The Dual Endothelin/Angiotensin II Receptor (ET_AR/AT_1R) Antagonist Sparsentan Slows Renal Disease, Improves Lifespan, and Attenuates Hearing Loss in Alport Mice: Comparison with Losartan and Atrasentan

Dominic Cosgrove[1], Brianna Dufek[1], Duane Delimont[1], Dan Meehan[1], Gina Samuelson[1], Jared Hartsock[2], Grady Phillips[2], Ruth Gill[2], James Hasson[3], Celia Jenkinson[3], Radko Komers[3], Michael Anne Gratton[2]

[1]Boys Town National Research Hospital, Omaha, NE, USA, [2]Washington University in St. Louis, St. Louis, MO, USA, [3]Retrophin, Inc., San Diego, CA, USA

Background

In Alport syndrome (AS), ET_AR activation is important in renal and inner ear pathologies.[1-3] Currently, angiotensin converting enzyme inhibitors (ACEi) or angiotensin II receptor (AT_1R) blockers are the standard of care for Alport patients; however, these drugs have not been shown to have an impact on hearing. Previously, we showed that sparsentan (SP) prevented increases in proteinuria, fibrosis, glomerulosclerosis, and glomerular basement membrane dysmorphology and prevented noise-induced hearing loss in AS mice (presentations at ASN 2018, 2019).

Objective

To compare in wild-type (WT) or $COL4A3^{-/-}$ autosomal AS mice the effect of SP, the AT_1R antagonist losartan (LS), and the ET_AR antagonist atrasentan (ATR) on lifespan and proteinuria in AS mice treated from 4 weeks (W), and the effect of SP and LS on noise-induced hearing loss and inner ear pathology in mice treated from 3 W to 8.75 W.

Methods

Study design

- $COL4A3^{-/-}$ (AS) or WT mice (male and female) on the 129/Sv background were treated with vehicle (V) or SP (60, 120, or 200 mg/kg) daily by oral gavage or LS (10 mg/kg) or ATR (7.5 mg/kg females; 10 mg/kg males) administered in the drinking water. In hearing studies where treatment was initiated at 3 W of age, LS was administered by oral gavage for the first week. For survival studies, treatment was initiated at 4 W and compared with survival in AS mice treated with V. A schematic of the study design is shown in **Figure 1**.

Figure 1. Study design in Alport mice

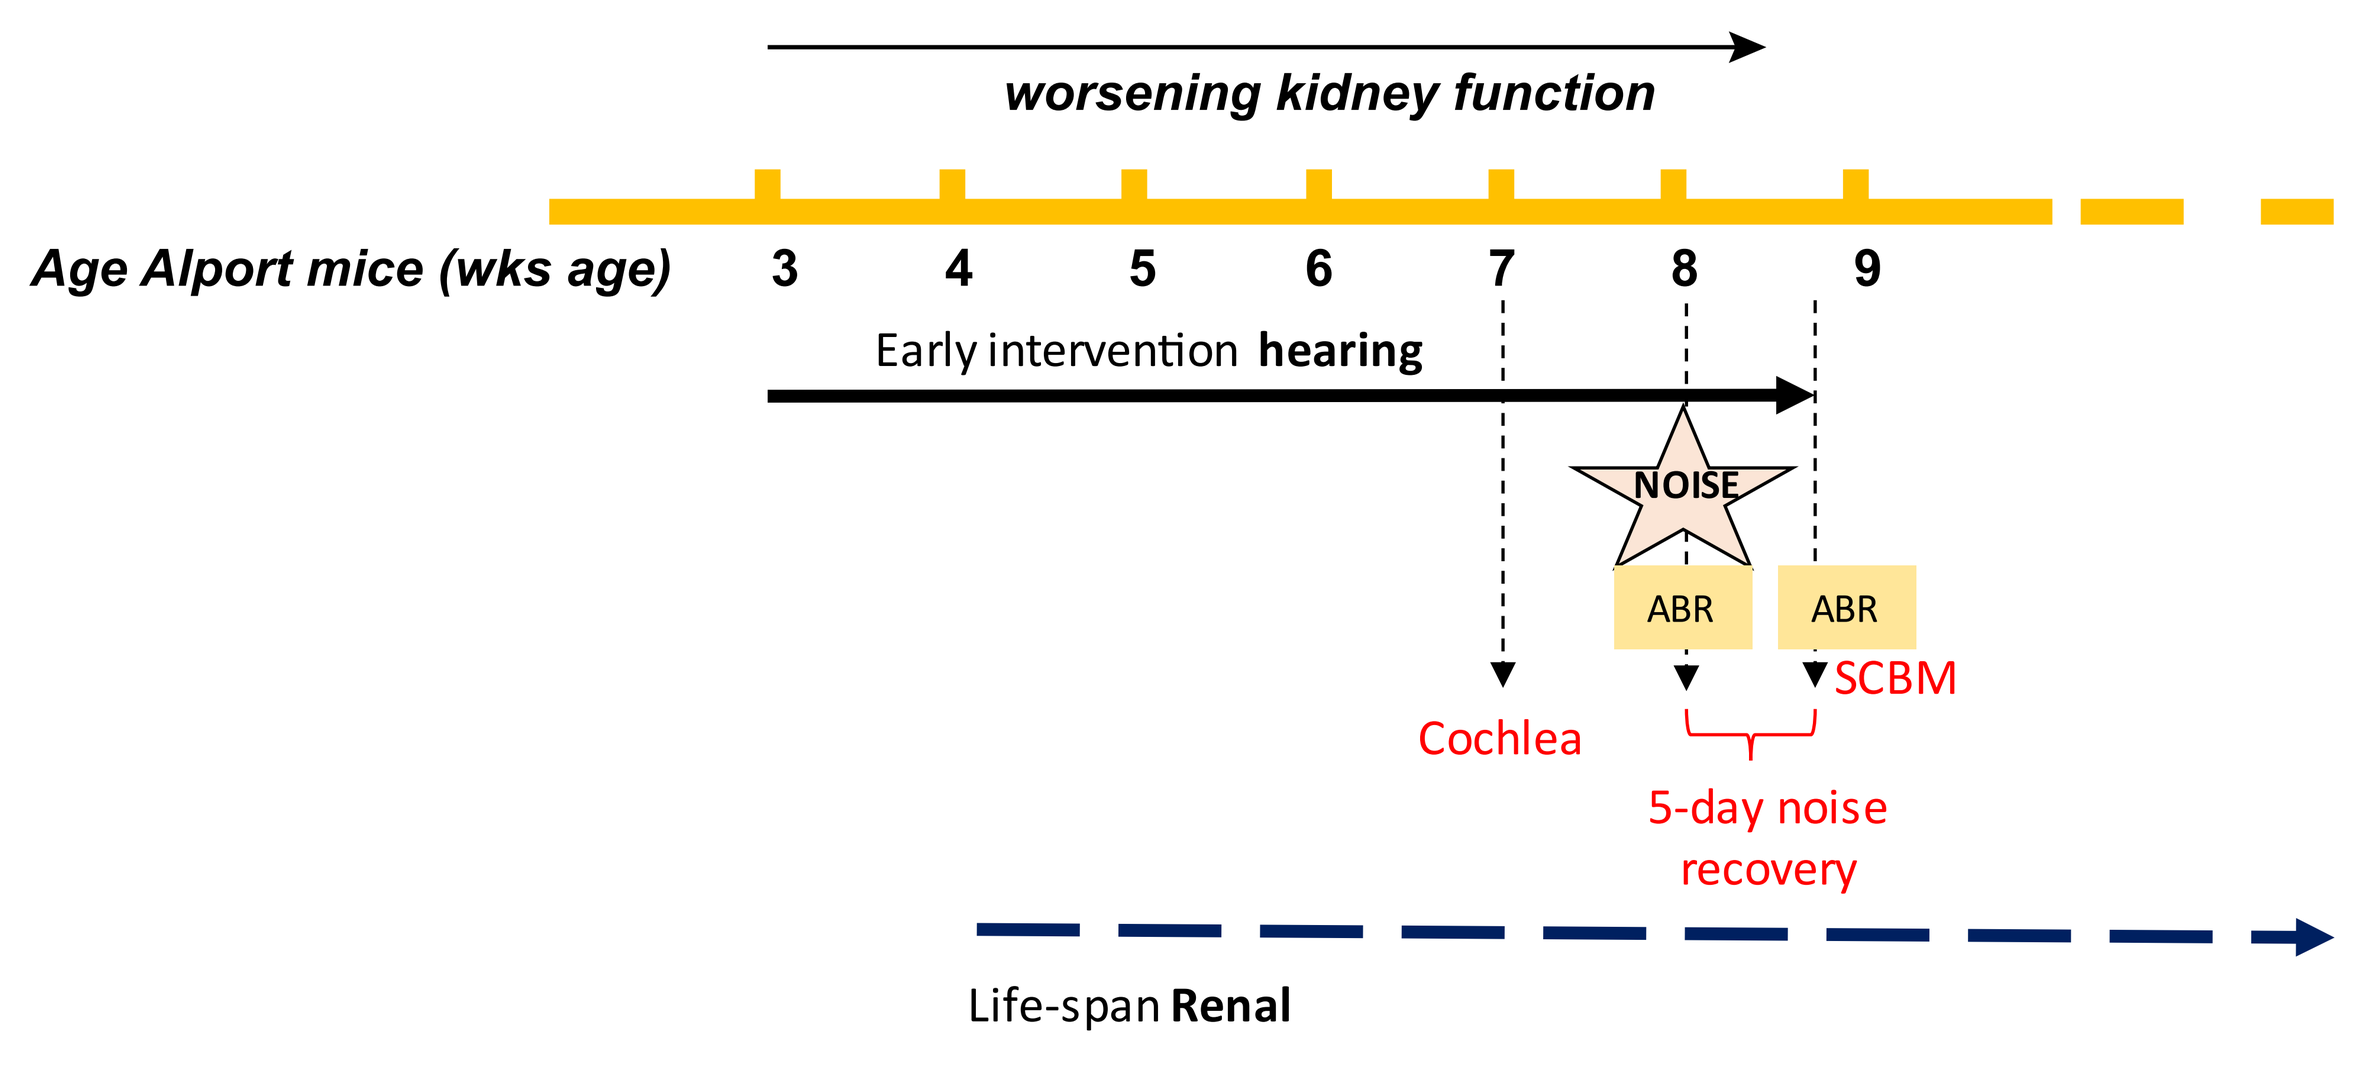
worsening kidney function
Age Alport mice (wks age)
3
4
5
6
7
8
9
Early intervention hearing
NOISE
ABR
ABR
SCBM
Cochlea
5-day noise recovery
Life-span Renal

