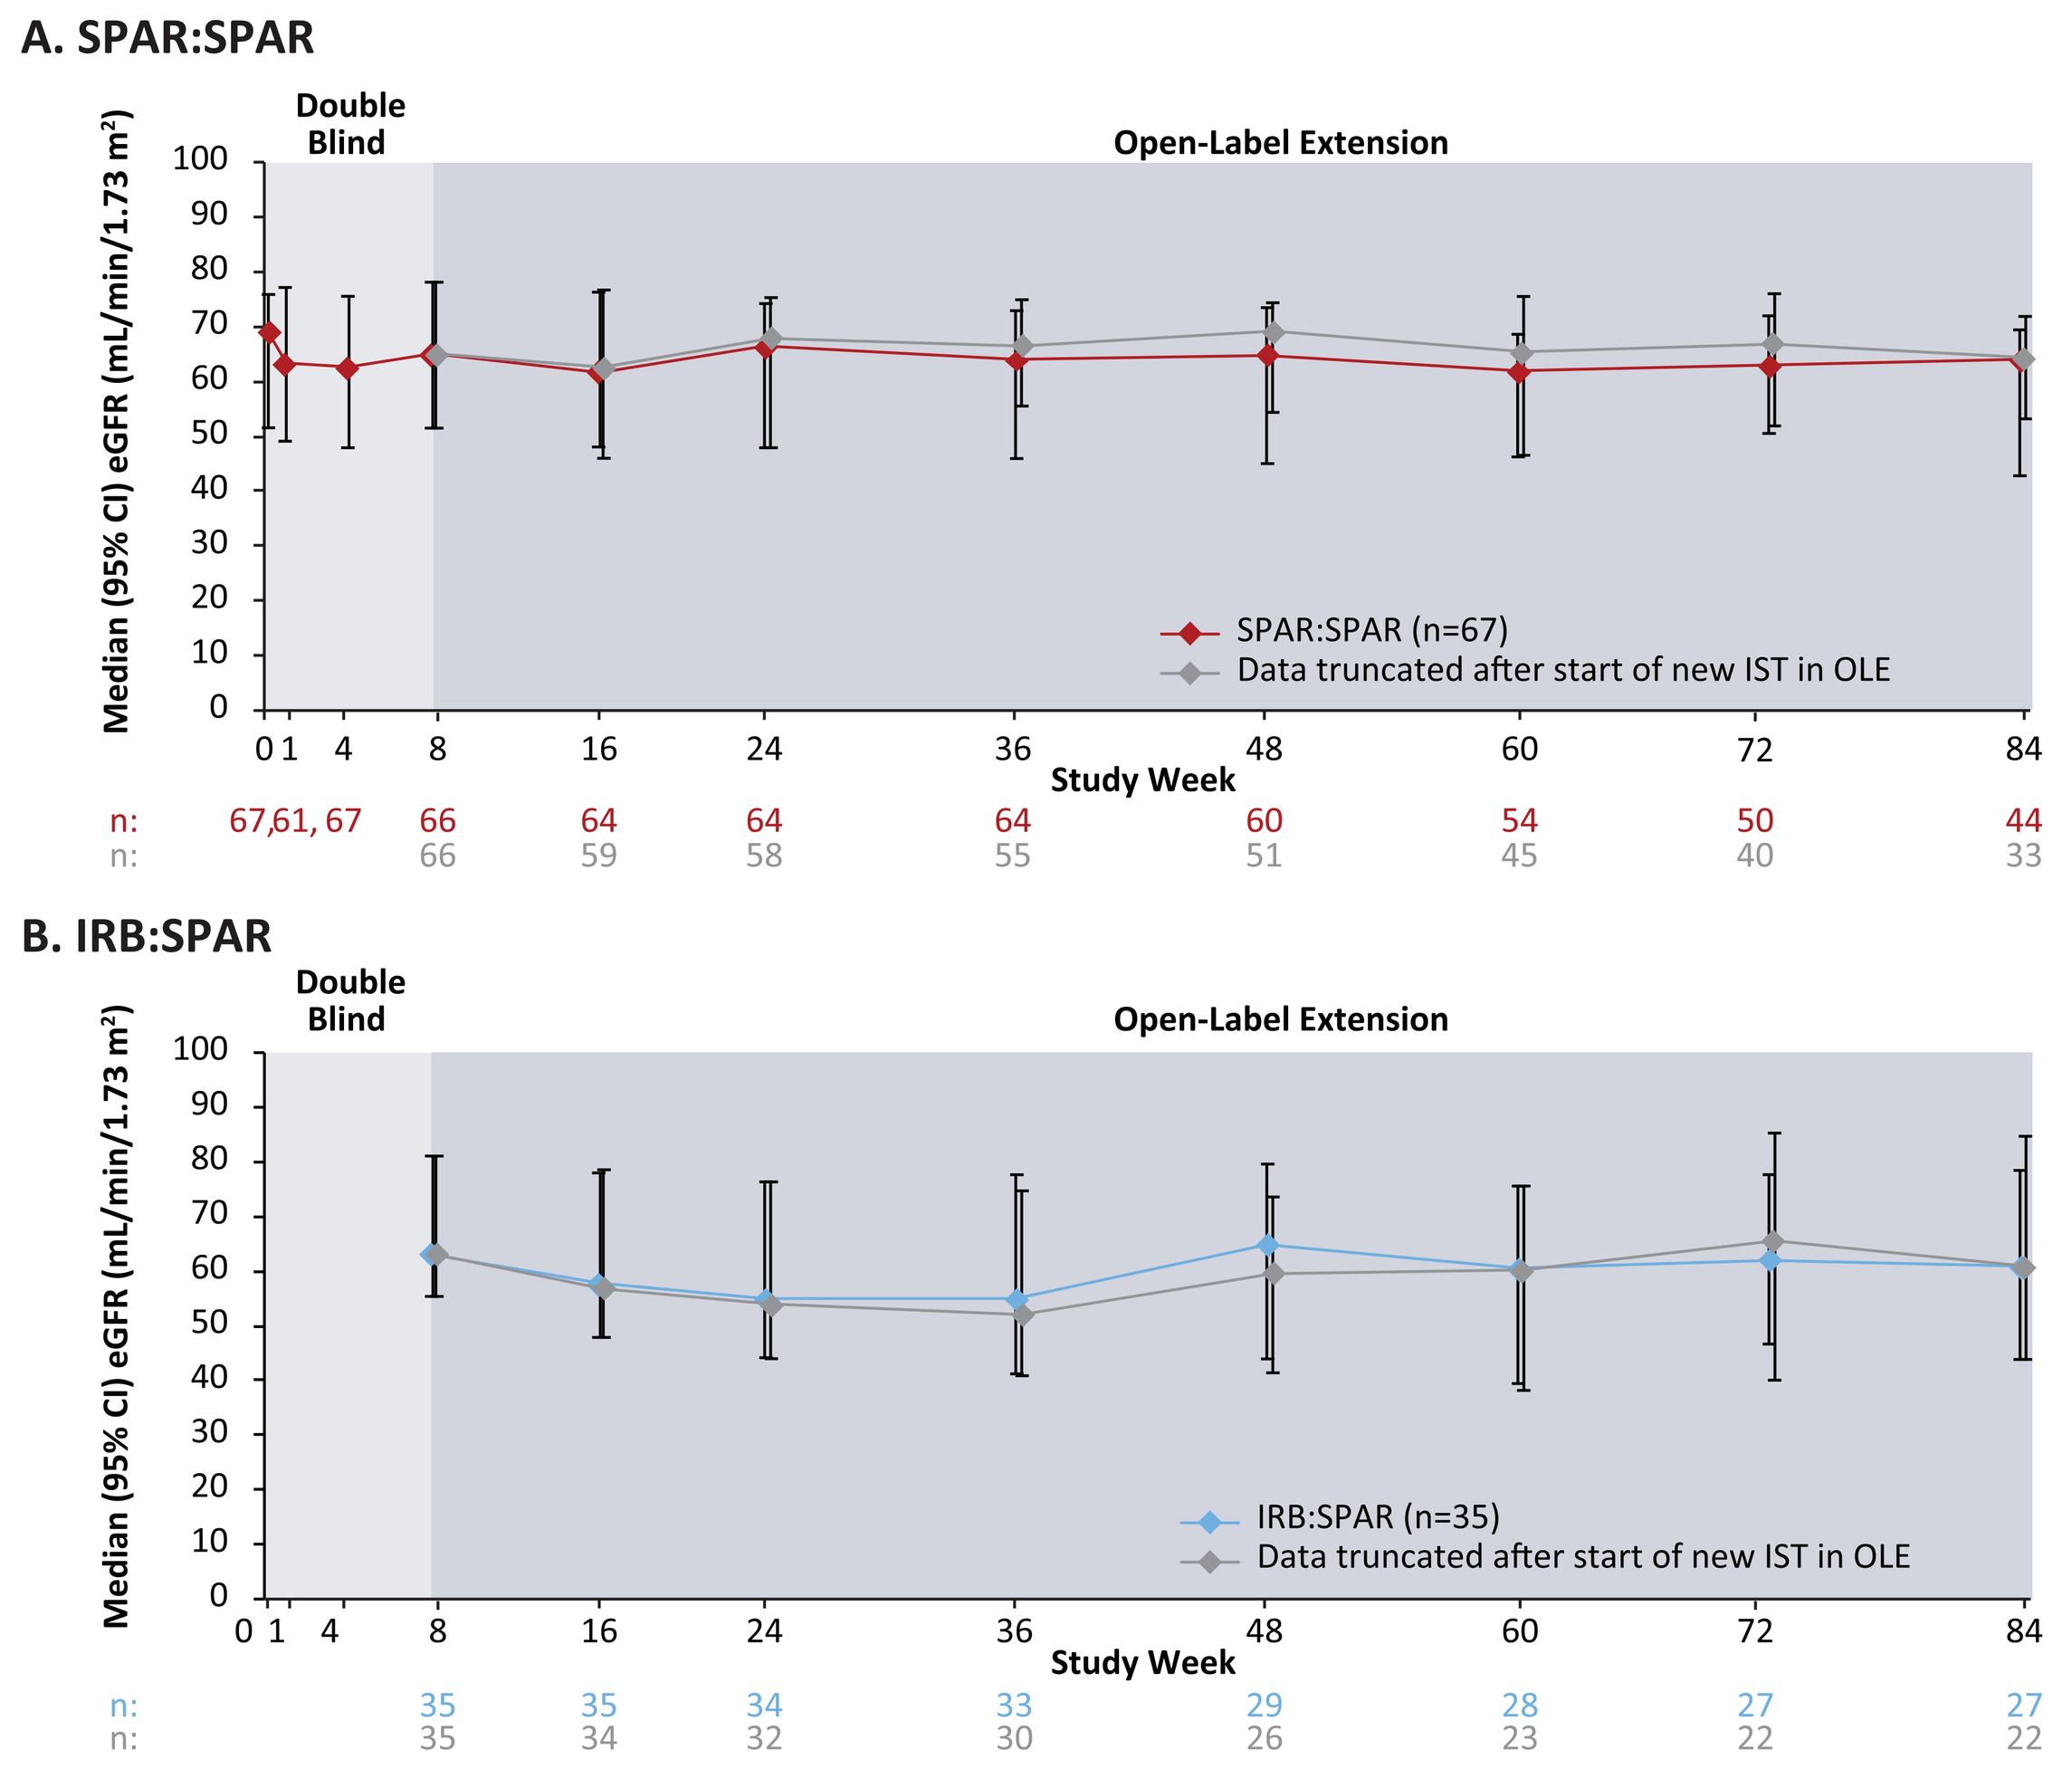
A. SPAR:SPAR
Double Blind
Open-Label Extension
Median (95% CI) eGFR (mL/min/1.73 m2)
SPAR:SPAR (n=67)
Data truncated after start of new IST in OLE
Study Week
n: 67,61, 67 66 64 64 64 60 54 50 44
n: 66 59 58 55 51 45 40 33
B. IRB:SPAR
Double Blind
Open-Label Extension
Median (95% CI) eGFR (mL/min/1.73 m2)
IRB:SPAR (n=35)
Data truncated after start of new IST in OLE
Study Week
n: 35 35 34 33 29 28 27 27
n: 35 34 32 30 26 23 22 22

