Genetic Testing Identified Causative Variants in Nearly 20% of Early-Onset Bilateral Cataracts Cases

Erin D. Stahl, MD[1]; Jamie C. Fox, PhD[2]; Dana M. Talsness, PhD[2]; Amy Ponte, PhD, MPH[3]*; Rana Dutta, PhD[3]; Sharon F. Freedman, MD[4]; Bharti R. Nihalani, MD[5]; Deborah K. VanderVeen, MD[5]

[1]Pediatric Ophthalmology, Children's Mercy Hospital, Kansas City, Missouri, USA; [2]PreventionGenetics, LLC, Marshfield, Wisconsin, USA; [3]Travere Therapeutics, Inc., San Diego, California, USA; [4]Department of Ophthalmology, Duke University Medical Center, Durham, North Carolina, USA; [5]Boston Children's Hospital, Harvard Medical School, Boston, Massachusetts, USA

*Former employee of Travere Therapeutics, Inc.

INTRODUCTION

- Early-onset cataracts occur in patients younger than 40 years of age
 - In children, the prevalence is 1-15 per 10,000[1]
- Cataracts can present in patients in both syndromic and non-syndromic forms
 - For example, cataracts can often occur as one of the initial symptoms of cerebrotendinous xanthomatosis (CTX), a treatable disorder of cholesterol metabolism
- Genetic testing for early-onset cataracts can clarify disease etiology, identify the disease earlier, inform patient management, and improve outcomes

OBJECTIVES

- To identify genetic variants responsible for early-onset idiopathic cataracts as part of Travere's "Behind the Blur" genetic testing program

METHODS

- We analyzed 374 individuals with acquired cataracts (ages 18 months to 35 years) via a next-generation sequencing (NGS) panel of 66 genes
 - Individuals >35 years old can qualify for testing with proof of childhood onset cataracts
- All testing was completed at PreventionGenetics, a CLIA-approved laboratory, using PGxome® test as part of Travere's sponsored testing program "Behind the Blur: No-Cost Patient Genetic Testing Program for Early-Onset Bilateral Cataracts" (https://www.preventiongenetics.com/sponsoredTesting/Travere_Cataracts)
- This panel typically provides ≥98% coverage of all coding exons of the genes listed, plus ~10 bases of flanking noncoding DNA
- Coverage was defined as ≥20× NGS or Sanger sequencing reads
- Sanger sequencing was used to confirm variants as necessary
- Copy number variants (CNVs) were also detected from NGS data and confirmed via a secondary method, such as microarray-based Comparative Genomic Hybridization (aCGH), multiplex ligation-dependent probe amplification (MLPA), or polymerase chain reaction (PCR), before they were reported

RESULTS

- Of the 347 patients tested, 43.2% were female, 55.3% were male, and 1.4% were sex unknown
- Baseline age categories are shown in **Table 1**

Table 1. Age categories of patients

Age Group	Female n=150	Male n=192	Unknown n=5	Total N=347
0 to <2 years	14 (9.3%)	11 (5.7%)	1 (20.0%)	26 (7.5%)
2 to <6 years	32 (21.3%)	52 (27.1%)	2 (40.0%)	86 (24.8%)
6 to <11 years	38 (25.3%)	73 (38.0%)	0	111 (32.0%)
11 to <19 years	48 (32.0%)	41 (21.4%)	1 (20.0%)	90 (25.9%)
19 to <36 years	15 (10.0%)	11 (5.7%)	1 (20.0%)	27 (7.8%)
≥36 years	3 (2.0%)	4 (2.1%)	0	7 (2.0%)

- A positive diagnostic outcome was found for 69 (19.9%) patients, 221 (63.7%) were indeterminate, and 57 (16.4%) were negative (**Figure 1**)

