Comorbid Diagnoses, Medications, and Healthcare Provider Types among Patients Diagnosed with Classical Homocystinuria

Marcia Sellos-Moura[1], Frank Glavin[1], David Lapidus[2], Patrick T. Horn[1], Kristin A. Evans[3], Carolyn R. Lew[3], Debra E. Irwin[3]

[1]Orphan Technologies, Lexington, MA, United States; [2]LapidusData Inc., Oklahoma City, OK, United States; [3]IBM Watson Health™, Bethesda, MD, United States

orphan technologies

IBM **Watson Health**™

Background

- Classical homocystinuria (HCU) is a rare genetic disorder in which the body is unable to process the amino acid homocysteine (HCY). HCU is caused by mutations in the cystathionine beta synthase (CBS) gene.[1]
- HCU may be underdiagnosed at birth due to screening that is inadequately sensitive. Newborn screening typically tests for HCU indirectly by measuring the concentration of methionine (a precursor to HCY), rather than HCY itself. As a result, newborn screening often produces false negatives.[2]
- Recent research has estimated the prevalence of HCU in the U.S. to be 1 in 10,000 people,[3] which is substantially higher than prior estimates of 1 in 100,000-200,000 in the U.S. and 1 in 200,000-335,000 worldwide.[4-6] Several studies have estimated the birth prevalence of HCU to be much higher based on genetic modeling.[7,8]
- Symptoms of HCU can be mistaken for those of other disorders and vary from patient to patient. Adults not diagnosed with HCU during childhood are frequently misdiagnosed later in life with other conditions associated with abnormally high blood levels of total homocysteine (tHcy).
- Delayed diagnosis of HCU results in delayed therapy and the development of potentially preventable comorbid conditions. Better physician understanding and awareness of the diagnosis and management of HCU could improve early detection and treatment of classical HCU patients.

Objectives

- To compare the prevalence of select diagnoses and medications between patients with and without diagnosed HCU
- To describe the types of healthcare providers seen on the healthcare claims of patients diagnosed with HCU

Methods

Data Source

- Administrative claims in the IBM® MarketScan® Commercial and Medicare Supplemental Databases from January 1, 2010 through December 31, 2016.

Patient Selection

- Patients were selected from among all patients enrolled in the MarketScan Databases any time between 1/1/2010 and 12/31/2016
 - HCU patients
 - ≥2 non-diagnostic claims (90+ days apart) with ICD-10 diagnosis code E72.11 (homocystinuria) between 1/1/2010 and 12/31/2016 **OR**
 - ≥1 non-diagnostic claim with ICD-9 diagnosis code 270.4 (disturbances of sulphur-bearing amino acid metabolism) AND ≥1 non-diagnostic claim with ICD-10 code E72.11between 1/1/2010 and 12/31/2016
 - Controls
 - For each HCU patient, one age- and sex-matched control was randomly selected who had no claims with ICD-9 code 270.4, ICD-10 code E72.11, or Current Procedural Terminology (CPT) code 83090 for homocysteine testing between 1/1/2010 and 12/31/2016
 - Note that the HCU patient sample represents a population with prevalent disease; patients may have been initially diagnosed prior to the study time period.
 - Patients' entire available data history between 1/1/2010 and 12/31/2016 was included, though the ICD-10 diagnosis code for homocystinuria was not introduced until 10/1/2015.

Analysis

- The proportion of HCU patients with any claims showing selected diagnoses (shown in Figure 1) any time during their enrollment was compared to the proportion of controls with those diagnoses; relative risks and 95% confidence intervals were calculated for each diagnosis.
- The prevalence of selected therapeutic classes of medications (shown in Figure 2) was compared between HCU patients and controls using chi-squared tests.
- The proportion of HCU patients with different types of healthcare providers (shown in Figure 3) on their first claim with an HCU diagnosis in the study period was examined.

Results

Sample Characteristics

- A total of 2,450 HCU patients and 2,450 age- and sex-matched controls were included.
- The mean age of HCU patients and matched controls was 56.8 +14.6 years (median=57 years).
- 90% of HCU patients were 40+ years old at the time of their first HCU diagnosis
- The distribution of males (52%) and females (48%) was nearly equal.

Risk of Comorbid Diagnoses Associated with Diagnosed HCU

- HCU patients had a significantly higher risk, compared to matched controls, of experiencing several of the diagnoses examined any time during their enrollment in the databases (Figure 1). HCU patients were:
 - 5 times as likely to have a coagulation disorder (RR [95% CI]: 5.3 [4.0-7.1]) or lens dislocation (5.0 [1.1-22.8])
 - More than twice as likely to have a developmental/intellectual disability (RR [95% CI]:2.4 [1.3-4.9]) or vitamin D deficiency (RR [95% CI]:2.7 [2.4-3.1])
 - Twice as likely to be diagnosed with a non-stroke thrombosis (RR [95% CI]:2.0 [1.7-2.3])
 - 60%-80% more likely to experience hemiplegia/paraplegia, foot/knee/chest deformity, stroke/TIA, or retinal detachment

Therapeutic Medication Class Usage Associated with HCU

- HCU patients were significantly more likely than matched controls to have at least 1 prescription claim for several of the therapeutic medication classes examined any time during their enrollment in the database. HCU patients were:
 - Nearly 5 times as likely to have a prescription for folic acid or another B vitamin (27.2% vs. 5.9%; p<0.001) though less than 30% were prescribed, suggesting potential undertreatment
 - More than twice as likely to be prescribed an antihypertensive medication (59.8% vs. 22.0%; p<0.001)
 - 20%-40% more likely to have a prescription for a thyroid hormone (23.1% vs. 16.5%), vitamin D (17.1% vs. 12.0%), or a lipid-lowering medication (56.1% vs. 47.8%; all p<0.001).
- HCU patients were significantly less likely to have a prescription for a blood thinner or thrombolytic (28.4% vs. 46.8%; p<0.001) despite a significantly elevated risk of coagulation disorders

