PO1454

The Dual Endothelin Angiotensin Receptor Antagonist (Deara) Sparsentan Protects from Glomerular Hypercellularity and Associated Immune/Inflammatory Gene-Network-Activity in a Model of Iga Nephropathy (Igan)

Colin Reily[1], Zina Moldoveanu[1], Tiziano Pramparo[2], Stacy Hall[1], Lea Novak[1], Radko Komers[2], Celia Jenkinson[2], Jan Novak[1]

[1]University of Alabama at Birmingham, Departments of Medicine, Pathology, and Microbiology, Birmingham, AL, USA; [2]Travere Therapeutics, Inc. San Diego, CA

Abstract

IgA nephropathy (IgAN) is an autoimmune glomerulonephritis wherein immune complexes (IC) composed of galactose-deficient IgA1 (Gd-IgA1; autoantigen) and Gd-IgA1-specific IgG autoantibodies (AuAb) deposit in the glomeruli and cause injury. In a mouse model of IgAN, induced by IC formed *in vitro* from human Gd-IgA1 and a recombinant IgG AuAb (rIgG), we used whole-kidney RNAseq profiling to assess how sparsentan affects the gene expression of pathways dysregulated by IC.

Introduction

IgAN is the most common primary glomerulonephritis worldwide and is characterized by mesangial deposition of Gd-IgA1 and AuAb IC. These IC induce glomerular injury with around 40% of IgAN patients progressing to kidney failure. IgA1 is unique to humans and higher order primates, which has hampered development of small-animal models of IgAN. Here we use a model where engineered IC (EIC) derived from rIgG and Gd-IgA1 is created *in vitro* and injected multiple times into nude mice to induce glomerular injury mimicking human IgAN.[1]

Sparsentan is a selective dual endothelin angiotensin receptor antagonist (DEARA) with affinity for endothelin (A type) and angiotensin II (Type 1) receptors. While endothelin A and angiotensin II are known to play a role in the pathobiology of IgAN, it is not known which molecular mechanisms sparsentan acts on to potentially reduce mesangial hypercellularity. We have previously shown that sparsentan attenuates the mesangial hypercellularity in this model.[2] This study used nude mice injected with EIC and dosed with vehicle or two different doses of sparsentan to assess the effects of treatment on glomerular cellularity and related changes in gene transcription.

Methods

EIC were injected into ~7-week-old nude mice every other day for a total of 6 doses (n=5/group) via tail vein. Vehicle or sparsentan (60 [S60] or 120 [S120] mg/kg) was given by gavage once daily from the first day of EIC injections. Negative-control mice received only vehicle. Kidney tissue for histopathology and RNAseq was harvested on day 12. RNAseq raw data processed using DESeq2 identified differentially expressed genes. Unsigned weighted gene correlation network analysis (WGCNA) was used for network-level profiling and to identify co-expressed genes associated with hypercellularity and Ki-67 positivity of glomeruli, as assessed by quantitative morphometry (Q-morphometry). Gene set enrichment analysis (GSEA) and eXpression2Kinases (X2K) assessed changes at the pathway level and imputed correlated upstream cell-signaling networks. Genes identified from WGCNA and DESeq2 were compared to IgAN biopsy RNA DESeq2 analysis. Pathway enrichment p-values were adjusted with FDR.