Figure 1. Diagnostic outcome by NGS

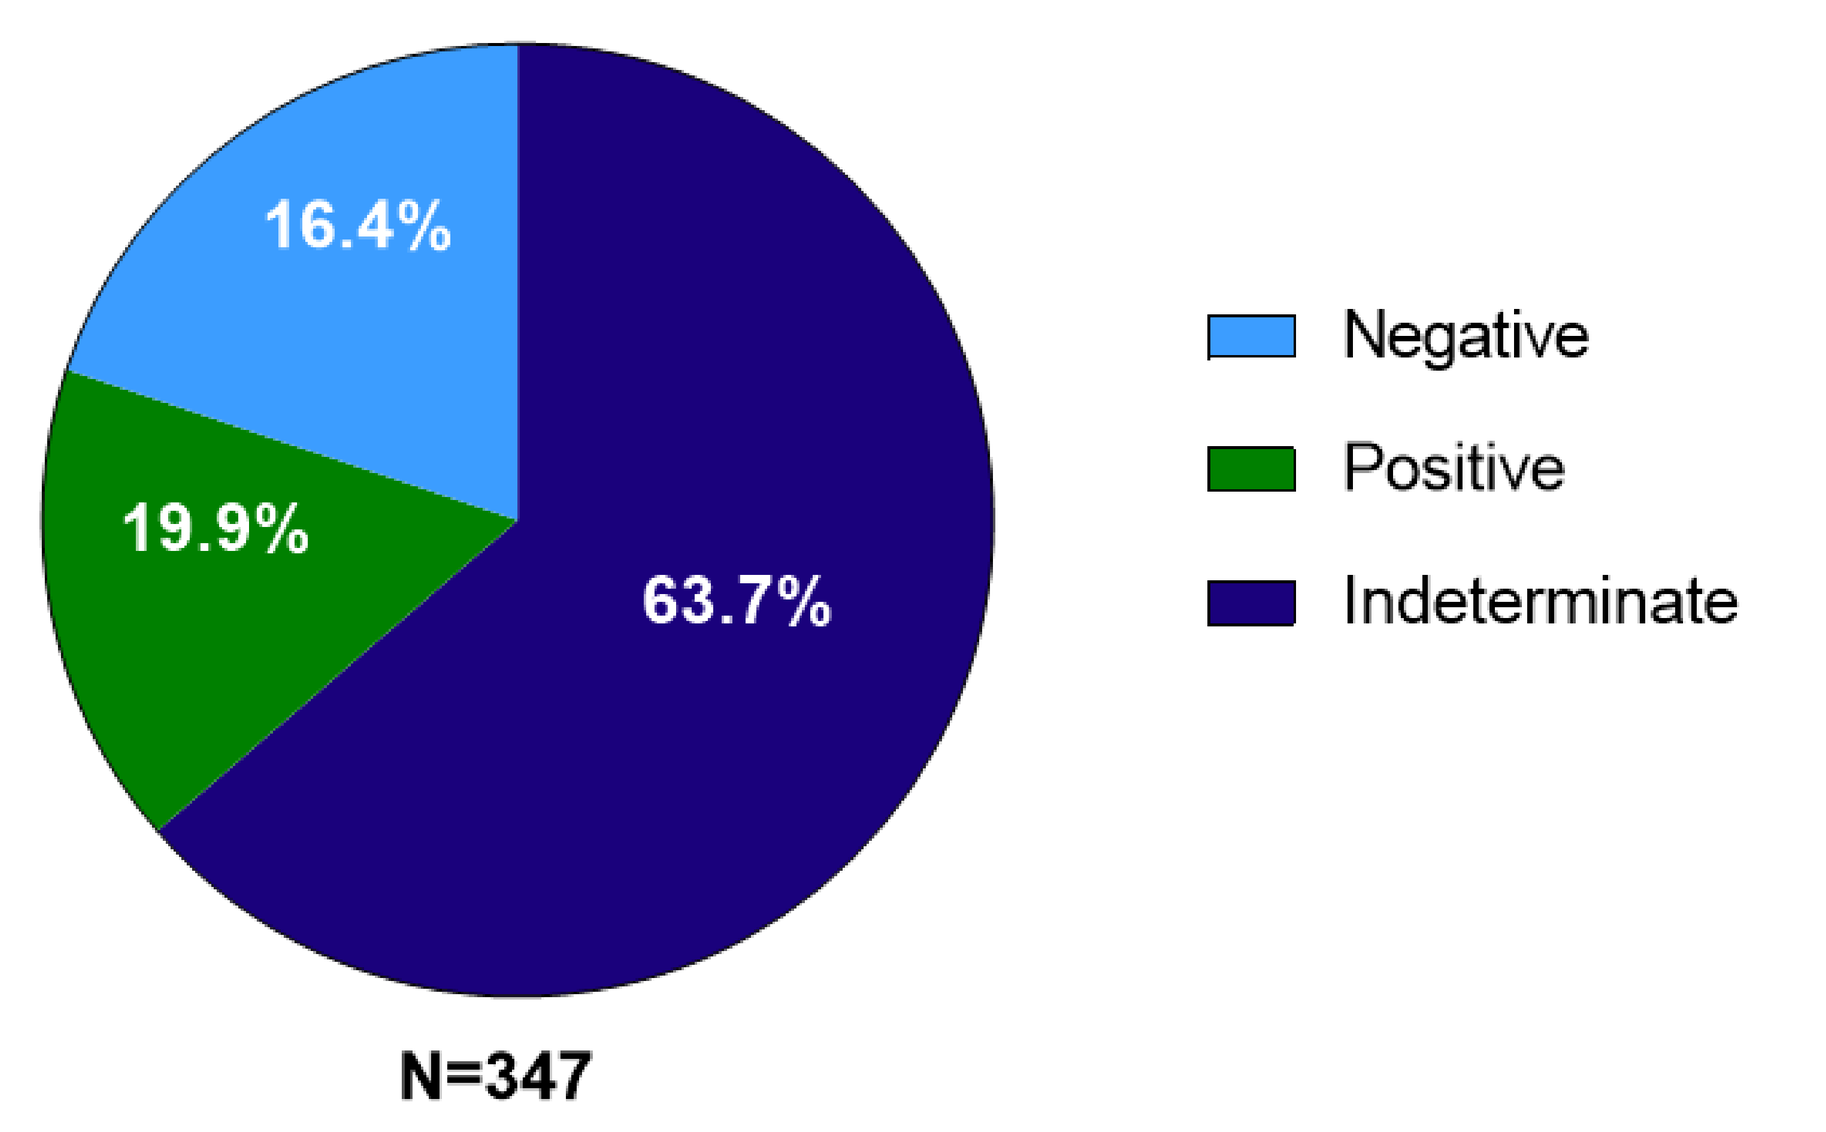
16.4%
19.9%
63.7%
Negative
Positive
Indeterminate
N=347