Figure 1. Relative Risk of Select Diagnoses: HCU Patients vs. Controls

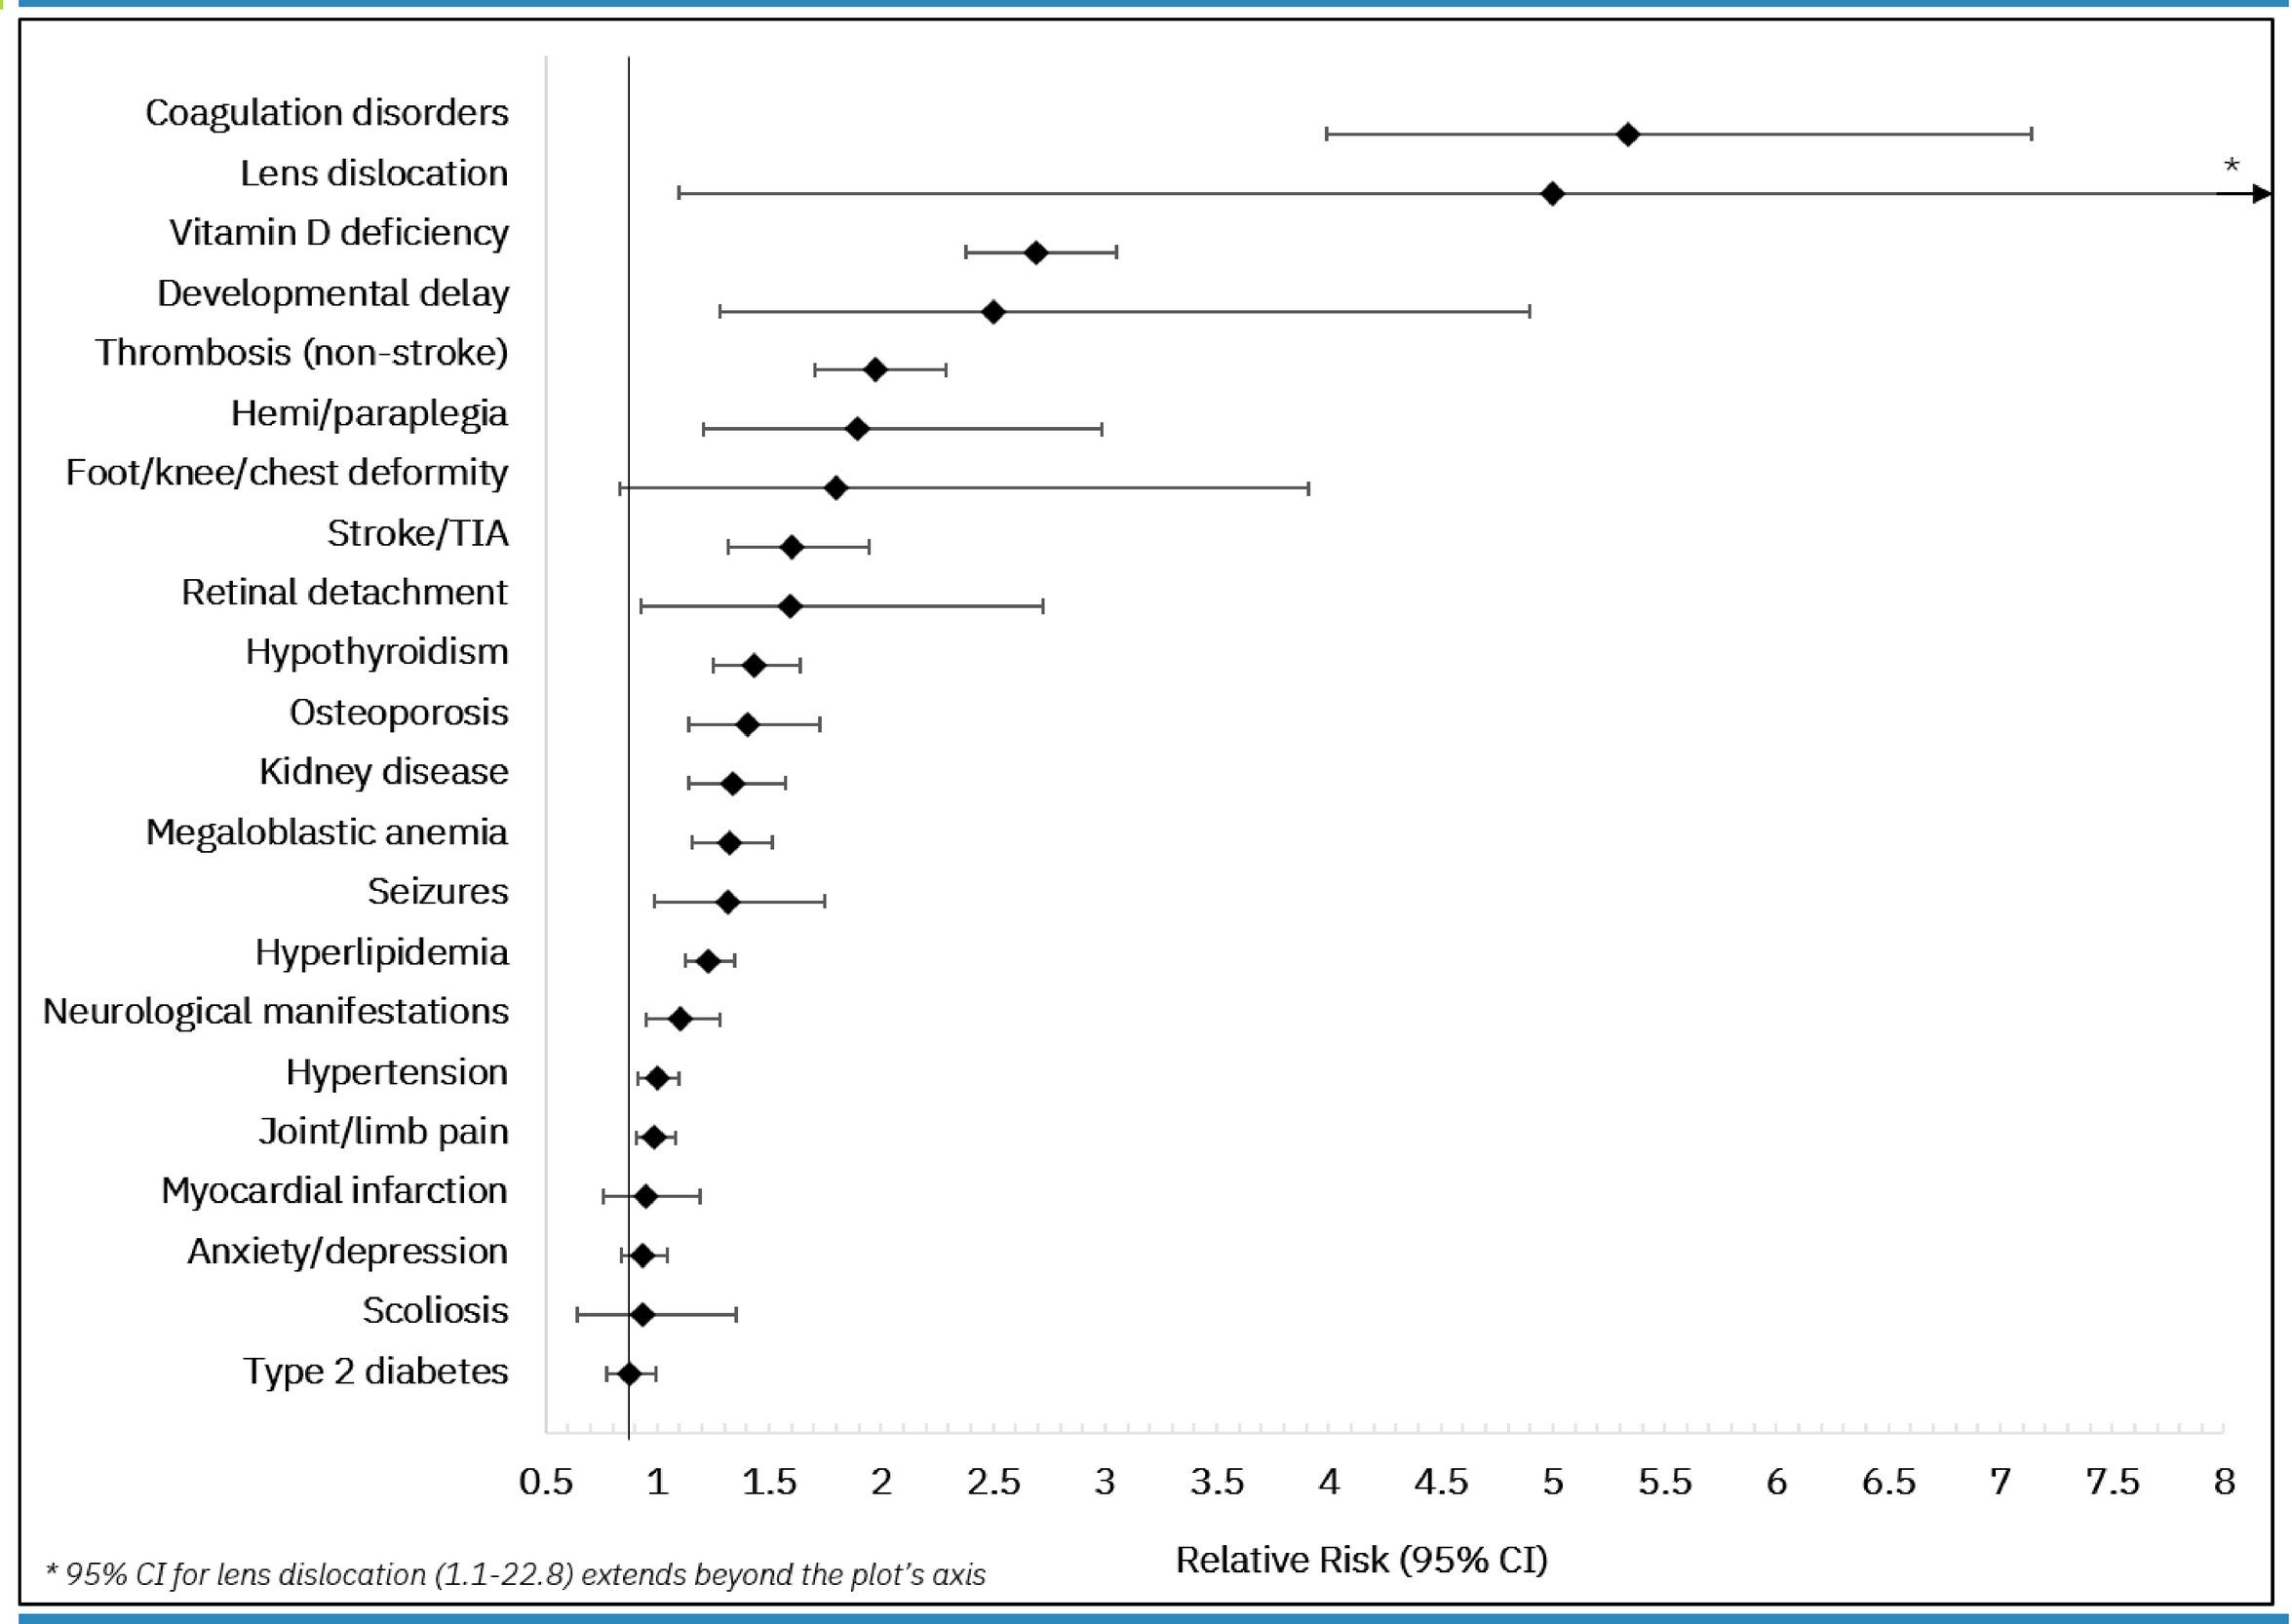
Coagulation disorders
Lens dislocation
Vitamin D deficiency
Developmental delay
Thrombosis (non-stroke)
Hemi/paraplegia
Foot/knee/chest deformity
Stroke/TIA
Retinal detachment
Hypothyroidism
Osteoporosis
Kidney disease
Megaloblastic anemia
Seizures
Hyperlipidemia
Neurological manifestations
Hypertension
Joint/limb pain
Myocardial infarction
Anxiety/depression
Scoliosis
Type 2 diabetes
*
0.5 1 1.5 2 2.5 3 3.5 4 4.5 5 5.5 6 6.5 7 7.5 8
Relative Risk (95% CI)