Experimental design and RNAseq analysis

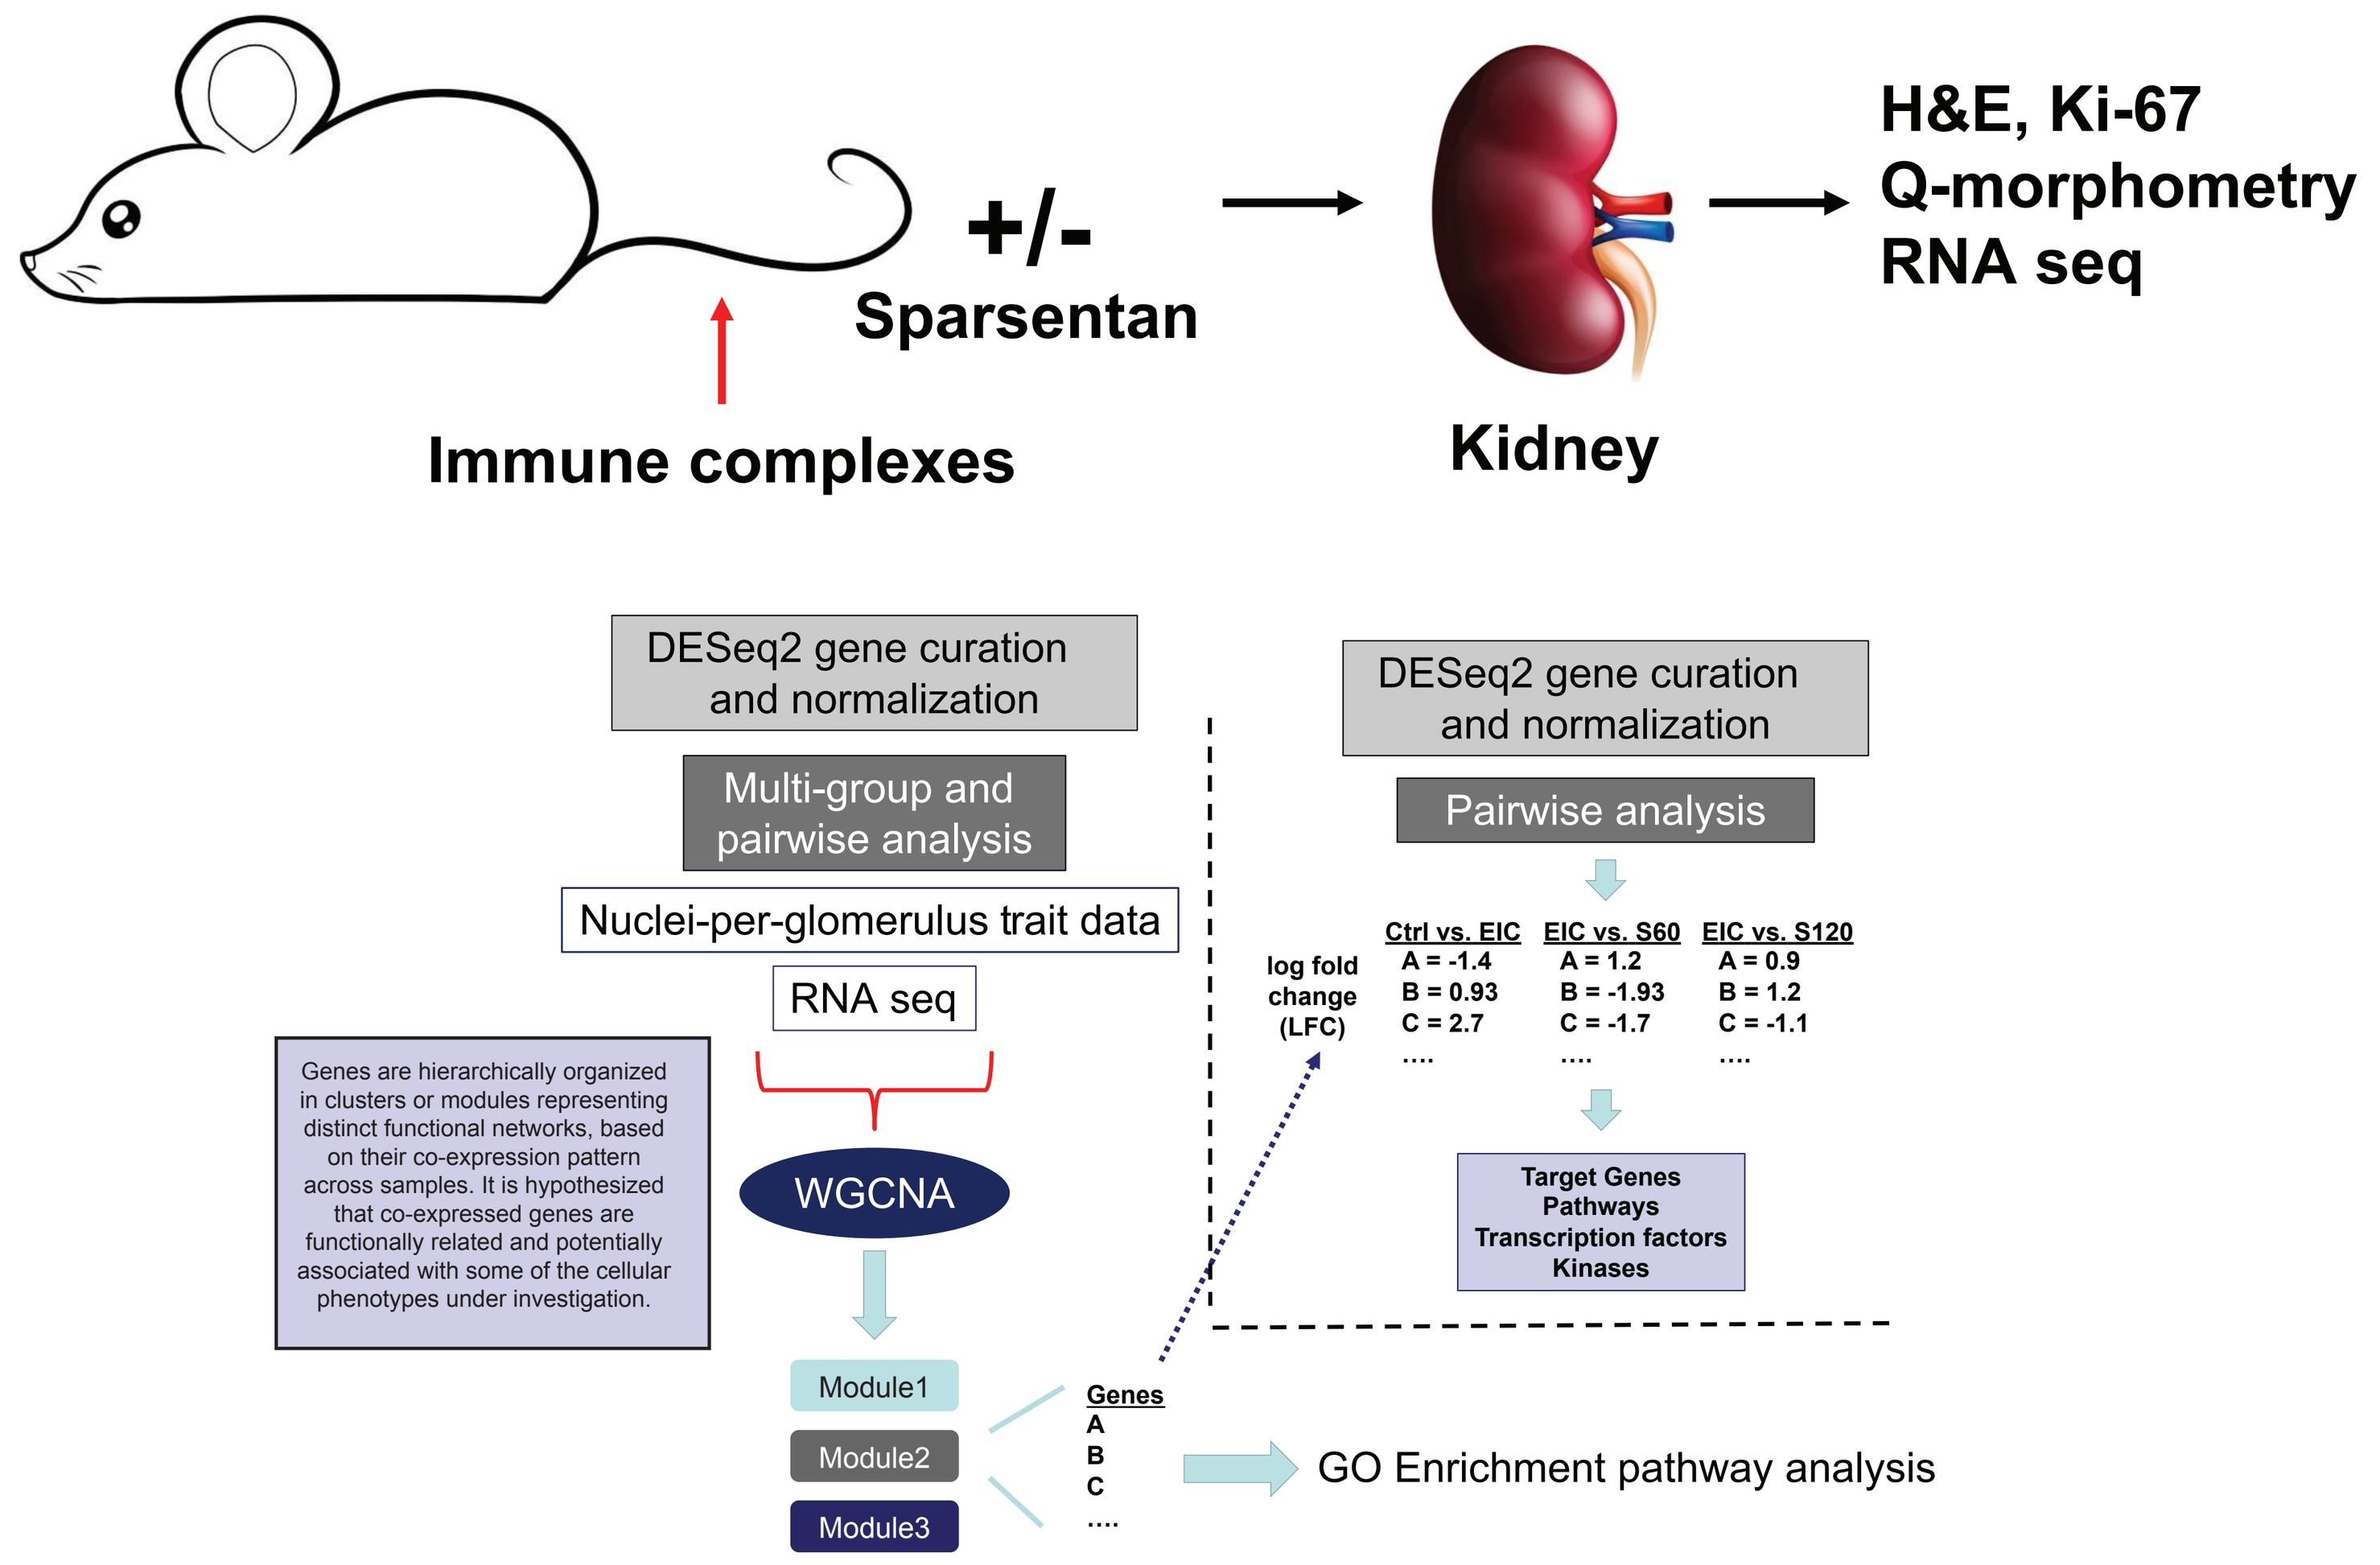
+/-
Sparsentan
Immune complexes
Kidney
H&E, Ki-67
Q-morphometry
RNA seq
DESeq2 gene curation and normalization
Multi-group and pairwise analysis
Nuclei-per-glomerulus trait data
RNA seq
Genes are hierarchically organized in clusters or modules representing distinct functional networks, based on their co-expression pattern across samples. It is hypothesized that co-expressed genes are functionally related and potentially associated with some of the cellular phenotypes under investigation.
WGCNA
Module1
Module2
Module3
Genes
A
B
C
....
GO Enrichment pathway analysis
DESeq2 gene curation and normalization
Pairwise analysis
log fold change (LFC)
Ctrl vs. EIC
A = -1.4
B = 0.93
C = 2.7
EIC vs. S60
A = 1.2
B = -1.93
C = -1.7
EIC vs. S120
A = 0.9
B = 1.2
C = -1.1
Target Genes
Pathways
Transcription factors
Kinases