ABR, auditory brainstem response; SCBM, strial capillary basement membrane.

Sample collection and analysis

Renal and survival studies

- Spot urine was sampled between 11:00 am-12:00 pm pre-study and weekly during treatment and analyzed for protein and creatinine.
- During the survival studies, mice were terminated when they had lost 10% of their peak body weight.
 - Glomerular filtration rate (GFR) was determined in 2 WT mice and 1 AS mouse at 8 W of age and in 1 AS mouse at 9 W of age using a Medibeacon device (MediBeacon, Mannheim, Germany).[4] Mice were anesthetized with isoflurane, and the transdermal device mounted via double-sided adhesive tape onto each shaved animal's neck. Background signal was recorded for 5 minutes prior to retro-orbital injection of 150 mg/kg FITC-Sinistrin. Animals were conscious during the recording (approximately 1.5 hours).

Hearing studies

- Hearing was assessed at 8 W (n=5/grp) by auditory brainstem response (ABR). The mice were exposed to a 10-hour moderate noise stress and 5 days post-noise underwent a second ABR analysis.
 - Cochlea were excised at 8.75 W of age after 5.75 W of treatment and strial capillary basement membrane (SCBM) width and ultrastructure were analyzed using transmission electron microscopy (TEM). Digital images (JEOL 1200 EX II, AMT 8 megapixel camera, AMT Image Capture Engine V602) were taken at 40,000x (n=5/grp).
- In a separate study, cochlea were also excised at 7 W after 4 W of treatment. Accumulation of extracellular matrix proteins (ECM) in the SCBM was determined from frozen mid-modiolar cochlear sections incubated with antibodies against laminin α2, laminin α5, and collagen α1(IV) and visualized using a Leica confocal-imaging system.

Data analysis

- GFR was analyzed using Mannheim Pharma and Diagnostics Lab Software (MediBeacon, Mannheim, Germany). The GFR (µl/min) was calculated from the decrease of fluorescence intensity over time (i.e., plasma $t_{1/2}$ of FITC-Sinistrin) using a two-compartment model, the body weight of the mouse, and an empirical conversion factor.
- Hearing loss was calculated by subtracting the ABR hearing threshold for pre-noise from that of post-noise hearing testing. Comparison of active dose to the AS vehicle used t-tests.
- Comparison of the SCBM thickness measures used one-way ANOVA and Tukey's multiple-comparison post-hoc test.
- Analysis of urinary protein-to-creatinine ratio (UP/C) was based on log transformed data and was analyzed by t-test for 4 W. Comparison of AS-V to AS treatments used mixed-model analysis with fixed effects for age, treatment, and treatment by age interaction, and week 4 values as a fixed covariate.
- For all statistical analyses, significance was set at $P<0.05$.

Results

Figure 2. Preliminary data suggest Alport mice tend to have decreased GFR by 8 W of age

