Poster# P-036

Population Pharmacokinetic Analysis of Sparsentan in Healthy Volunteers and Subjects with Focal Segmental Glomerulosclerosis (FSGS)

Russell Wada,[1] Huub Jan Kleijn,[1] Lu Zhang,[1] Shang-Chiung Chen[2]

[1]Certara, Inc., Menlo Park, CA, USA; [2]Travere Therapeutics, Inc., San Diego, CA, USA.

RESULTS

- 10,957 samples from a total of 446 healthy volunteers/patients (1,692 samples from 195 FSGS patients) were used in the population PK analysis after applying exclusion criteria (eg, PK samples that were below the lower limit of quantitation, missing time of dosing, or had conditional weighted residuals [CWRES] >5)

Model Summary

- Sparsentan PK was best described by a 2-compartment model with first-order absorption and absorption lag time (Tlag)
 - Relative bioavailability (Frel) decreased with increasing sparsentan dose (at doses ≥200 mg), reflecting less than dose-proportional exposure
 - At the target dose of 800 mg for FSGS patients, the typical value of apparent clearance (CL/F) was 5.47 L/h after a single dose, increasing to 7.21 L/h at steady state
 - The typical value of apparent central volume of distribution (Vc/F) was 69.5 L
 - The terminal half-life was 9.6 hours at steady state
- Diagnostic plots and parameter estimates for the final population PK model are shown in **Figure 1 and Table 2**, respectively

Covariate Relationships

- 6 covariate parameter relationships were retained in the final model
 - Effects of alkaline phosphatase, creatinine clearance (CrCL), co-administration with moderate/strong CYP3A4 inhibitors, and sex on clearance (CL)
 - Effects of race on central volume of distribution (Vc)
 - Effects of formulation on Tlag and on absorption rate constant (KA)