A positive diagnostic result indicated the presence of 1 pathogenic or likely pathogenic variant in an autosomal dominant gene or 2 pathogenic or likely pathogenic variants in an autosomal recessive gene. An indeterminate result indicated the presence of 1 variant of unknown significance in an autosomal dominant gene, 1 pathogenic or likely pathogenic variant found in an autosomal recessive gene, or 1 pathogenic or likely pathogenic variant and a variant of unknown significance in an autosomal recessive gene. Negative results indicated no presence of pathogenic, likely pathogenic, or variant of unknown significance.
NGS, next-generation sequencing.

- Of the 74 variants identified in 25 genes, 64 (86.5%) were single nucleotide variants (SNVs) and 10 (13.5%) were CNVs (**Figure 2**)
- **Crystallin genes**: 20 (27.0%) variants were identified in a crystallin gene (*CRYAA, CRYAB, CRYBA1, CRYBA2, CRYBA4, CRYBB1, CRYBB2, CRYGC, CRYGD,* and *CRYGS* were analyzed on this panel)
- **Collagen genes**: 5 (6.8%) variants were identified in a collagen gene (*COL2A1, COL18A1, COL4A1,* and *COL11A1* were analyzed on this panel)

Figure 2. NGS variant results

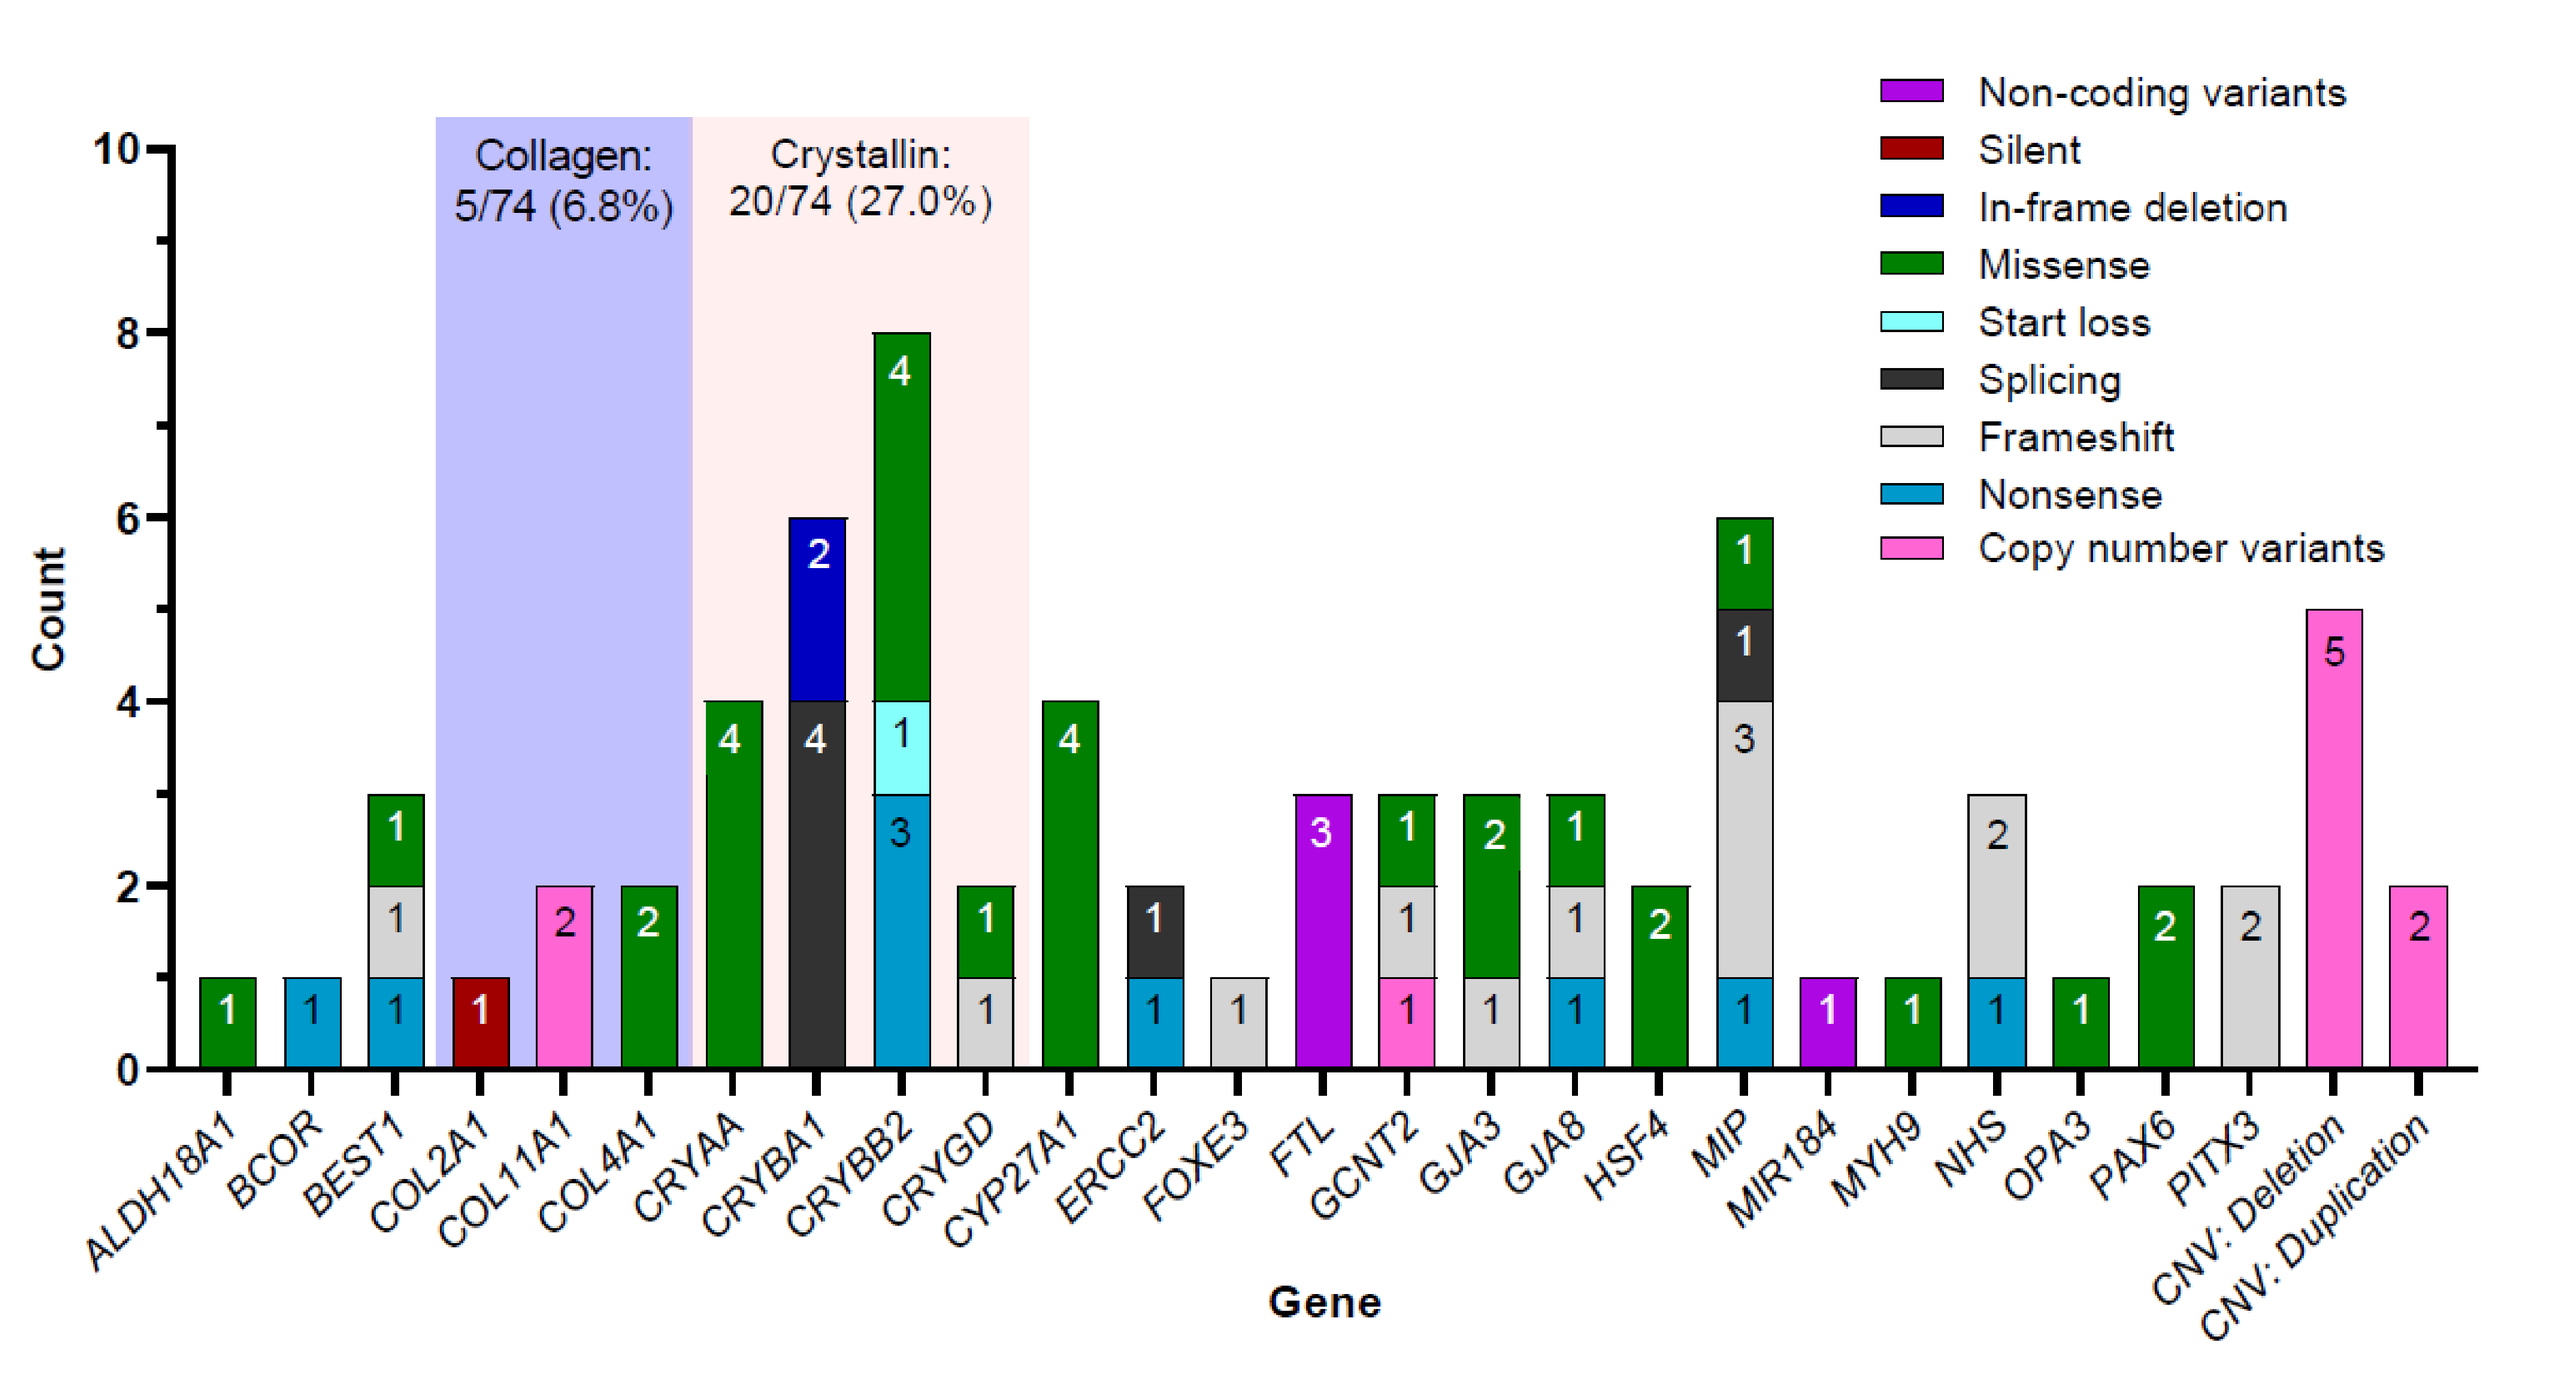
Collagen: 5/74 (6.8%)
Crystallin: 20/74 (27.0%)
Non-coding variants
Silent
In-frame deletion
Missense
Start loss
Splicing
Frameshift
Nonsense
Copy number variants
Count
Gene
ALDH18A1
BCOR
BEST1
COL2A1
COL11A1
COL4A1
CRYAA
CRYBA1
CRYBB2
CRYGD
CYP27A1
ERCC2
FOXE3
FTL
GCNT2
GJA3
GJA8
HSF4
MIP
MIR184
MYH9
NHS
OPA3
PAX6
PITX3
CNV: Deletion
CNV: Duplication