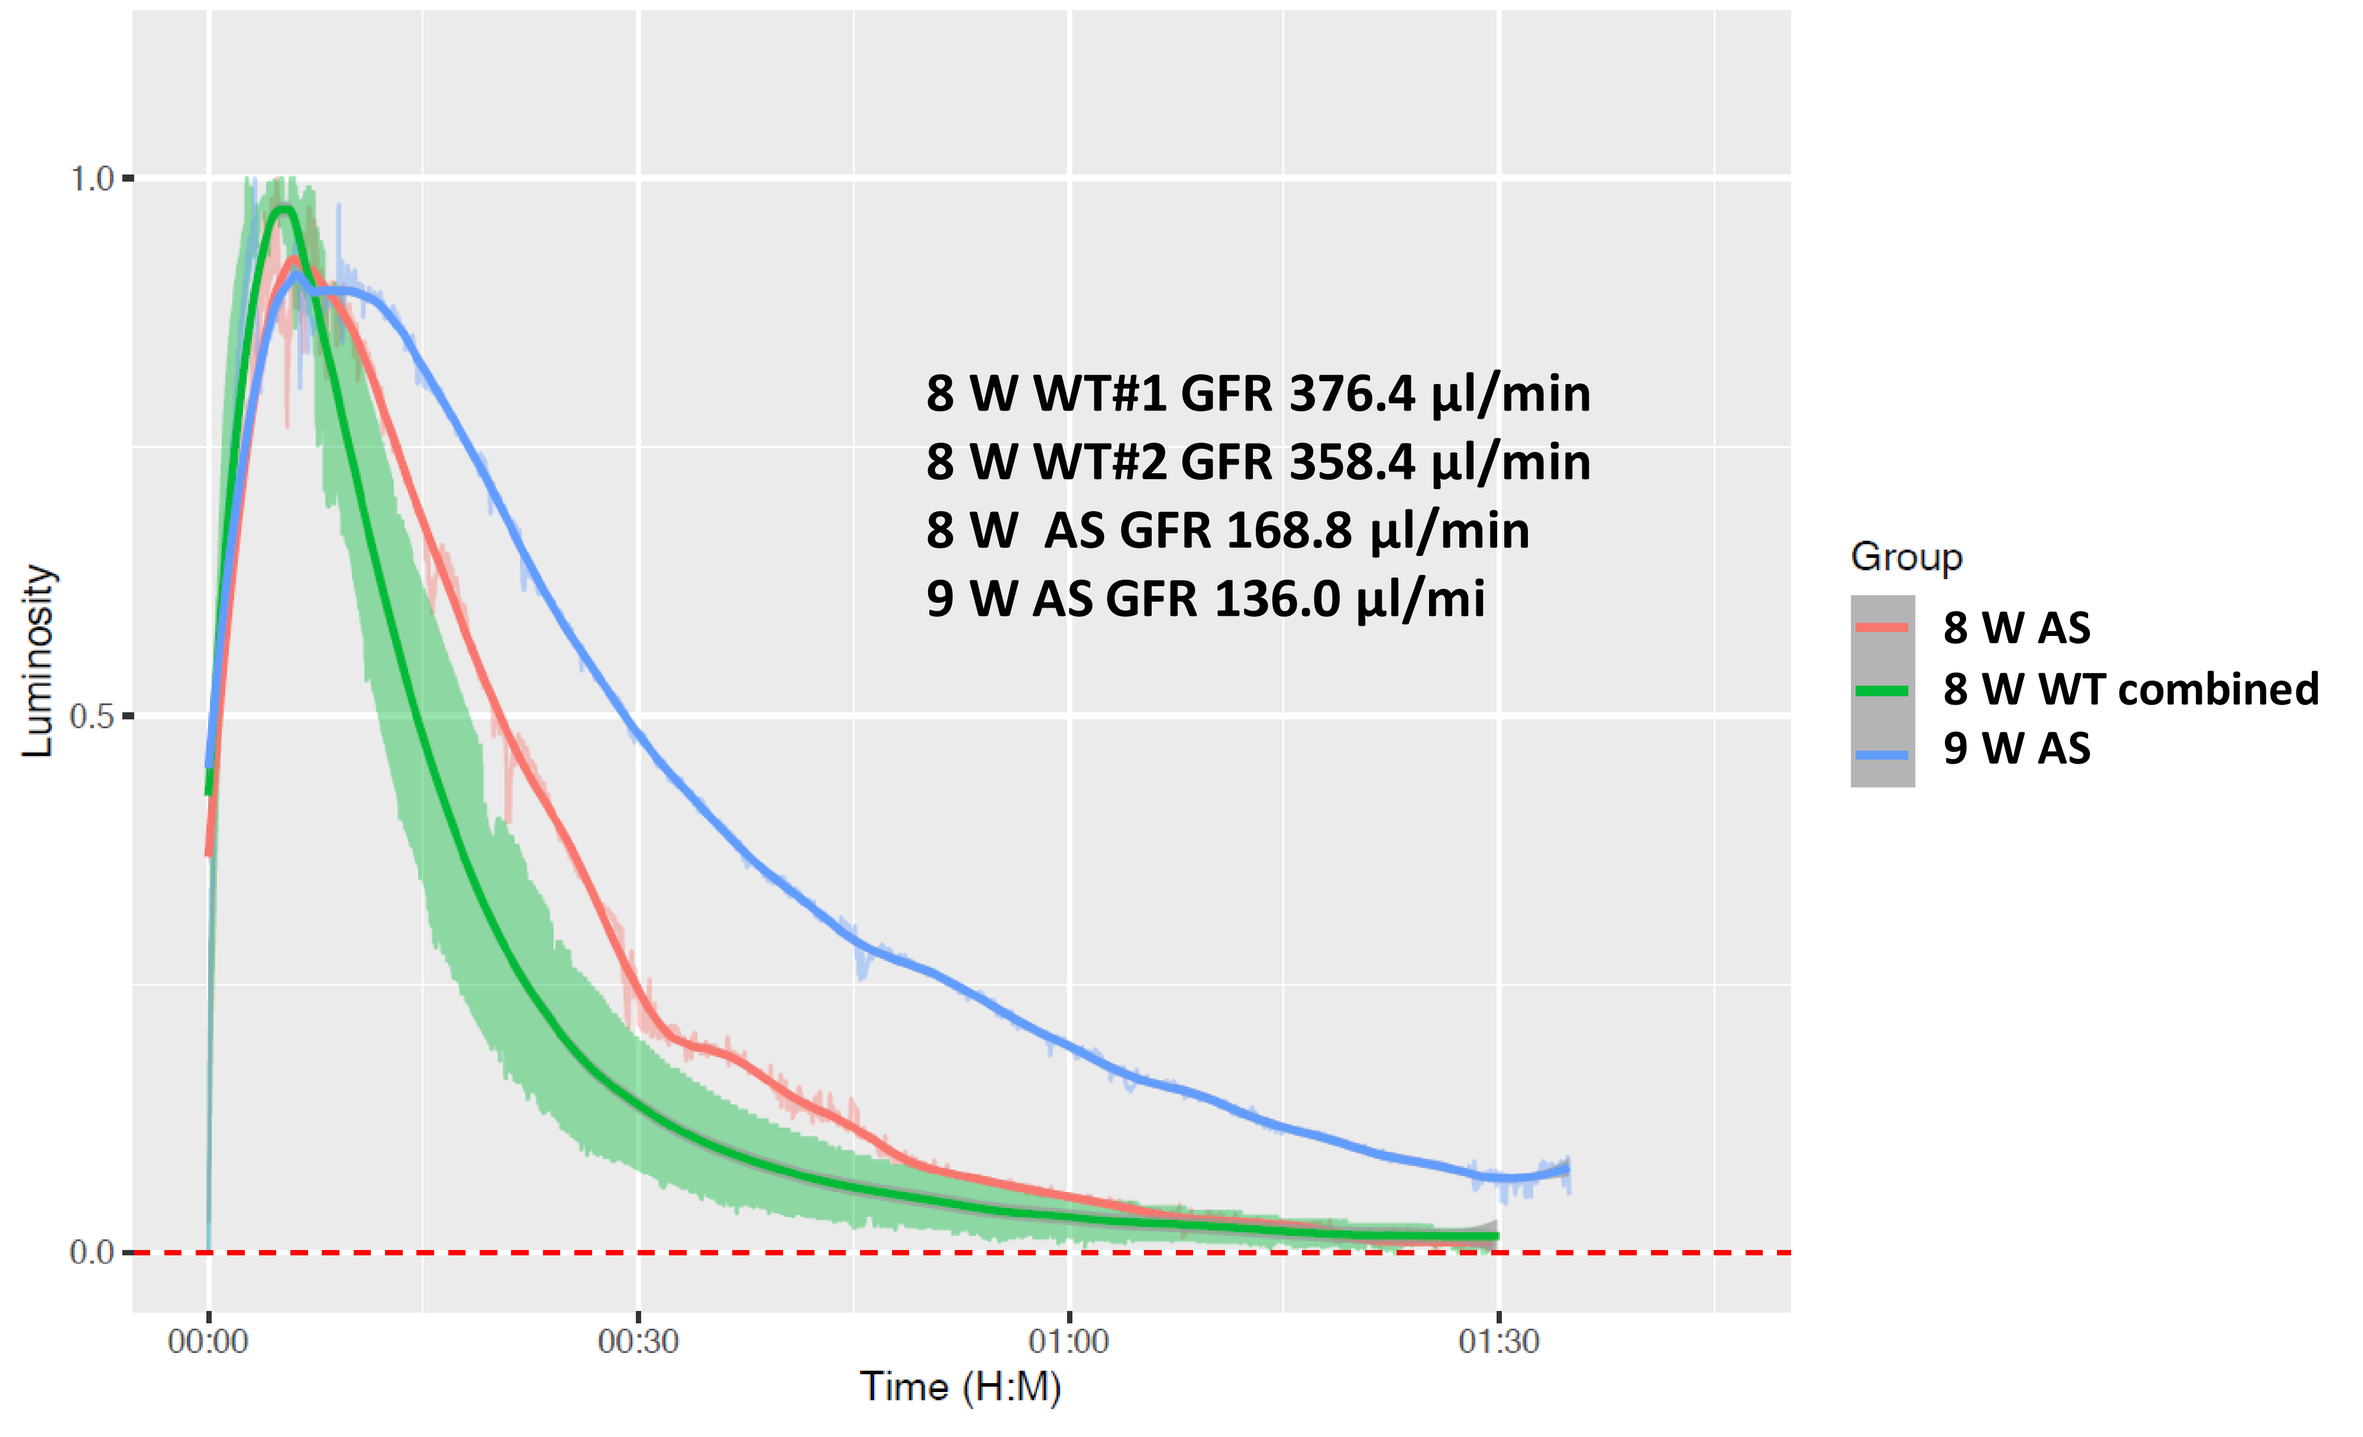
8 W WT#1 GFR 376.4 µl/min
8 W WT#2 GFR 358.4 µl/min
8 W AS GFR 168.8 µl/min
9 W AS GFR 136.0 µl/mi
Group
8 W AS
8 W WT combined
9 W AS
Luminosity
Time (H:M)

- The ability to detect a decrease in GFR with age in AS mice highlights the opportunity to study the effect of therapeutics on a clinically translatable end-point.