Results

Sparsentan attenuates EIC-induced mesangial hypercellularity

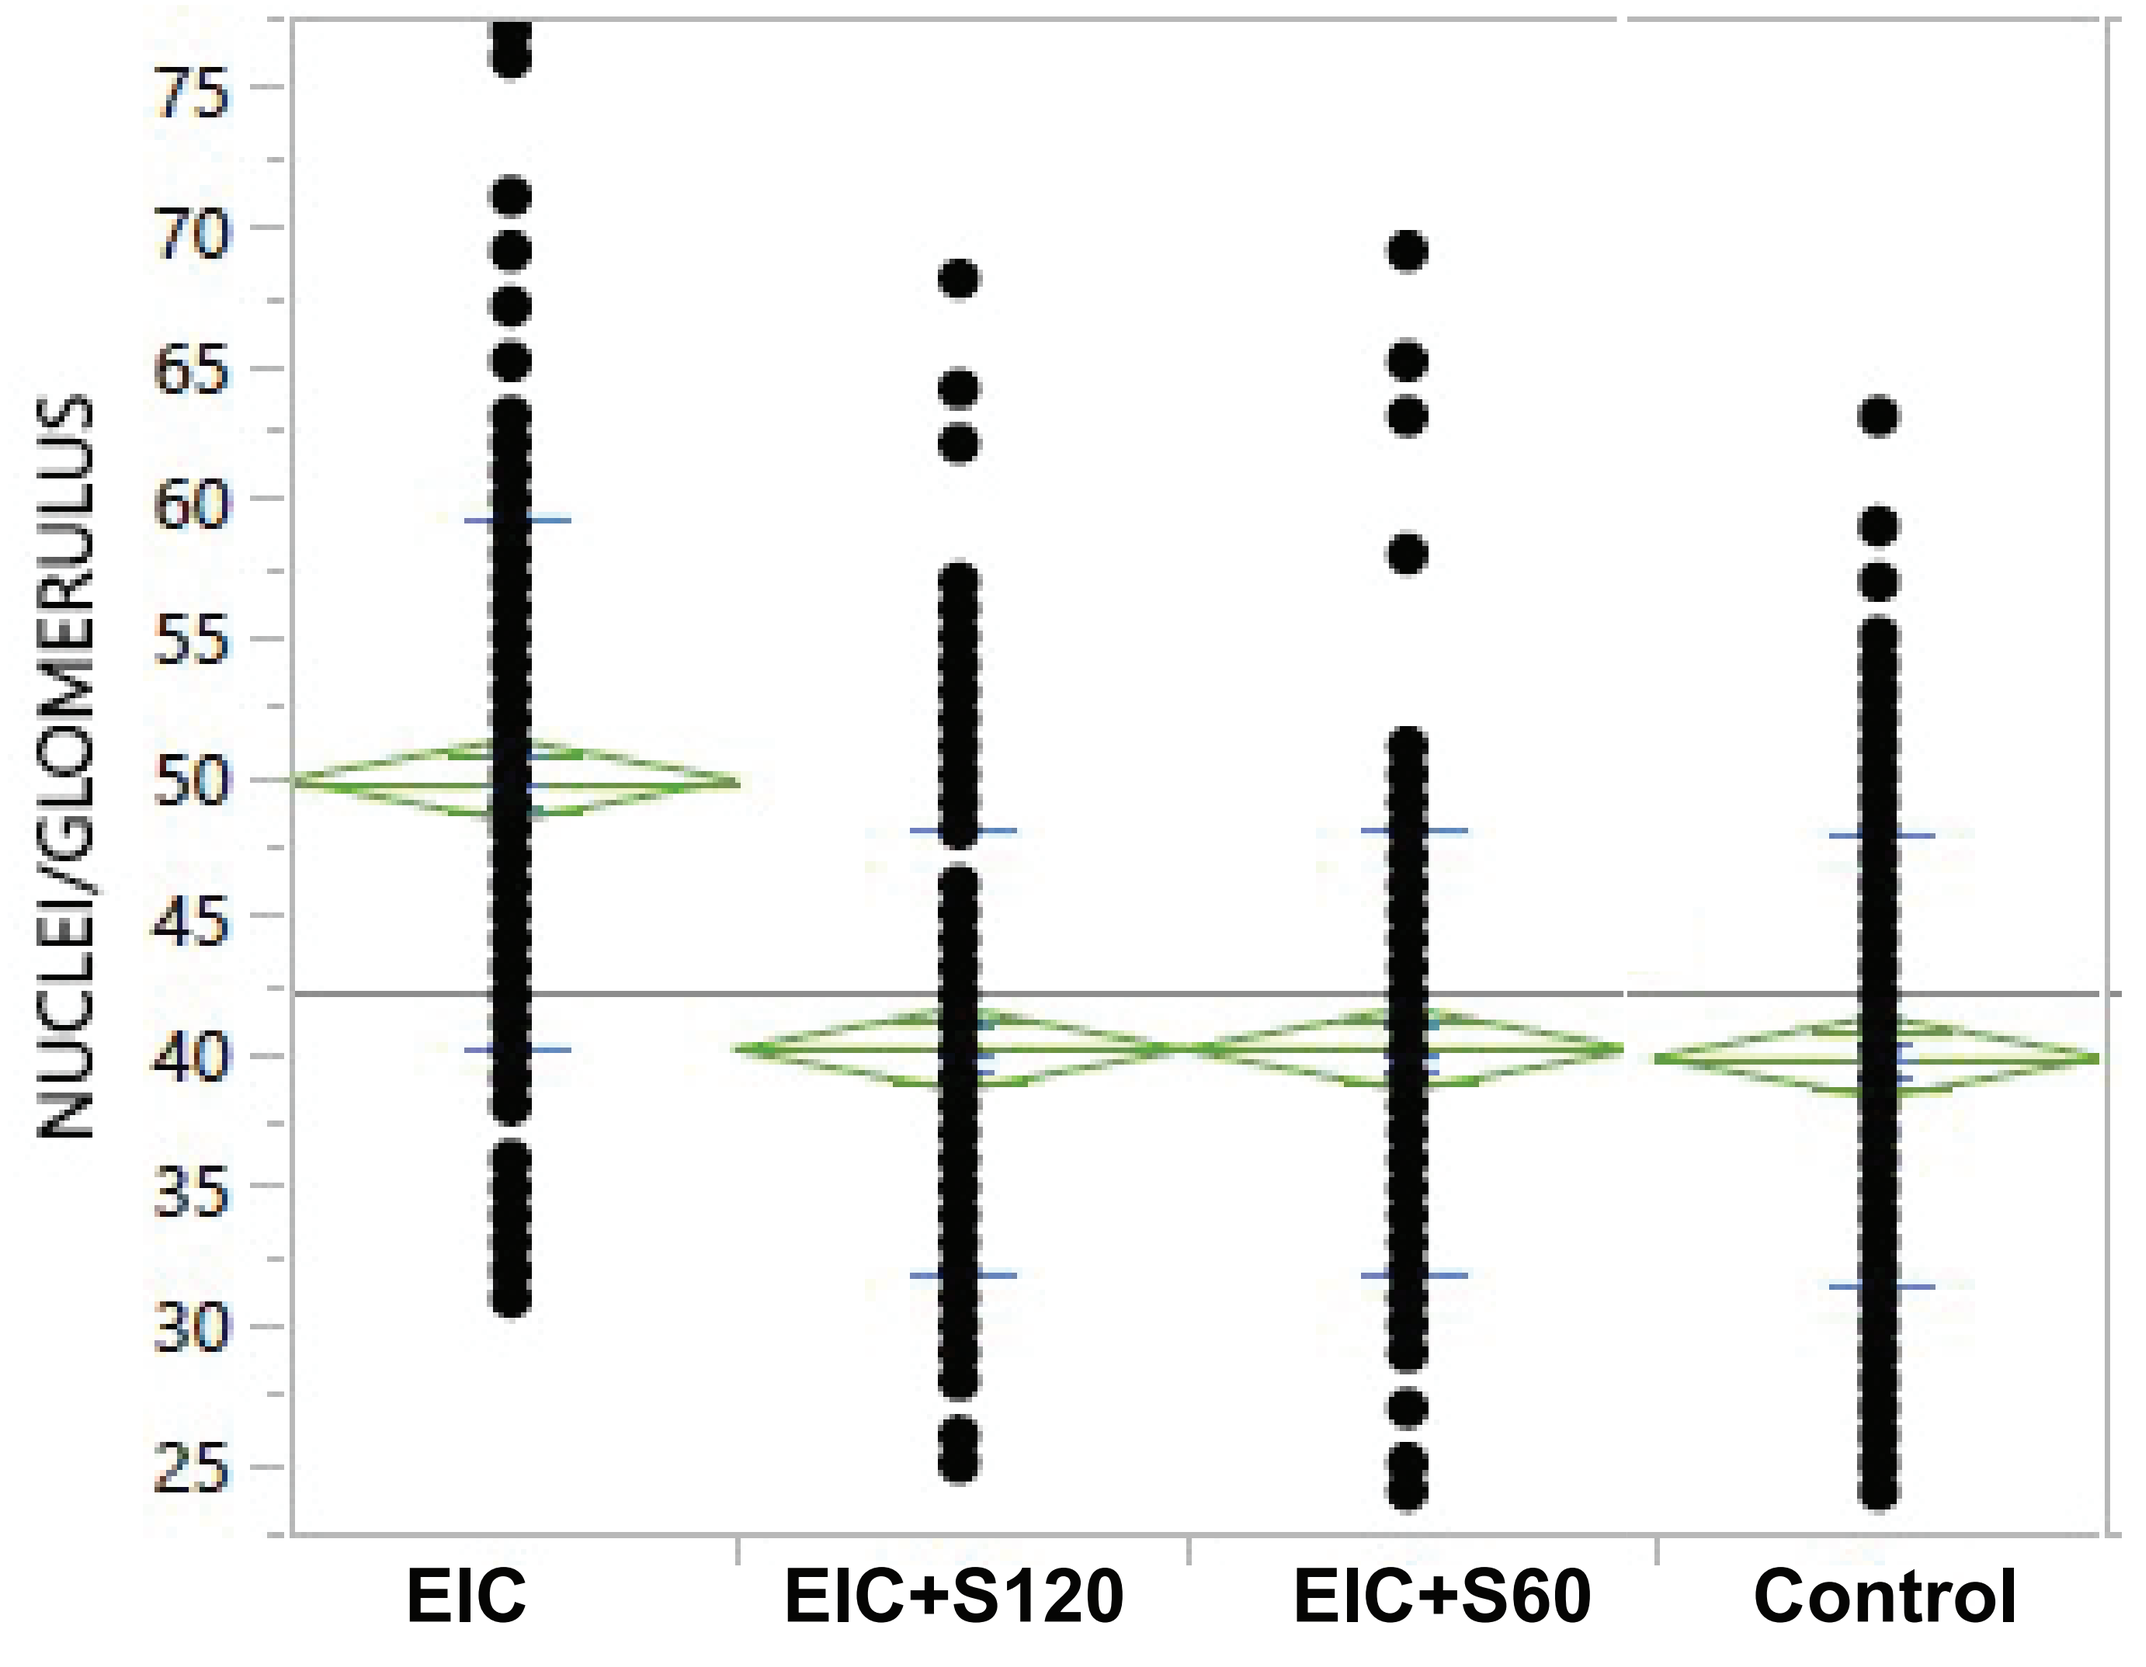
NUCLEI/GLOMERULUS
75
70
65
60
55
50
45
40
35
30
25
EIC
EIC+S120
EIC+S60
Control

