Poster# P202

Sparsentan Improves Glomerular Blood Flow and Augments Protective Tissue Remodeling in Mouse Models of Focal Segmental Glomerulosclerosis (FSGS)

Georgina Gyarmati[1], Urvi Shroff[1], Audrey Izuhara[1], Radko Komers[2], Patricia W. Bedard[2], Janos Peti-Peterdi[1]

[1]Departments of Physiology and Neuroscience, and Medicine, Zilkha Neurogenetic Institute, University of Southern California, Los Angeles, CA, USA; [2]Travere Therapeutics, San Diego, CA, USA

RESULTS

Healthy-Tg Mice: Basal Renal Hemodynamic Responses

- Sparsentan treatment significantly increased both AA and EA diameters and SNGFR vs. control after 6 weeks (**Figure 1**)
- ET-1+Ang II-induced AA vasoconstriction and vascular smooth muscle cell (VSMC) Ca^{2+} elevation was almost completely blocked in sparsentan-treated mice; losartan-treated mice had only Ca^{2+} restoration (**Figure 2**)
 - Sparsentan-treated mice had non-significant improvements in glomerular diameter and tuft area
- Sparsentan-treatment almost completely restored ET-1-induced elevations in AA vasoconstriction, VSMC Ca^{2+}, and glomerular tuft area reduction (**Figure 2**)
 - Losartan had no effect on any of the ET-1-induced glomerular hemodynamic alterations

Figure 1. Healthy-Tg mouse kidney: basal physiologic response to 6 weeks of daily sparsentan or losartan treatment

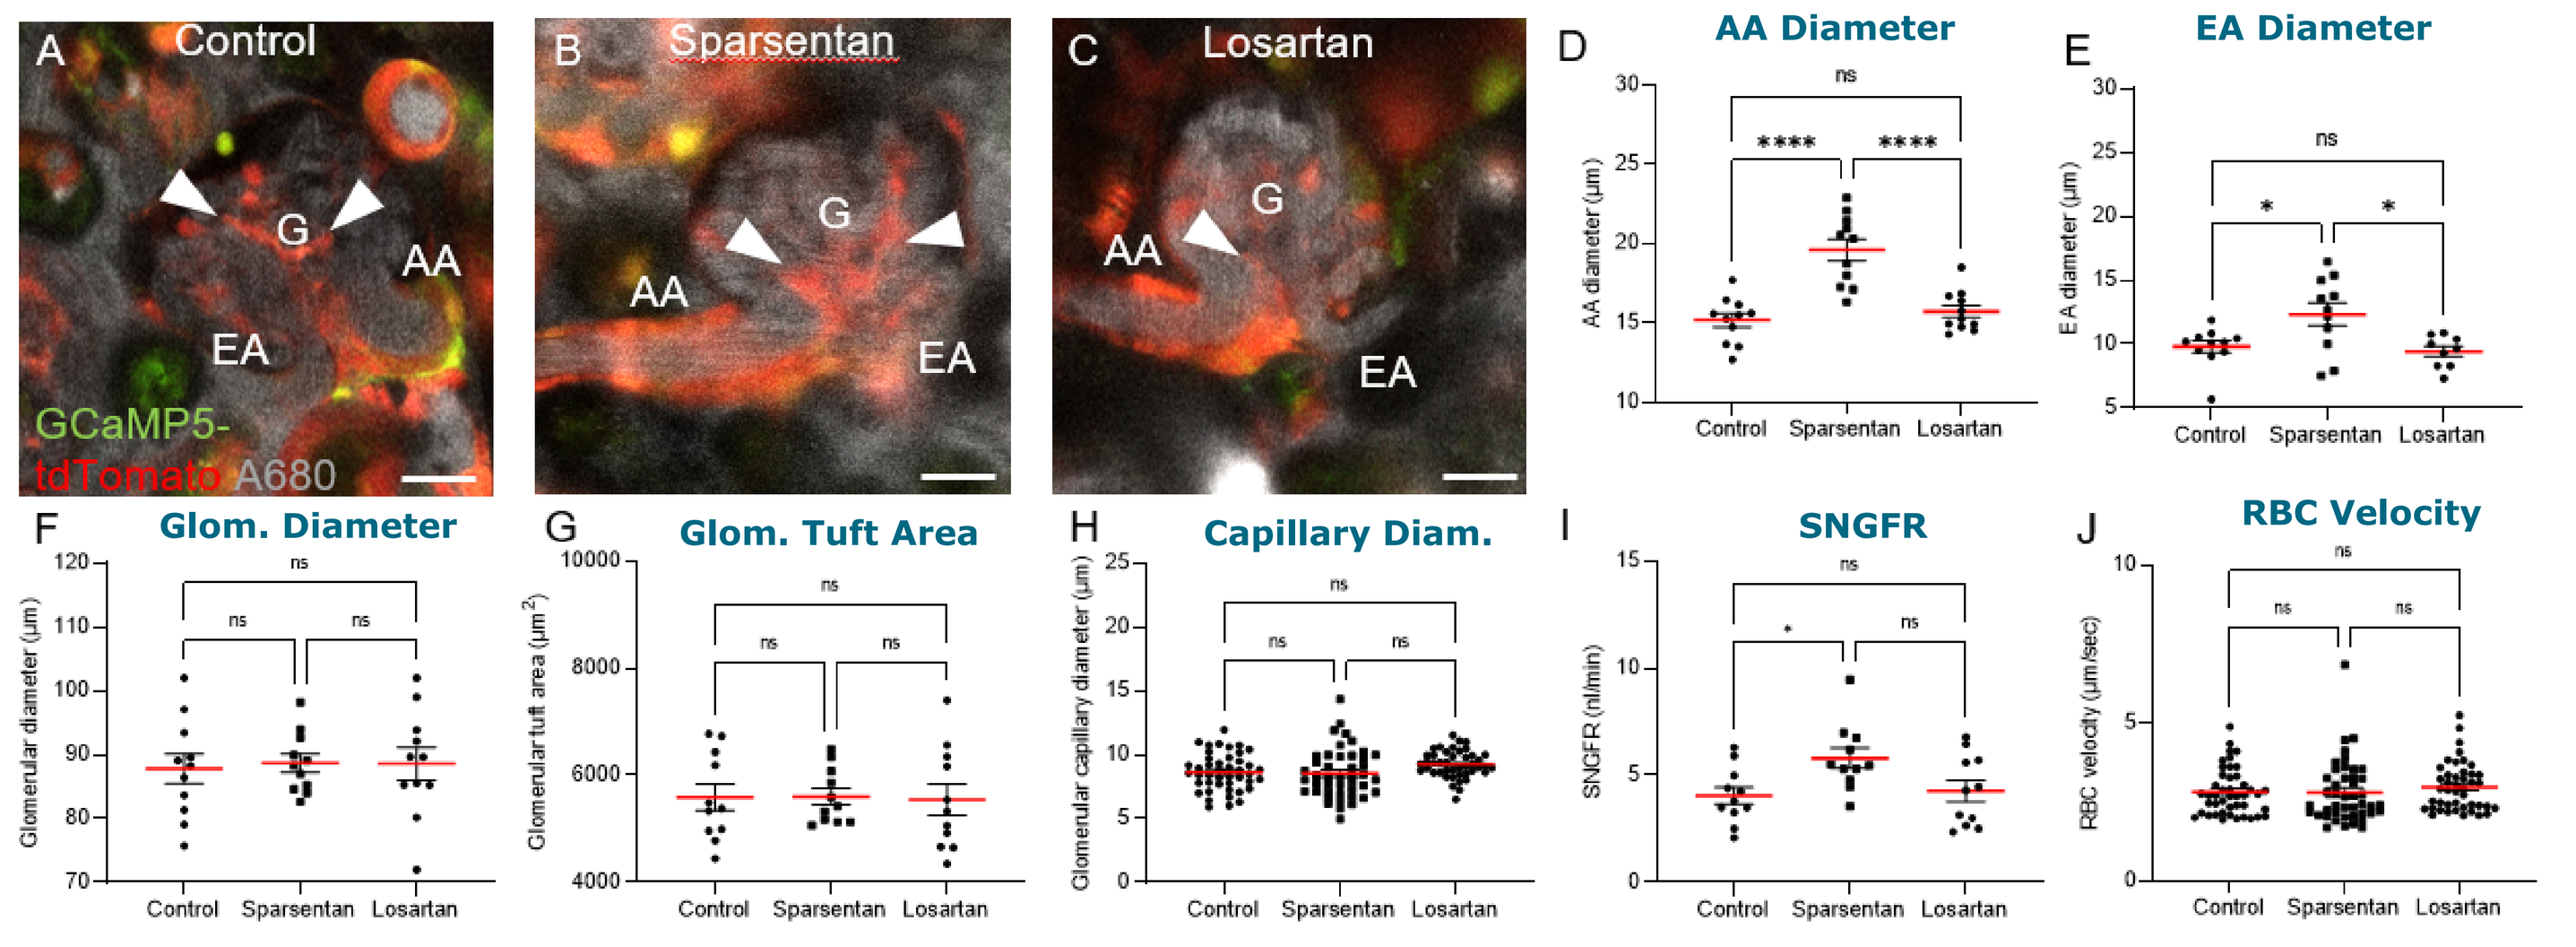
A Control
B Sparsentan
C Losartan
GCaMP5-tdTomato A680
D AA Diameter
E EA Diameter
F Glom. Diameter
G Glom. Tuft Area
H Capillary Diam.
I SNGFR
J RBC Velocity
Control Sparsentan Losartan