Figure 1. Goodness-of-Fit Plots for Final Model

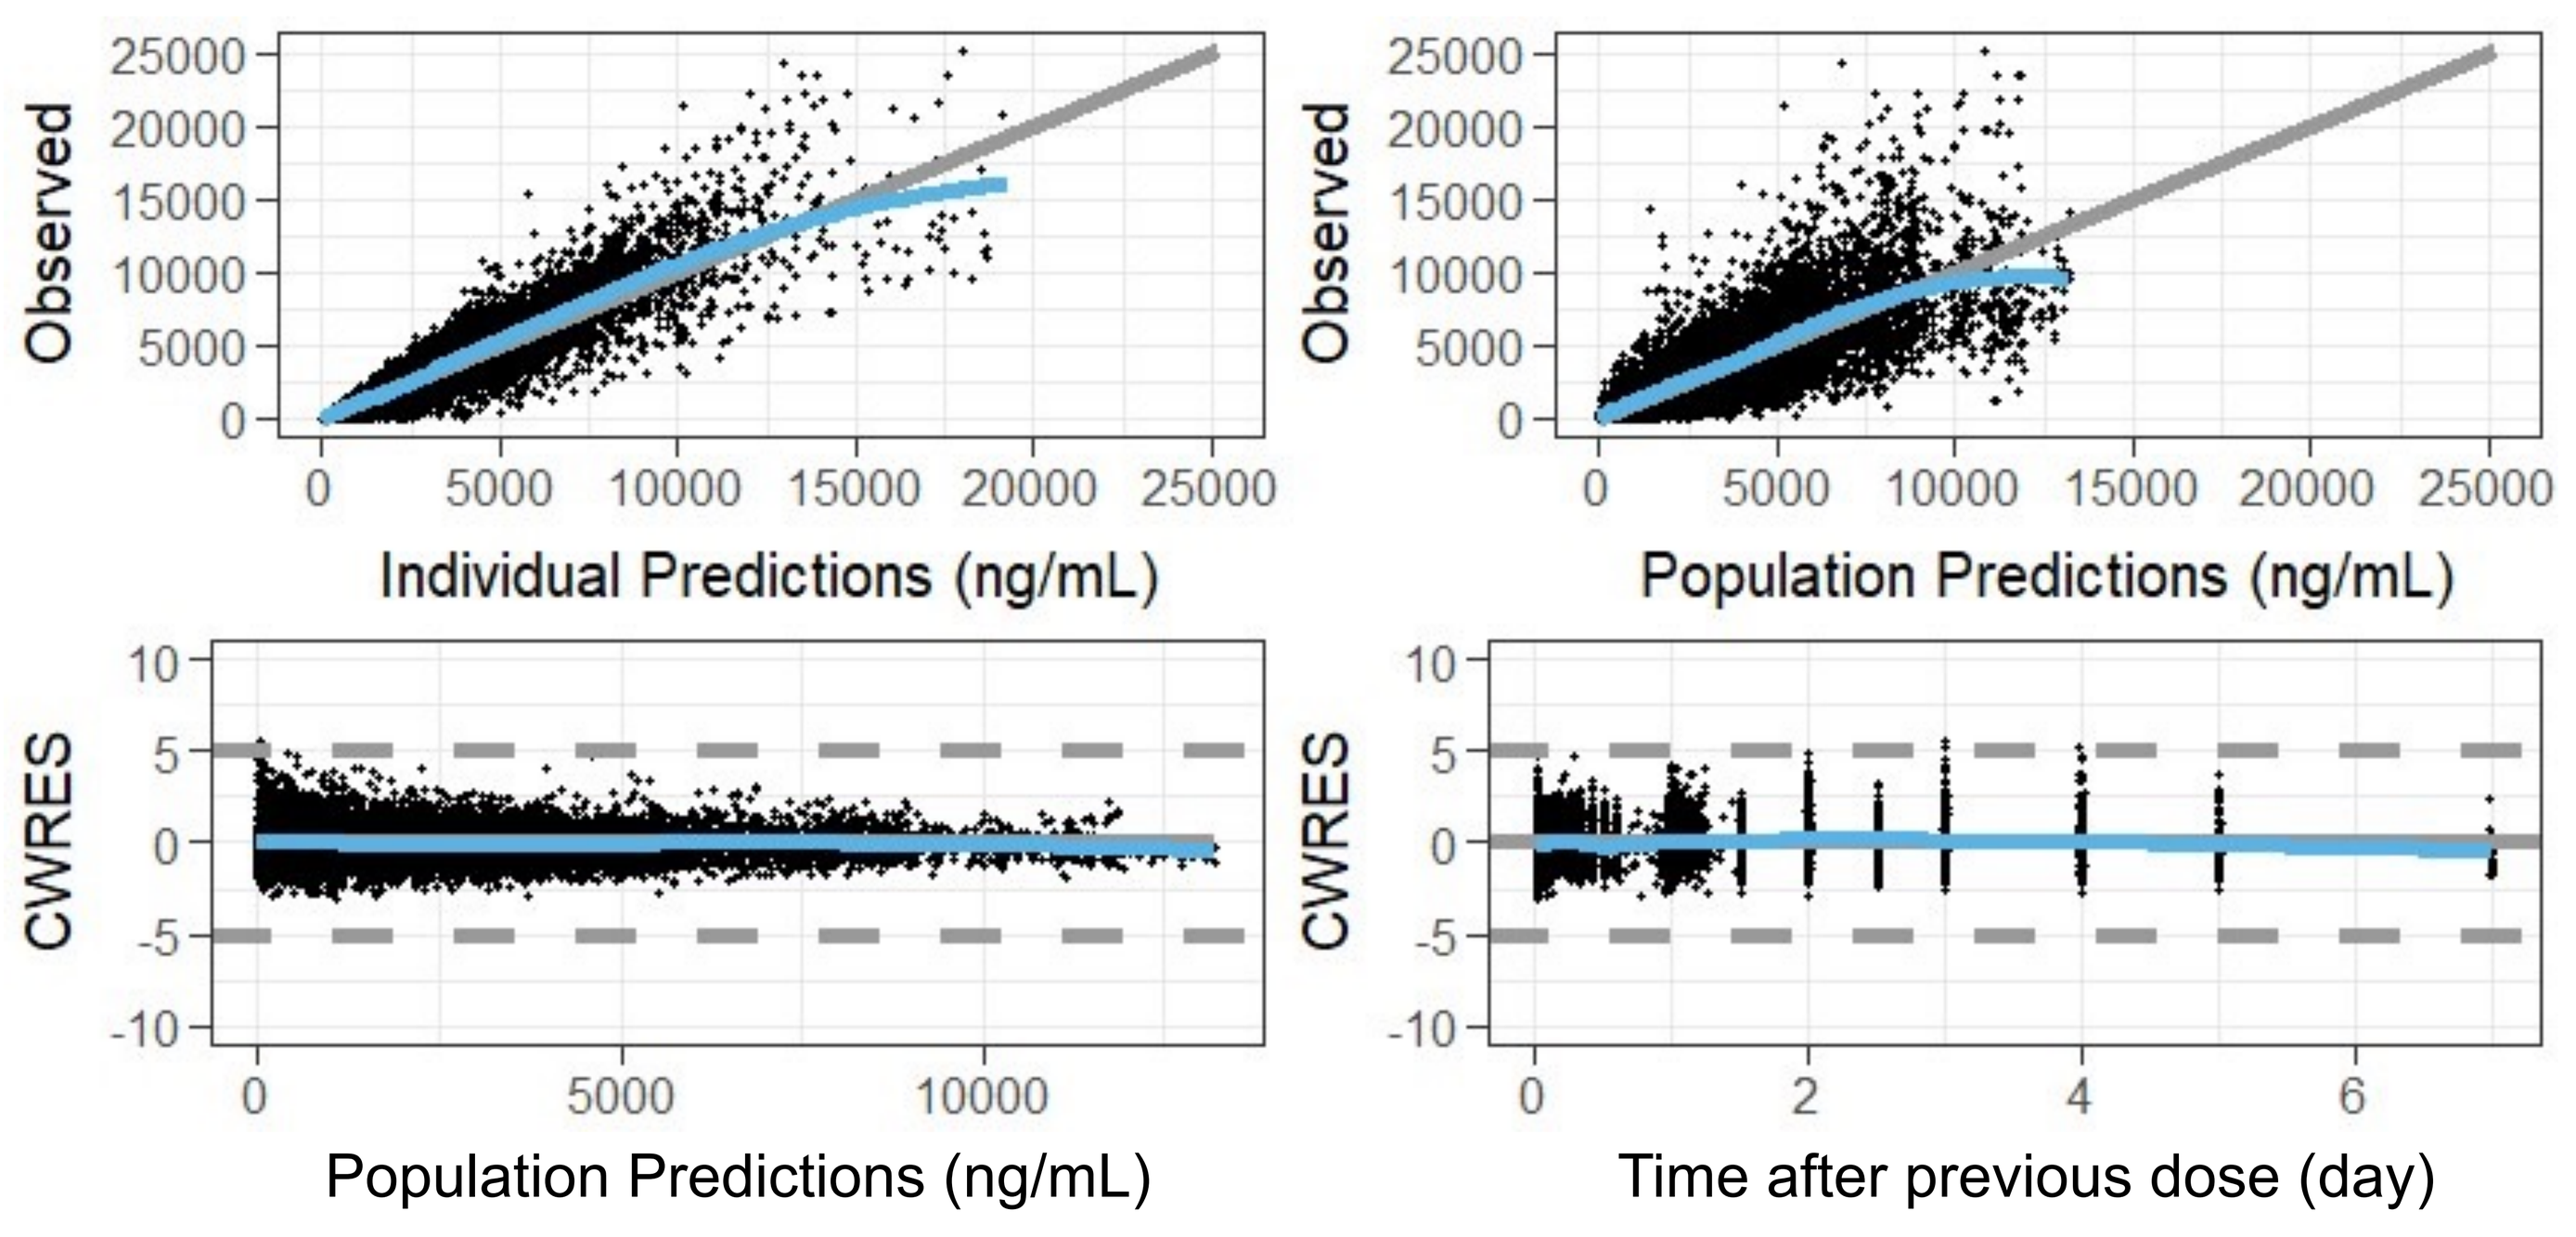
Observed
Individual Predictions (ng/mL)
Observed
Population Predictions (ng/mL)
CWRES
Population Predictions (ng/mL)
CWRES
Time after previous dose (day)