Sparsentan-treated mice showed a statistically significant attenuation in number of nuclei per glomerulus compared to EIC-injected mice alone, and similar to control mice (n=5); p<0.001 for EIC+S60 and EIC+S120 vs. EIC one-way ANOVA, Dunnett's post-hoc.

Pathway-level analysis shows downregulated expression in immune/inflammatory processes after sparsentan treatment

Up Pathways		Down Pathways			
Control vs. EIC		EIC vs. EIC+S120		EIC vs. EIC+S60	
Pathway	p value	Pathway	p value	Pathway	p value
Cytokine signaling	2.53E-08	Cytokine signaling	2.97E-09	Cellular response to INF1	1.49E-08
Cellular response to IFN1	2.53E-08	Cellular response to INF1	3.87E-09	IFN1 signaling	1.49E-08
IFN1 signaling	2.53E-08	IFN1 signaling	3.87E-09	Cytokine signaling	2.90E-07
Neutrophil response	3.52E-05	Neutrophil response	1.24E-05	Neutrophil response	0.001
Neutrophil degranulation	6.37E-05	Neutrophil degranulation	2.40E-05	Neutrophil degranulation	0.002
Neutrophil immunity	8.22E-05	Neutrophil immunity (GO:0002446)	3.08E-05	MHC1-mediated Ag response	0.002
Cellular response to cytokine	7.33E-04	Cellular response to cytokine	0.001	Neutrophil immunity	0.002
MHC1-mediated Ag response	0.003	MHC1-mediated Ag response	0.001	Response to IFNγ	0.003
Response to IFNγ	0.004	Response to IFNγ	0.001	Response to IFNβ	0.005
Cytokine response	0.004	MHC1-mediated, TAP-dependent Ag response	0.003	MHC1-mediated, TAP-dependent Ag response	0.006

WGCNA analysis between grouped pairs (control vs. EIC, EIC vs. EIC+S120, EIC vs. EIC+S60) was performed to find module eigengenes that correlate with nuclei per glomerulus and Ki-67 positivity. Genes from the modules significantly associated with the cellular phenotypes were then grouped and subjected to GSEA GO biological process pathway analysis. Top 10 pathways for each pairwise analysis are shown above. LFC of genes within each pathway were identified from DESeq2 analysis, and found to be decreased compared to the LFC of the upregulated genes in control vs. EIC. Ag, antigen; INF, interferon; MCH1, major histocompatibility complex class 1.