Survival studies with sparsentan (from 4 W): Renal

Figure 3. Sparsentan increases survival and delays increases in the UP/C of AS mice compared to losartan or atrasentan

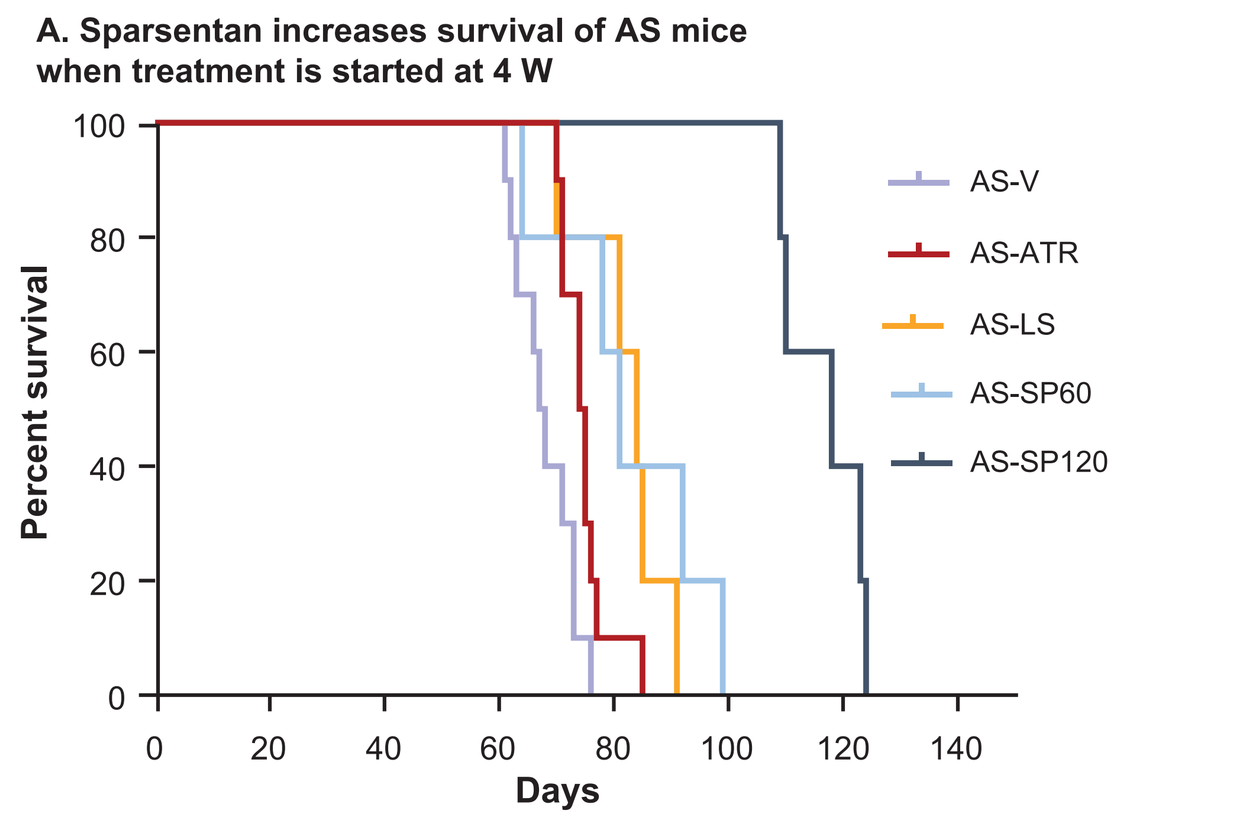
A. Sparsentan increases survival of AS mice when treatment is started at 4 W
Percent survival
Days
AS-V
AS-ATR
AS-LS
AS-SP60
AS-SP120

Treatment	Median Life Span , days (CI)[a]
AS-V	67.5 (61,73)
AS-ATR	74.5 (70,76)
AS-LS	84 (70, 91)
AS-SP60	81 (64,99)
AS-SP120	118.0 (109, 124)*

Data are presented as median (CI). *$P<0.05$ vs AS vehicle.
[a]At study start, AS-V and AS-ATR n=10; AS-LS, AS-SP60, AS-SP120 n=5.

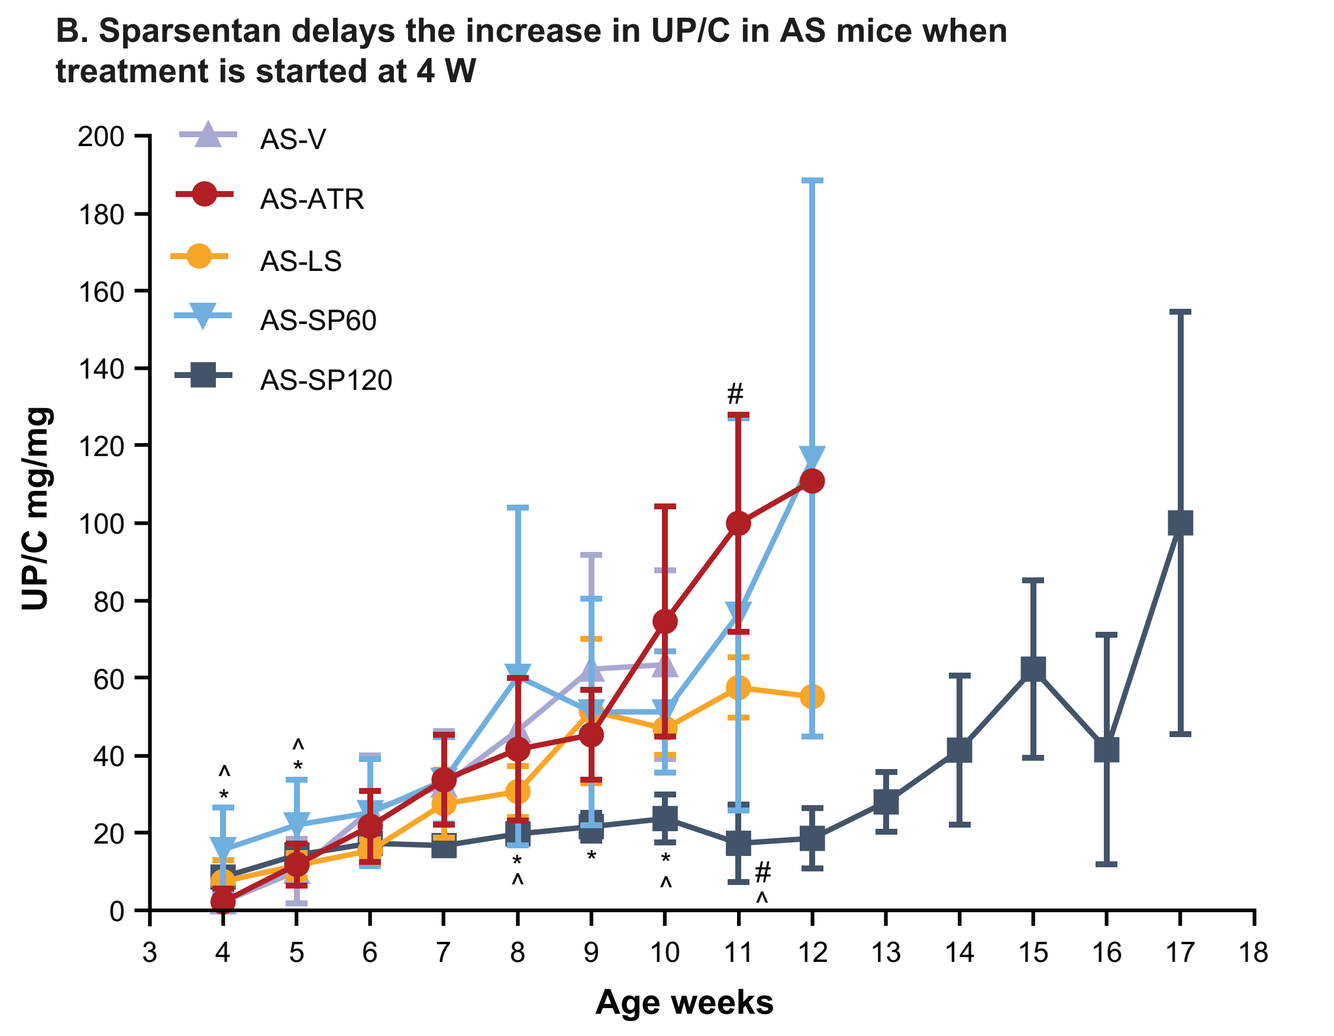
B. Sparsentan delays the increase in UP/C in AS mice when treatment is started at 4 W
UP/C mg/mg
Age weeks
AS-V
AS-ATR
AS-LS
AS-SP60
AS-SP120

Data presented as mean ± SD. *$P<0.05$ vs AS-V. #$P<0.05$ vs AS-LS. ^$P<0.05$ vs AS-ATR.
AS-V: n=10 for 4-8 W, n=7 for 9 W, n=4 for 10 W; AS-ATR: n=10 for 4-9 W, n=7 for 10 W, n=2 for 11 W, n=1 for 12 W; AS-LS: n=5 for 4-10 W, n=4 for 11 W, n=1 for 12 W; SP60: n=5 for 4-9 W, n=4 for 10-11 W, n=3 for 12 W; SP120: n=5 for 4-15 W, n=3 for 16 W, n=2 for 17 W.

