The Dual ET_AR/AT_1R Blocker Sparsentan Attenuates Noise-Induced Hearing Loss in Alport Mice

Michael Anne Gratton,[1] Dominic Cosgrove,[2] Jared Hartsock,[1] Grady Phillips,[1] Ruth Gill,[1] Brianna Dufek,[2] Duane Delimont,[2] Daniel Meehan,[2] Celia Jenkinson,[3] Radko Komers[3]

[1]Washington University in St. Louis, St. Louis, MO, USA; [2]Boys Town National Research Hospital, Omaha, NE, USA; [3]Retrophin, Inc., San Diego, CA, USA

Introduction

Most emergent therapies for Alport syndrome (AS) address the renal disease but not the cochlear pathology and hearing loss frequently associated with the disease. Endothelin-1-mediated activation of the endothelin type A receptor (ET_AR) on strial marginal cells results in strial pathology, namely, thickening of the strial capillary basement membranes (SCBM), in the cochlea, and activation on glomerular mesangial cells contributes to glomerulosclerosis.[1,2] Currently, angiotensin-converting enzyme inhibitors (ACEi) or angiotensin II type 1 receptor (AT_1R) blockers are the standard of care for patients with AS; however, these drugs have not been shown to improve hearing. Previously we have reported that sparsentan (SP), a dual ET_AR/AT_1R antagonist, is nephroprotective in an AS mouse model and extends lifespan.[3,4] Therefore this report (1) shows that SP, but not losartan (LS), prevents noise-induced hearing loss in the AS mouse model and (2) details the changes in underlying pathology that may contribute to this difference.

Objective

To assess the effects of dual ET_AR/AT_1R inhibition with sparsentan and the blockade of AT_1R with losartan on noise-induced hearing loss and cochlear pathology in wild-type (WT) or COL4A3$^{-/-}$ autosomal Alport (KO) mice.

Methods

Study Design and Sample Collection

KO and WT 129/Sv littermates were treated with vehicle (V) or SP (120 mg/kg) by oral gavage from 3-8.75 weeks (wks) of age or with LS (20 mg/kg) by daily oral gavage from 3-4 wks of age and in drinking water (10 mg/kg) from 4-8.75 wks of age. As illustrated in **Figure 1**:

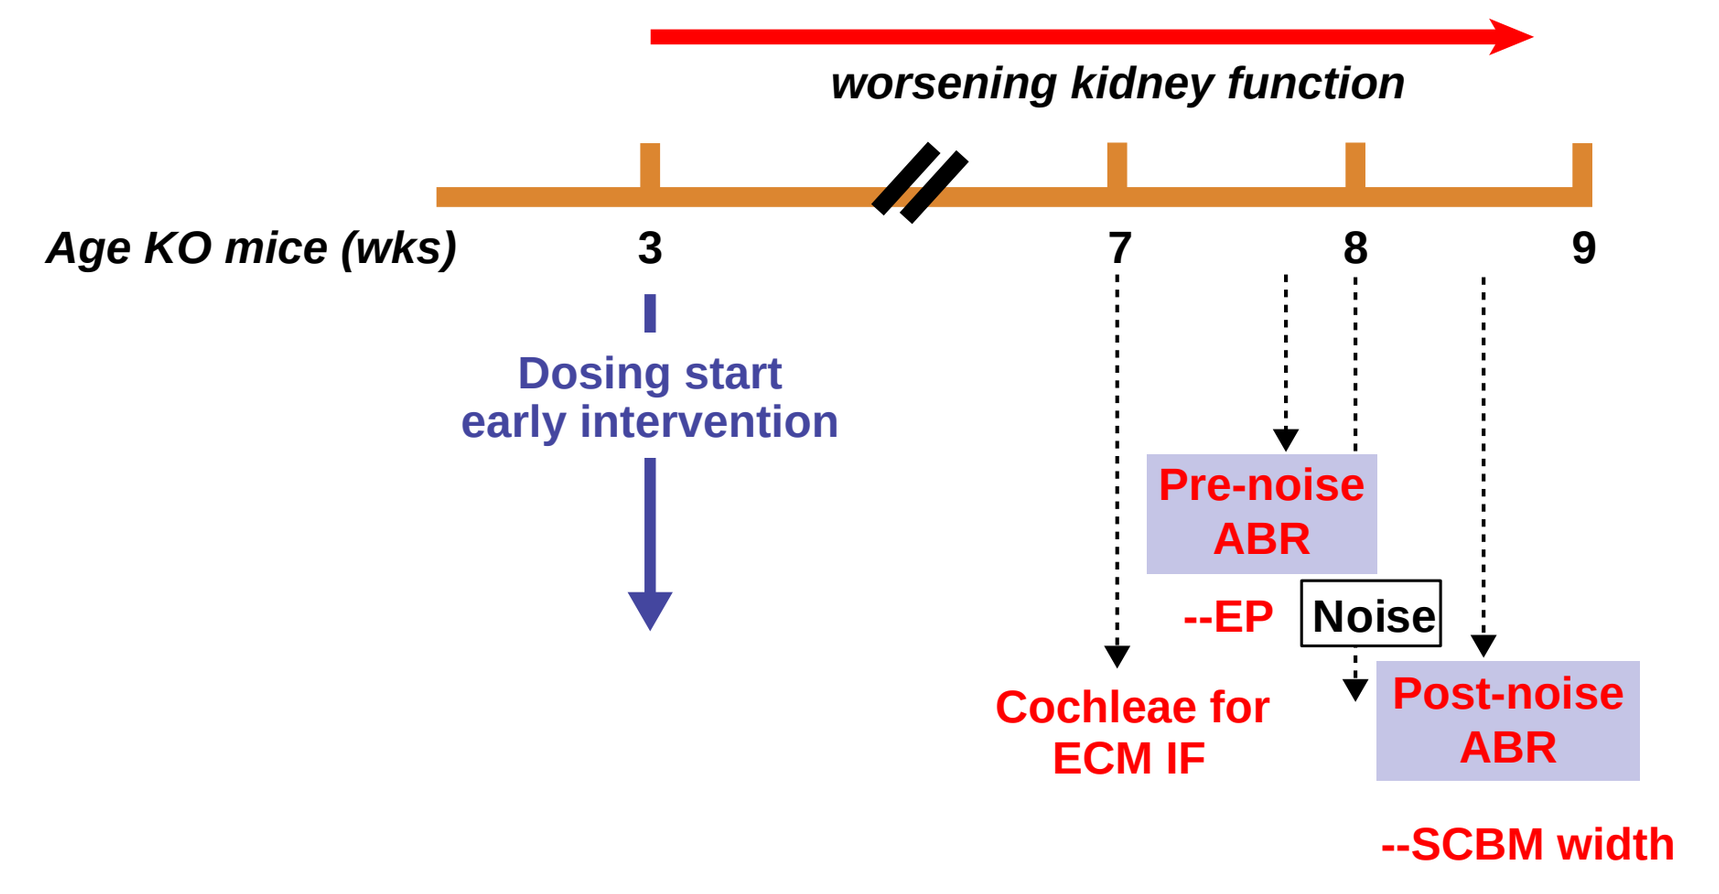
worsening kidney function
Age KO mice (wks)
3
7
8
9
Dosing start
early intervention
Pre-noise ABR
--EP
Noise
Cochleae for ECM IF
Post-noise ABR
--SCBM width

Figure 1. Study design in KO mice.

- Hearing was assessed between 7.5-8 wks (n=5-7/grp) in WT and KO mice treated with V, SP, or LS using the auditory brainstem response (ABR). The mice were then exposed to a 10-hour noise stress (106 dB SPL, 10kHz OBN), and 5 days post-noise, at 8.5 wks of age, underwent a second ABR analysis.
- Cochleae were excised after the final ABR test and the stria vascularis examined using transmission electron microscopy (TEM).
- WT or KO mice (n=10-13/grp) were treated with V or SP and underwent measurement of the endocochlear potential (EP) at 7.75 wks of age.
- Cochleae, obtained at 7 wks of age from additional WT or KO mice treated with V, SP (200 mg/kg), or LS, were prepared as frozen sections for immunofluorescence microscopy. Mid-modiolar cryosections were immunostained with one of the following antibodies: rat anti-mouse laminin $\alpha2$ antibody (L0663, Sigma-Aldrich, St. Louis, MO, USA), rabbit anti-mouse laminin $\alpha5$ antibody (a gift from Jeff Miner, Washington University), or rabbit anti-mouse collagen $\alpha1$(IV) antibody (T40261R, Biodesign, Saco, ME, USA). All sections were stained at the same time and microscope settings standardized to the V-treated KO cochleae.