CWRES, conditional weighted residuals.
Dots are individual data points; gray solid lines are either lines of unity or zero lines; dashed gray lines demarcate residual levels of -5 to +5; blue lines are locally weighted smoothing lines.

- Other covariates, including population (FSGS patient versus healthy volunteer), age, weight, serum aspartate aminotransferase, serum alanine aminotransferase, total bilirubin, albumin, and total protein, had no significant effect on the apparent clearance or apparent volume of distribution of sparsentan
- No effects of concomitant acid reducer, mild CYP3A4 inhibitor, CYP3A4 inducer, or P-glycoprotein (P-gp) inhibitor were detected
- Prediction-corrected visual predictive checks (pcVPC) after the first dose and at steady state are shown in **Figure 2** for healthy volunteers (**A and B**) and for patients with FSGS (**C and D**)
- The univariate effects of each covariate included in the final model on exposure at steady state after administration of once-daily sparsentan 800 mg in FSGS patients are shown in **Figure 3**
 - Co-administration of CYP3A4 inhibitors increased steady-state area under the plasma concentration-time curve (AUC) by 31% for moderate CYP3A4 inhibitors and 191% for strong CYP3A4 inhibitors
 - The effects of covariates were not considered to be clinically meaningful, except for that of strong CYP3A4 inhibitors

Figure 2. Prediction-corrected Visual Predictive Checks of Final Model in Healthy Volunteers and Those With Hepatic Impairment (A), Healthy Volunteers (B), and in Patients with FSGS (C and D)