(A-C) MPM images of Healthy-Tg glomeruli (G) from treated mice. Circulating plasma was labeled by IV injected albumin-Alexa Fluor 680 (grey). The Ca^{2+}-sensitive reporter GCaMP5 (green) and Ca^{2+}-insensitive tdTomato (red) highlight cells of the renin lineage including AA and EA vascular smooth muscle cells (VSMCs), and extra- and intra-glomerular mesangial cells (arrowheads). (D-J) measured hemodynamic parameters.

Figure 2. Healthy-Tg mouse kidney: hemodynamic response to acute agonist-induced vasoconstriction after 6 weeks of daily sparsentan or losartan treatment

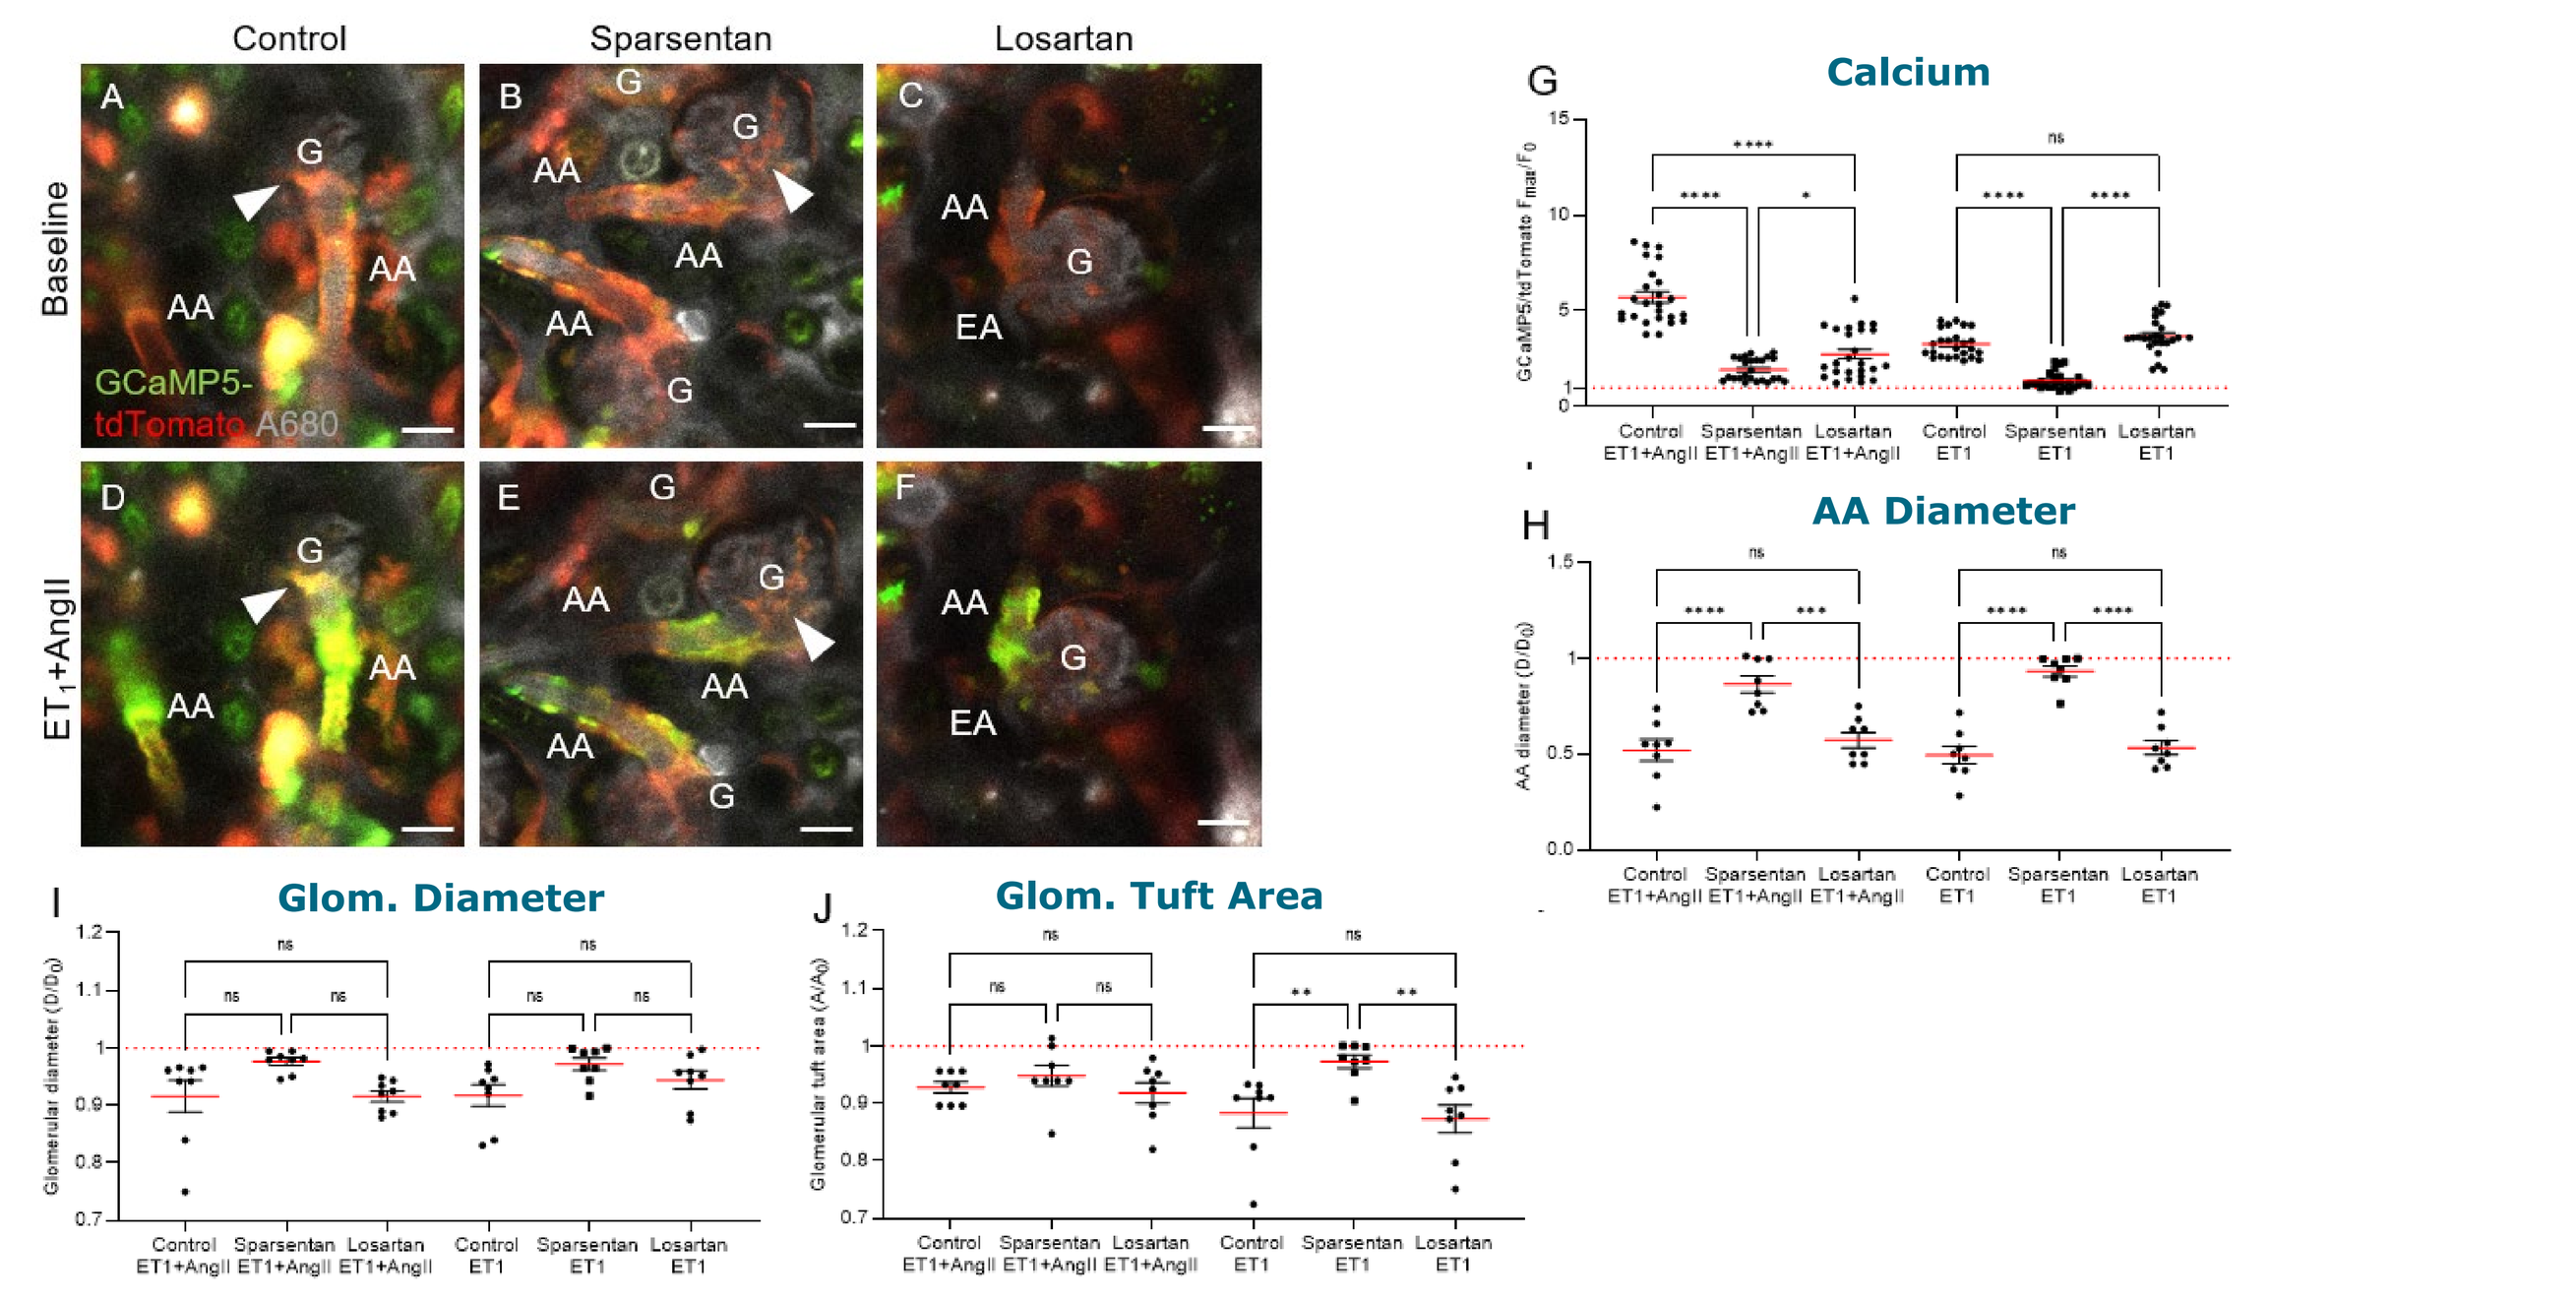
Control
Sparsentan
Losartan
Baseline
ET_1+AngII
GCaMP5-tdTomato A680
G Calcium
H AA Diameter
I Glom. Diameter
J Glom. Tuft Area