Data Collection

- Hearing ability was assessed via the ABR technique using frequency-specific tone pips.
- The EP was measured in the basal turn of the right cochlea using a 150 KCl-filled glass pipette coupled to a DC amplifier/electrometer. The stable (1 min) DC voltage was referenced to an Ag/AgCl ground electrode in the flank tissue.
- The thickness of SCBM was measured in TEM digital images (JEOL 1200 EX II, AMT 8 megapixel camera, AMT Image Capture Engine V602) taken at 40,000x.
- Accumulation of extracellular matrix proteins (ECM) in the SCBM was determined from frozen mid-modiolar cochlear sections incubated with antibodies against laminin $\alpha2$, laminin $\alpha5$, and collagen $\alpha1$(IV), and visualized using a Leica confocal-imaging system.

Data Analysis

- Hearing loss was calculated by subtracting the ABR hearing threshold for pre-noise from that of post-noise hearing testing. Comparison of active dose to the KO vehicle used t-tests.
- Comparison of the SCBM thickness measures as well as the EP values among groups used one-way ANOVA and Tukey's multiple-comparison post-hoc test.
- For all statistical analyses, significance was set at $p<0.05$.

Results

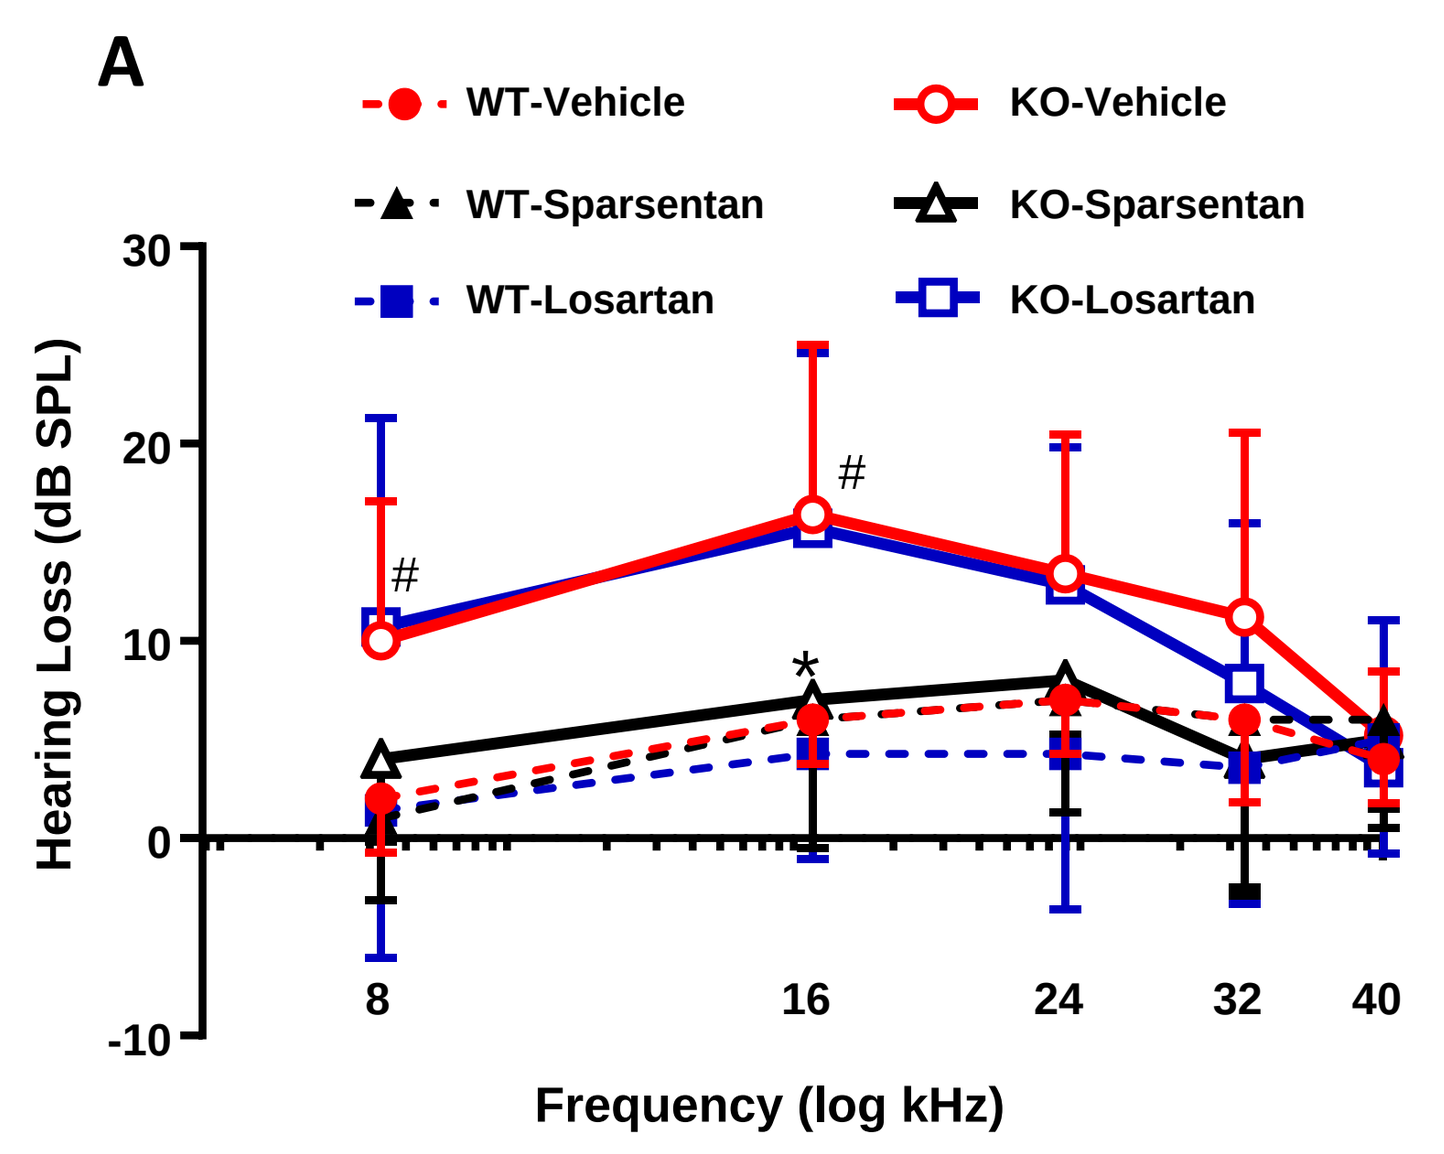
A
WT-Vehicle
WT-Sparsentan
WT-Losartan
KO-Vehicle
KO-Sparsentan
KO-Losartan
Hearing Loss (dB SPL)
Frequency (log kHz)
8
16
24
32
40

Figure 2A. Sparsentan (open black triangle) but not losartan (open blue square) attenuates noise-induced hearing loss in KO mice. Data are presented as mean ± SD (n=5). #$p<0.05$ KO-Vehicle vs WT-Vehicle. *$p<0.05$ KO-Vehicle vs KO-SP.