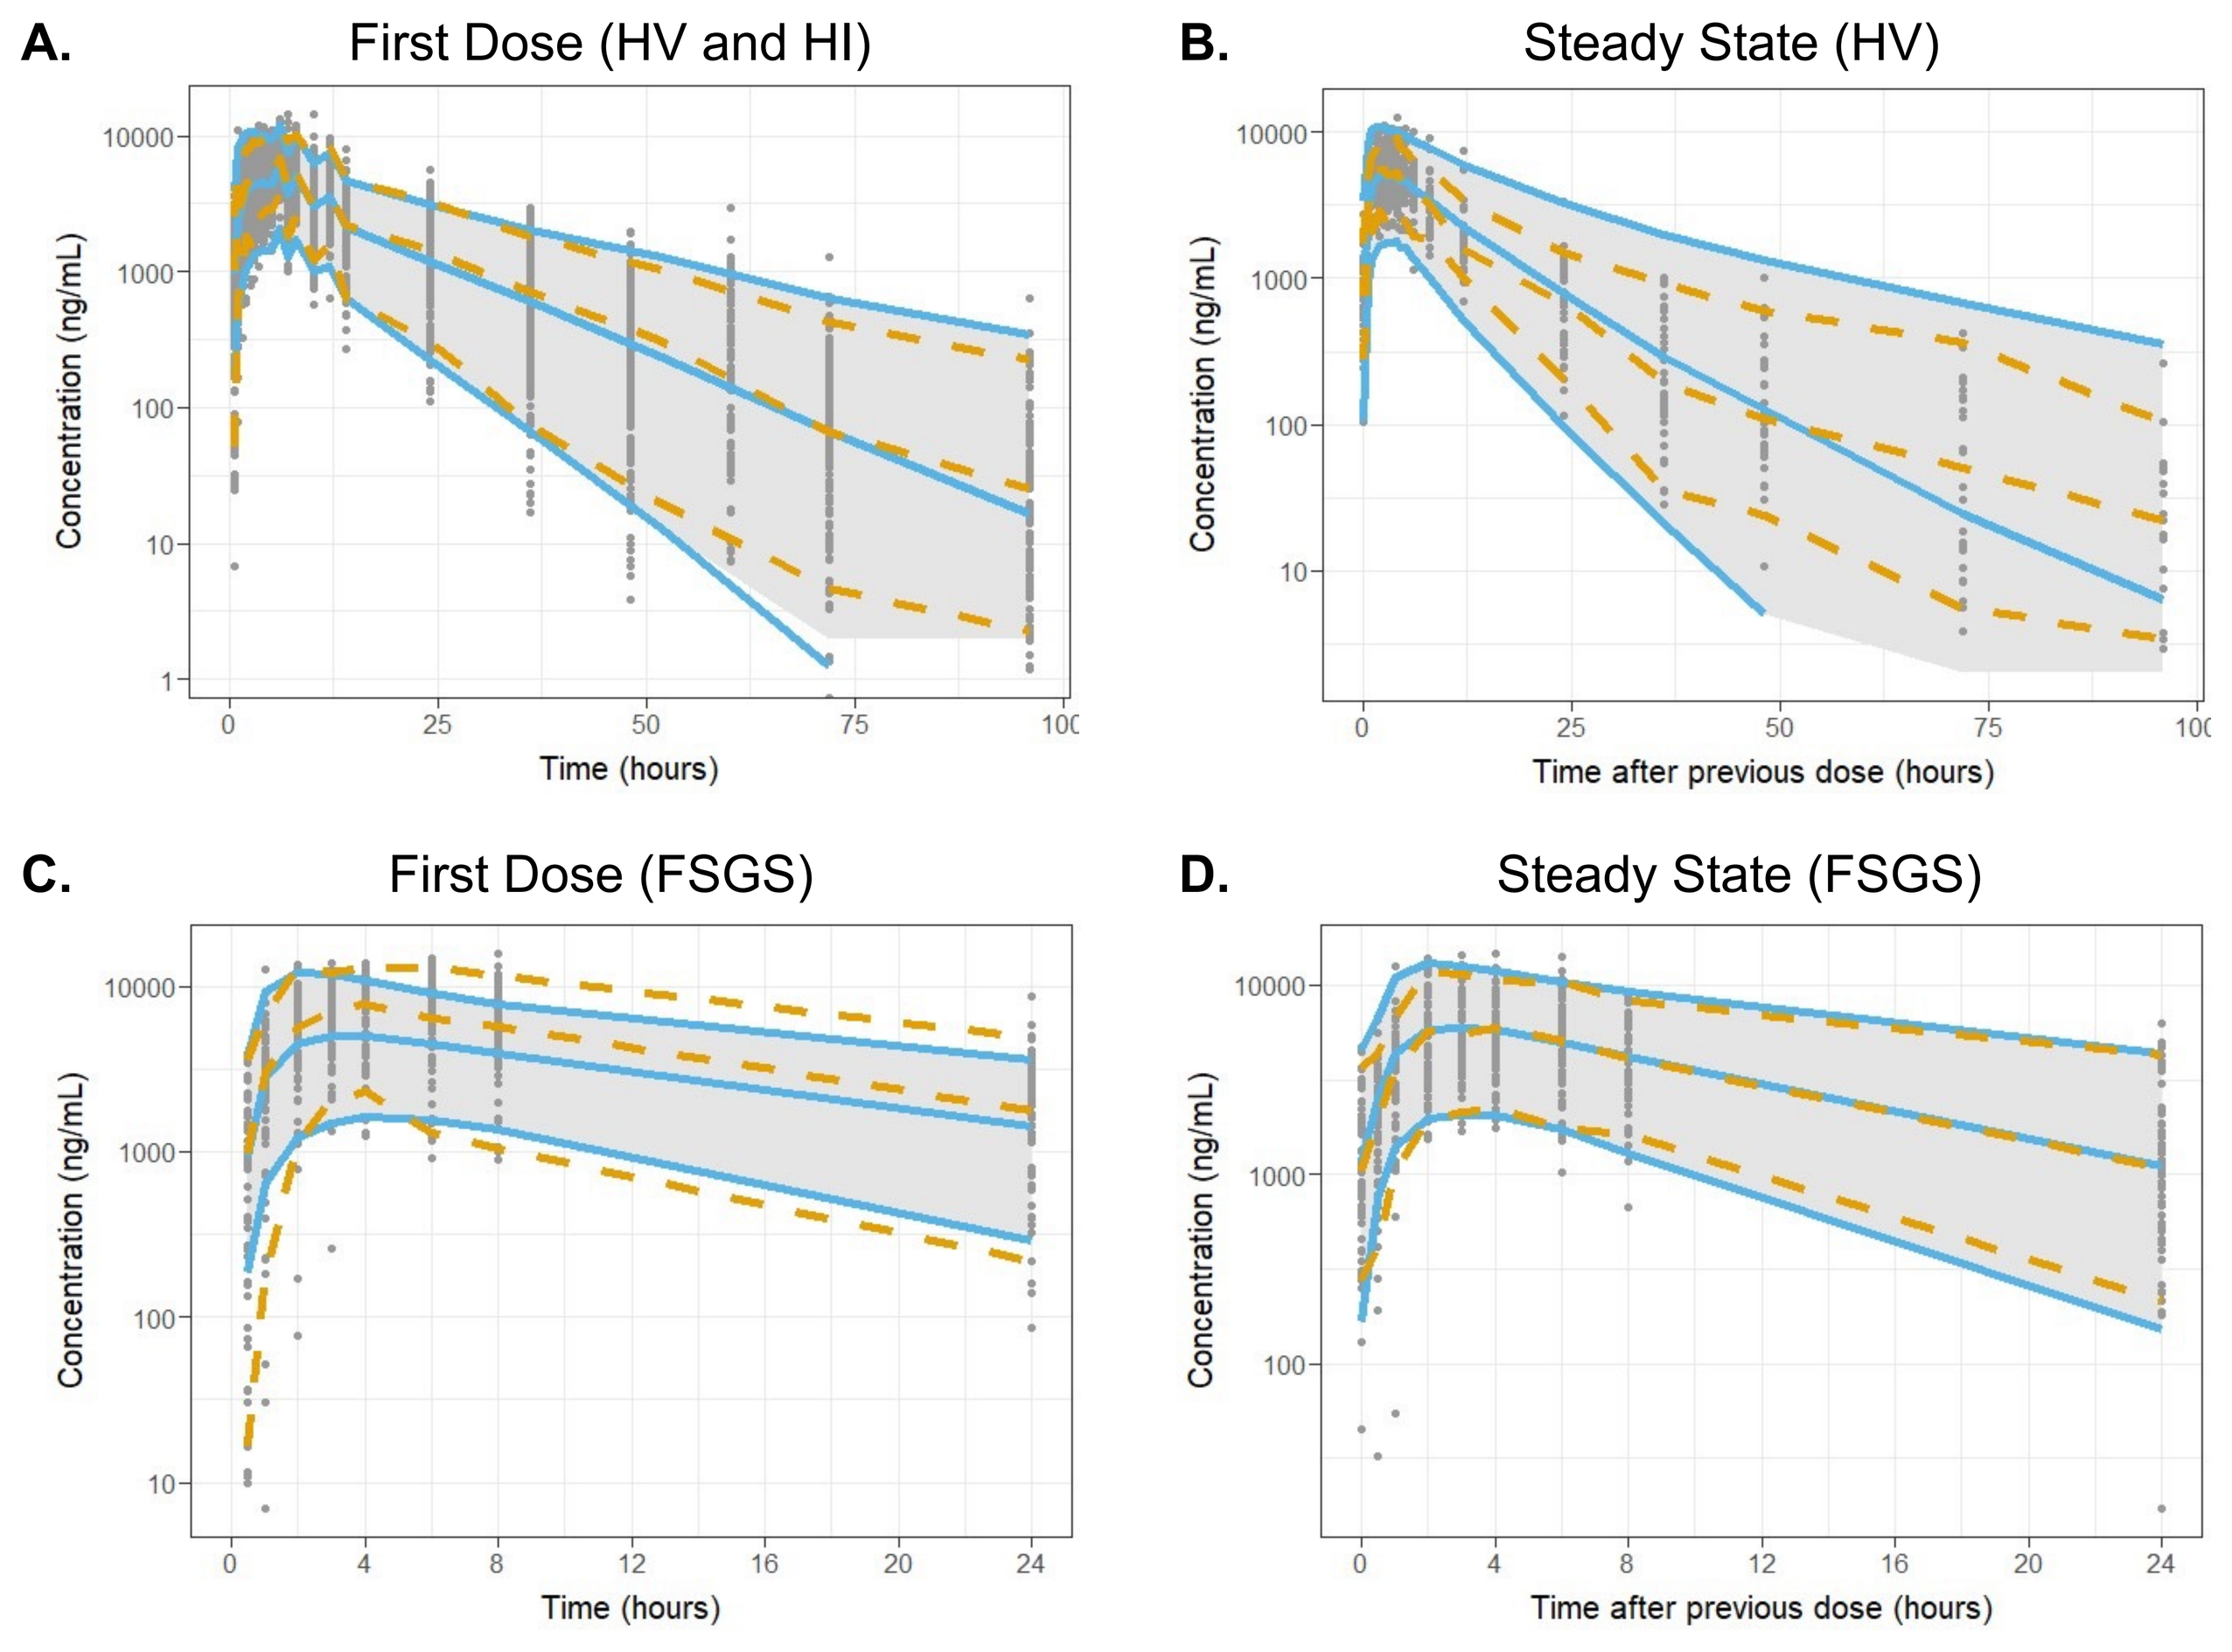
A. First Dose (HV and HI)
Concentration (ng/mL)
Time (hours)
B. Steady State (HV)
Concentration (ng/mL)
Time after previous dose (hours)
C. First Dose (FSGS)
Concentration (ng/mL)
Time (hours)
D. Steady State (FSGS)
Concentration (ng/mL)
Time after previous dose (hours)