MPM images of Healthy-Tg glomeruli (G) before (A-C) and after (D-F) the bolus agonist injection. Circulating plasma was labeled by albumin-Alexa Fluor 680 (grey). GCaMP5 (green) and tdTomato (red) highlight cells of the renin lineage including AA and EA VSMCs, and extra- and intra-glomerular mesangial cells (arrowheads). (G-J) Measured hemodynamic parameters normalized to baseline (ratio of maximum effect/before injection).

FSGS-Tg Mice

- Six-week sparsentan treatment significantly improved podocyte Ca^{2+}, AA/EA diameter, GSC, SNGFR, and ACR vs. control while losartan improved podocyte Ca^{2+} and blood flow (**Figure 3**)
 - Sparsentan-treated mice had non-significant improvements in glomerular diameter and tuft area
- ET-1+Ang II-induced AA vasoconstriction, increased Ca^{2+}, decreased glomerular diameter and tuft area was blocked in sparsentan-treated mice while losartan improved Ca^{2+} (**Figure 4**)
- Sparsentan treatment significantly improved podocyte number, glomerular sclerosis and tissue fibrosis indexes vs. control, and more than losartan (**Figure 5**)

Figure 3. FSGS-Tg mouse chronic disease kidney: basal physiologic response to 6 weeks of daily sparsentan or losartan treatment