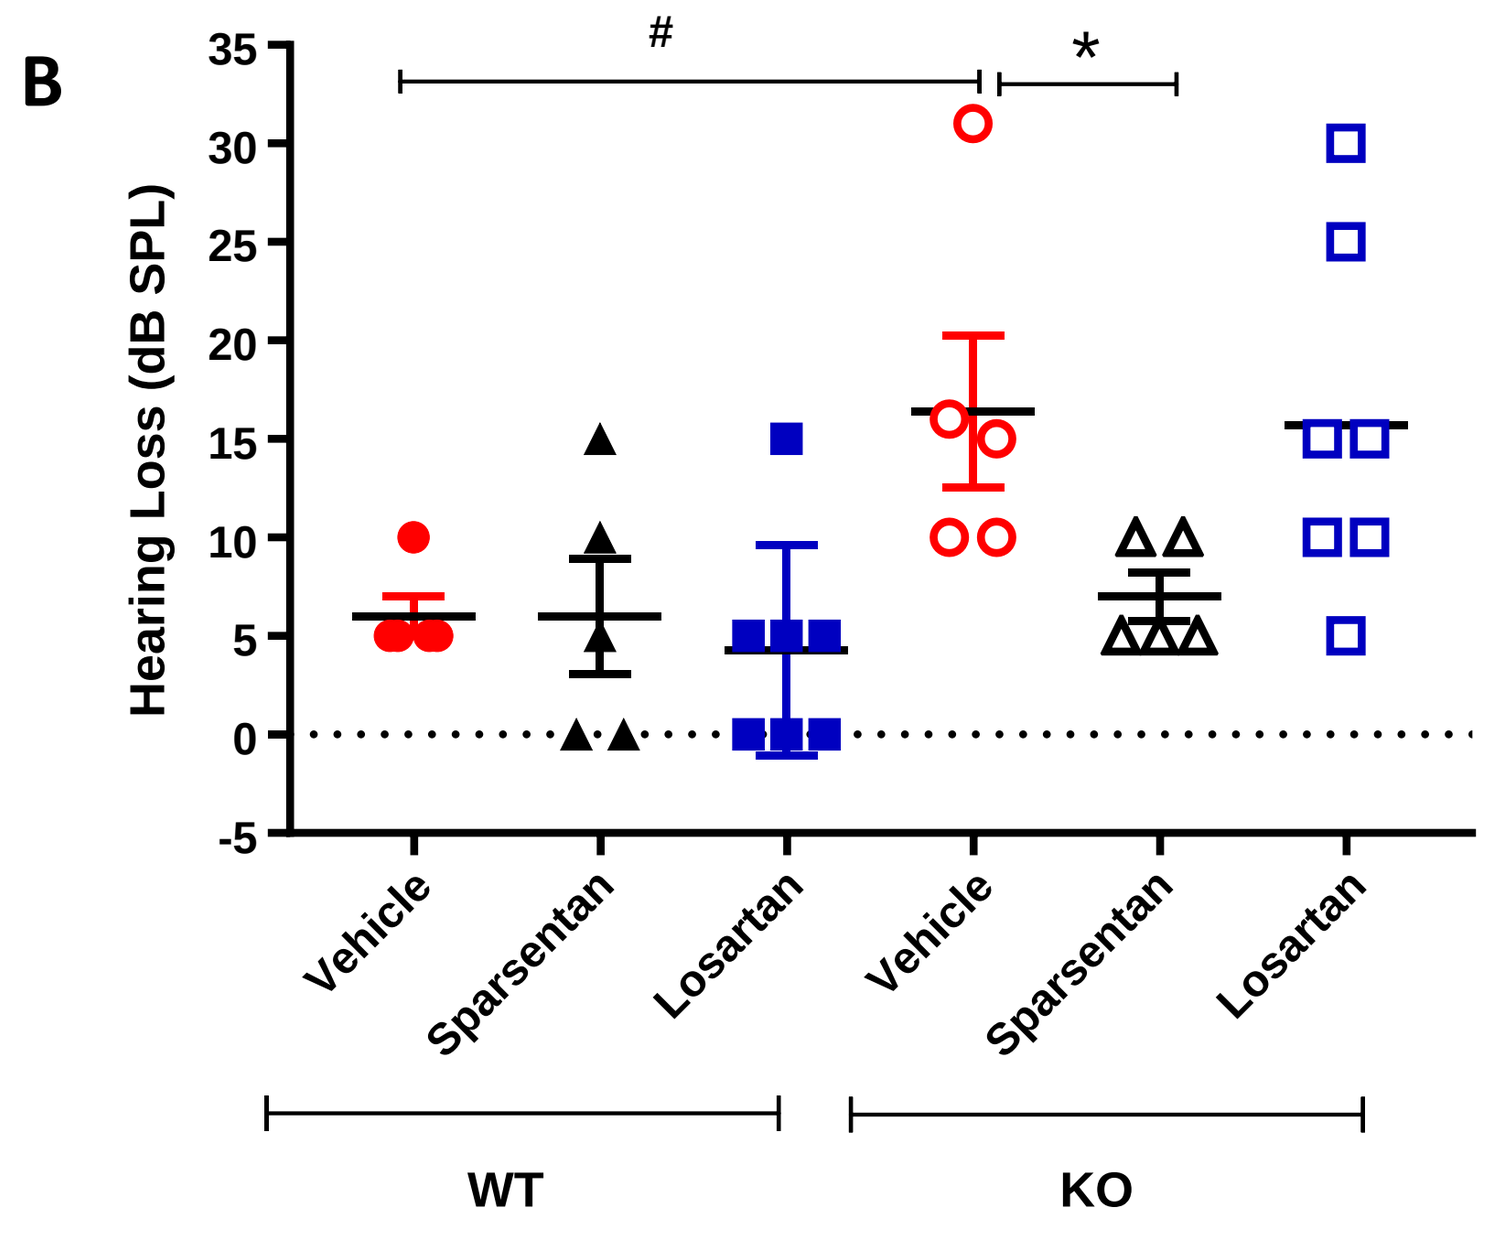
B
Hearing Loss (dB SPL)
Vehicle
Sparsentan
Losartan
Vehicle
Sparsentan
Losartan
WT
KO

Figure 2B. The individual ABR thresholds, group mean ± SD, are depicted for each group at 16 kHz. Note that variability in the