FSGS, focal segmental glomerulosclerosis; HI, hepatic impairment; HV, healthy volunteers.
Dots are prediction-corrected concentrations.
Gray shaded area shows the 5th to 95th percentile range of the model prediction.
Blue lines show the 5th, median, and 95th percentile predictions.
Gold dashed lines show the 5th, median, and 95th percentile range of the concentrations.

Table 2. Final Model Parameter Estimates

Parameter	Estimate	RSE (%)	IIV (%)	Shrinkage (%)
Apparent clearance (CL/F, L/h)	3.88	4.6	39.5	4.7
Apparent central volume (Vc/F, L)	49.3	4.3	48.4	17.2
Apparent distribution clearance (Q/F, L/h)	2.03	12.0	--	--
Apparent peripheral volume (Vp/F, L)	12.1	10.5	--	--
Absorption rate (KA, 1/h)	0.740	6.9	68.9	22.2
Absorption lag time (Tlag, h)	0.32	4.0	NA	NA
Induction change in clearance (L/h)	1.23	13.6		
Induction half-life ($T_{1/2}$, day)	0.001	FIXED		
Dose on relative bioavailability	-0.495	5.1		
Moderate CYP3A4 inhibitor on CL	-0.273	18.8		
Strong CYP3A4 inhibitor on CL	-1.069	10.0		
Alkaline phosphatase on CL	-0.208	27.5		
Creatinine clearance on CL	0.222	26.5		
Male on CL	0.139	32.8		
Black or African American on Vc	0.309	18.4		
Asian on Vc	0.265	48.4		
Tablet on KA	-0.306	34.8		
Crushed tablet on KA	0.080	159.1		
Tablet on Tlag	-0.269	29.1		
Crushed tablet on Tlag	-1.175	28.7		
Variance in CL	0.156	8.7		
Variance in Vc	0.234	11.3		
Variance in KA	0.474	10.2		
SD of additive error (ng/mL)	2	FIXED		
SD of proportional error	0.365	1.9		

ALKP, alkaline phosphatase; CL, clearance; CL/F, apparent clearance; CrCL, creatinine clearance; CYP, cytochrome P450; F, bioavailability; Frel, relative bioavailability; IIV, interindividual variance; KA, absorption rate constant; NA, not available; Q, distribution clearance; RSE, residual squared error; SD, standard deviation; $T_{1/2}$, half-life; Tlag, absorption lag time; Vc, central volume of distribution; Vc/F, apparent central volume of distribution, Vp, peripheral volume of distribution.
Frel=(Dose/400)$^{-0.495}$ if Dose ≥200 mg, F=(200/400)$^{-0.495}$ if Dose <200 mg. Steady-state CL=CL/F + induction change in CL; the RSE is not estimated for this parameter as well as other derived parameters. Induction half-life and SD of additive error are fixed. The reference subject is a white female receiving a 400-mg capsule, no CYP3A4 inhibitor, with CrCL of 112 mL/min, and with ALKP of 68 U/L.

Figure 3. Tornado Plots Showing the Effects of Covariates on Steady State AUC and C_{max}