* 95% CI for lens dislocation (1.1-22.8) extends beyond the plot's axis

Figure 3. Prevalence of Therapeutic Medication Classes

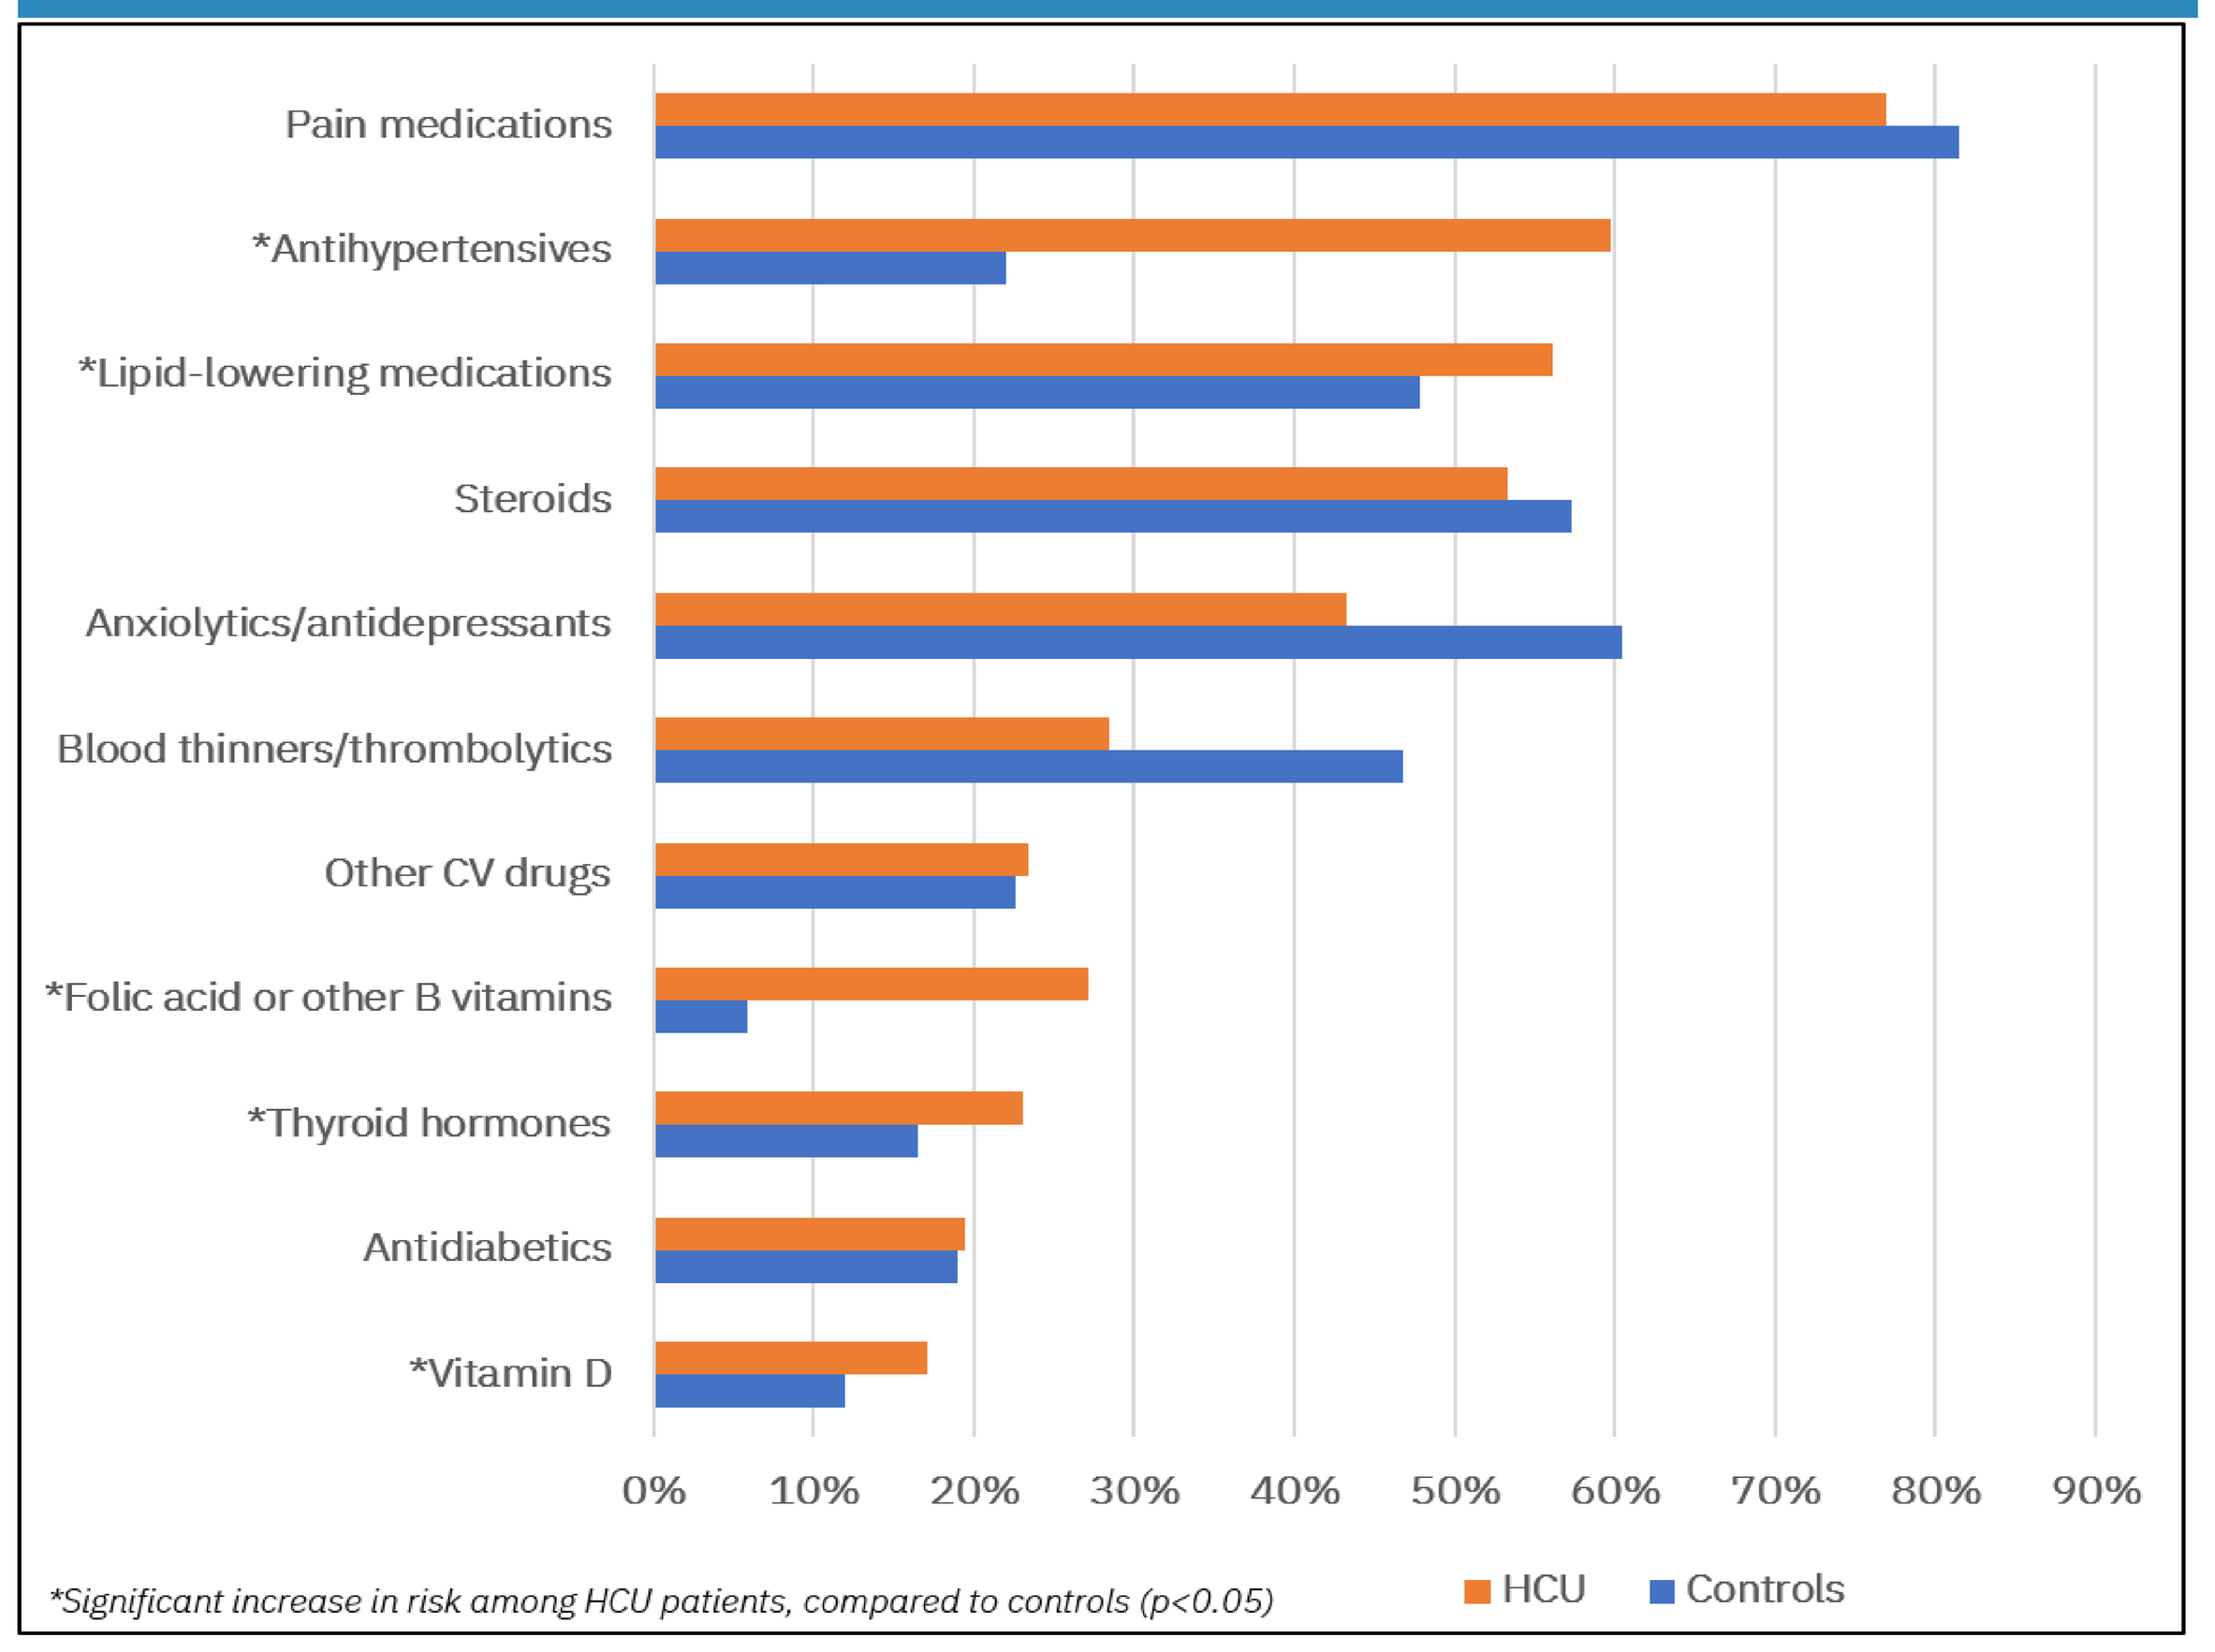
Pain medications
*Antihypertensives
*Lipid-lowering medications
Steroids
Anxiolytics/antidepressants
Blood thinners/thrombolytics
Other CV drugs
*Folic acid or other B vitamins
*Thyroid hormones
Antidiabetics
*Vitamin D
0% 10% 20% 30% 40% 50% 60% 70% 80% 90%
HCU
Controls