KO-LS mice is similar to the threshold spread common in the KO-Vehicle mice. #$p<0.05$ KO-Vehicle vs WT-Vehicle. *$p<0.05$ KO-Vehicle vs KO-SP.

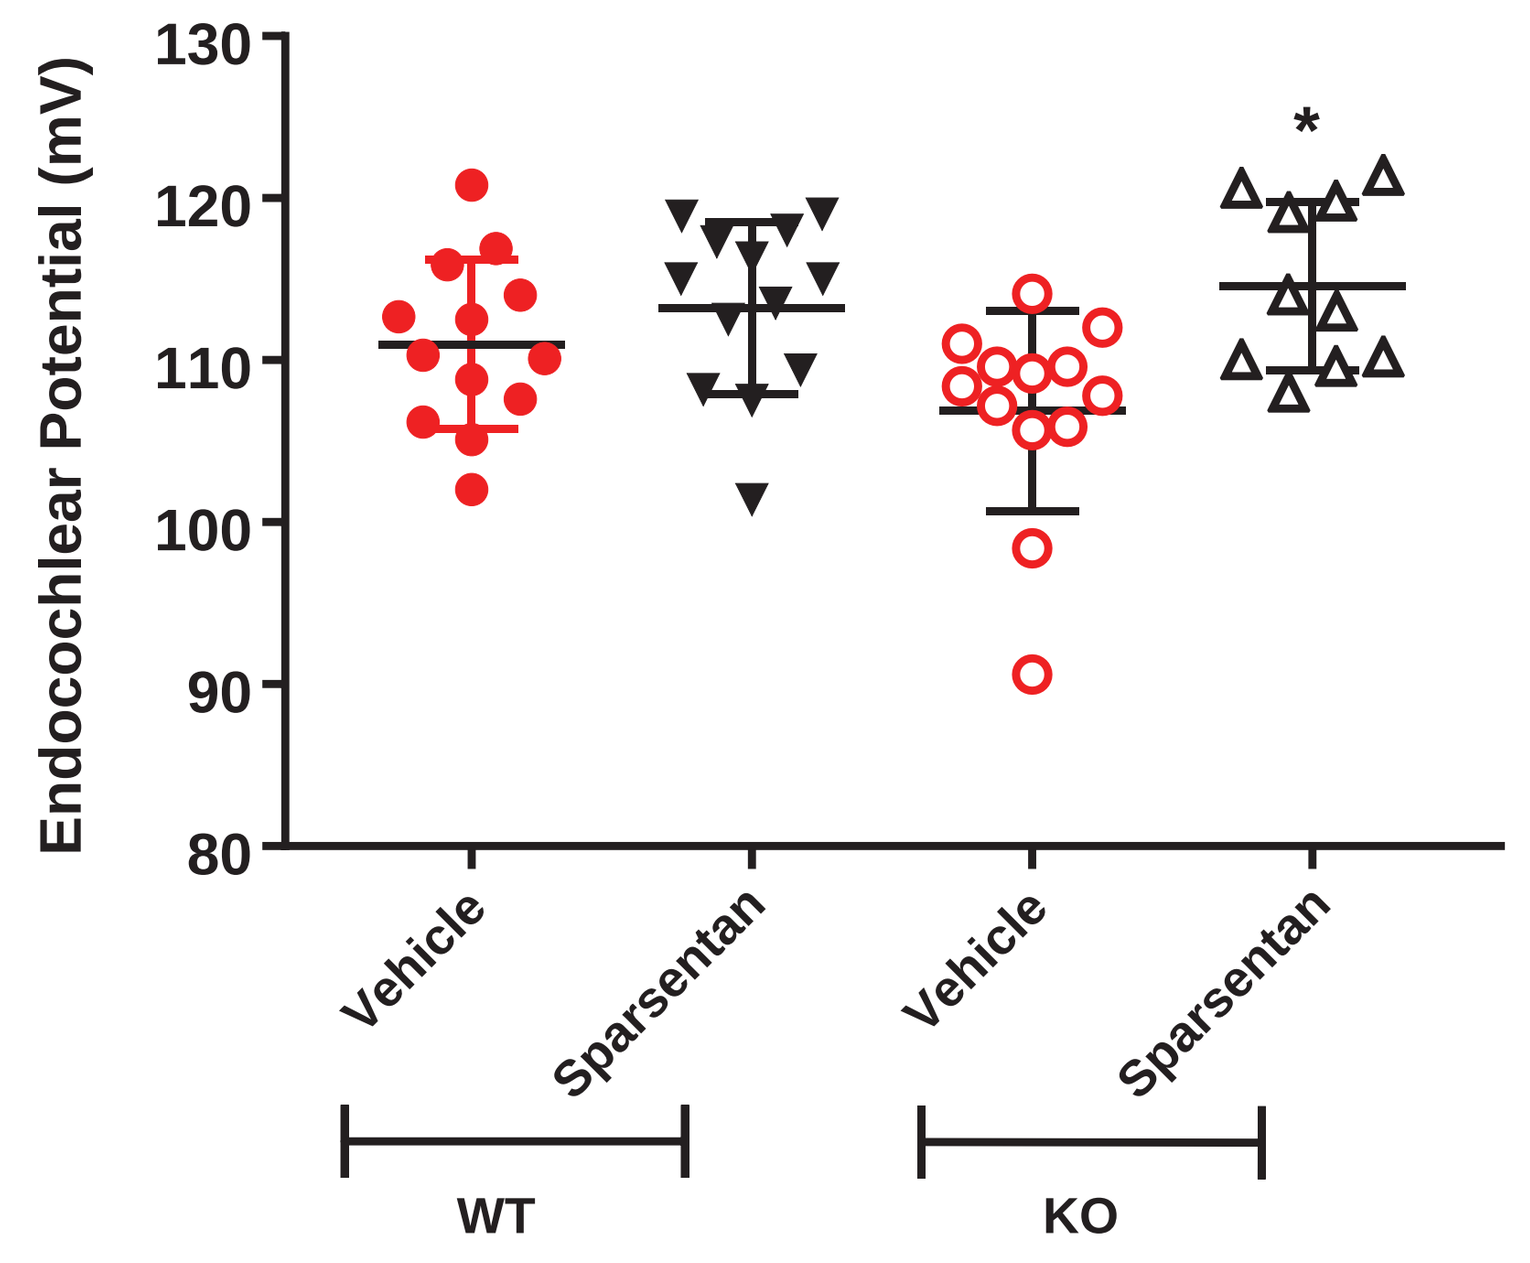
Endocochlear Potential (mV)
Vehicle
Sparsentan
Vehicle
Sparsentan
WT
KO