Top Log Fold Change (LFC) gene changes in DESeq2 identified from top modules in WGCNA

Control vs. EIC		EIC vs. EIC+S60		EIC vs. EIC+S120	
Gene	LFC	Gene	LFC	Gene	LFC
Z-DNA binding protein 1 (Zbp1)	2.78	***Tgtp2***	-1.63	***Fcgr4***	-1.90
C-type lectin domain family 4, member a1 (Clec4a1)	2.46	***Gm4951***	-1.62	***Myo1g***	-1.70
Fcgr4	2.05	***Tgtp1***	-1.51	***Nlrc5***	-1.68
Iigp1	2.04	***Iigp1***	-1.39	*Lyz2*	-1.65
T-cell-specific guanine nucleotide triphosphate-binding protein 2 (Tgtp2)	2.02	***Fcgr4***	-1.35	*Gbp6*	-1.61
Lysozyme 2 (Lyz2)	1.90	*Nlrc5*	-1.34	***Clec4a1***	-1.51
Myosin 1G (Myo1G)	1.86	*Guanylate binding protein family member 6 (Gbp6)*	-1.26	***Iigp1***	-1.46
NOD-like receptor family CARD domain containing 5 (Nlrc5)	1.81	*C-type lectin domain family 12 member A (Cleck12a)*	-1.23	*Neutrophil cytosolic factor 4 (Ncf4)*	-1.43
T-cell-specific guanine nucleotide triphosphate-binding protein 1 (Tgtp1)	1.79	***Zbp1***	-1.12	*Mitogen-activated protein kinase kinase kinase kinase 1 (Map4k1)*	-1.36