Early intervention studies with sparsentan (3-8.75 W treatment): Hearing

Figure 4. Sparsentan but not losartan prevents noise-induced hearing loss in AS mice

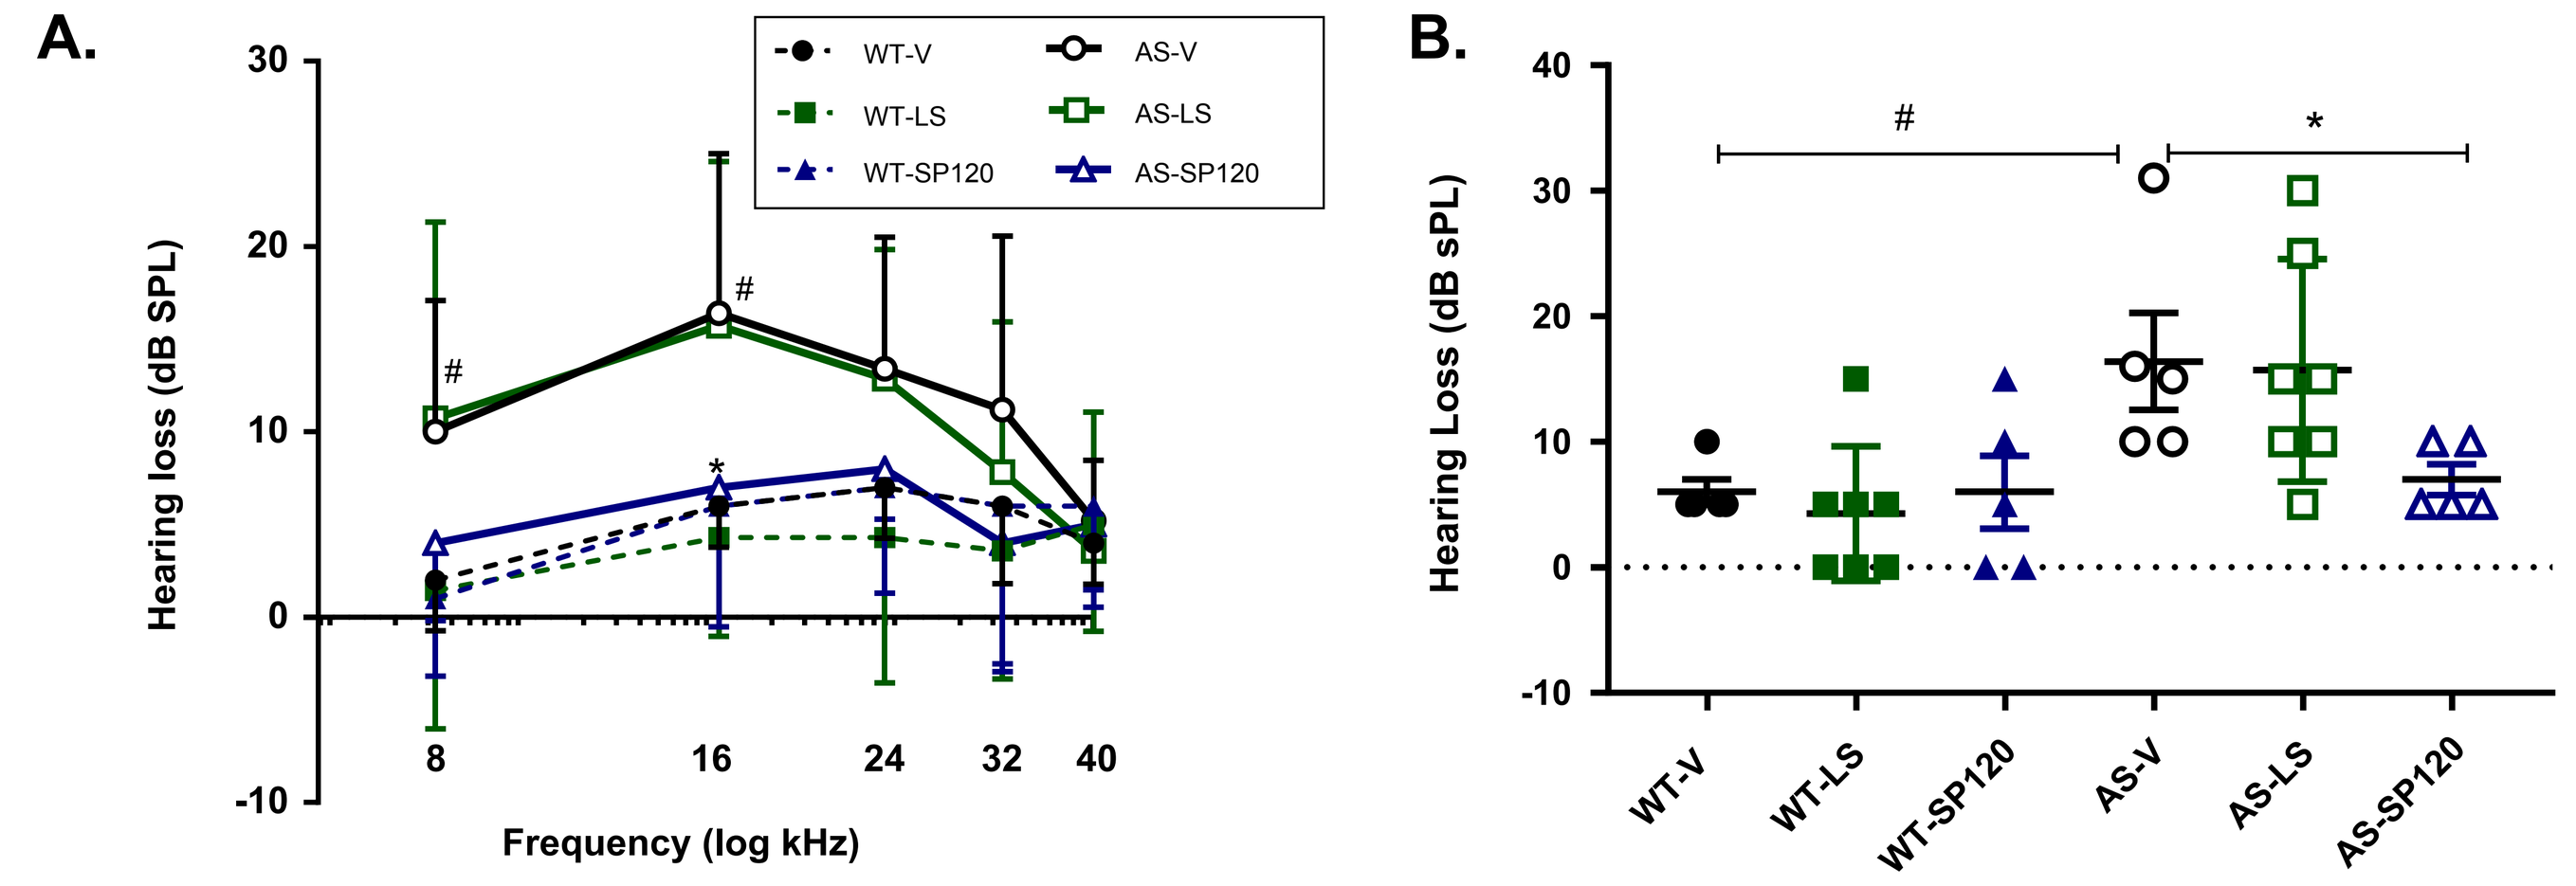
A.
Hearing loss (dB SPL)
Frequency (log kHz)
WT-V
WT-LS
WT-SP120
AS-V
AS-LS
AS-SP120
B.
Hearing Loss (dB sPL)
WT-V
WT-LS
WT-SP120
AS-V
AS-LS
AS-SP120

(A) Hearing loss determined at test frequencies from 8 to 40 kHz equivalent to 0.5 to 8 kHz in humans.
(B) Hearing loss at 16 kHz. Data are presented as mean ± SD (WT-LS, AS-LS n=7; other groups n=5).
*$P<0.05$ AS-SP120 vs AS-V. #$P<0.05$ AS-V vs WT-V. dB, decibel; SPL, sound pressure level.