Figure 4. Sparsentan significantly increases the endocochlear potential in KO mice. *$p<0.05$ KO-Vehicle vs KO-SP. Data are mean ± SD. n=13: WT-Vehicle, WT-SP, KO-Vehicle. n=10: KO-SP. Comparisons with WT-Vehicle and KO-Vehicle in LS-treated mice will be available at a future date.

Figure 6: Sparsentan (200 mg/kg, left panels), but not losartan (10 mg/kg, right panels), treatment prevents accumulation of ECM in the KO mice SCBMs. KO mice were treated with sparsentan, losartan, or vehicle from 3-7 weeks of age. Note that vessels in the spiral ligament do not show changes in staining intensity upon treatment, and thus serve as a control.

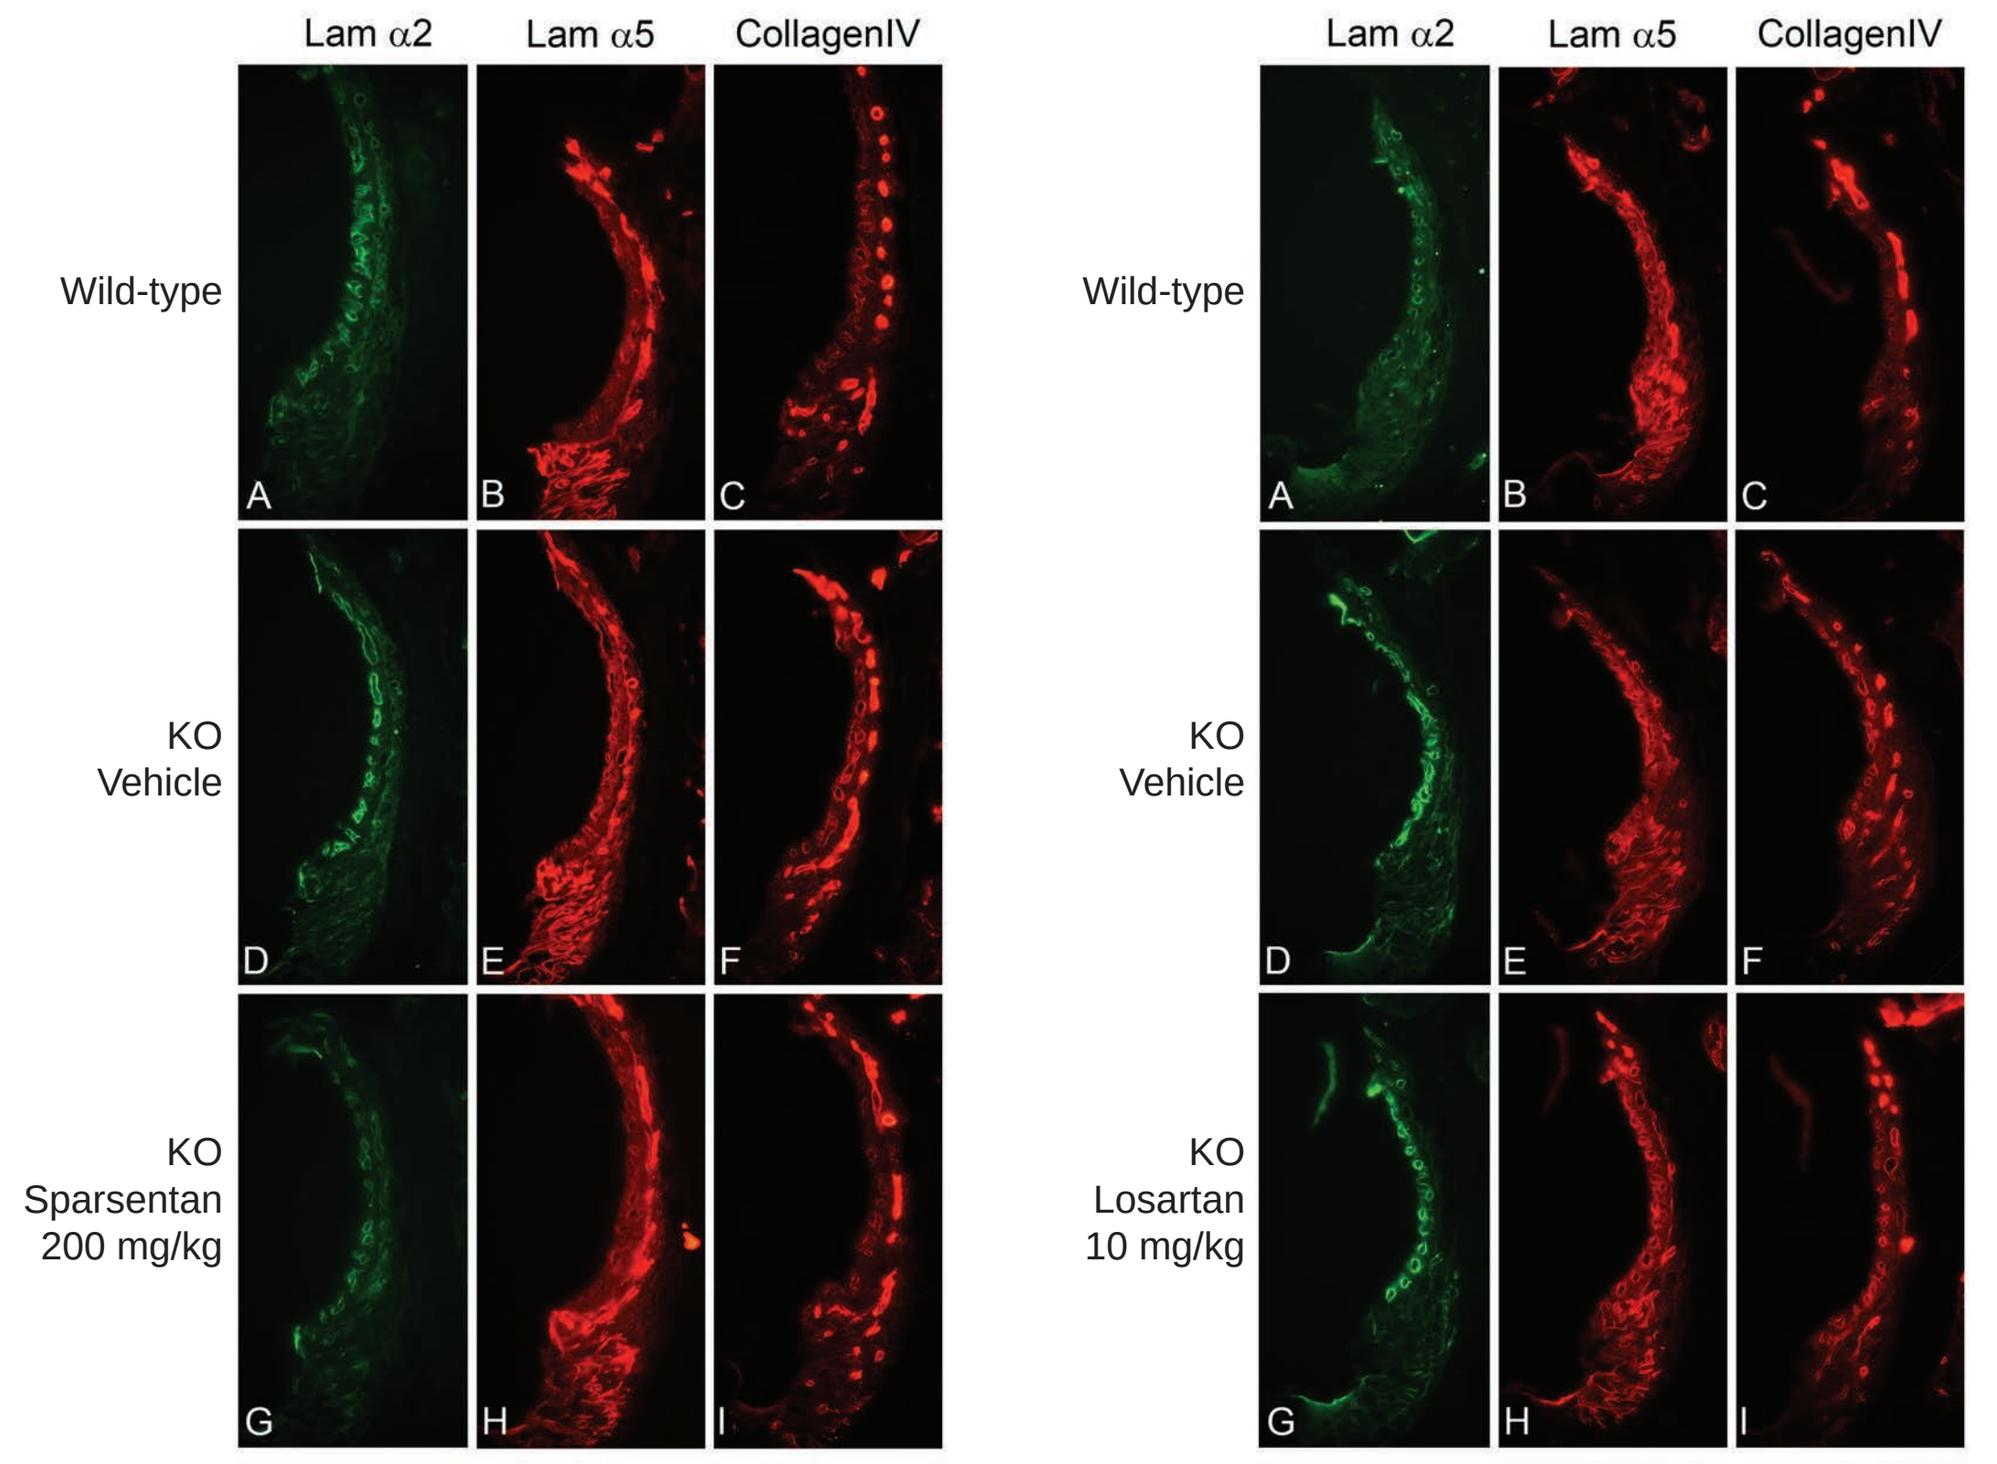
Lam α2
Lam α5
CollagenIV
Wild-type
KO Vehicle
KO Sparsentan 200 mg/kg
KO Losartan 10 mg/kg