Based on the full analysis set in the OLE. 95% CIs were calculated for the median value at each time point. Note: There were 9 study withdrawals due to renal causes during the follow-up period, including 1 case of end-stage kidney disease. CI, confidence interval; eGFR, estimated glomerular filtration rate; IRB:SPAR, patients who received irbesartan in the DB and sparsentan in the OLE; IST, immunosuppressive therapy; OLE, open-label extension; SPAR:SPAR, patients who received sparsentan in the DB and OLE.

- Newly initiated IST had no effect on the course of BP over 84 weeks in the OLE in the SPAR:SPAR (**Figure 6A**) or IRB:SPAR (**Figure 6B**) groups

Figure 6. Blood pressure over 84 weeks

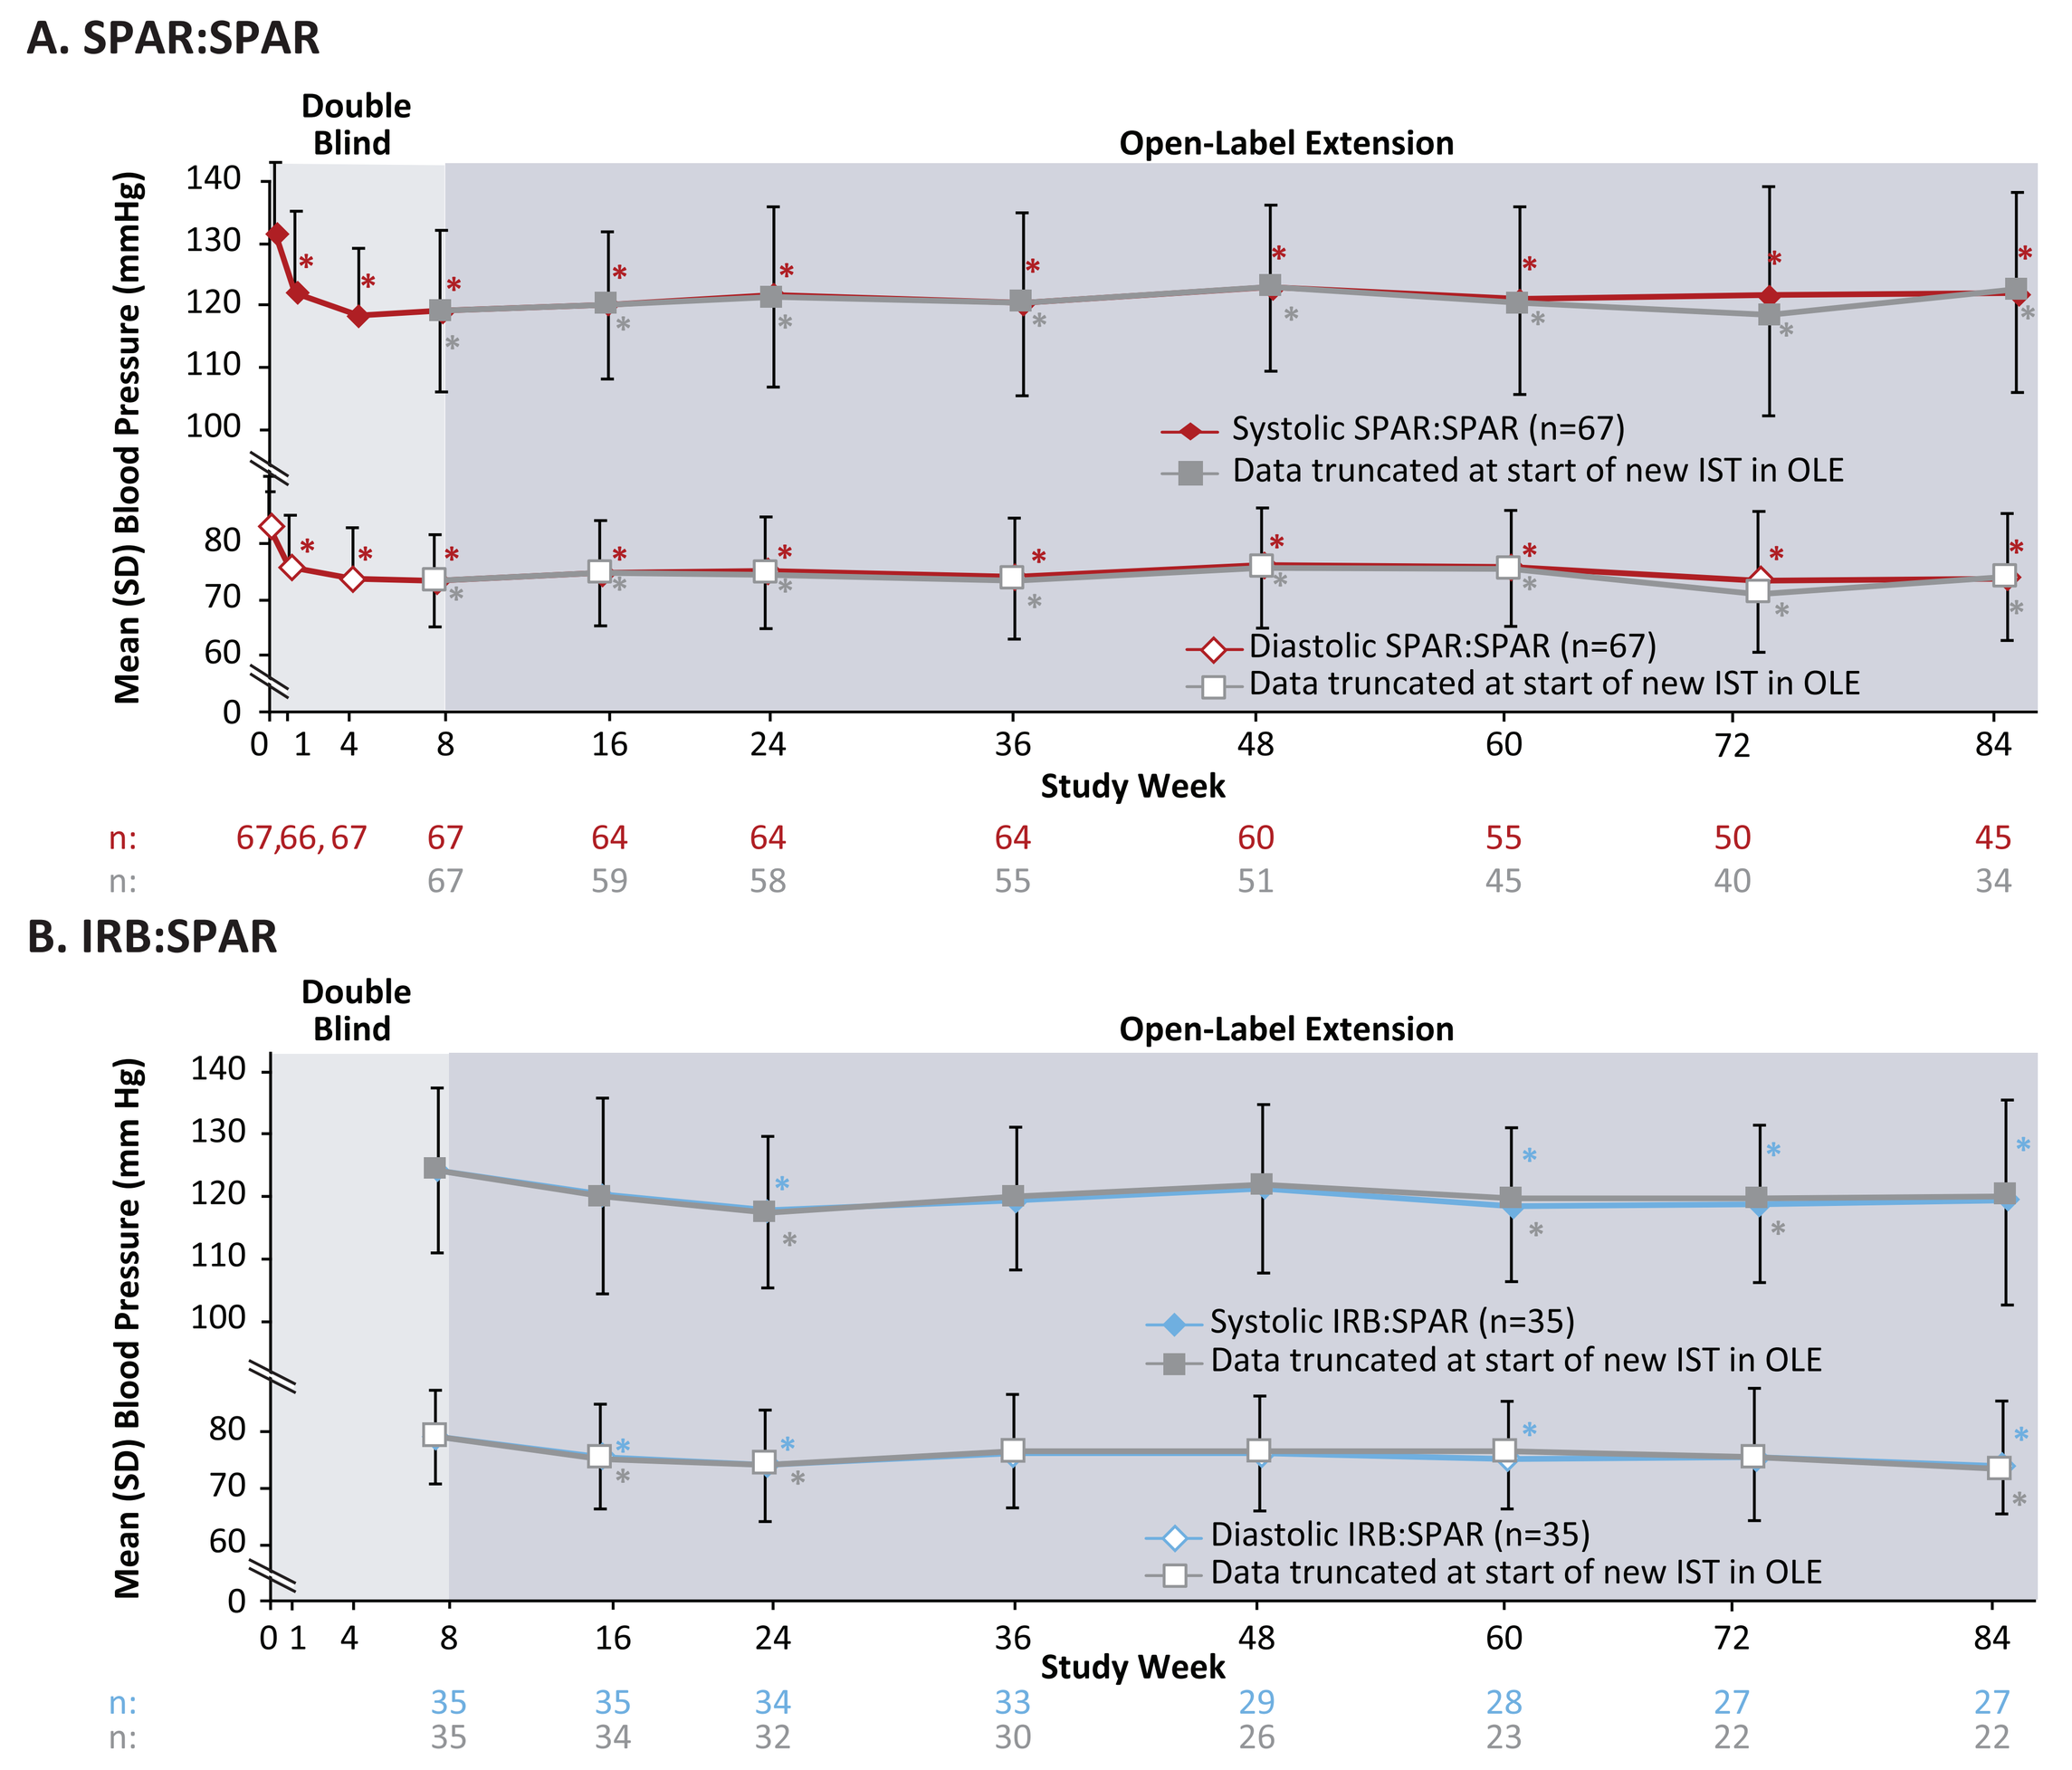
A. SPAR:SPAR
Double Blind
Open-Label Extension
Mean (SD) Blood Pressure (mmHg)
Systolic SPAR:SPAR (n=67)
Data truncated at start of new IST in OLE
Diastolic SPAR:SPAR (n=67)
Data truncated at start of new IST in OLE
Study Week
n: 67,66,67 67 64 64 64 60 55 50 45
n: 67 59 58 55 51 45 40 34
B. IRB:SPAR
Double Blind
Open-Label Extension
Mean (SD) Blood Pressure (mm Hg)
Systolic IRB:SPAR (n=35)
Data truncated at start of new IST in OLE
Diastolic IRB:SPAR (n=35)
Data truncated at start of new IST in OLE
Study Week
n: 35 35 34 33 29 28 27 27
n: 35 34 32 30 26 23 22 22