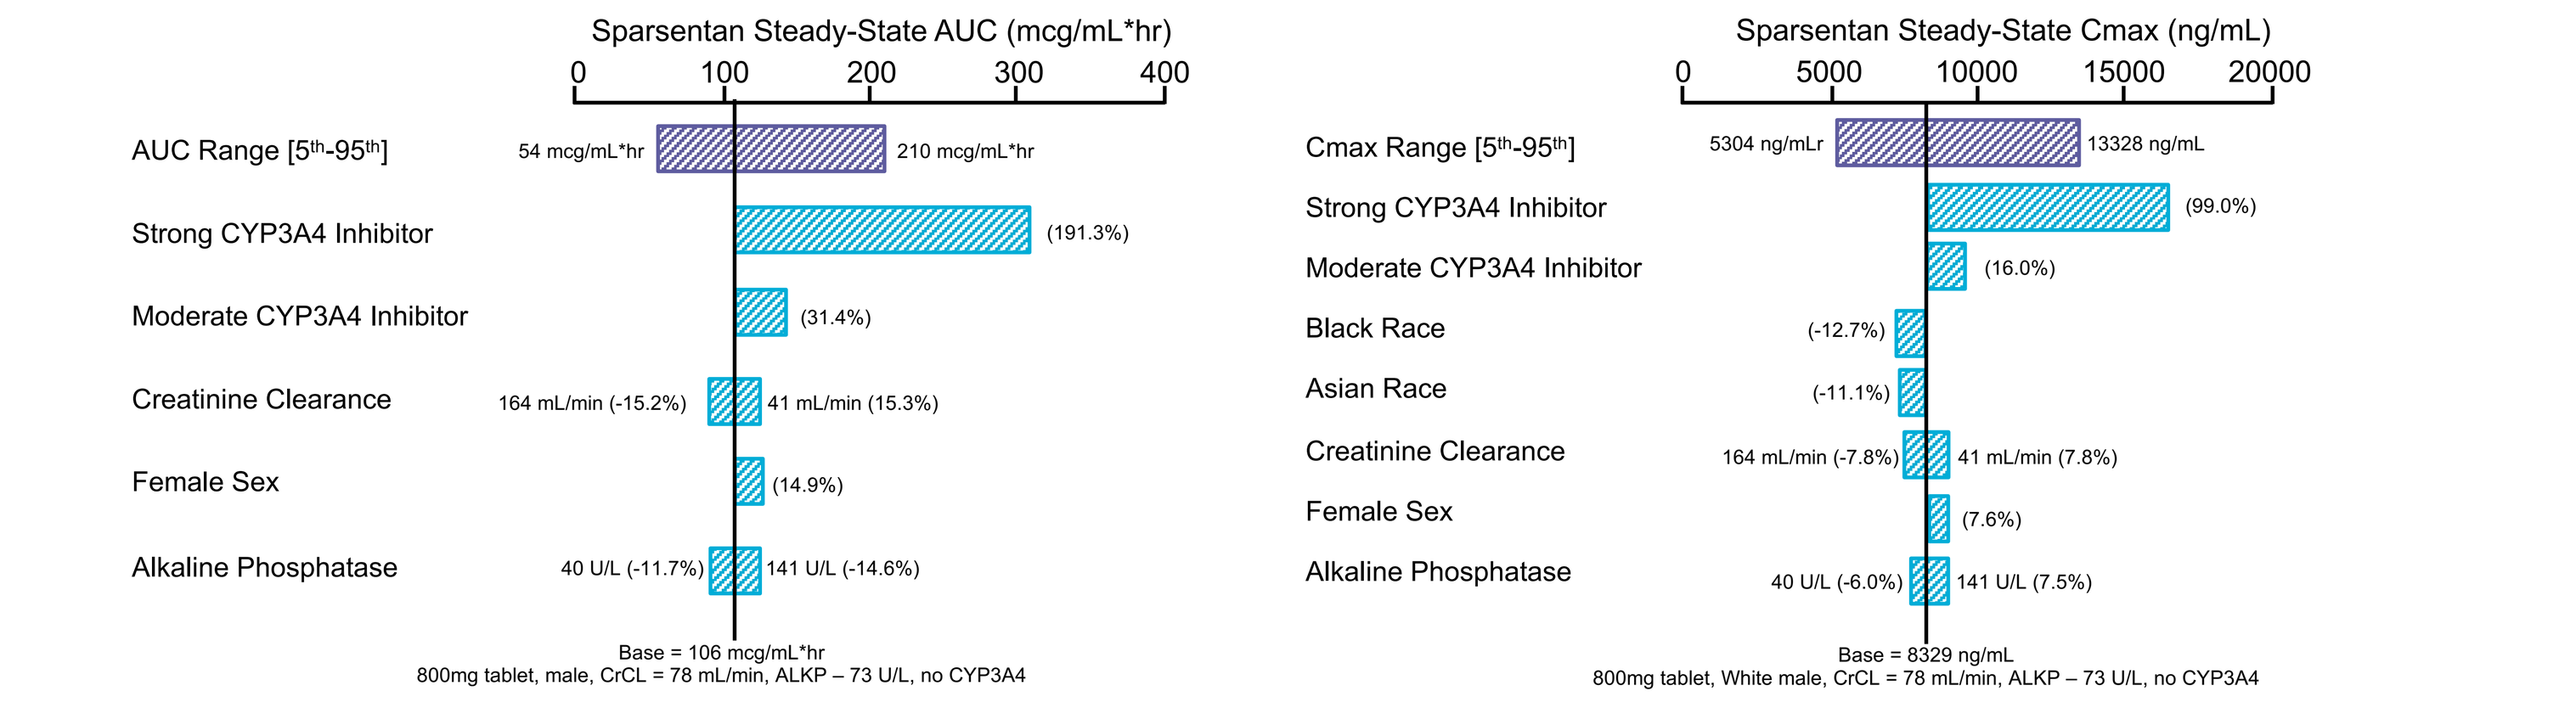
Sparsentan Steady-State AUC (mcg/mL*hr)
AUC Range [5th-95th]
54 mcg/mL*hr
210 mcg/mL*hr
Strong CYP3A4 Inhibitor
(191.3%)
Moderate CYP3A4 Inhibitor
(31.4%)
Creatinine Clearance
164 mL/min (-15.2%)
41 mL/min (15.3%)
Female Sex
(14.9%)
Alkaline Phosphatase
40 U/L (-11.7%)
141 U/L (-14.6%)
Base = 106 mcg/mL*hr
800mg tablet, male, CrCL = 78 mL/min, ALKP – 73 U/L, no CYP3A4
Sparsentan Steady-State Cmax (ng/mL)
Cmax Range [5th-95th]
5304 ng/mLr
13328 ng/mL
Strong CYP3A4 Inhibitor
(99.0%)
Moderate CYP3A4 Inhibitor
(16.0%)
Black Race
(-12.7%)
Asian Race
(-11.1%)
Creatinine Clearance
164 mL/min (-7.8%)
41 mL/min (7.8%)
Female Sex
(7.6%)
Alkaline Phosphatase
40 U/L (-6.0%)
141 U/L (7.5%)
Base = 8329 ng/mL
800mg tablet, White male, CrCL = 78 mL/min, ALKP – 73 U/L, no CYP3A4

ALKP, alkaline phosphatase; AUC, area under the plasma concentration-time curve; C_{max}, maximum plasma concentration; CrCL, creatinine clearance; CYP, cytochrome P450; FSGS, focal segmental glomerulosclerosis.
Base, as represented by the black vertical line, refers to the predicted steady-state AUC of sparsentan in a typical FSGS male subject receiving an 800 mg tablet with ALKP of 73 U/L, with CrCL of 78 mL/min, and not receiving a moderate or strong CYP3A4 inhibitor. Red shaded bar shows the 5th to 95th percentile exposure range in the FSGS population. Blue shaded bars represent the influence of a single covariate (listed left) on the steady state exposure after once-daily sparsentan 800 mg. Blue bars are ranked in decreasing order of largest deviation from the base. Upper and lower values for each covariate capture 90% of the range in the population.