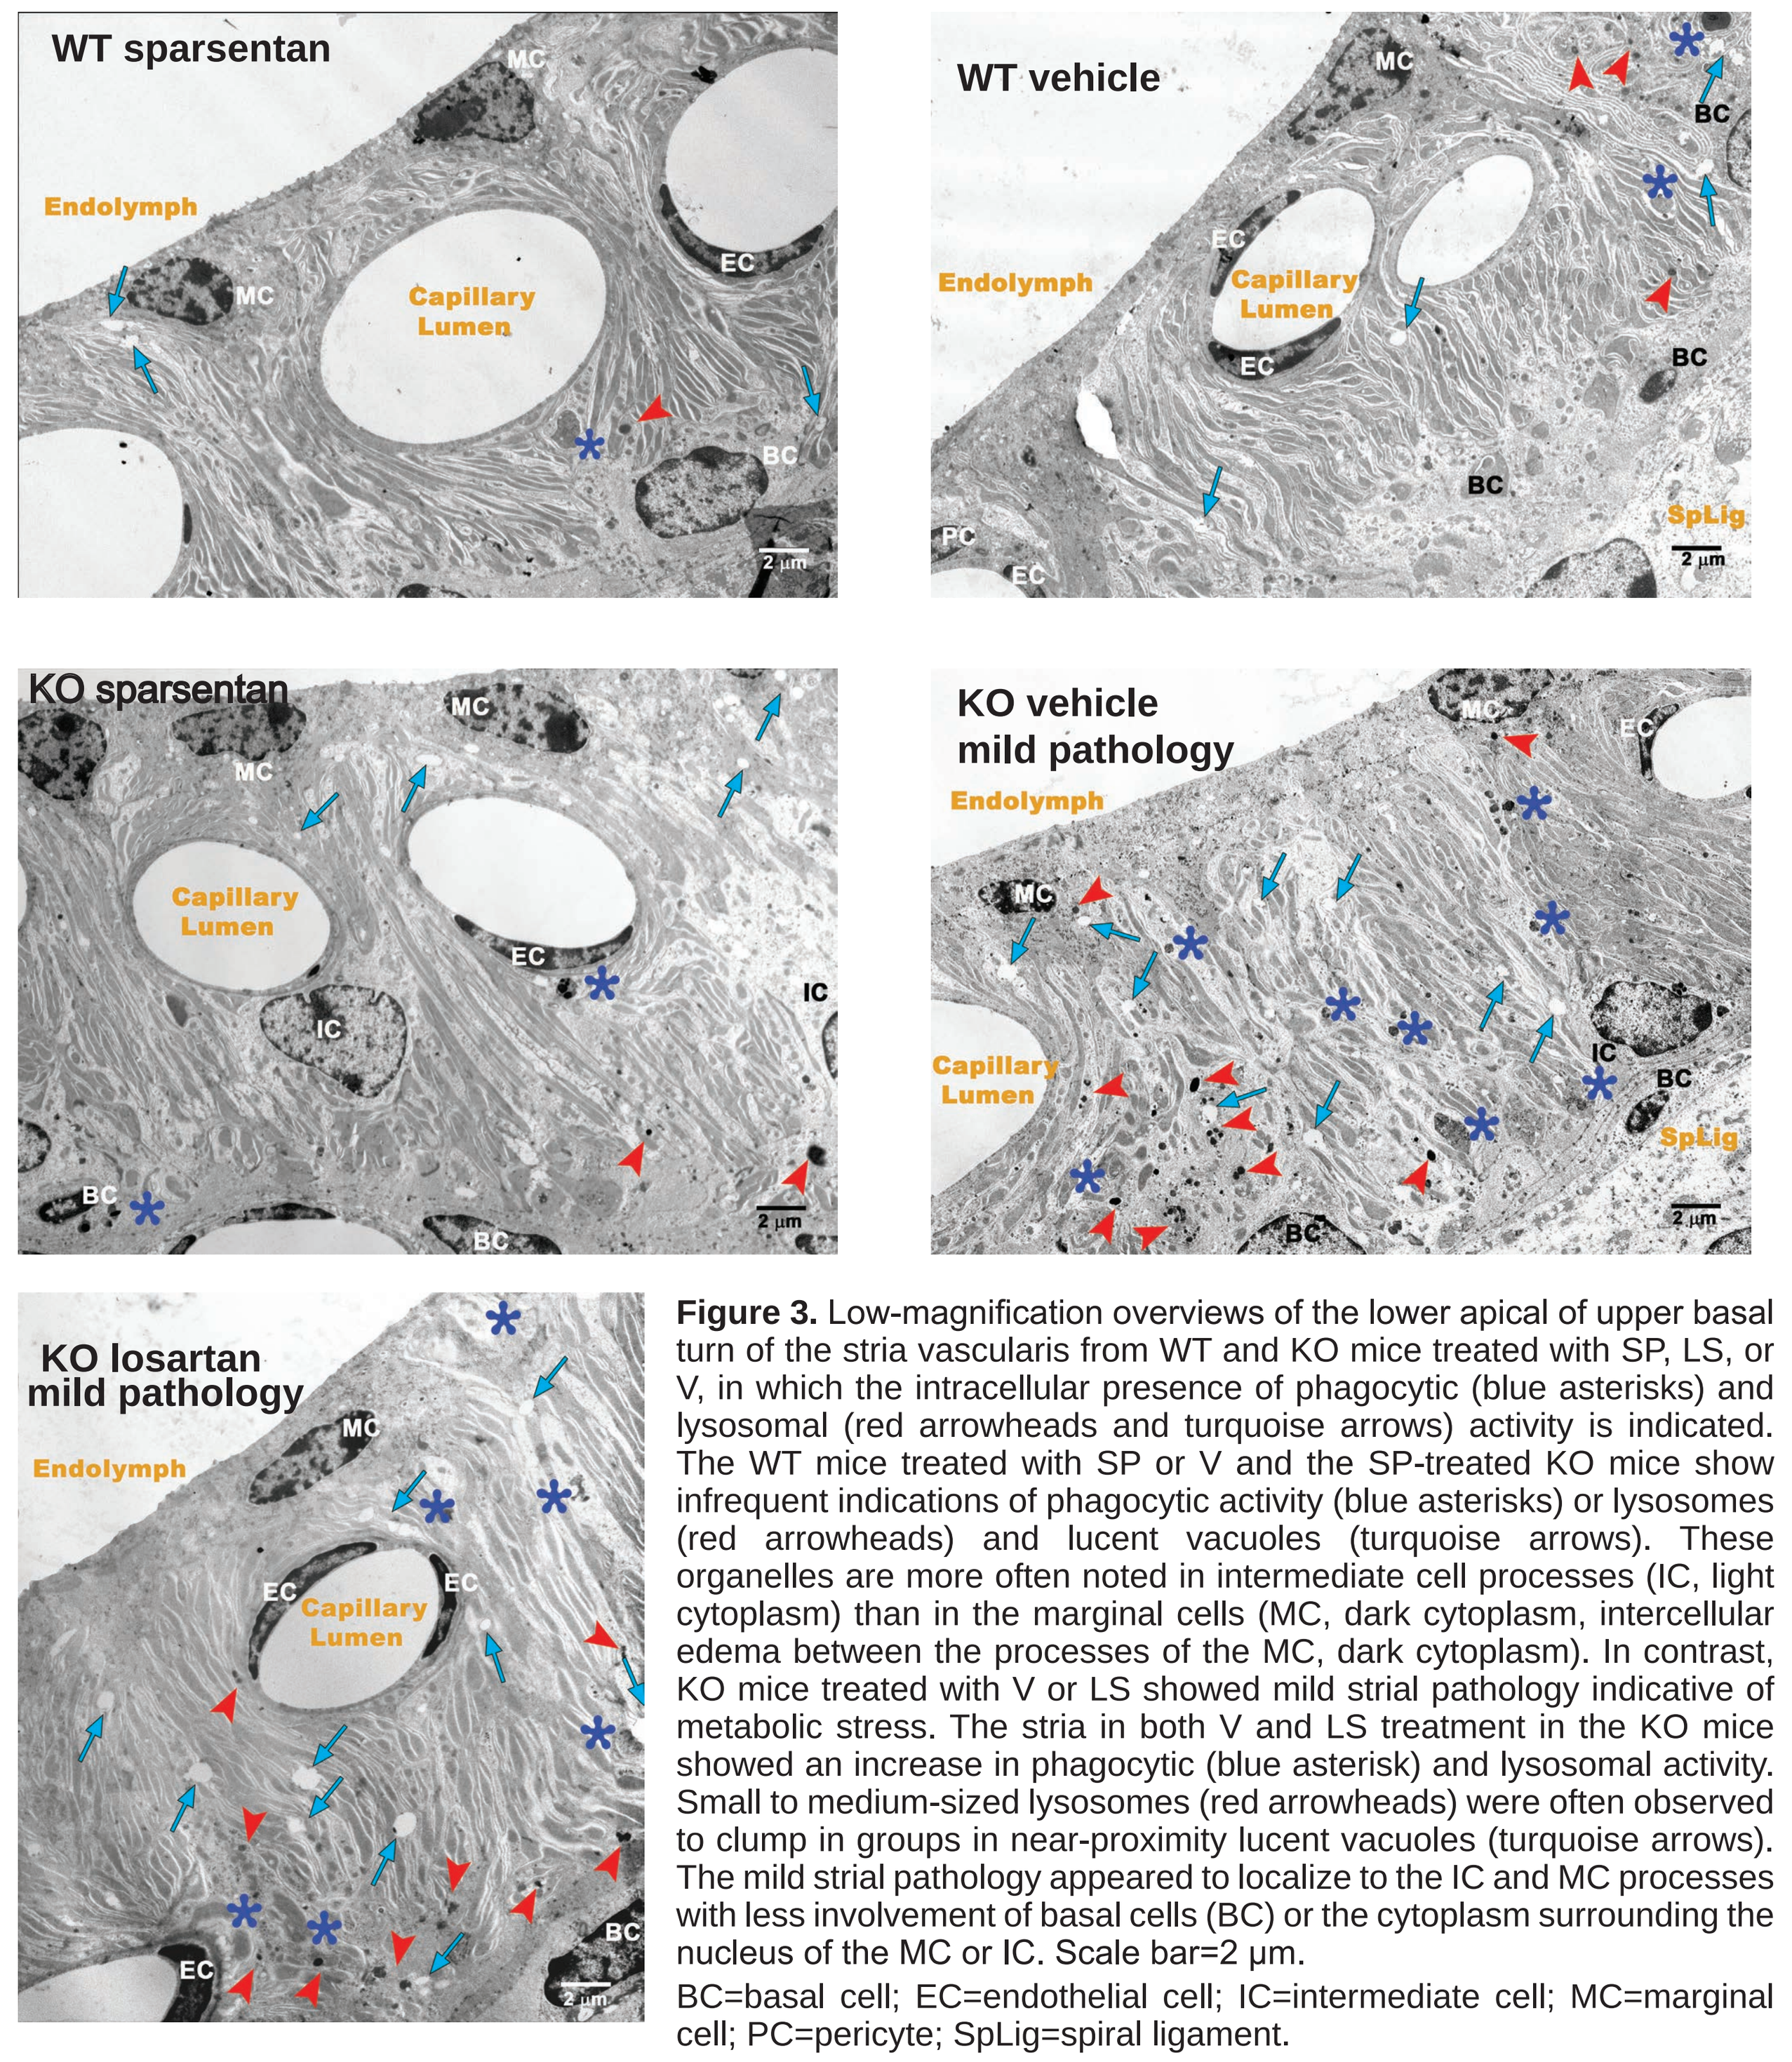
WT sparsentan
WT vehicle
KO sparsentan
KO vehicle mild pathology
KO losartan mild pathology
Endolymph
Capillary Lumen