CNV, copy number variant; NGS, next-generation sequencing.

- 49 variants were identified in various other genes involved with other cellular structures and pathways (**Figure 3**)
 - 5 patients had a CNV overlapping with a genomic region involved with 1q21.1 microdeletion syndrome
 - 3 patients had a variant in ferritin light chain (*FTL*), a gene associated with iron storage and disruption that can lead to autosomal dominant hyperferritinemia-cataract syndrome (OMIM® #600886)
 - 1 patient had two pathogenic variants in *ERCC2*, a gene associated with helicase activity and important for nucleotide excision repair and disruption; these variants can lead to autosomal recessive (AR) xeroderma pigmentosum (OMIM® #126340)
- We identified 2 patients who were confirmed to have CTX after identifying known pathogenic missense mutations in *CYP27A1* (both patients had p.T339M.c.1016C>T and p.R127Q.c.380G>A)

Figure 3. Biological role of noncrystallin or collagen variants identified via NGS

Transcription Factors & Repressors
- *BCOR*, XL
- *ERCC2*, AR
- *HSF4*, AD
- *PAX6*, AD
- *PITX3*, AD, AR

Protein Channels and Junctions
- *BEST1*, AD, AR
- *GJA8*, AD
- *GJA3*, AD, AR

Mitochondrial Support
- *OPA3*, AD, AR

Nuclear Localization
- *NHS*, XL

Non-coding RNA
- *MIR184*, AD

Iron Storage
- *FTL*, AD

Glycosylation
- *MIP*, AR

AD, autosomal dominant; AR, autosomal recessive; NGS, next-generation sequencing; XL, X-linked.