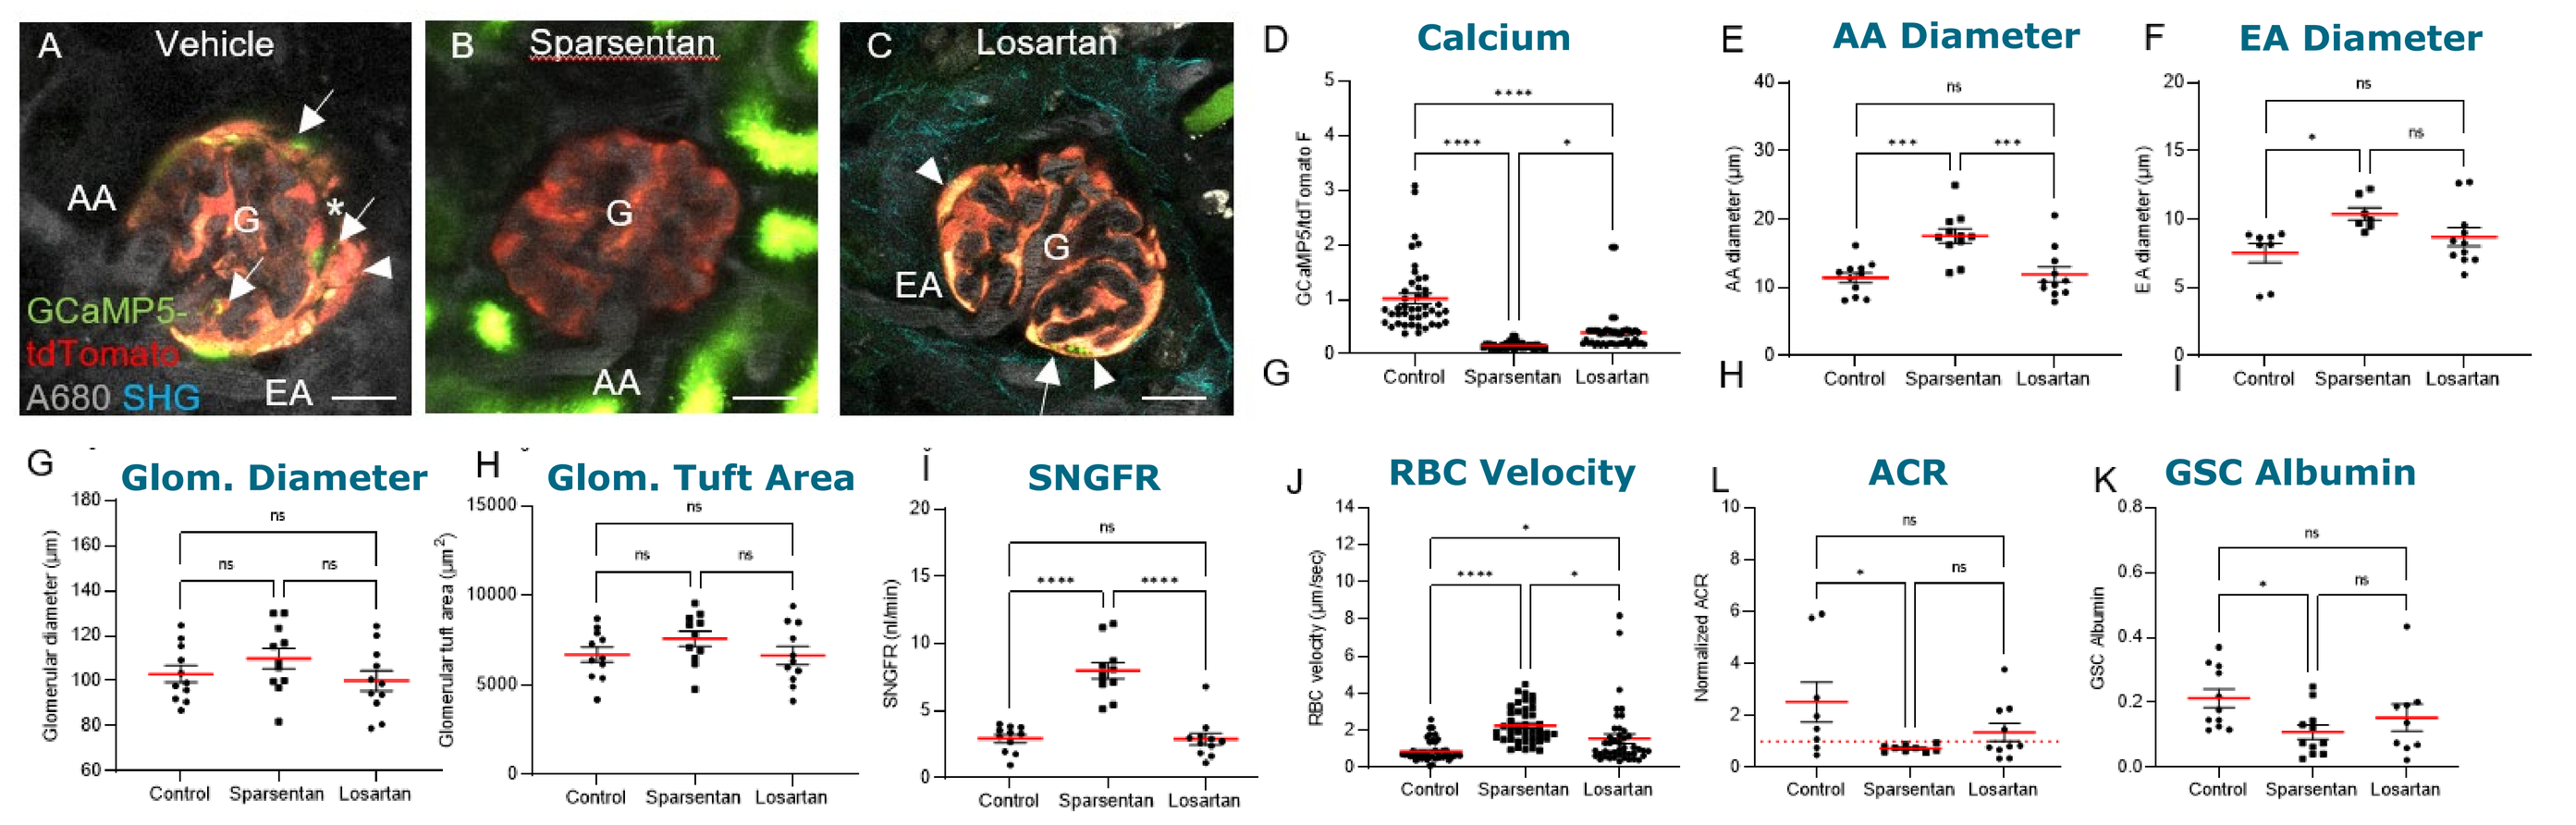
A Vehicle
B Sparsentan
C Losartan
GCaMP5-tdTomato A680 SHG
D Calcium
E AA Diameter
F EA Diameter
G Glom. Diameter
H Glom. Tuft Area
I SNGFR
J RBC Velocity
L ACR
K GSC Albumin