Interferon-gamma-inducible GTPase Ifgga2 protein (Gm4951)	1.79	*Ms4a6b*	-1.11	***Tgtp2***	-1.35
Adhesion G protein-coupled receptor E1 (Adgre1)	1.74	***Lyz2***	-1.10	*Integrin subunit alpha L (Itgal)*	-1.33
C-type lectin domain family 4, member a2 (Clec4a2)	1.74	*Guanylate-binding protein 9 (Gbp9)*	-1.09	***Clec4a2***	-1.27
Membrane-spanning 4-domains subfamily A member 6B (Ms4a6b)	1.70	*Tryptase beta 2 (Tpsb2)*	-1.04	***Lyz1***	-1.25
Lysozyme C-1 (Lyz1)	1.67	*XIAP-associated factor 1 (Xaf1)*	-1.03	*Itgb2*	-1.24
Allograft Inflammatory Factor 1 (Aif1)	1.66	*Schlafen family member 8 (Slfn8)*	-1.03	*Xaf1*	-1.24

Combined genes from WGCNA top modules were cross-referenced to DESeq2 LFC data of those groups. Comparison of the top genes LFC found statistically significant reversion of expression (in bold).

Imputed kinase and transcription factor significantly correlated with WGCNA top module genes

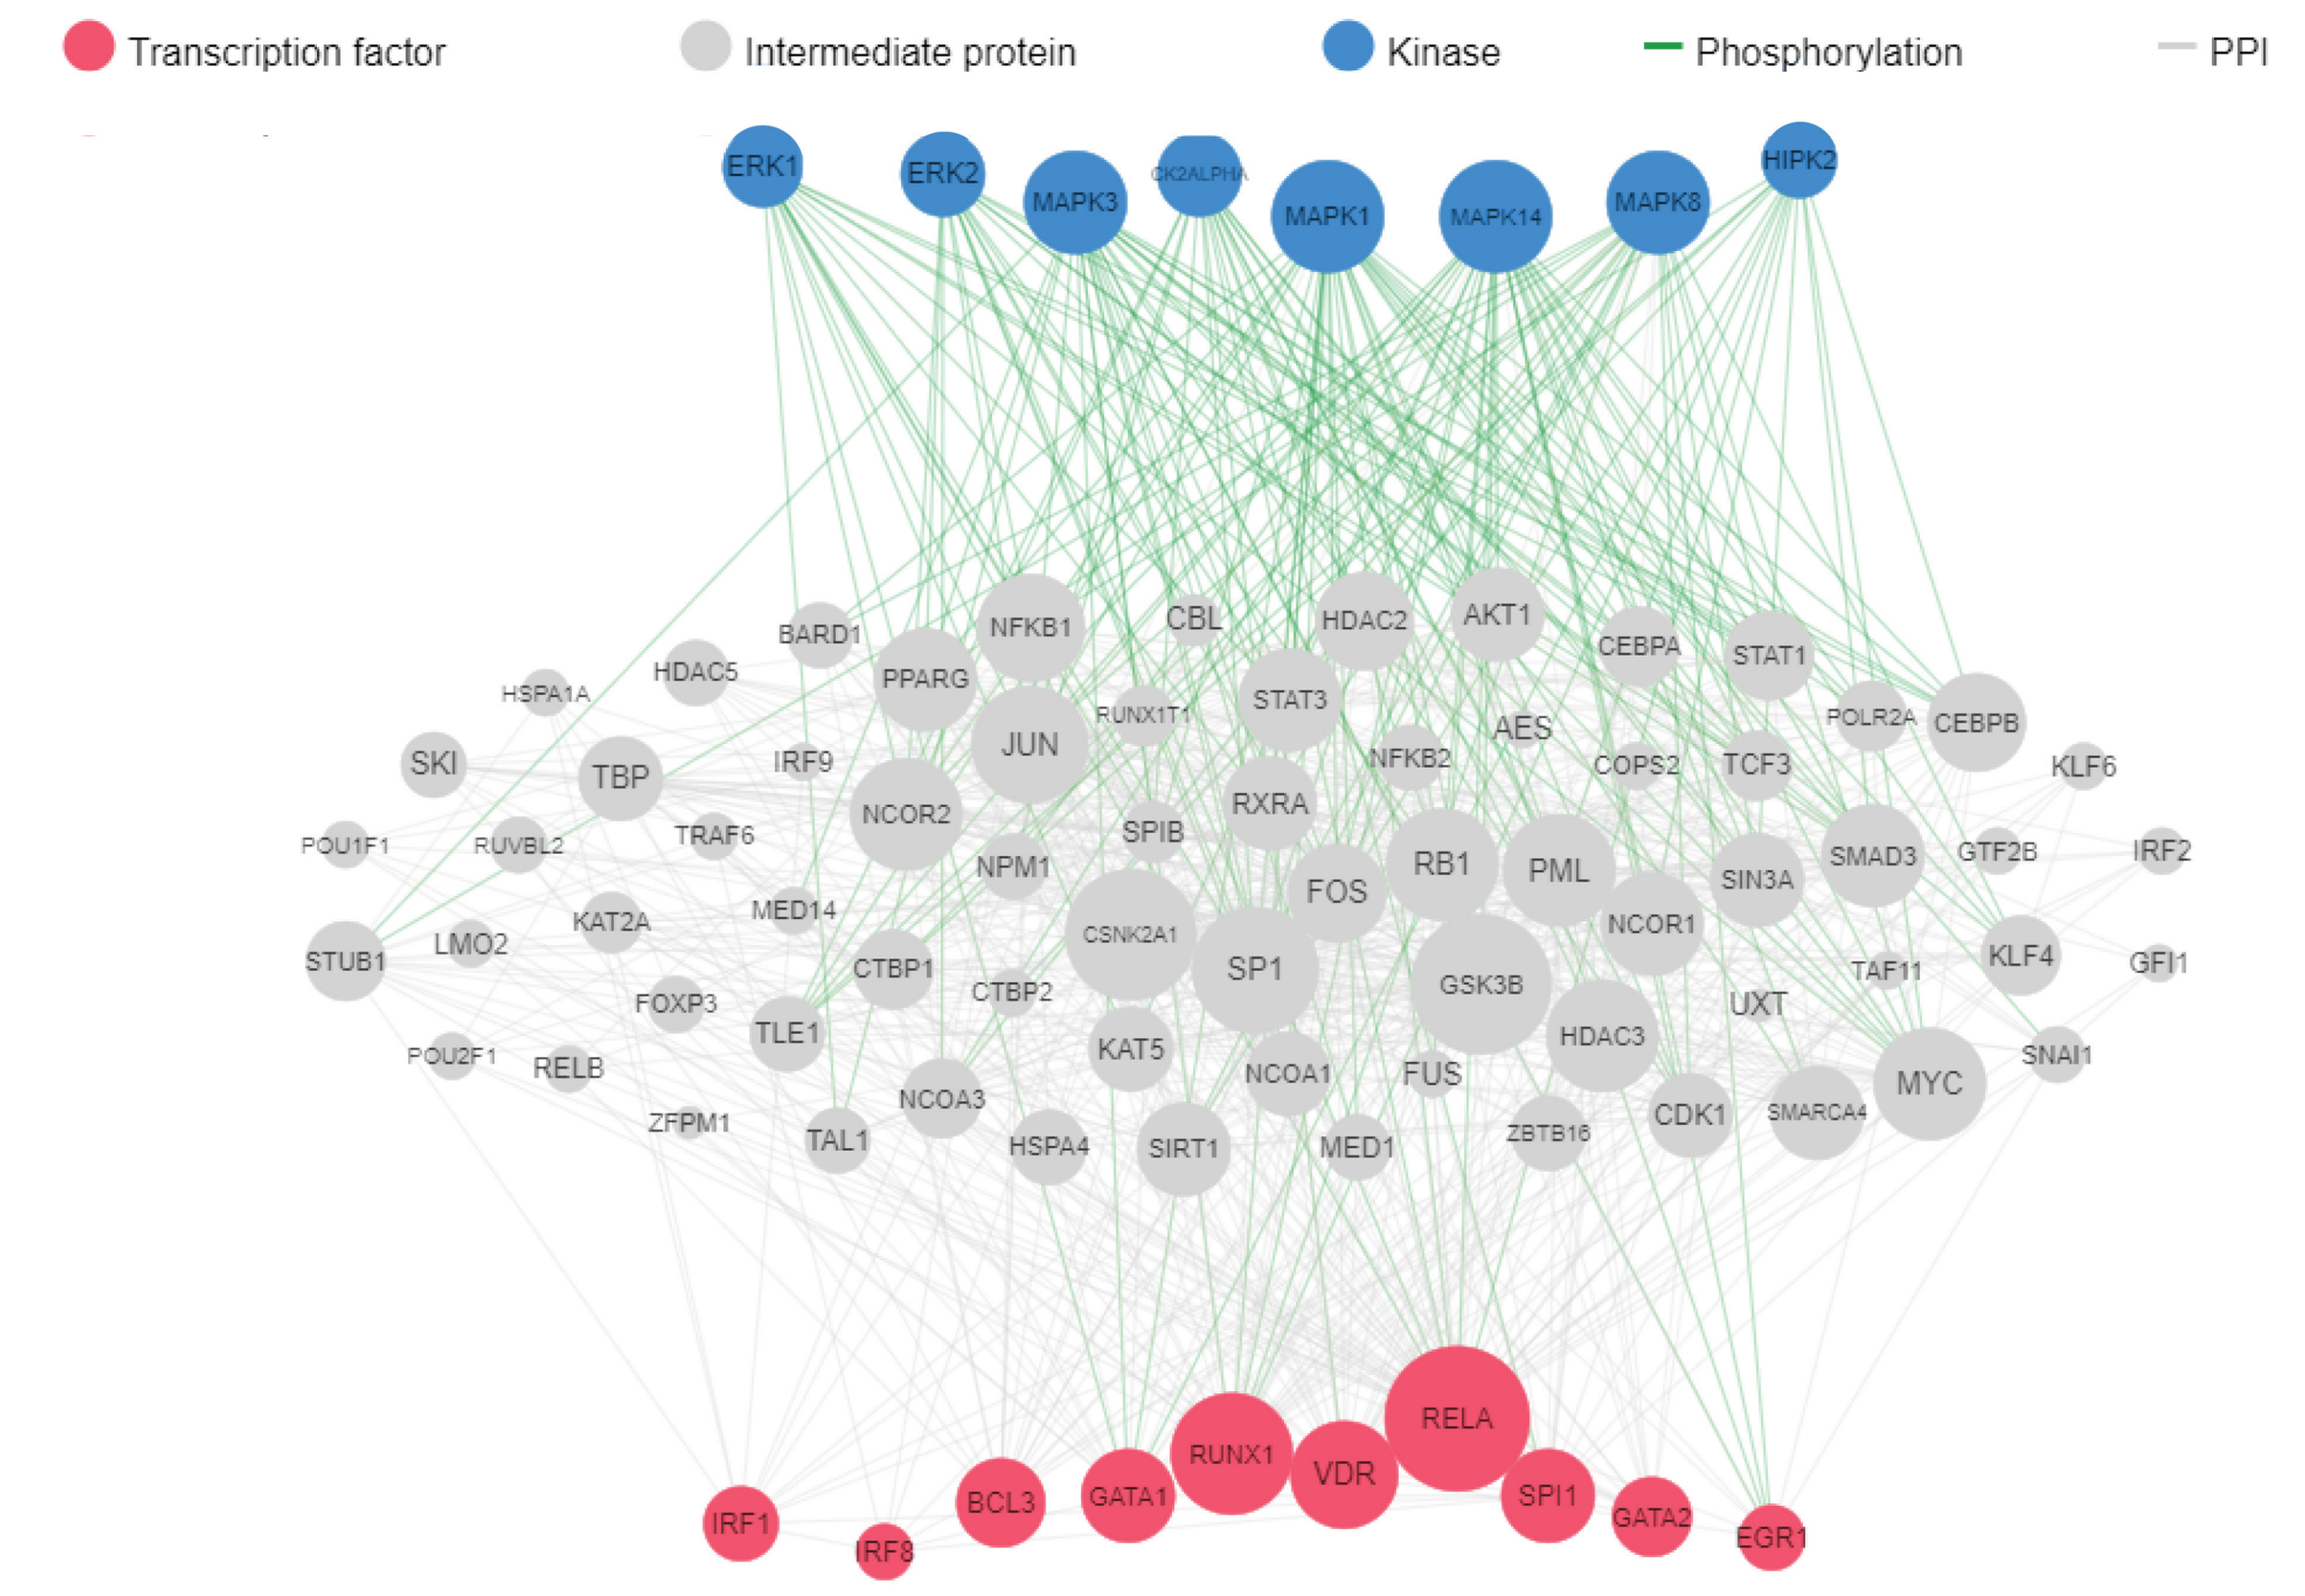
Transcription factor
Intermediate protein
Kinase
Phosphorylation
PPI
ERK1
ERK2
MAPK3
MAPK1
MAPK14
MAPK8
HIPK2
RELA
RUNX1
VDR
GATA1
BCL3
IRF1
SPI1
GATA2
EGR1