Discussion

Crystallins

- Crystallins are a major protein component of the lens (~90%), responsible for maintaining the integrity and refractive index of the lens[2]
- Variants in these genes lead to an increase in protein instability that can result in protein aggregation and clouding of the normally transparent lens (**Figure 4**)
- Variants can lead to both autosomal dominant and autosomal recessive forms of cataracts[2]

Collagens

- Collagens are a major component of connective tissues and organization of the collagen fibril (**Figure 4**) and are largely responsible for the transparency and refraction of the cornea, lens, vitreous humor, and other parts of the eye[3]
- Variants that perturb the collagen fibril can lead to cataracts as well as other eye and non-eye manifestations[3]

Figure 4. Cataract viewed as a protein aggregation disease

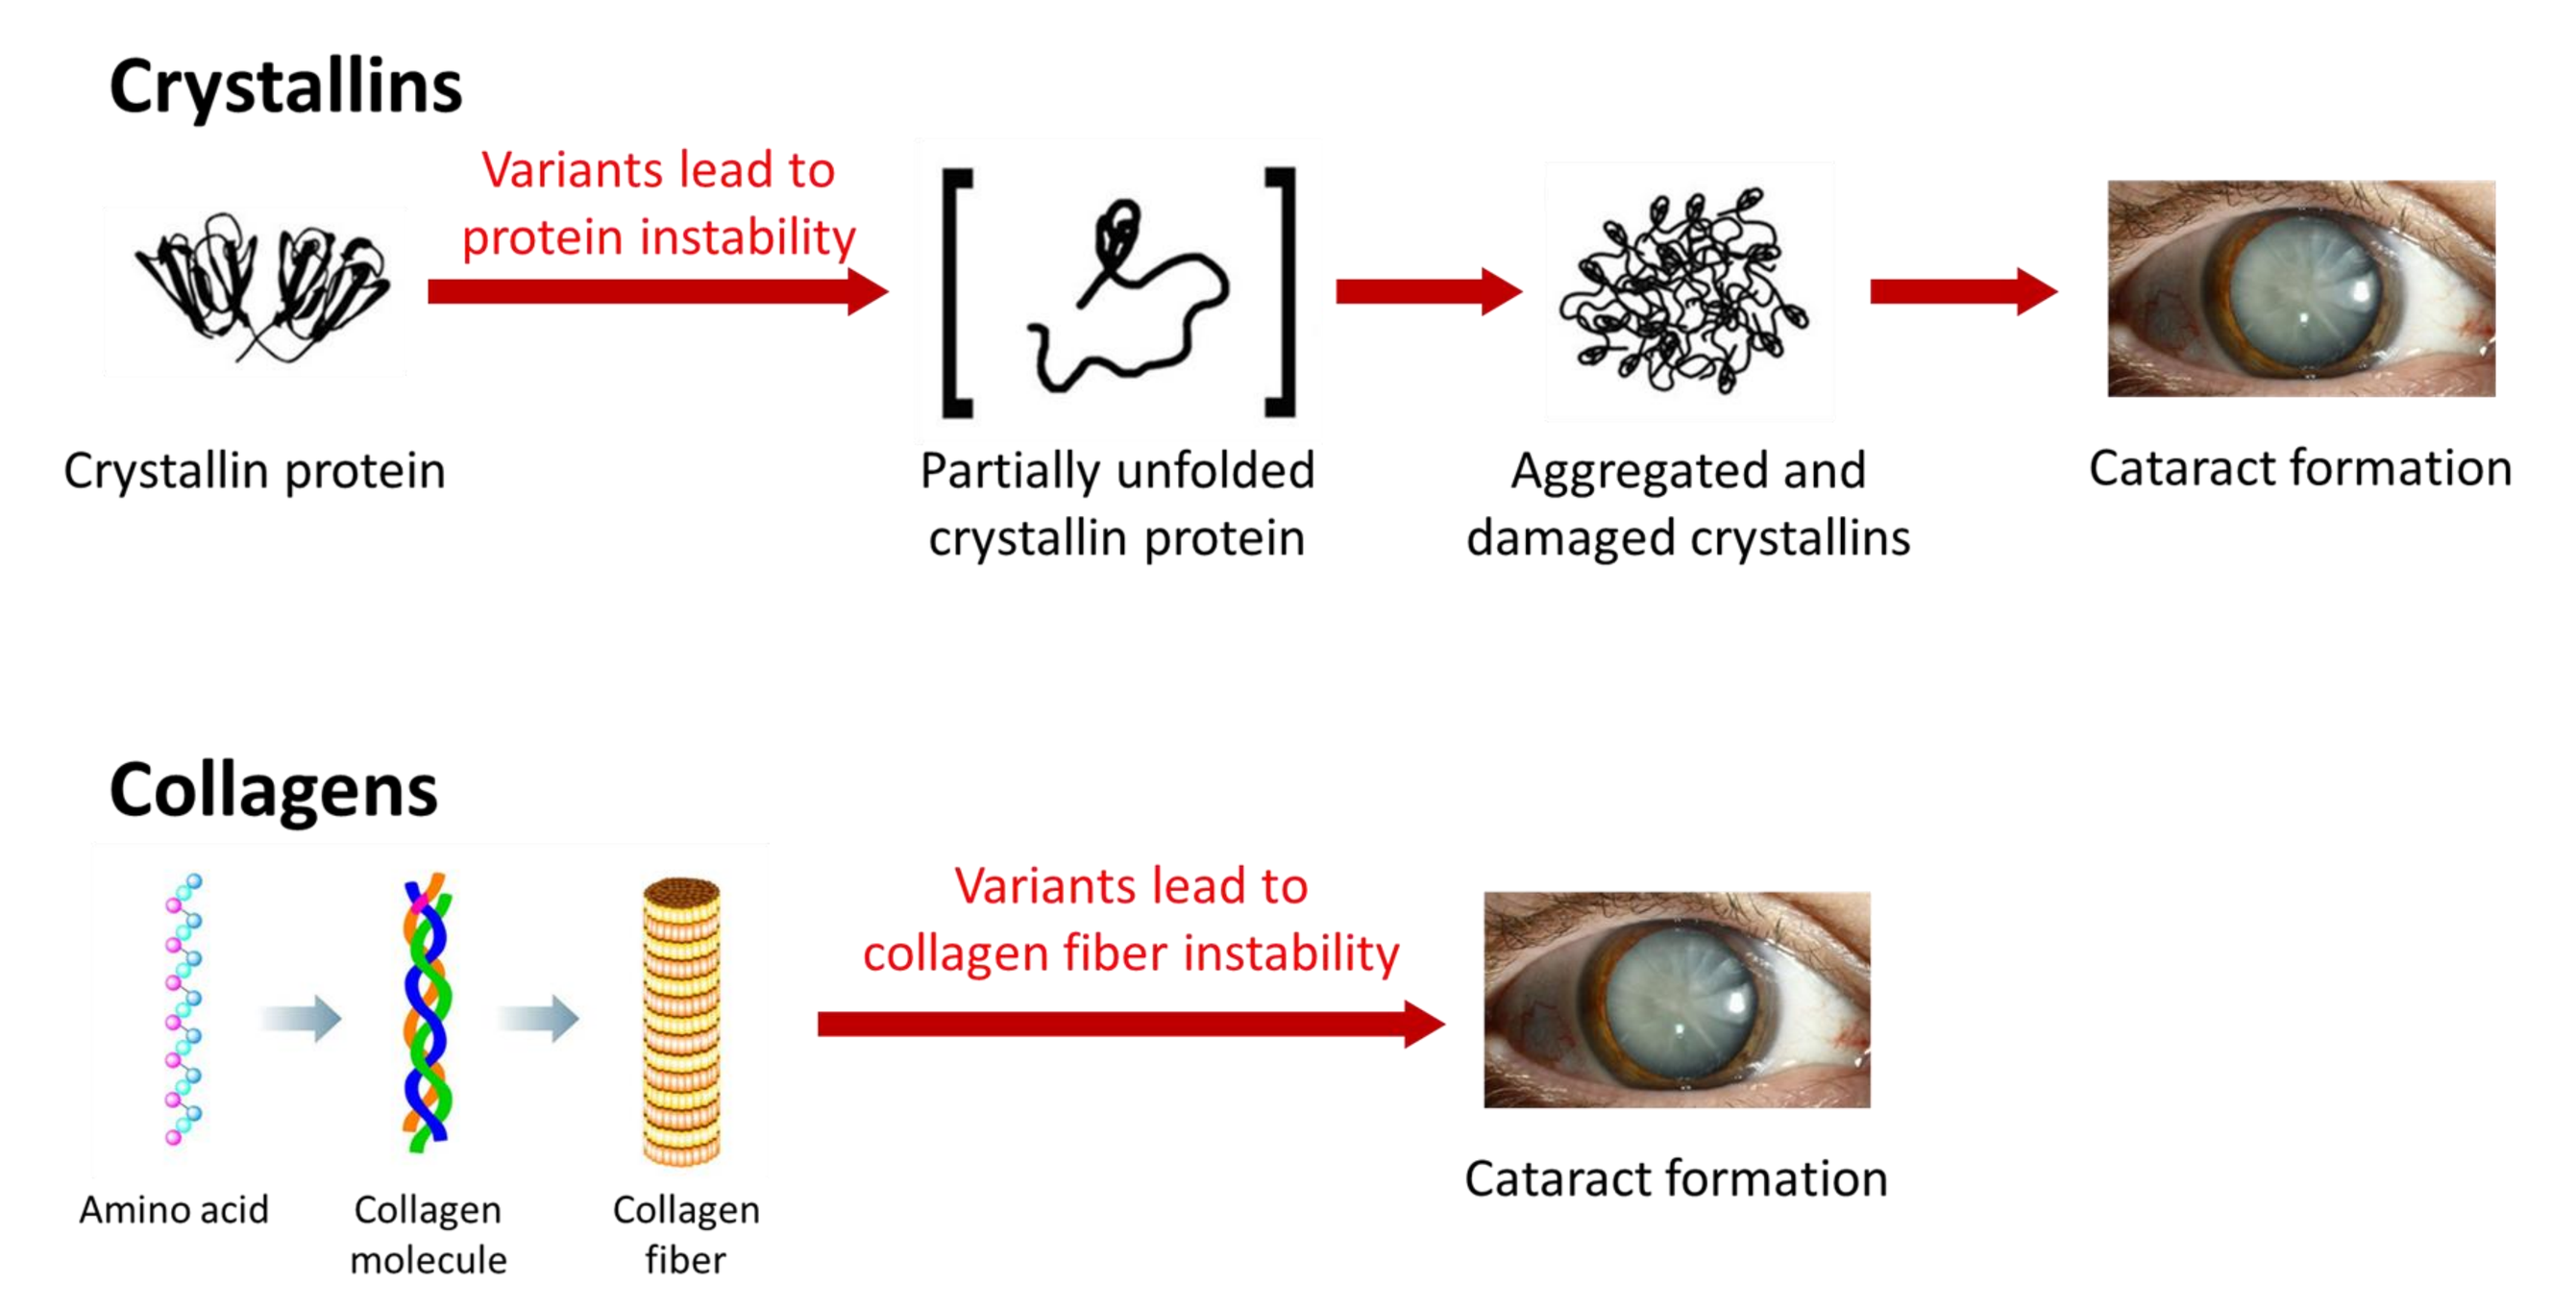
Crystallins
Variants lead to protein instability
Crystallin protein
Partially unfolded crystallin protein
Aggregated and damaged crystallins
Cataract formation
Collagens
Amino acid
Collagen molecule
Collagen fiber
Variants lead to collagen fiber instability
Cataract formation