Figure 3. Low-magnification overviews of the lower apical of upper basal turn of the stria vascularis from WT and KO mice treated with SP, LS, or V, in which the intracellular presence of phagocytic (blue asterisks) and lysosomal (red arrowheads and turquoise arrows) activity is indicated. The WT mice treated with SP or V and the SP-treated KO mice show infrequent indications of phagocytic activity (blue asterisks) or lysosomes (red arrowheads) and lucent vacuoles (turquoise arrows). These organelles are more often noted in intermediate cell processes (IC, light cytoplasm) than in the marginal cells (MC, dark cytoplasm, intercellular edema between the processes of the MC, dark cytoplasm). In contrast, KO mice treated with V or LS showed mild strial pathology indicative of metabolic stress. The stria in both V and LS treatment in the KO mice showed an increase in phagocytic (blue asterisk) and lysosomal activity. Small to medium-sized lysosomes (red arrowheads) were often observed to clump in groups in near-proximity lucent vacuoles (turquoise arrows). The mild strial pathology appeared to localize to the IC and MC processes with less involvement of basal cells (BC) or the cytoplasm surrounding the nucleus of the MC or IC. Scale bar=2 µm.

BC=basal cell; EC=endothelial cell; IC=intermediate cell; MC=marginal cell; PC=pericyte; SpLig=spiral ligament.

Turquoise arrows=lucent vacuoles; blue asterisks=phagocytic whorls or multivesicular bodies; red arrowheads=lysosomes.

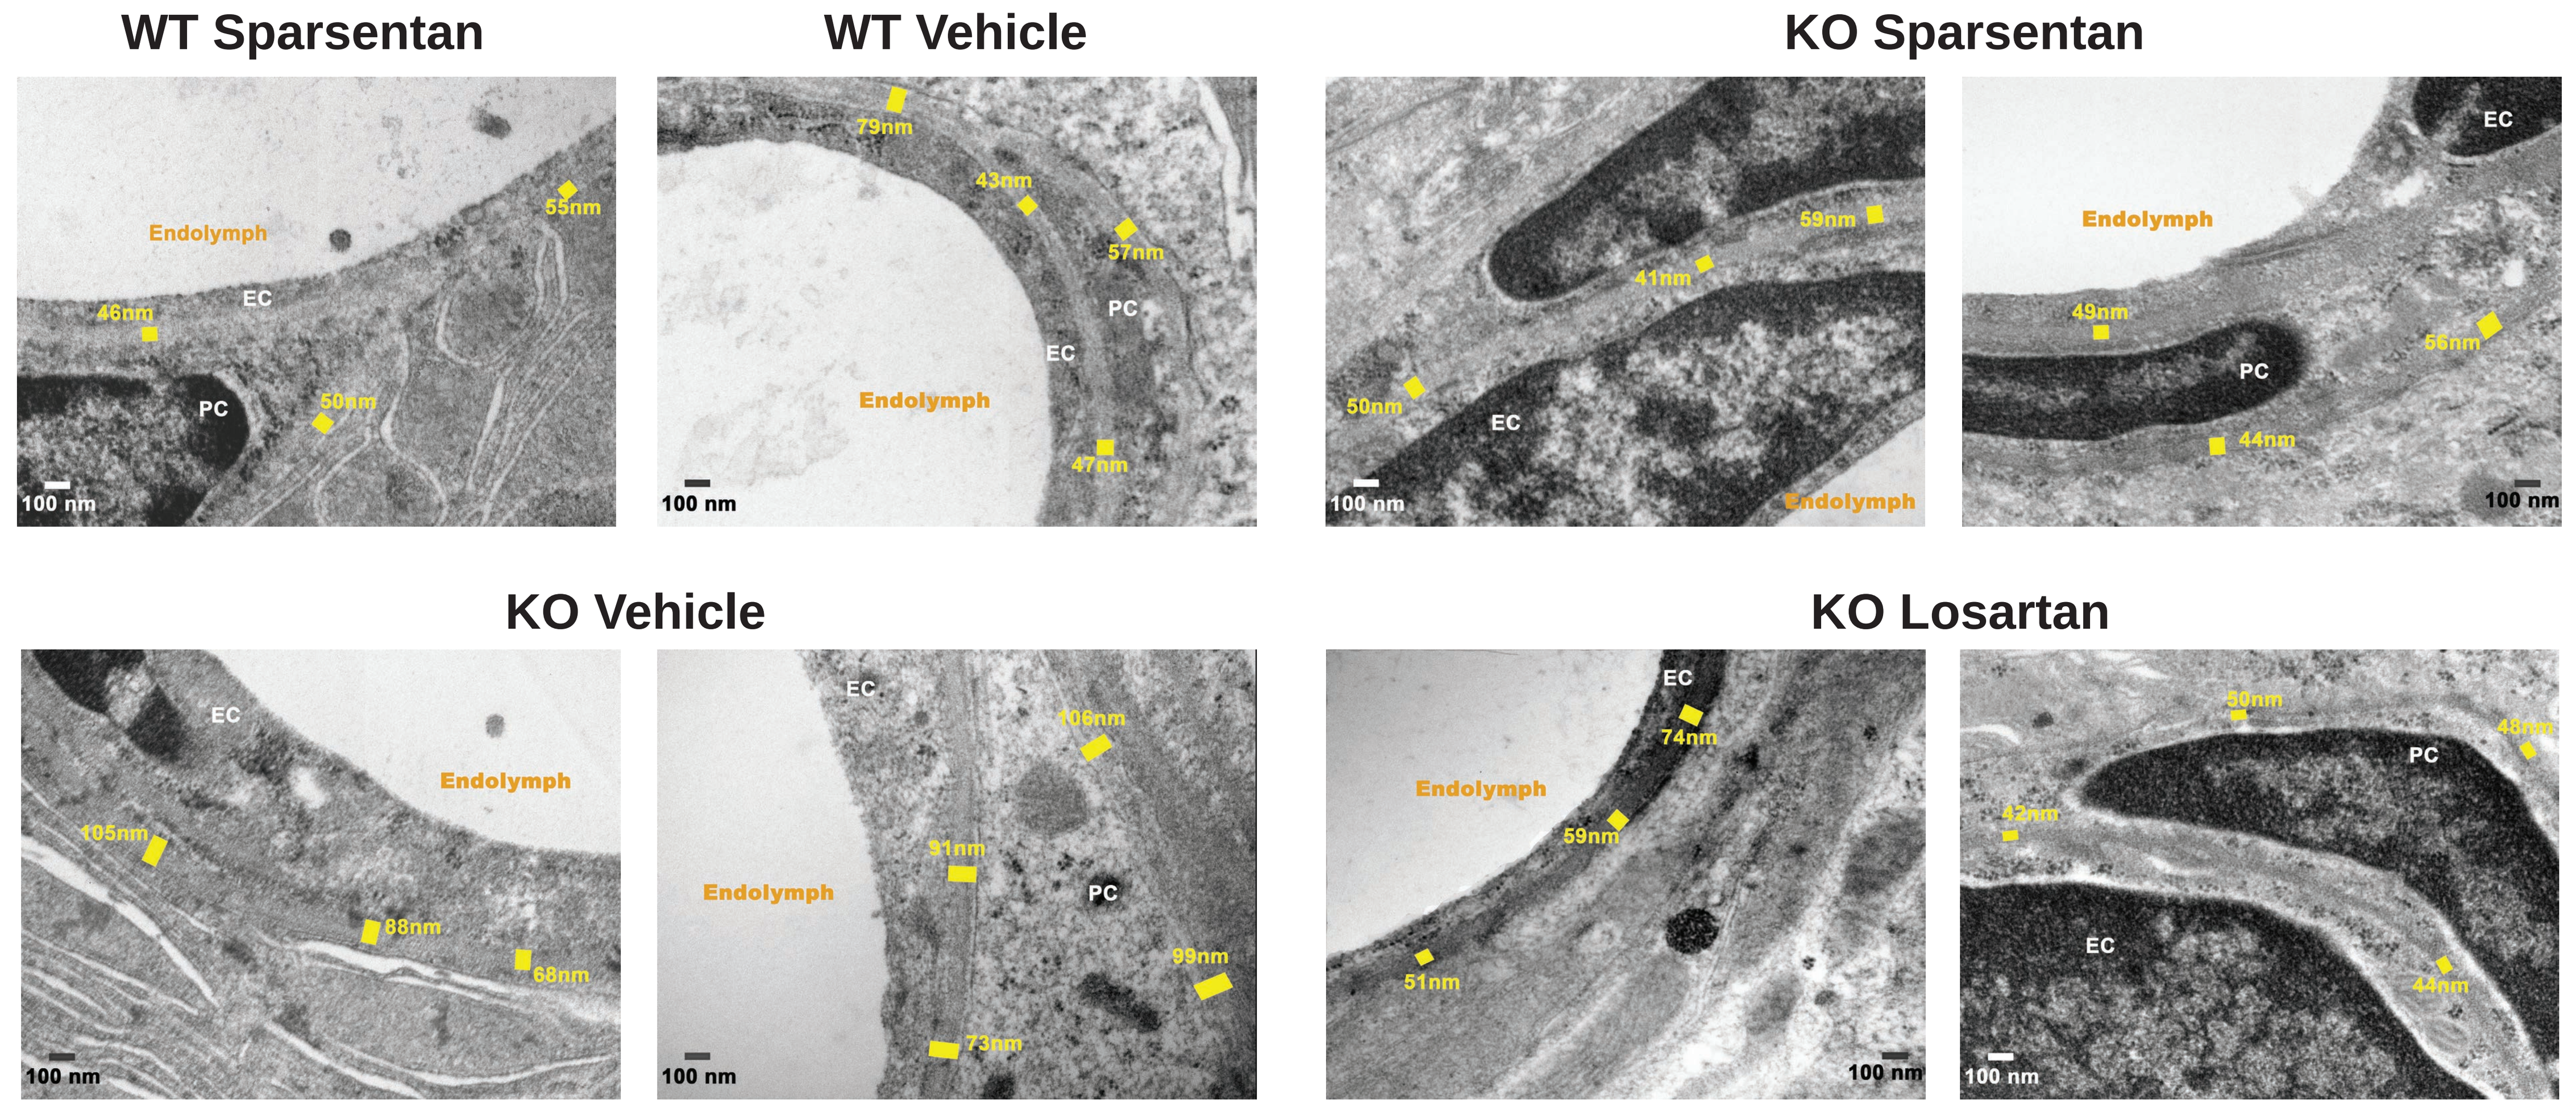
WT Sparsentan
WT Vehicle
KO Sparsentan
KO Vehicle
KO Losartan
Endolymph
100 nm