*95% CI of the mean change from baseline (Week 0 for SPAR:SPAR; Week 8 for IRB:SPAR) excludes 0.
Based on the safety analysis set for OLE. One-sided error bars represent 1 SD.
IRB:SPAR, patients who received irbesartan in the DB and sparsentan in the OLE; OLE, open-label extension; SD, standard deviation; SPAR:SPAR, patients who received sparsentan in the DB and OLE.

- Newly initiated IST had no impact on TEAE incidence (**Table 2**)

Table 2. TEAEs during treatment with sparsentan for patients who entered OLE[a]

	Full Safety Population for OLE		New IST Excluded Safety Population for OLE	
	SPAR:SPAR (n=67)	IRB:SPAR[b] (n=35)	SPAR:SPAR (n=67)[c]	IRB:SPAR[b] (n=35)[c]
Patients with ≥1 TEAE, n (%)	63 (94.0)	32 (91.4)	63 (94.0)	31 (88.6)
TEAEs occurring in ≥10%, n (%)				
Headache	20 (29.9)	6 (17.1)	18 (26.9)	5 (14.3)
Edema, peripheral	16 (23.9)	5 (14.3)	16 (23.9)	5 (14.3)
Hypotension	15 (22.4)	3 (8.6)	14 (20.9)	3 (8.6)
Nausea	13 (19.4)	0 (0.0)	13 (19.4)	0 (0.0)
Dizziness	12 (17.9)	2 (5.7)	12 (17.9)	2 (5.7)
Anemia[d]	10 (14.9)	6 (17.1)	10 (14.9)	6 (17.1)
Diarrhea	10 (14.9)	3 (8.6)	8 (11.9)	3 (8.6)
Hypertension	5 (7.5)	5 (14.3)	3 (4.5)	5 (14.3)
Sinusitis	3 (4.5)	5 (14.3)	2 (3.0)	4 (11.4)
Cough	9 (13.4)	2 (5.7)	8 (11.9)	2 (5.7)
Hyperkalemia	9 (13.4)	2 (5.7)	5 (7.5)	2 (5.7)
Pyrexia	9 (13.4)	1 (2.9)	8 (11.9)	0 (0.0)
Vomiting	9 (13.4)	1 (2.9)	9 (13.4)	1 (2.9)
Blood creatinine increased	7 (10.4)	3 (8.6)	7 (10.4)	3 (8.6)
Fatigue	7 (10.4)	0 (0.0)	7 (10.4)	0 (0.0)
Nasal congestion	7 (10.4)	3 (8.6)	7 (10.4)	3 (8.6)
Nasopharyngitis	7 (10.4)	2 (5.7)	6 (9.0)	2 (5.7)
Oropharyngeal pain	7 (10.4)	1 (2.9)	7 (10.4)	1 (2.9)
Upper respiratory tract infection	7 (10.4)	5 (14.3)	7 (10.4)	5 (14.3)

[a]Total duration of sparsentan exposure in patients who entered the OLE was 124.5 patient-years for SPAR:SPAR and 61.1 patient-years for IRB:SPAR; [b]Does not include TEAEs reported during 8 weeks of double-blind treatment with irbesartan; [c]Adverse events occurring on or after the date of the first new IST use in OLE are excluded. The percentage of patients reporting TEAEs uses the patient sample size at the start of the OLE as the denominator; [d]Includes anemia, iron-deficiency anemia, and decreased blood hemoglobin.
IRB:SPAR, patients who received irbesartan in the DB and sparsentan in the OLE; IST, immunosuppressive therapy; OLE, open-label extension; SPAR:SPAR, patients who received sparsentan in the DB and OLE; TEAEs, treatment-emergent adverse events.

Conclusions

- Ongoing sparsentan treatment in patients with FSGS over 84 weeks in the DUET OLE resulted in sustained and progressive antiproteinuric effects, and an increasing proportion of patients who achieved FPRE
- Sparsentan was well tolerated during the entire OLE
- Newly initiated IST had no meaningful impact on the efficacy and safety measures of sparsentan treatment over the 84 weeks of follow-up in in the DUET OLE

References

1. Komers R, et al. *Kidney Int Rep.* 2017;2:254-64.
2. Trachtman H, et al. *J Am Soc Nephrol.* 2018;29:2745-54.
3. Hogan J, et al. Presented at American Society of Nephrology Kidney Week, October 23-28, 2018, San Diego, CA.
4. Troost JP, et al. *Clin J Am Soc Nephrol.* 2018;13:414-21.

Disclosures

VT: Consultancy fees from AbbVie, Amgen, Bayer, Boehringer-Ingelheim, ChemoCentryx, and Fresenius Medical Care.

HT: Consultancy fees from ChemoCentryx, Kaneka, and Otsuka, and previously a consultant to Genzyme and Optherion. HT is an unpaid consultant to Retrophin, Inc. and has an agreement with Goldfinch Biopharma through NYU.

EM, BF, and RK: Employees of Retrophin, Inc. and may have an equity or other financial interest in Retrophin, Inc.

This study was funded by Retrophin, Inc. Writing support was provided by Lynanne McGuire, PhD, CMPP, of MedVal Scientific Information Services, LLC (Princeton, NJ) and was funded by Retrophin, Inc.

Presented at the International Society of Nephrology World Congress of Nephrology, April 12-15, 2019, Melbourne, Australia