Figure 5. Sparsentan and losartan prevent the increase in SCBM width but only sparsentan maintains strial ultrastructure

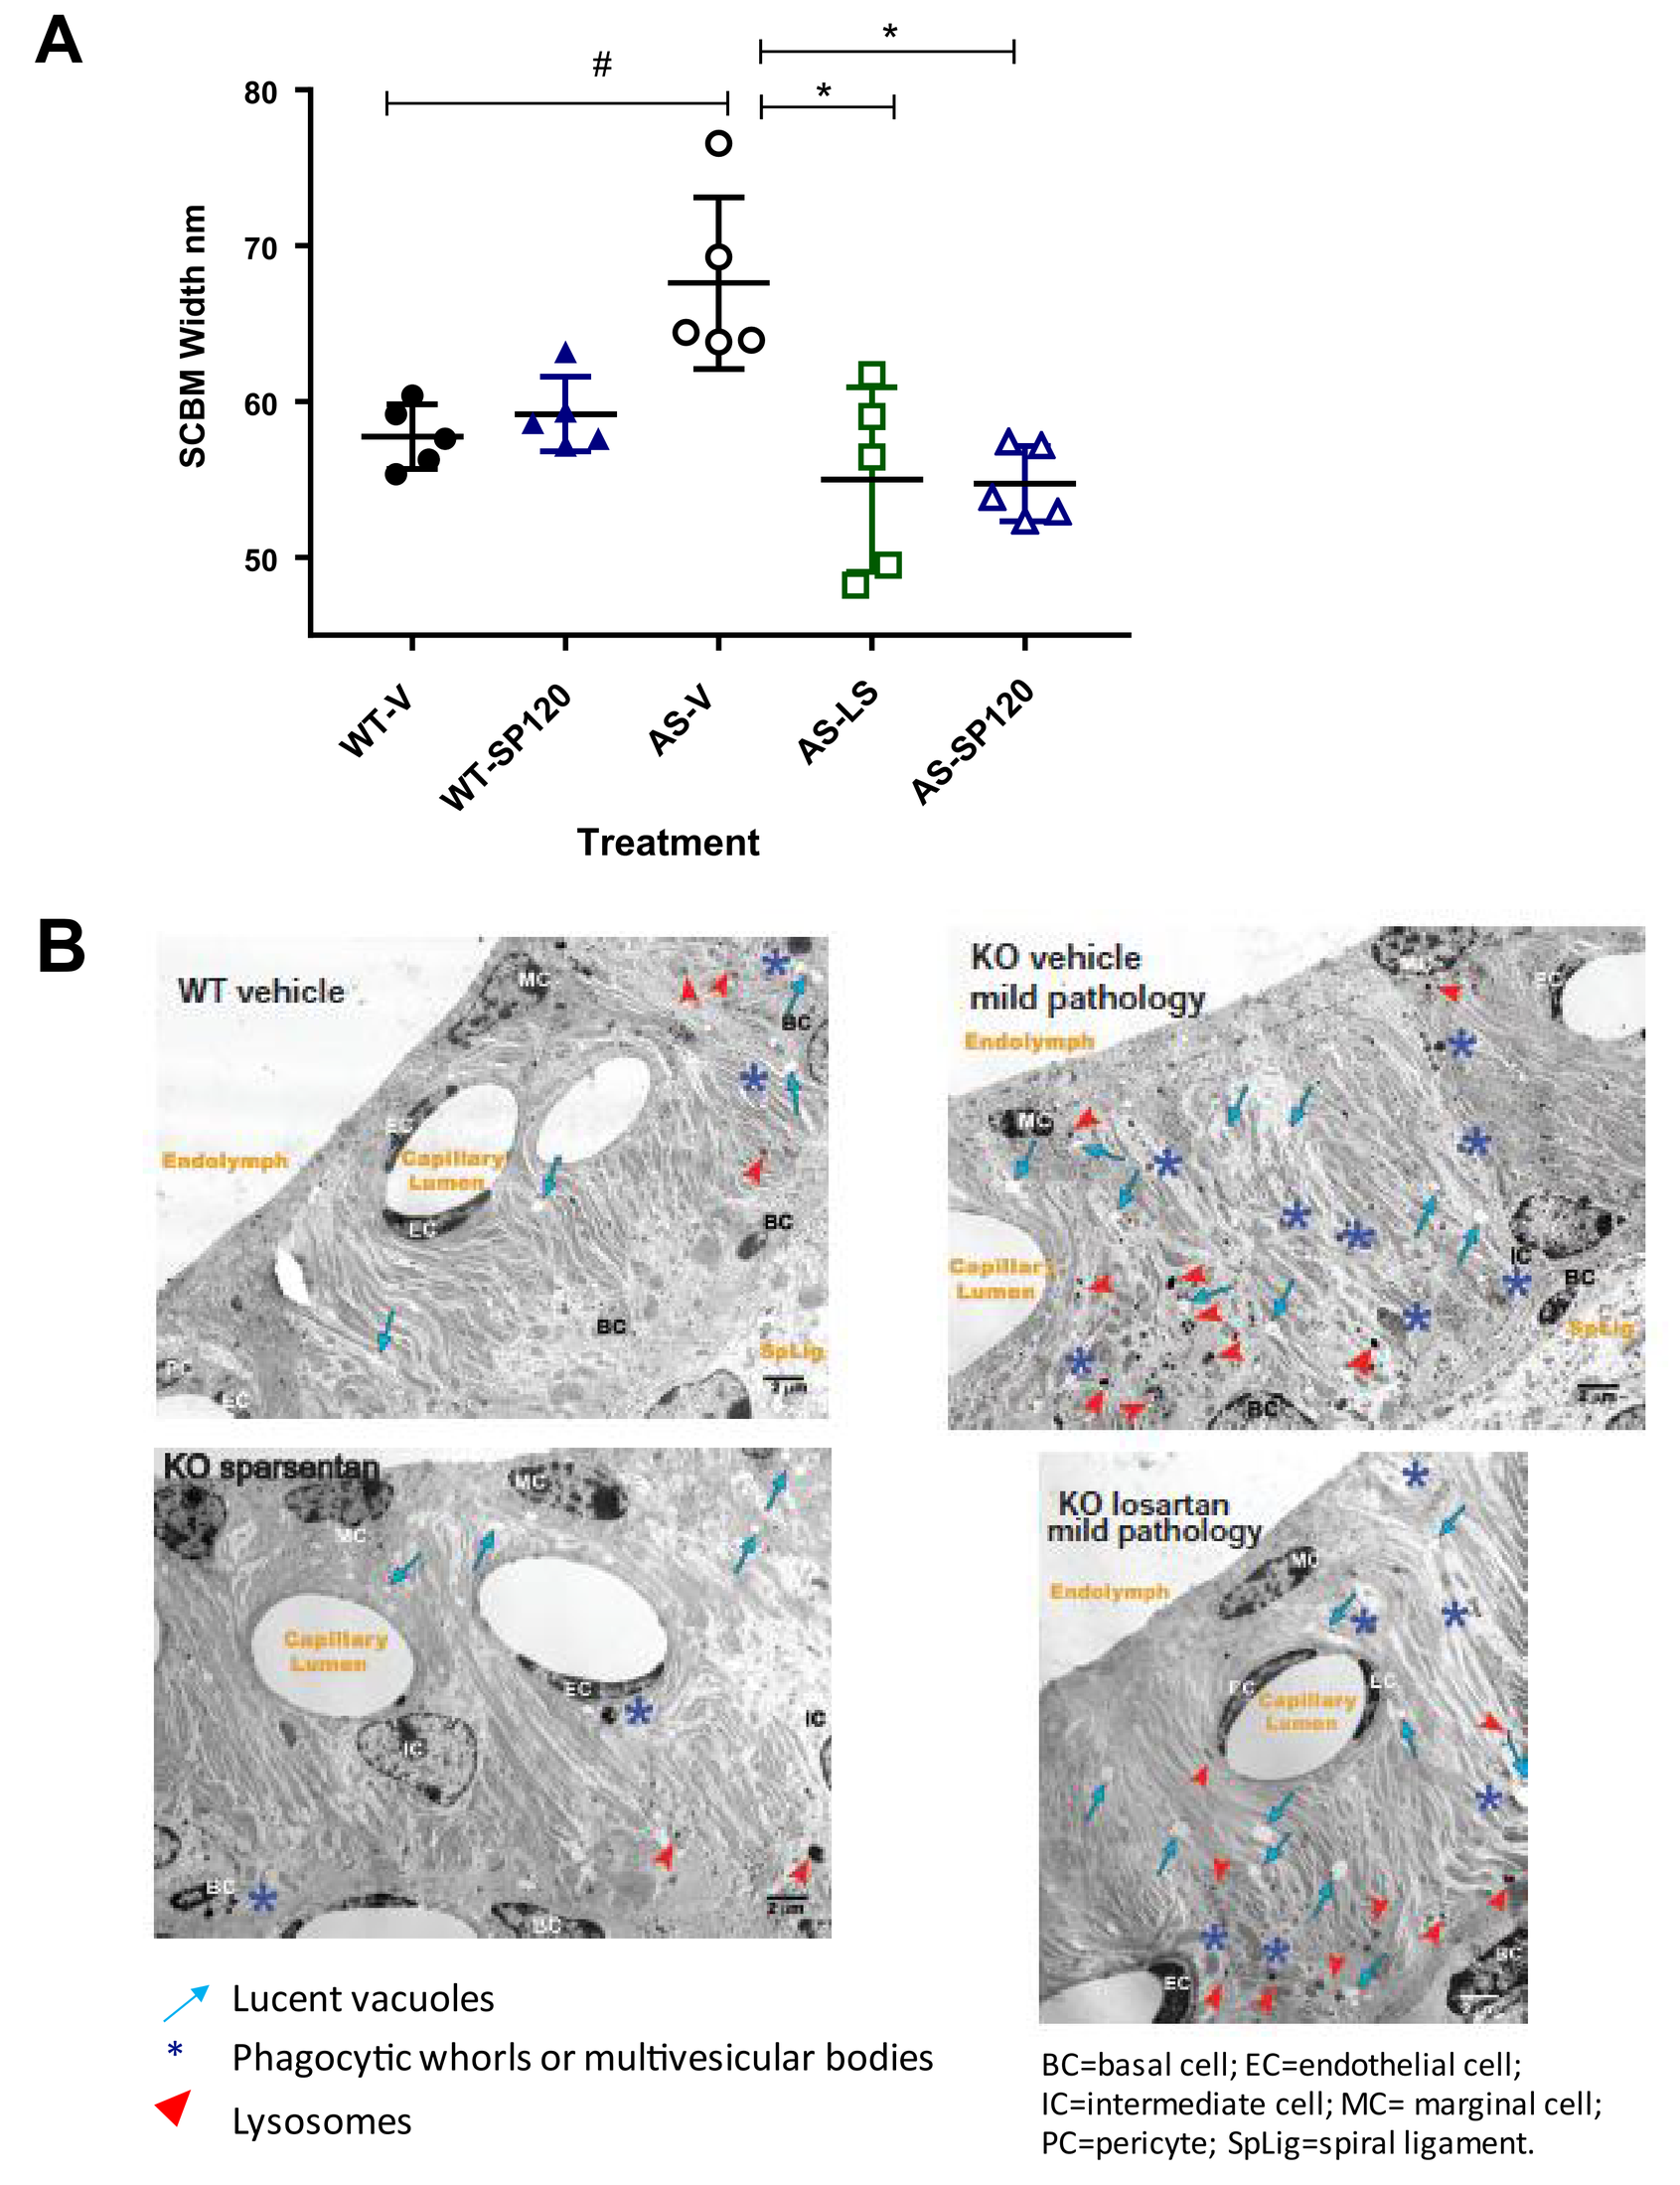
A
SCBM Width nm
WT-V
WT-SP120
AS-V
AS-LS
AS-SP120
Treatment
B
WT vehicle
KO vehicle mild pathology
KO sparsentan
KO losartan mild pathology
Lucent vacuoles
Phagocytic whorls or multivesicular bodies
Lysosomes
BC=basal cell; EC=endothelial cell; IC=intermediate cell; MC= marginal cell; PC=pericyte; SpLig=spiral ligament.