Genes identified from top modules in the control vs. EIC group were uploaded to X2K, and kinase and transcription factors were imputed from the gene list. PPI, protein–protein interaction.

Top WGCNA module genes show normalization or reversal of expression after sparsentan treatment

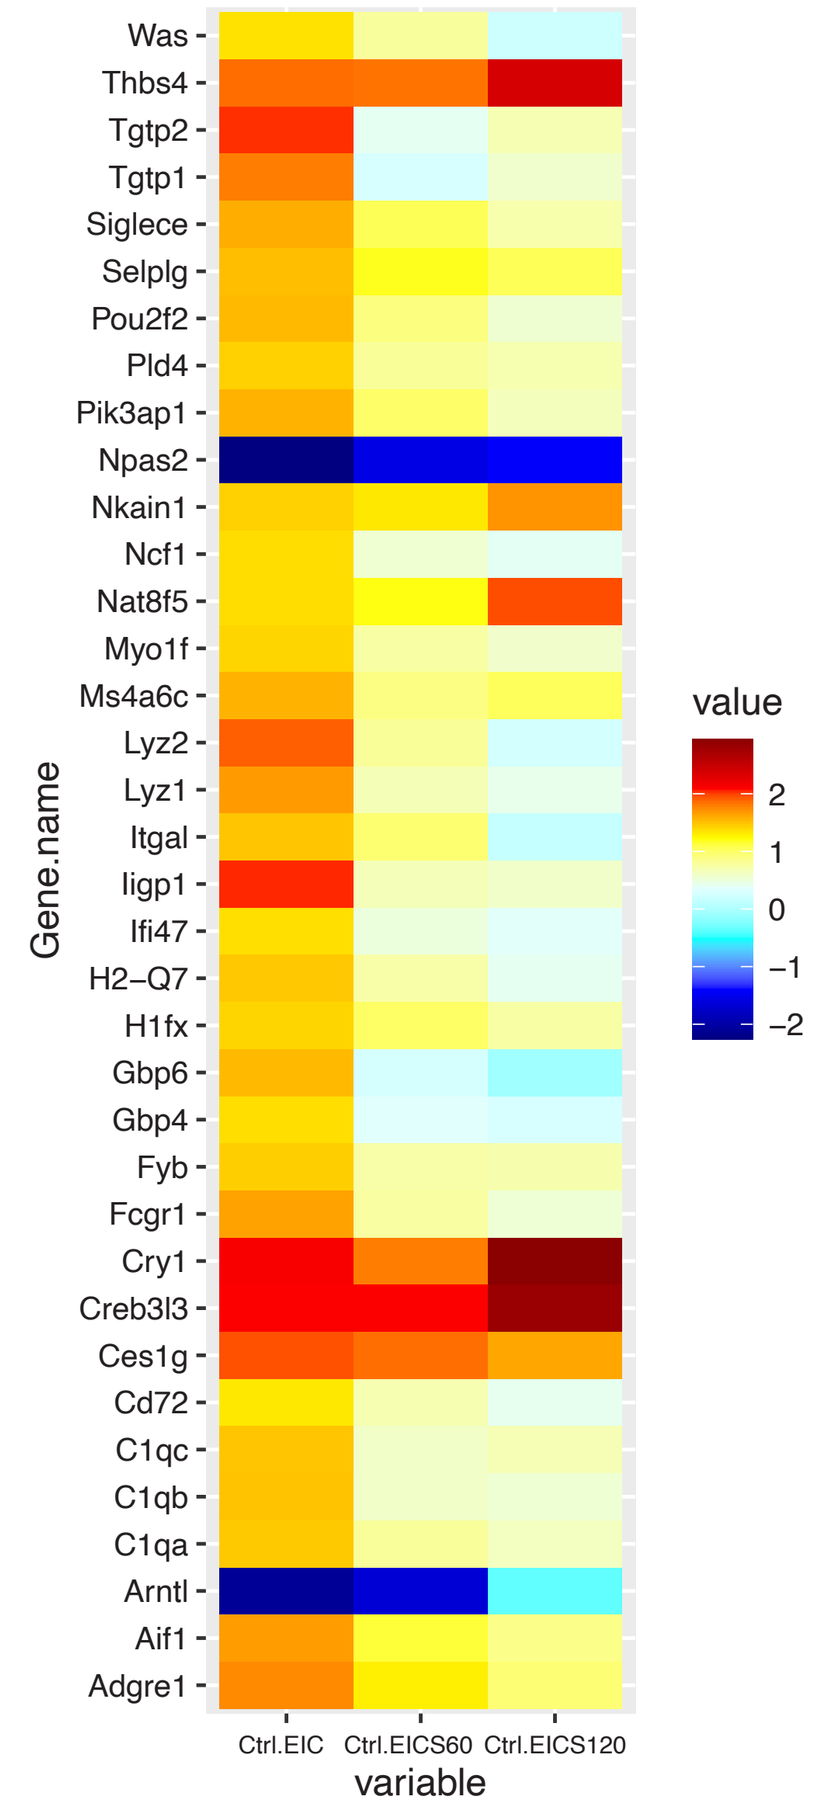
Was
Thbs4
Tgtp2
Tgtp1
Siglece
Selplg
Pou2f2
Pld4
Pik3ap1
Npas2
Nkain1
Ncf1
Nat8f5
Myo1f
Ms4a6c
Lyz2
Lyz1
Itgal
Iigp1
Ifi47
H2−Q7
H1fx
Gbp6
Gbp4
Fyb
Fcgr1
Cry1
Creb3l3
Ces1g
Cd72
C1qc
C1qb
C1qa
Arntl
Aif1
Adgre1
Gene.name
Ctrl.EIC
Ctrl.EICS60
Ctrl.EICS120
variable
value
2
1
0
−1
−2

WGCNA analysis between grouped pairs (control vs. EIC, control vs. EIC+S120, control vs. EIC+S60) was performed to find module eigengenes that correlate with nuclei per glomerulus and Ki67 positivity. Genes from modules significantly associated with cellular phenotypes (p-adj. <0.05), were then cross-compared to DESeq2 LFC analysis.

Gene networks in IgAN human tissue partially overlap with dysregulation in EIC animal model treated with sparsentan