INTRODUCTION

- Focal segmental glomerulosclerosis (FSGS) is a rapidly progressive kidney disease that is often associated with kidney failure requiring dialysis or transplantation[1,2]
- Sparsentan is a novel, first-in-class, single-molecule Dual Endothelin Angiotensin Receptor Antagonist (DEARA) in evaluation for the treatment of FSGS
- Here we report results from a population pharmacokinetic (PK) analysis conducted to characterize the PK of sparsentan in healthy volunteers and FSGS patient populations and to evaluate the impact of disease characteristics and concomitant medications on its PK

METHODS

- The population PK analysis was conducted using PK data collected from 9 clinical studies, including 6 phase 1 studies in healthy volunteers (N=224), 1 phase 1 study in volunteers with hepatic impairment vs healthy volunteers (N=28), and the phase 2 DUET[3] (N=71) and phase 3 DUPLEX (N=125) studies in patients with primary and genetic FSGS
 - Across the phase 1 studies, volunteers were administered sparsentan doses ranging from 50 mg to 1,600 mg
 - For the DUET and DUPLEX studies, patients received 200 mg, 400 mg, or 800 mg of once-daily sparsentan
- Blood samples were collected to determine sparsentan concentrations using validated liquid chromatography-tandem mass spectrometry methods
- The population PK analysis was performed using NONMEM Version 7.5 with the first-order conditional estimation with eta-epsilon interaction (FOCE-I) method
 - Model quality was assessed by inspection of model parameters and their confidence intervals, residual-based goodness-of-fit plots, distributions of random effects, and prediction-corrected visual predictive checks
- The covariates assessed in this population PK model are summarized in **Table 1**
 - Model covariates were selected using a forward addition and backward elimination method based on significance levels of p<0.01 and p<0.001, respectively

Table 1. Covariates Evaluated in the Population PK Model

Category	Covariate	PK Parameter
Demographics	WT	CL/F, Vc/F
	Age	CL/F, Vc/F
	Sex (Male/Female)	CL/F, Vc/F
	Race	CL/F, Vc/F
Hepatic function	Serum albumin	CL/F
	AST	CL/F
	ALT	CL/F
	Total bilirubin	CL/F
	Alkaline phosphatase	CL/F
Renal function	Creatinine clearance	CL/F
Food	Food (fast/fed/nonspecified)	Frel, KA, Tlag
Formulation	Dose	Frel, KA, Tlag
	Formulation (capsule/tablet/crushed tablet)	Frel, KA, Tlag
Population	FSGS vs non-FSGS	CL/F, Vc/F
Concomitant medications	Acid-reducing agent	Frel, KA, Tlag
	CYP3A4 inducer	CL/F
	CYP3A4 inhibitors	CL/F
	P-gp inhibitor	CL/F

ALT, alanine transaminase; AST, aspartate transaminase; CL/F, apparent clearance; CYP, cytochrome P450; Frel, relative bioavailability; FSGS, focal segmental glomerulosclerosis; KA, absorption rate constant; P-gp, P-glycoprotein; PK, pharmacokinetic; Tlag, absorption lag time; Vc/F, apparent central volume of distribution; WT, body weight.

CONCLUSIONS

- The PK of sparsentan was well characterized by a two-compartment model with first order absorption with lag time, dose-dependent bioavailability, and first order elimination from the central compartment; a modest increase in clearance at steady state was observed
- Covariates associated with statistically significant but not clinically meaningful PK variability included formulation, sex, race, creatinine clearance, and serum alkaline phosphatase; however, the lack of clinically meaningful effects suggest that dose adjustments are not needed for each of these variables
- This population PK analysis suggests that dose reductions for sparsentan might be warranted for patients taking strong CYP3A4 inhibitors, but not across the range of other concomitant medications, patient characteristics, and disease characteristics analyzed

DISCLOSURES

RW: Currently with QuanTx Consulting. Formerly with Certara, a paid consultant to Travere Therapeutics, Inc. in connection with this work.

HJK: Employee of Certara, a paid consultant to Travere Therapeutics, Inc. in connection with this work.

LZ: Employee of Certara, a paid consultant to Travere Therapeutics, Inc. in connection with this work.

S-CC: Employee and equity ownership, Travere Therapeutics, Inc.

ACKNOWLEDGMENTS

This study was supported by Travere Therapeutics, Inc. (San Diego, CA). Medical writing assistance was provided by MedVal Scientific Information Services, LLC (Princeton, NJ) and was funded by Travere Therapeutics, Inc.

REFERENCES

1. Rosenberg AZ, et al. *Clin J Am Soc Nephrol*. 2017;12:502-517.
2. D'Agati VD, et al. *New Engl J Med*. 2011;365:2398-2411.
3. Trachtman H, et al. *J Am Soc Nephrol*. 2018;29:2745-2754.

Contact information: Shang-Chiung Chen, charles.chen@travere.com

Presented at the American Society for Clinical Pharmacology and Therapeutics (ASCPT) Online Annual Meeting; March 16-18, 2022