Figure 5. The measured SCBM width (in yellow for adjacent yellow bar) is shown in representative images from each treatment group. Measures were taken for the SCBM that surrounds the EC and PC as well as for the BM between the two cell types. As depicted in the images above and in the graph (right), treatment with SP attenuated the progressive SCBM thickening that occurs in KO mice. Treatment with LS also attenuated the SCBM thickness; however, the attenuation was less consistent (see graph: data are presented as mean ± SD [n=5]. #$p<0.05$ KO-Vehicle vs WT-Vehicle. *$p<0.05$ KO-Vehicle vs KO-SP or KO-LS. Comparisons with WT-Vehicle in LS-treated mice will be available at a future date). The average SCBM in two LS-treated mice had thinned to less than the normal BM width of 50-60 nm while the remaining three mice showed average SCBM widths consistent with those of WT mice. Note that the variability in the SCBM thickness measures in both LS- and V-treated KO mice is also present in the 16-kHz threshold hearing loss post-noise recovery (Fig 2B). Scale bar=100 nm.

EC=endothelial cell; PC=pericyte.

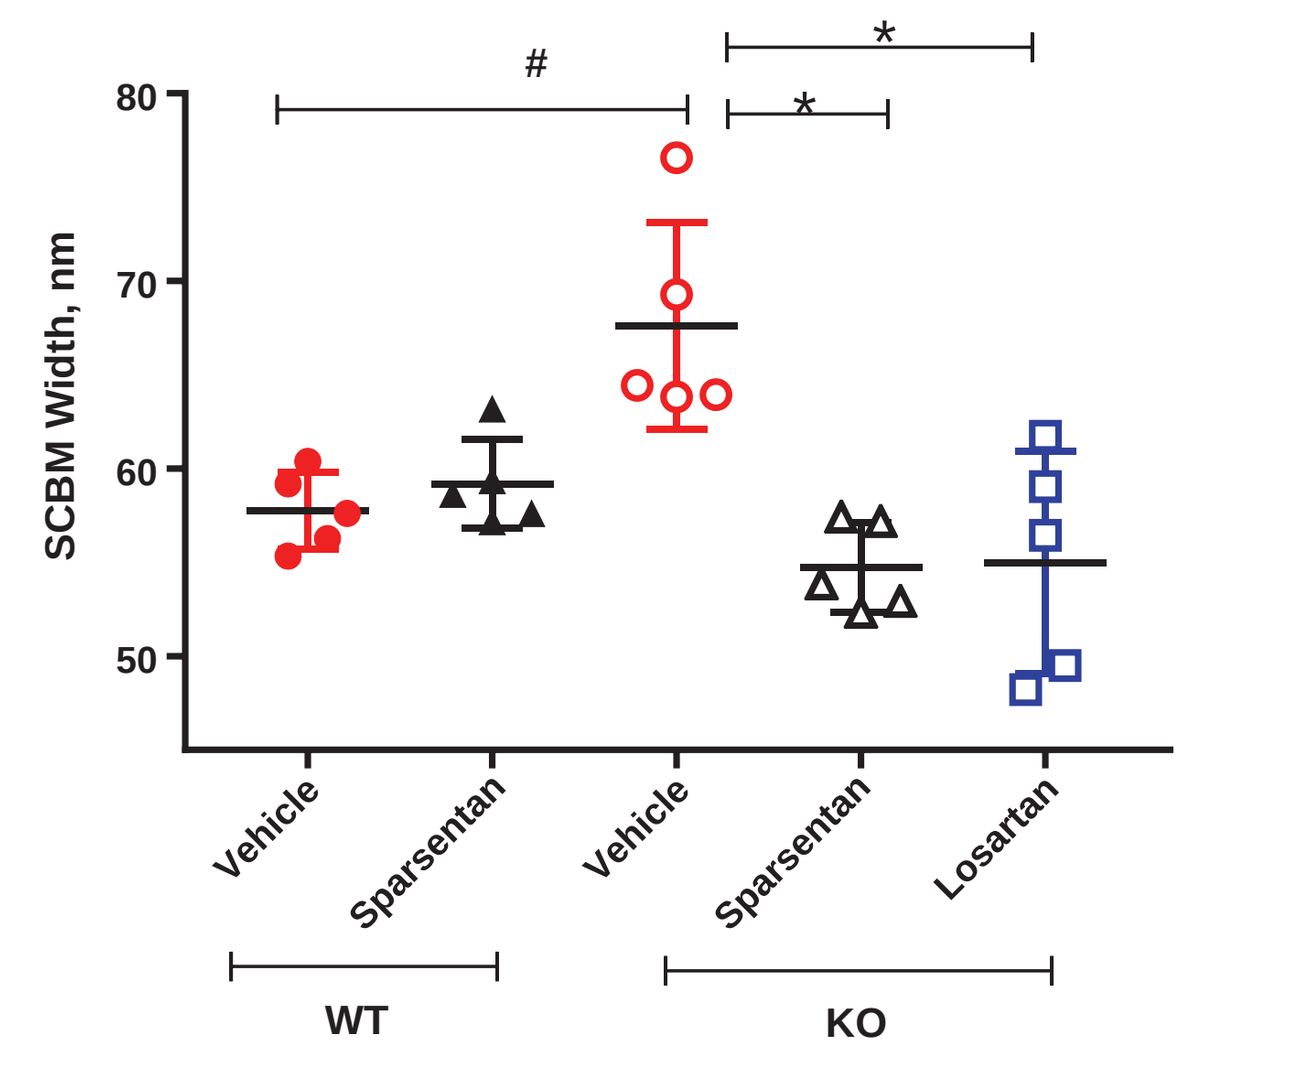
SCBM Width, nm
Vehicle
Sparsentan
Vehicle
Sparsentan
Losartan
WT
KO

Conclusions

- Sparsentan attenuated cochlear pathology and noise-induced hearing loss, indicating that sparsentan is capable of delaying the structural and functional auditory changes in Alport (KO) mice.
 - Losartan does not protect Alport mice from noise-induced hearing loss.
 - Sparsentan and losartan both prevent SCBM thickening, but this is not translated into a functional hearing improvement for losartan.
 - Sparsentan but not losartan tends to ameliorate the increase in ECM proteins and improve strial morphology.
- Results of the current sparsentan studies in Alport mice in conjunction with previous results on renal pathology,[3,4] if translated to the clinic, may present a novel therapeutic approach for reducing both hearing loss and renal injury in patients with Alport syndrome.

References

1. Dufek B, et al. *Kidney Int*. 2016;90(2):300-310.
2. Meehan DT, et al. *Hearing Res*. 2016;341:100-108.
3. Cosgrove D, et al. Presented at the American Society of Nephrology Kidney Week, October 23-28, 2018, San Diego, CA. Poster FR-PO995.
4. Cosgrove D, et al. Presented at the American Society of Nephrology Kidney Week, November 5-10, 2019, Washington, DC. Poster SA-PO576.

Disclosures

CJ and RK: Employees of Retrophin, Inc., and may have an equity or other financial interest in Retrophin, Inc.

MAG, DC, JH, GP, RG, BD, DD, DM: Received a research grant from Retrophin, Inc.

DC and MAG were funded by co-PI R01-DC-015385.

The mouse Alport studies were conducted in the laboratories of Michael Anne Gratton at Washington University in St. Louis and Dominic Cosgrove at Boys Town National Research Hospital and were funded by Retrophin, Inc. Editorial support was provided by Courtney Breuel, ELS, of MedVal Scientific Information Services, LLC (Princeton, NJ), and was funded by Retrophin, Inc.

Presented at the Association for Research in Otolaryngology, January 25-29, 2020, San Jose, CA, USA.