CONCLUSIONS

- Through NGS sequencing, we observed a positive diagnostic rate of 19.9%
 - CNVs were responsible for 13.5% of variants identified
 - Crystallin gene variants were the most common (27.0%)
 - Variants were also detected in genes involved in other cellular processes, such as transcription, transport, and iron storage
- We identified 1 patient with CTX using the NGS panel and another patient, a family member of the former, through targeted family follow up genetic testing
- This study demonstrates the utility of genetic testing in individuals with early-onset idiopathic bilateral cataracts and can aid in early diagnosis and treatment initiation, and thus better outcomes

DISCLOSURES

EDS: Investigator for Travere Therapeutics, Inc.; honoraria from Travere Therapeutics, Inc; consultant for Sydnexis and Santen; grant support from National Eye Institute
JCF: Employee of PreventionGenetics LLC, a wholly owned subsidiary of Exact Sciences Corporation; stockholder of Exact Sciences Corporation
DMT: Employee of PreventionGenetics LLC, a wholly owned subsidiary of Exact Sciences Corporation; stockholder of Exact Sciences Corporation
AP: Employee and stockholder of Travere Therapeutics, Inc. during this work
RD: Employee and stockholder of Travere Therapeutics, Inc.
SFF: Investigator for Travere Therapeutics, Inc.; compensated for attendance to the CTX workshop; scientific advisor for Qlaris Bio, Inc.
BRN: Investigator for Travere Therapeutics, Inc.
DKV: Investigator for Travere Therapeutics, Inc.; grant support from National Eye Institute and Ocular Therapeutix; consultant/advisor and grant support from OPHTEC

ACKNOWLEDGMENTS

This study is funded by Travere Therapeutics, Inc. Writing and editorial support was provided by Eric Scocchera, PhD and Courtney Breuel, ELS, of MedVal Scientific Information Services, LLC (Princeton, NJ), and was funded by Travere Therapeutics, Inc.

REFERENCES

1. Foster A, et al. *J Cataract Refract Surg.* 1997;23(Suppl 1):601.
2. Moreau KL, et al. *Trends Mol Med.* 2012;18(5):273-282.
3. Shiels A, et al. *Exp Eye Res.* 2021;209:108662.

To obtain a PDF of this poster:

Scan the QR code.
Charges may apply.
No personal information is stored.

Contact information: Medinfo@Travere.com

Presented at the American Association for Pediatric Ophthalmology and Strabismus (AAPOS) 48th Annual Meeting, New York, NY, March 29-April 2