(A) SCBM width. Data are presented as mean ± SD, n=5 per group. *$P<0.05$ AS-SP120 and AS-LS vs AS-V. #$P<0.05$ AS-V vs WT-V. All measurements were taken at 8.75 W of age.
(B) Transmission electron micrograph images of stria.

Figure 6. Sparsentan but not losartan prevents an increase in ECM in the SCBM of AS Mice

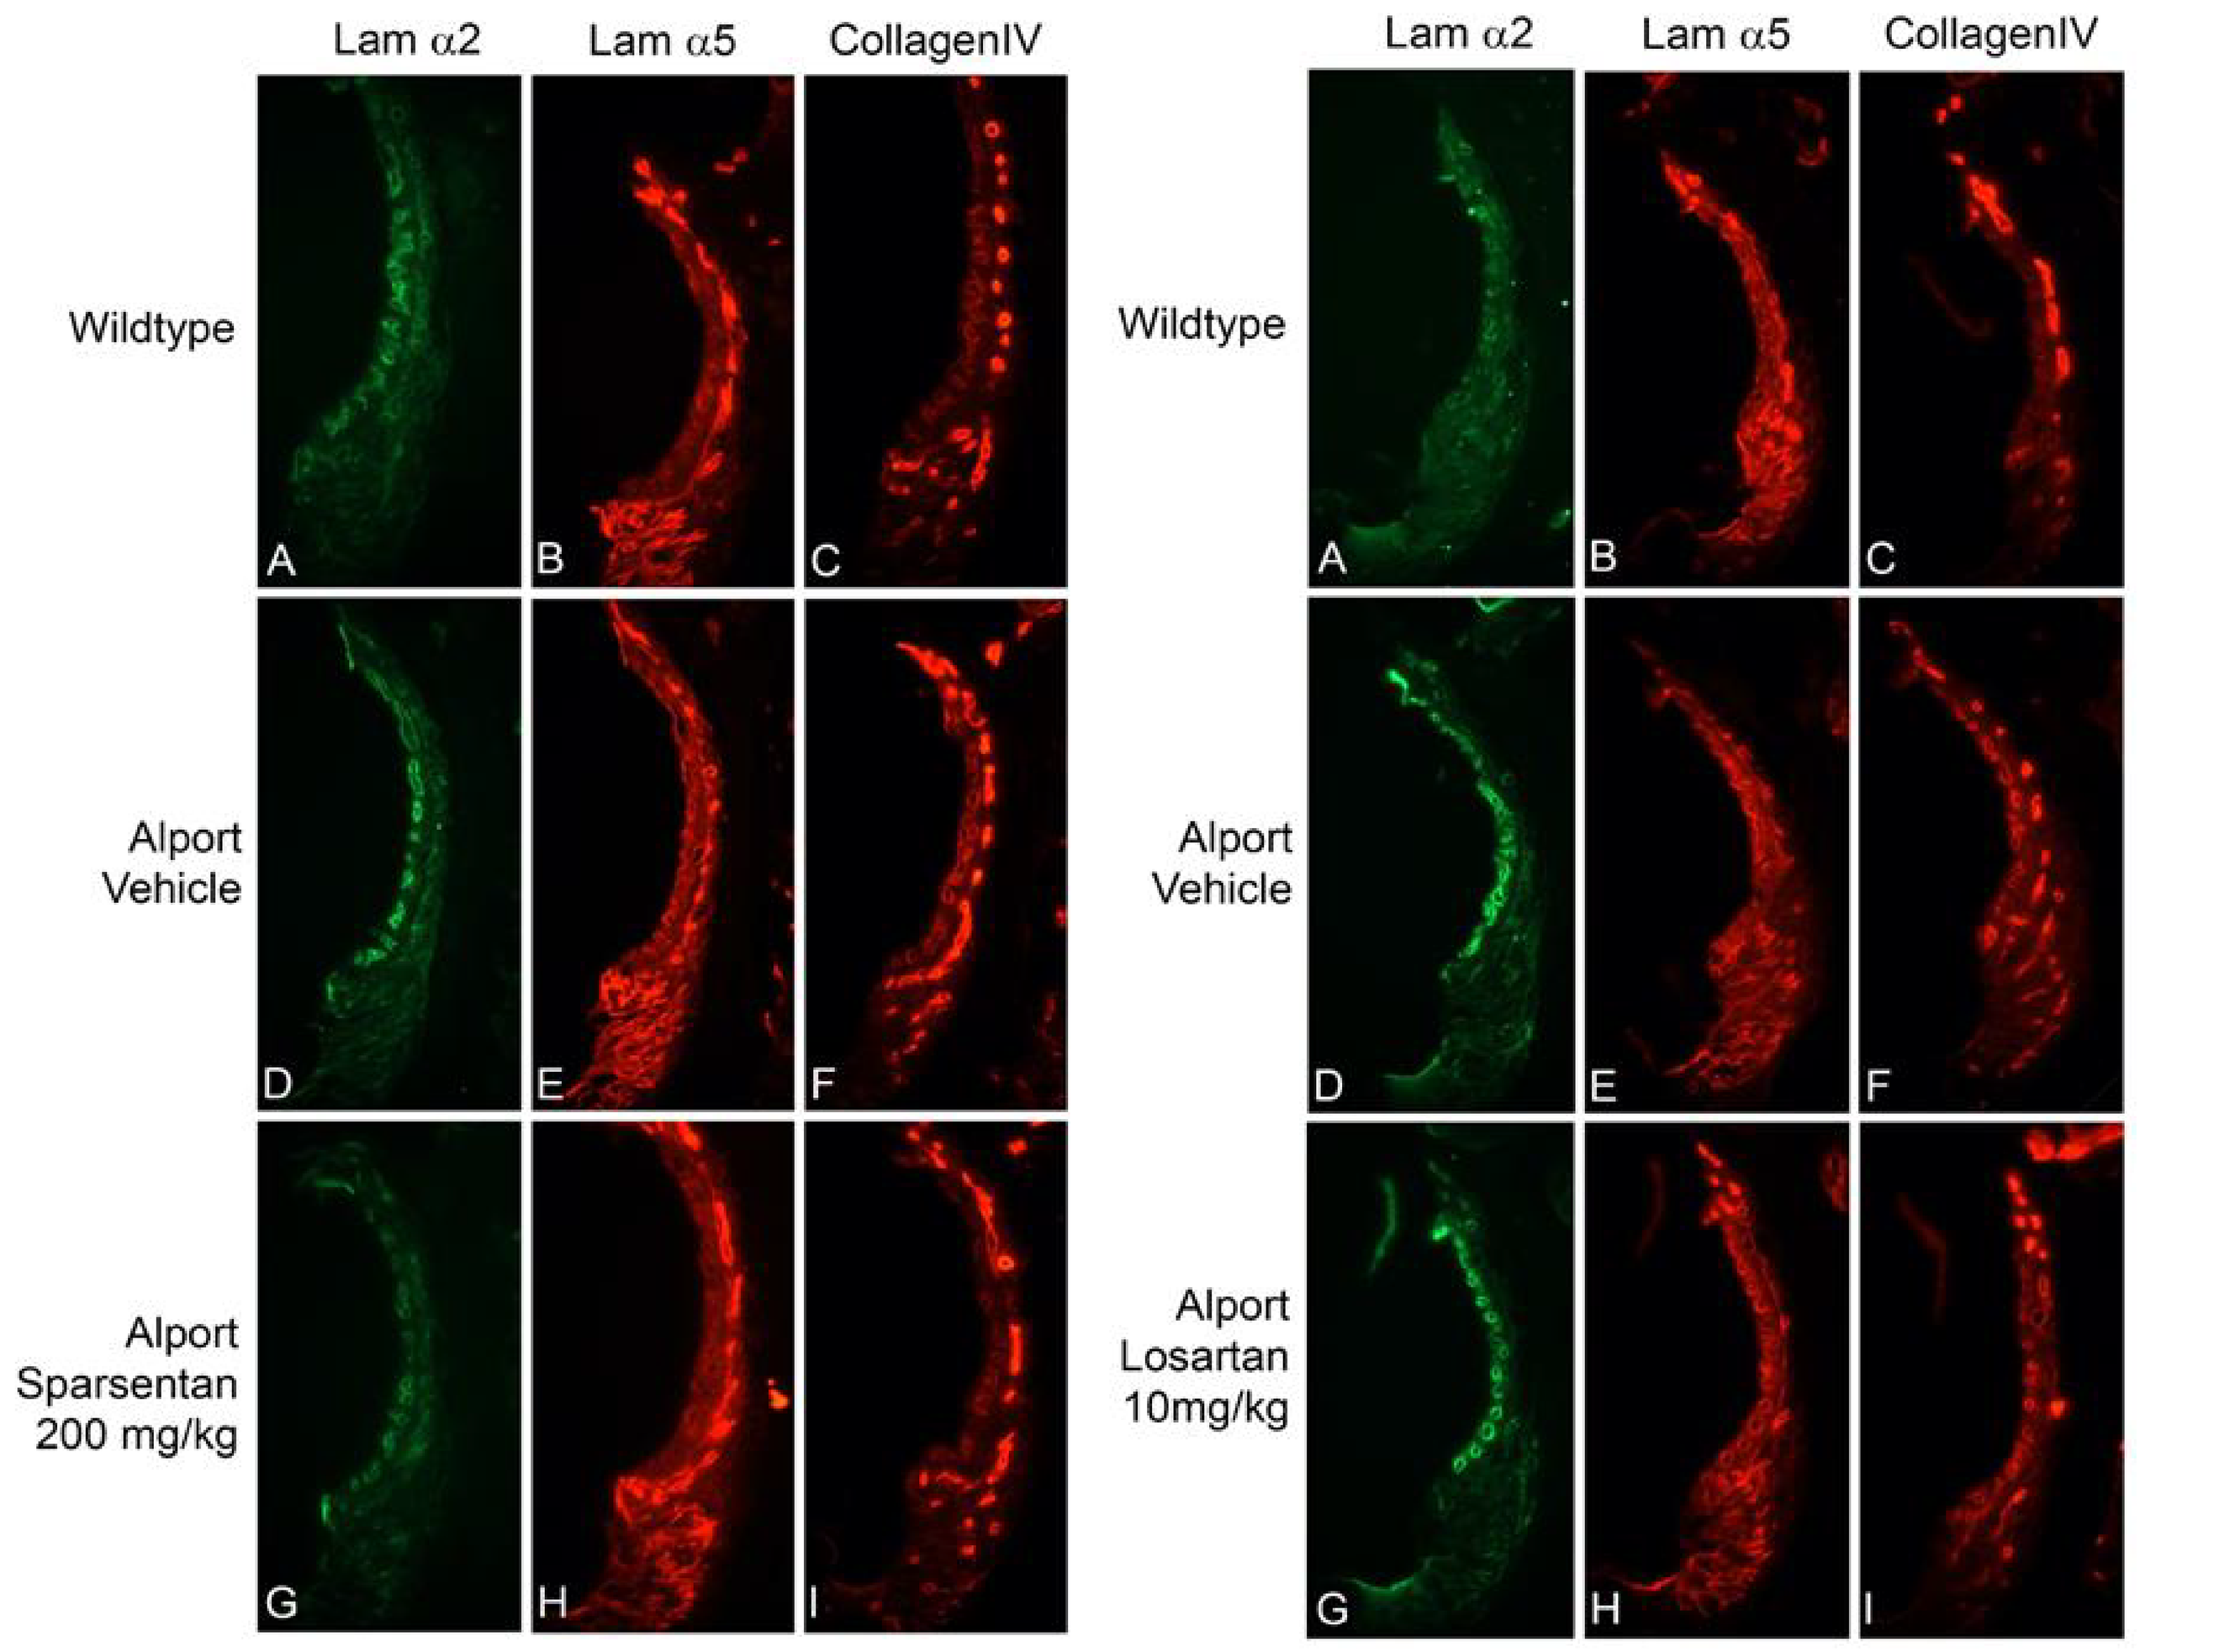
Lam α2
Lam α5
CollagenIV
Wildtype
Alport Vehicle
Alport Sparsentan 200 mg/kg
Alport Losartan 10mg/kg

AS mice were treated with sparsentan (200 mg/kg), losartan (20/10 mg/kg), or vehicle from 3 to 7 W. Images shown following immunofluorescence using anti-laminin α2, anti-laminin α5, and collagen IV antibodies. Vessels in the spiral ligament do not show changes in staining intensity upon treatment and thus serve as a control.

Conclusions

- Sparsentan (120 mg/kg) extends lifespan in Alport mice and significantly delays the increase in UP/C at 11 W compared to losartan (10 mg/kg) and at 8, 10 and 11 W compared to atrasentan (7.5 mg/kg female; 10 mg/kg male) when treatment was initiated at 4 W.
 - Neither losartan nor atrasentan was able to increase lifespan or prevent the increase in UP/C compared to Alport mice treated with vehicle when initiated at 4 W.
- Sparsentan (120 mg/kg) is capable of mitigating the structural and functional auditory changes in Alport mice when administered prophylactically from 3-8.75 W.