MPM images (A-C) of FSGS-TG mice glomeruli (G). Color Key: circulating plasma (albumin-Alexa Fluor 680, grey); fibrillar collagen (fibrosis by second harmonic generation, SHG, cyan); GCaMP5 (green) and tdTomato (red) highlight podocytes. Podocytes with high Ca^{2+} (intense green) are visible in a segmental pattern (arrows) in direct contact (adhesion) with parietal podocytes (arrowheads). GFB plasma albumin leakage is visible in the Bowman's space (asterisk). (D-K) Measured hemodynamic and GFB parameters including podocyte Ca^{2+} (based on the ratio of GCaMP5/tdTomato fluorescence intensity (D), ACR normalized to baseline (L), and glomerular sieving coefficient of serum albumin (K).

Figure 4. FSGS-Tg mouse chronic disease kidney hemodynamic response to acute agonist-induced vasoconstriction after 6 weeks of daily sparsentan or losartan treatment

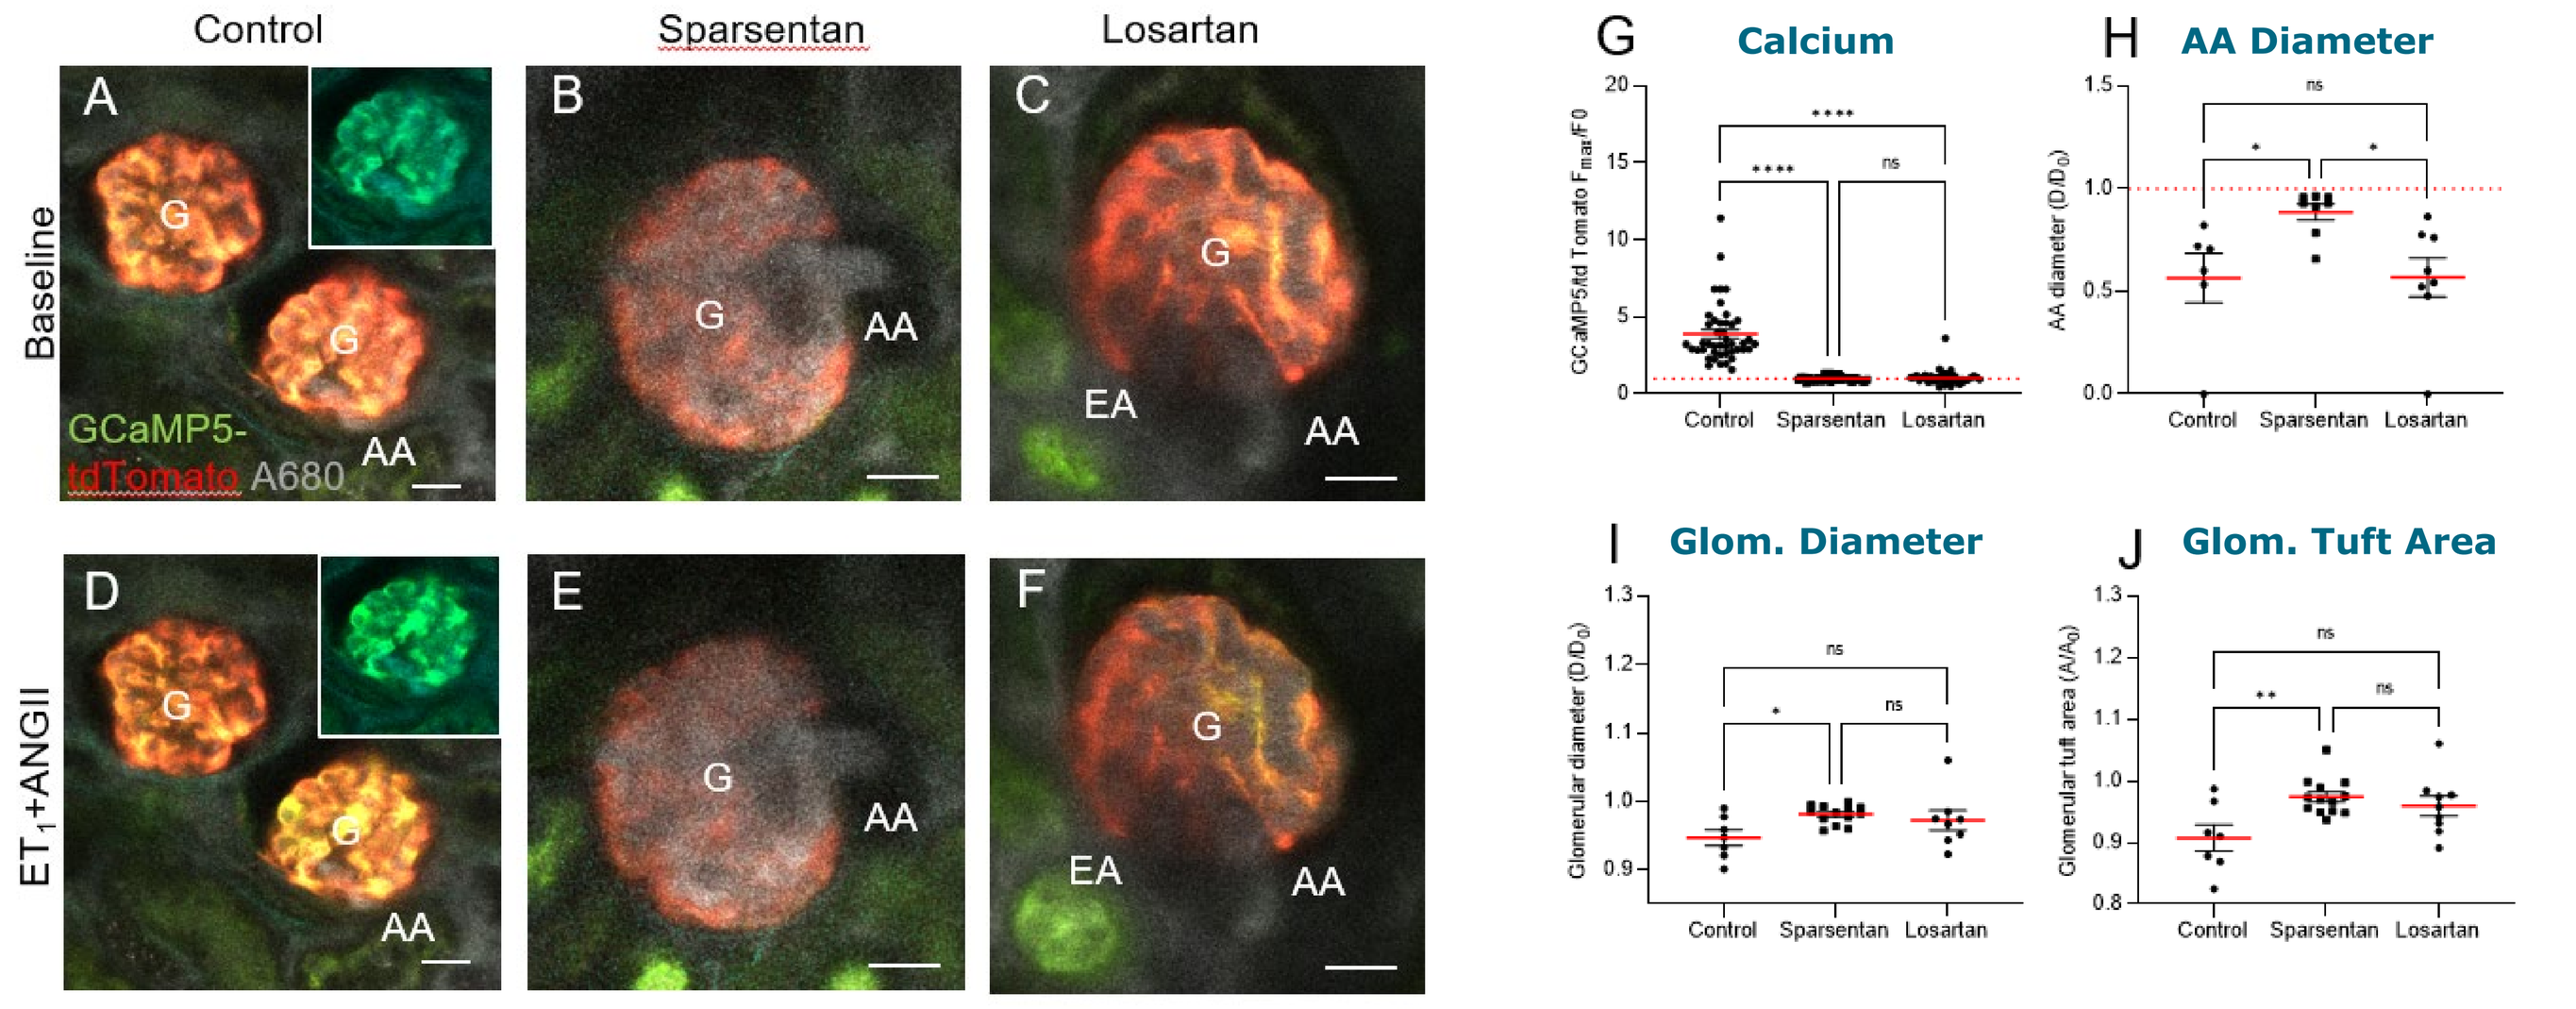
Control
Sparsentan
Losartan
Baseline
ET_1+ANGII
GCaMP5-tdTomato A680
G Calcium
H AA Diameter
I Glom. Diameter
J Glom. Tuft Area