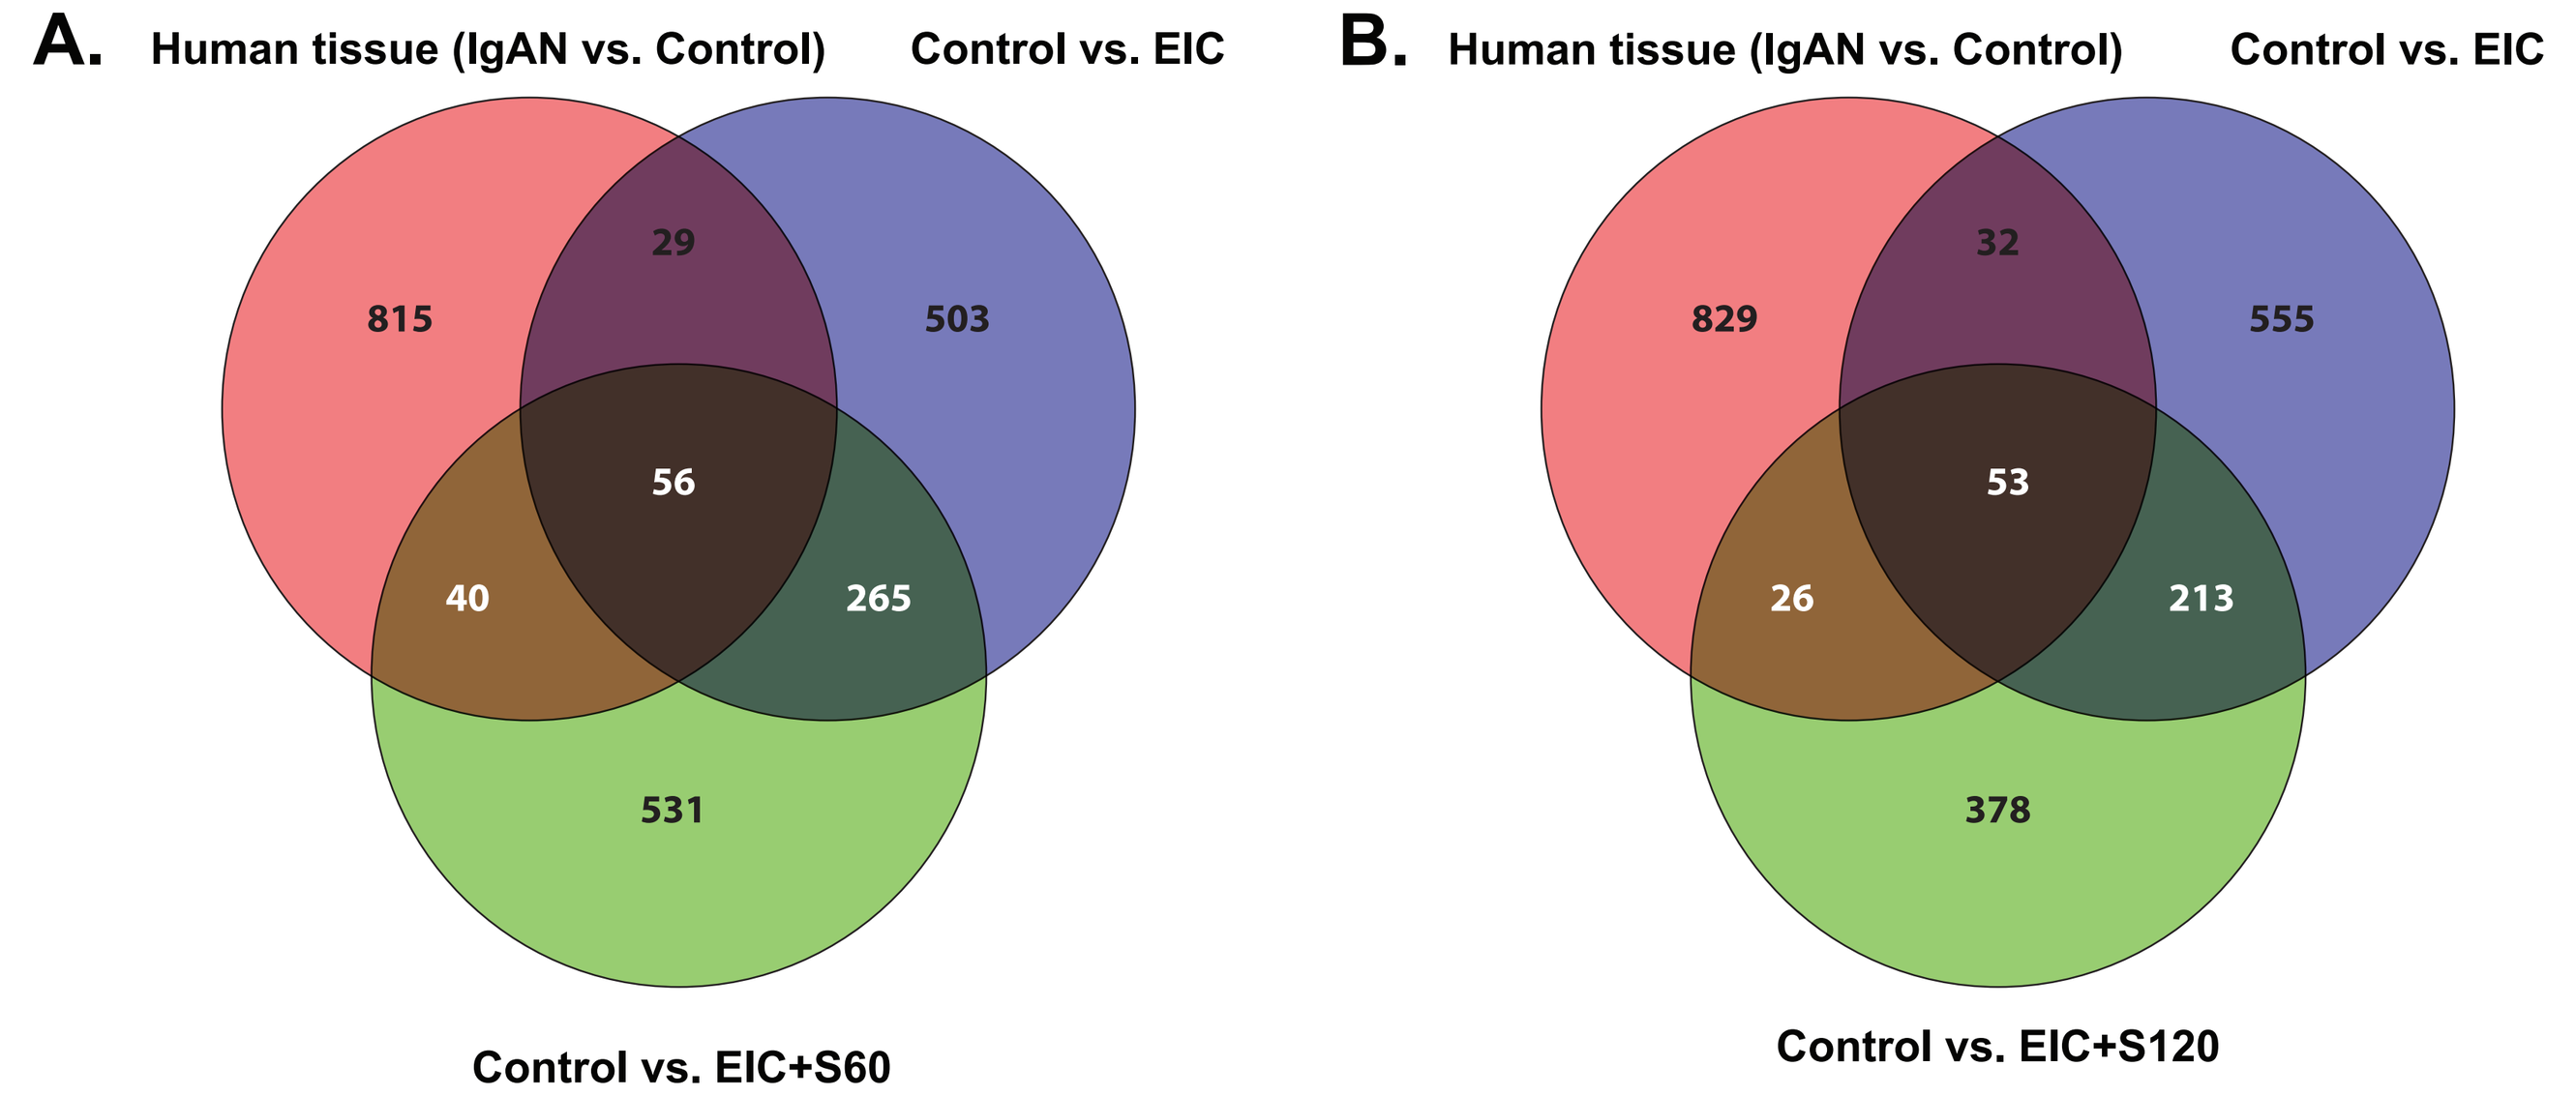
A.
Human tissue (IgAN vs. Control)
Control vs. EIC
815
29
503
56
40
265
531
Control vs. EIC+S60
B.
Human tissue (IgAN vs. Control)
Control vs. EIC
829
32
555
53
26
213
378
Control vs. EIC+S120

Transcriptional data of micro-dissected glomeruli from IgAN and control renal biopsies were analyzed using DESeq2. Statistically significant genes were compared to top module WGCNA genes from multiple pairwise analysis.[3]

Summary and Conclusions

Nude mice injected with EIC exhibited glomerular injury manifested by hypercellularity; this injury was prevented by treatment with sparsentan at 60 and 120 mg/kg doses. Changes at the transcriptional level for EIC-injected mice showed significant upregulation in pathways associated with cytokine stimulation and immune and cellular activation. These changes were reduced by sparsentan, as indicated by LFC of those critical genes in the treatment groups. Analysis of differential gene expression data from glomeruli of IgAN patients and comparison with the mouse model using EIC and EIC with sparsentan indicate that a subset of the genes with altered expression is located in the glomeruli.

Disclosure Information

Celia Jenkinson, Radko Komers, and Tiziano Pramparo are full-time employees of Travere Therapeutics, Inc., and may have an equity or other financial interest in Travere Therapeutics, Inc. The mouse IgAN studies were performed in the laboratory of Dr. Jan Novak at the University of Alabama at Birmingham and were funded by Travere Therapeutics, Inc. All authors had access to the data and participated in the development, review, and approval of the poster during the development process. Zina Moldoveanu and Jan Novak are co-inventors on US patent application 14/318,082 (assigned to UAB Research Foundation). Jan Novak is a co-founder and co-owner of and consultant for Reliant Glycosciences, LLC.

References

1. Moldoveanu Z, et al. *J Autoimmun*. 2021;118:102593. doi: 10.1016/j.jaut.2021.102593.
2. Jenkinson C, et al. Poster presented at WCN 2019, Melbourne, Australia (poster #SAT-010).
3. Berthier CC, et al. *J Immunol*. 2012;189:988-1001.

Presented at the American Society of Nephrology Kidney Week, November 4-7, 2021, San Diego, CA