*Significant increase in risk among HCU patients, compared to controls (p<0.05)

Results

Common Healthcare Provider Types

- The most common healthcare provider types on patients' first claim (during the study period) with an HCU diagnosis were primary care providers:
 - Family medicine: 36.7%
 - Internal medicine: 21.4%
- Only 6 HCU patients (0.2%) had a geneticist on their first HCU claim
- 10.7% of initial HCU diagnoses in the study period were from claims billed by a healthcare facility, rather than a specific type of provider
- Neurologists were the most common specialists on patients' first HCU claims during the study period (6.7%).
- Patients may have seen other types of providers before or after the date of the first HCU diagnosis in the study period
- Patients with >1 claim with an HCU diagnosis code on the same day may be represented in >1 provider type category (Figure 3).

Figure 3. Prevalence of Healthcare Provider Types: First Observed HCU Claim

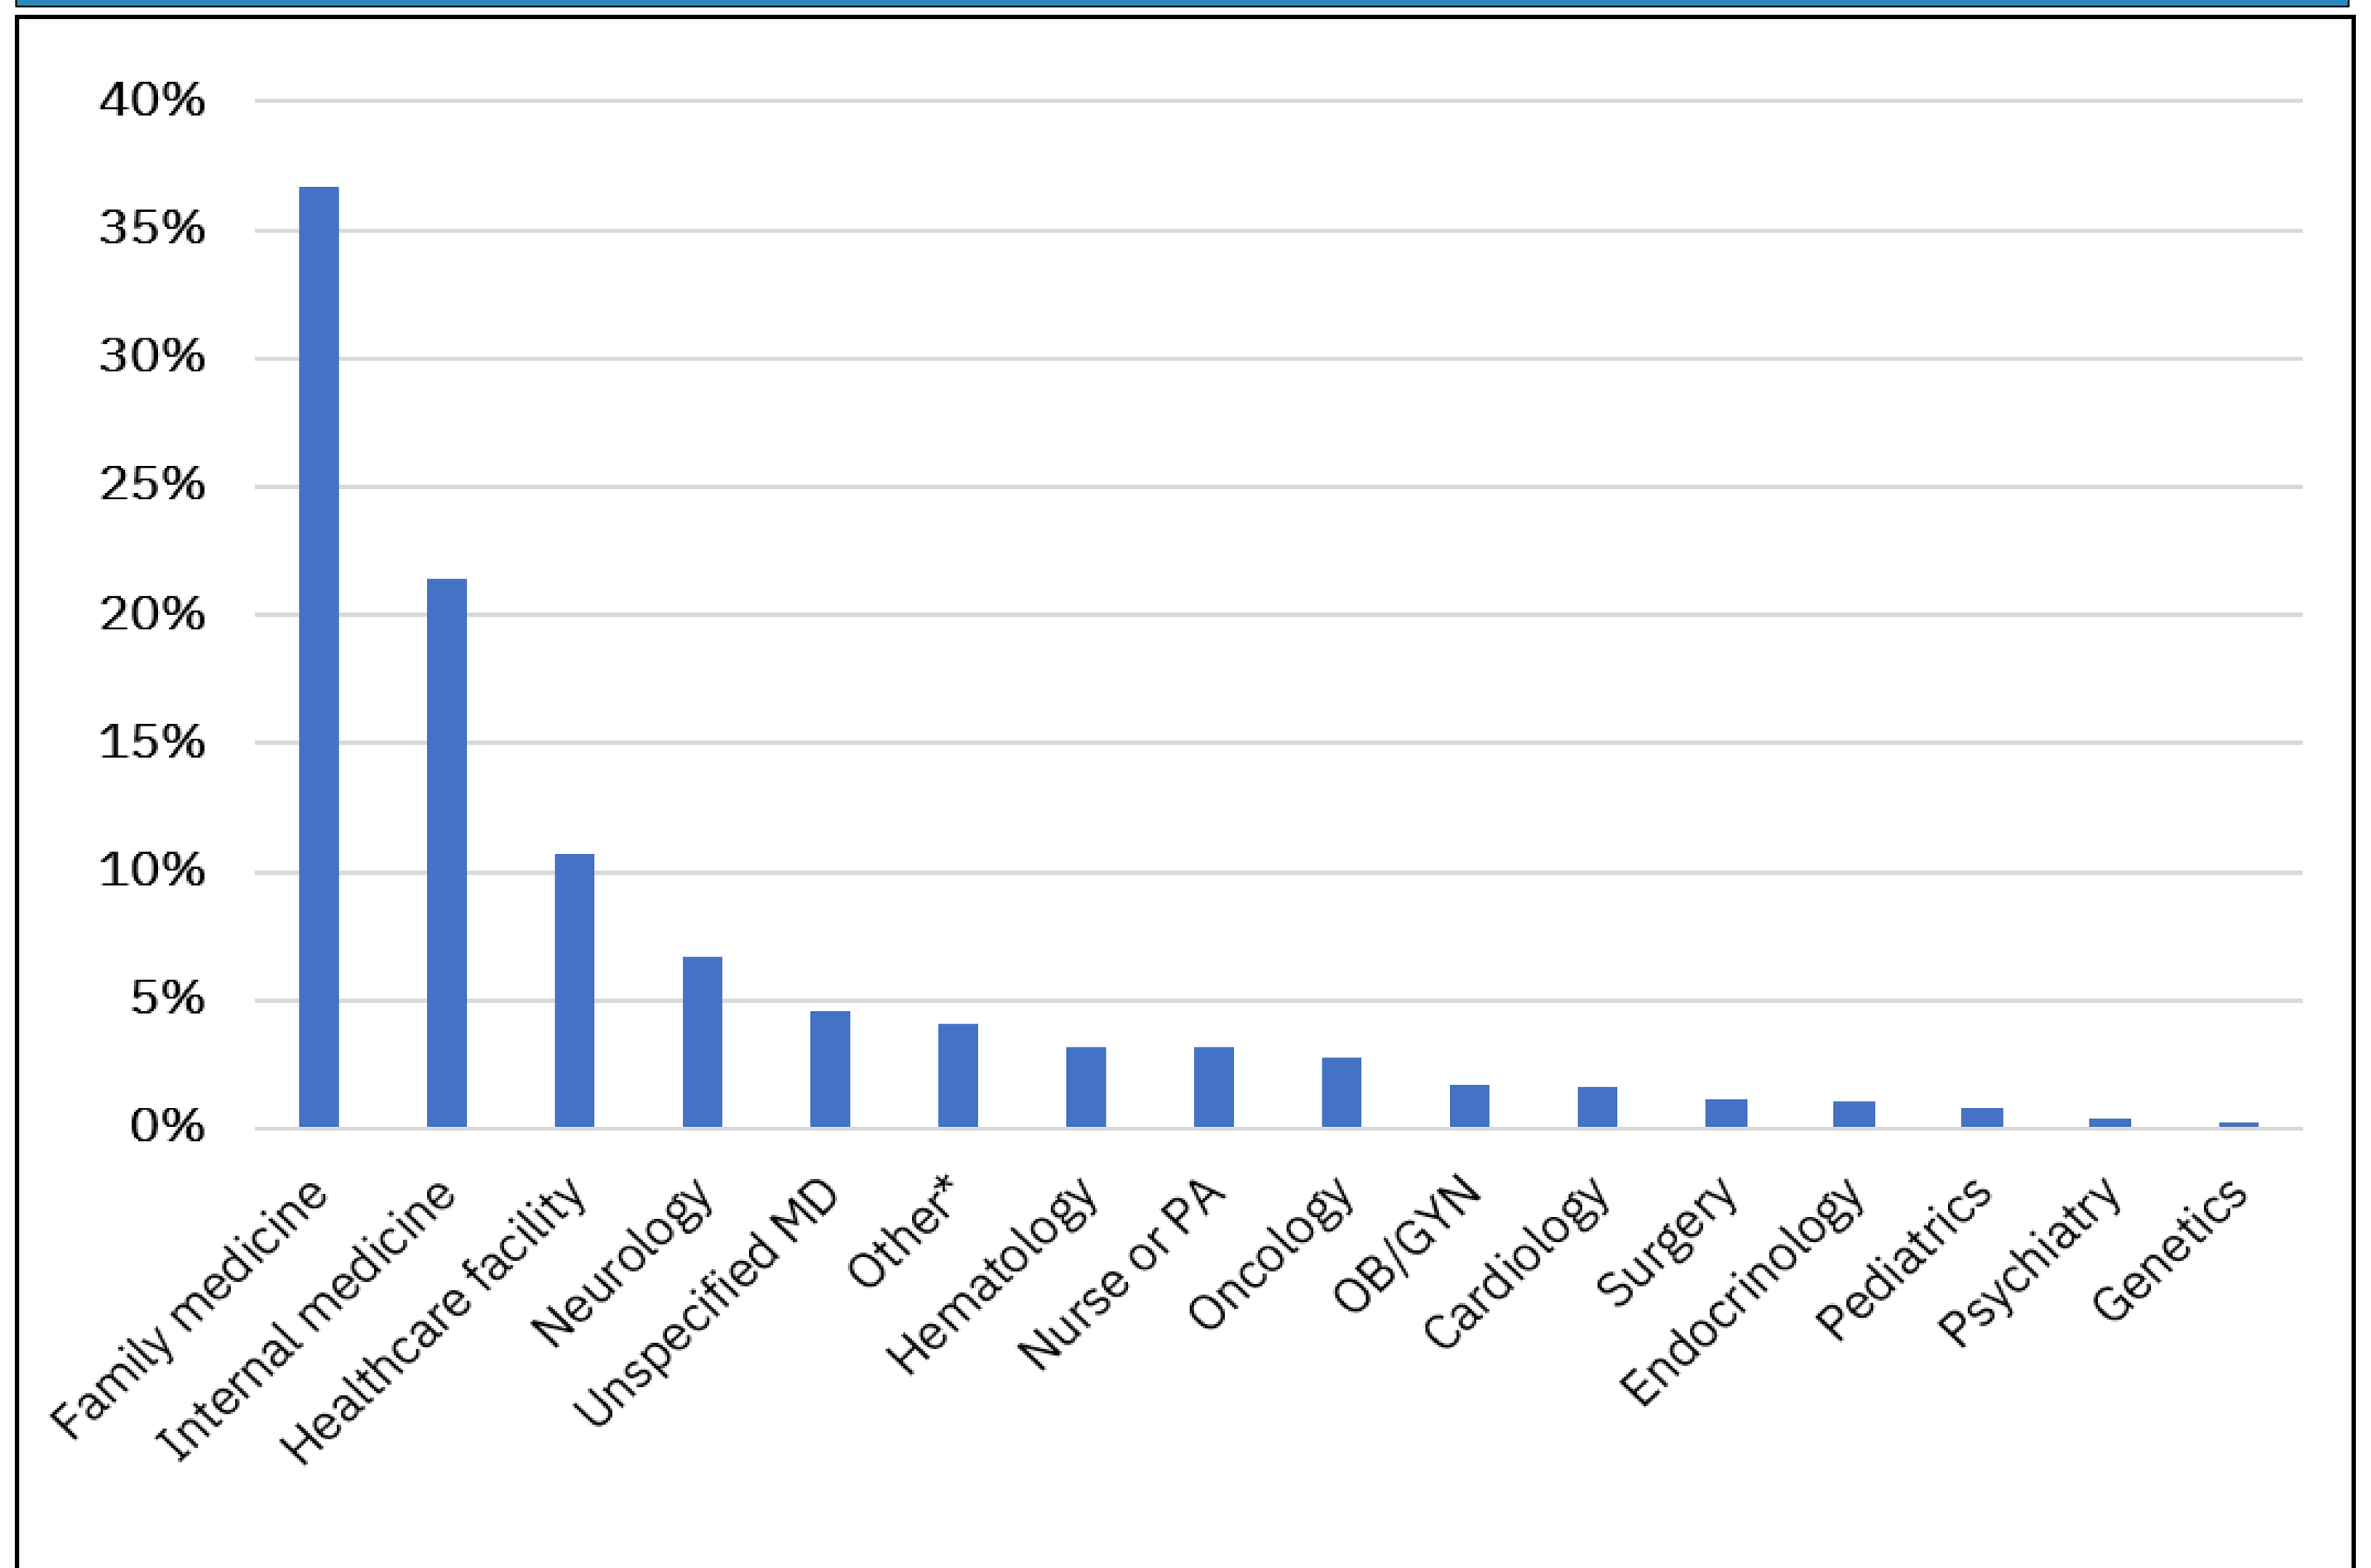
40%
35%
30%
25%
20%
15%
10%
5%
0%
Family medicine
Internal medicine
Healthcare facility
Neurology
Unspecified MD
Other*
Hematology
Nurse or PA
Oncology
OB/GYN
Cardiology
Surgery
Endocrinology
Pediatrics
Psychiatry
Genetics