- Losartan (10 mg/kg) did not improve strial ultrastructure nor protect Alport mice from noise-induced hearing loss.
- Results from this study, if translated to the clinic, suggest that inhibition of both ET_AR and AT_1R with sparsentan may provide a treatment option for both the renal damage and hearing loss in Alport Syndrome.

References

1. Delimont D, et al. *PLOSone*. 2014;9(6):e99083.
2. Dufek B, et al. *Kidney Int*. 2016;90(2):300-10.
3. Meehan DT, et al. *Hearing Res*. 2016;341:100-08.
4. Scarfe L, et al. *J Vis Exp*. 2018;140:e58520.

Acknowledgments

The authors would like to thank Denise Vosik, MD for her help with the GFR assessment. Editorial support was provided by Courtney Breuel, ELS, of MedVal Scientific Information Services, LLC (Princeton, NJ) and was funded by Retrophin, Inc.

Disclosures

DC, BD, DD, DM, GS, JH, GP, RG, MAG: Received a research grant from Retrophin, Inc.

CJ, JH, and RK: Employees of Retrophin, Inc., and may have an equity or other financial interest in Retrophin, Inc.

The mouse Alport studies were conducted in the laboratories of Dominic Cosgrove at Boys Town National Research Hospital and of Michael Anne Gratton at Washington University and were funded by Retrophin, Inc.

Presented at the German Society of Nephrology 12th Annual Virtual Congress 2020 (DGFN 2020), October 1-4, 2020