** Other types of healthcare professionals (e.g., dieticians, chiropractors, physical therapists, psychologists)*

Conclusions

- These patients have a high risk for the known negative clinical outcomes associated with classical HCU, including coagulation disorders, lens dislocation, thrombosis, developmental delays, and vitamin D deficiency.
- These data suggest that the great majority of patients with an HCU diagnosis may be first diagnosed by primary care providers rather than by medical geneticists or specialists in HCU management.
- Ideally, patients with HCU should be diagnosed as early as possible to prevent these outcomes, yet this analysis suggests that many patients are not diagnosed until they are already experiencing sequela of elevated tHcy. Improvements to newborn screening and detection in the pediatric population may be warranted.
- When diagnosed, therapeutics prescribed appear to be focused on symptomatic treatment rather than on lowering tHcy levels.
- Enhanced education of physicians regarding the diagnosis and management of HCU may be necessary.

Limitations

- By relying on medical codes and pharmacy prescriptions in healthcare claims, study findings were subject to data coding limitations and data entry errors. A potential for misclassification of diagnoses or other study outcomes was therefore present.
- This analysis utilized data between January 1, 2010 and December 31, 2016. Any temporal differences that may exist were not captured in this study.

References

1. Sacharow SJ, et al. Homocystinuria caused by cystathionine beta-synthase deficiency. *NCBI Gene Reviews.* Retrieved from https://www.ncbi.nlm.nih.gov/books/NBK1524.
2. Walter JH, et al. *Cochrane Database Syst Rev* 2015;10. Art. No.: CD008840.
3. Sellos-Moura M, et al. Comparison of the estimated prevalence of diagnosed homocystinuria and phenylketonuria in the United States. Poster session presented at: ISPOR Europe; 2018 Nov 10-14; Barcelona, Spain.
4. Homocystinuria. U.S. National Library of Medicine. Retrieved from https://ghr.nlm.nih.gov/condition/homocystinuria#sourcesforpage.
5. Mandava P. (updated June 14, 2017). Homocystinuria/Homocysteinemia. Retrieved from https://emedicine.medscape.com/article/1952251-overview#showall.
6. Mudd SH, et al. *Am J Hum Genet* 1985; 37(1): 1-31.
7. Janosik M, et al. *J. Pediatr* 2009; 154:431-437.
8. Refsum H, et al. *J. Pediatr* 2004; 144:830-832.

Presented at ACMG Annual Clinical Genetics Meeting, April 2-6, 2019, Seattle, Washington