MPM images of FSGS-TG glomeruli (G) before (A-C) and after (D-F) the bolus injection of agonists. The circulating plasma was labeled with albumin-Alexa Fluor 680 (grey), illuminating the AA and EA. GCaMP5 (green) and tdTomato (red) in podocytes. Insets show the GCaMP5 (green) channel separately to better visualize podocyte Ca^{2+} changes. (G-J) Measured hemodynamic and GFB parameters normalized to baseline (ratio of maximum effect/before injection), including GCaMP5/tdTomato fluorescence ratio (F_{max}/F0) in podocytes (G).

Figure 5. FSGS-Tg mouse chronic disease kidney: Effects of sparsentan and losartan on podocyte number, glomerulosclerosis, and tissue fibrosis

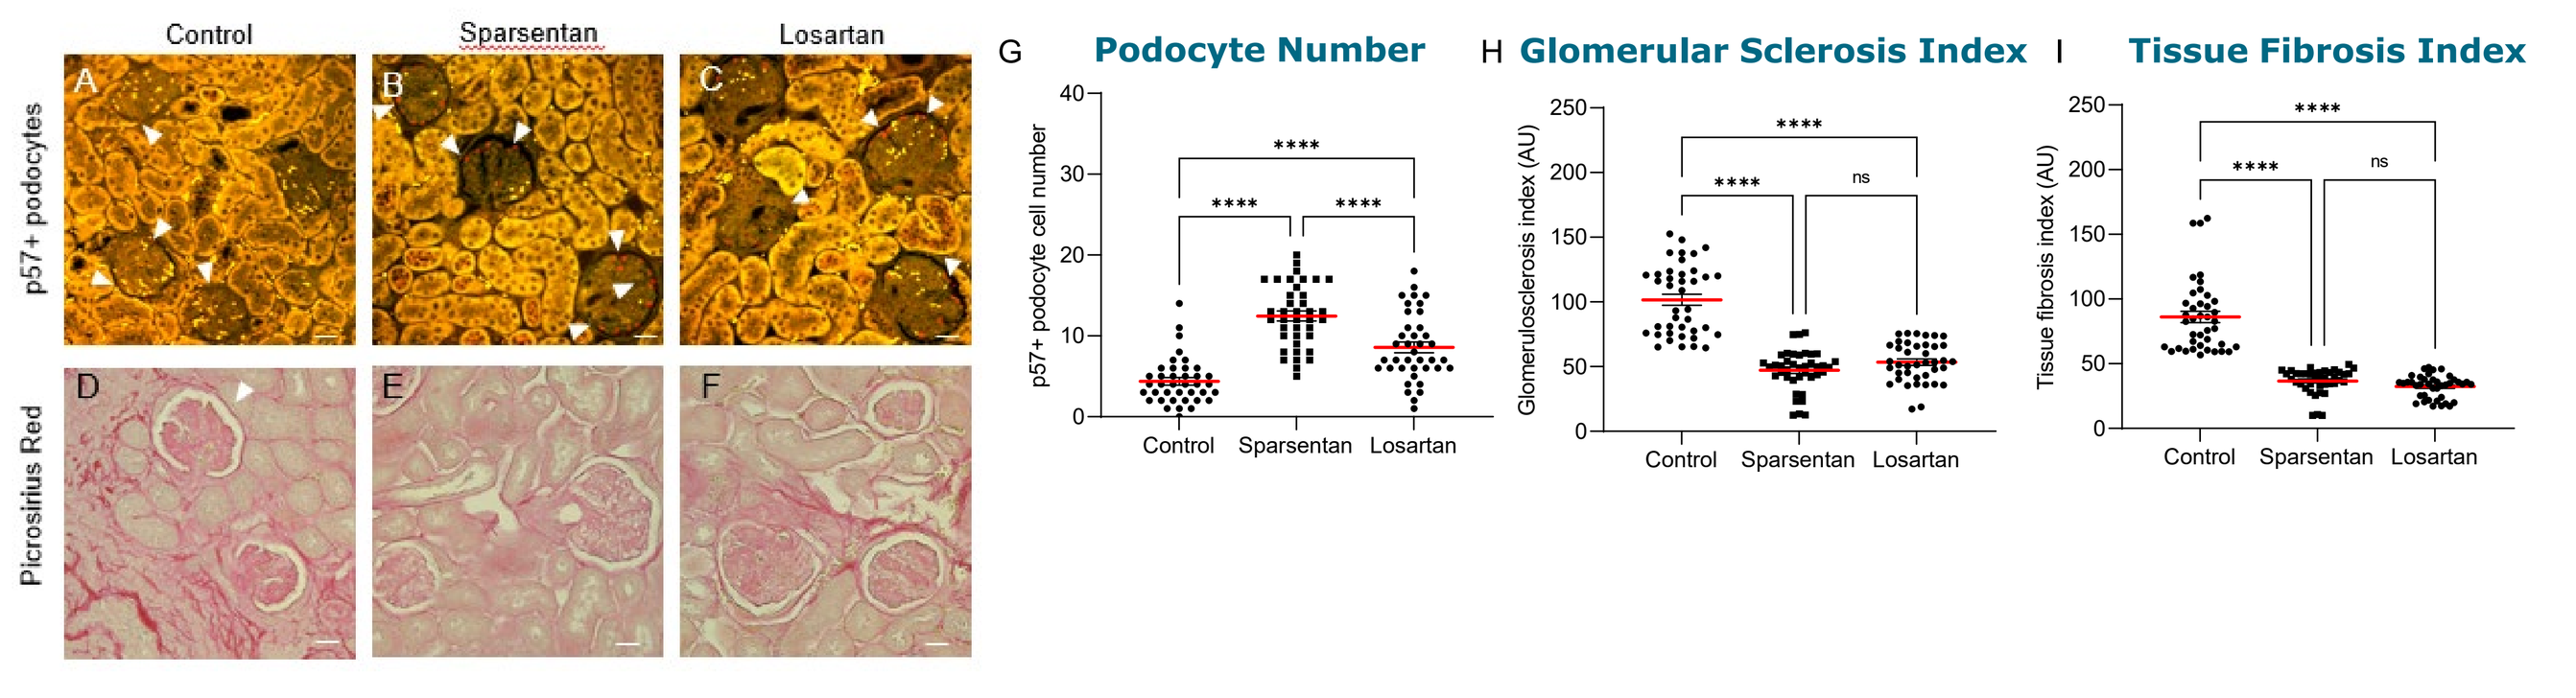
Control
Sparsentan
Losartan
p57+ podocytes
Picrosirius Red
G Podocyte Number
H Glomerular Sclerosis Index
I Tissue Fibrosis Index

FSGS-Tg-Fixed histological kidney sections. (A-C) p57+ podocyte nuclei (red, arrowheads), tissue autofluorescence in bright yellow. (D-F) Picrosirius red (red) staining. (G-I) Statistical summaries of podocyte number, glomerulosclerosis index (picrosirius red density per glomeruli) and tissue fibrosis index (picrosirius red density per full image frame).

Healthy Ren1d-Confetti-Tg Mice

- Sparsentan-treatment increased the number of Confetti+ cells, identical Confetti color cell groups (clones), and individual Confetti+ cells per clone in the glomerular tuft, at the glomerular vascular pole and terminal AA segment vs. control, and more than losartan treatment (**Figure 6**)
- Cells of the proximal tubule, the distal convoluted tubule, and the collecting duct showed active cellular (clonal) remodeling in response to sparsentan; losartan response was modest compared to sparsentan

Figure 6. Healthy Ren1d-Confetti-Tg mouse: basal physiologic response to 6 weeks of daily sparsentan or losartan treatment

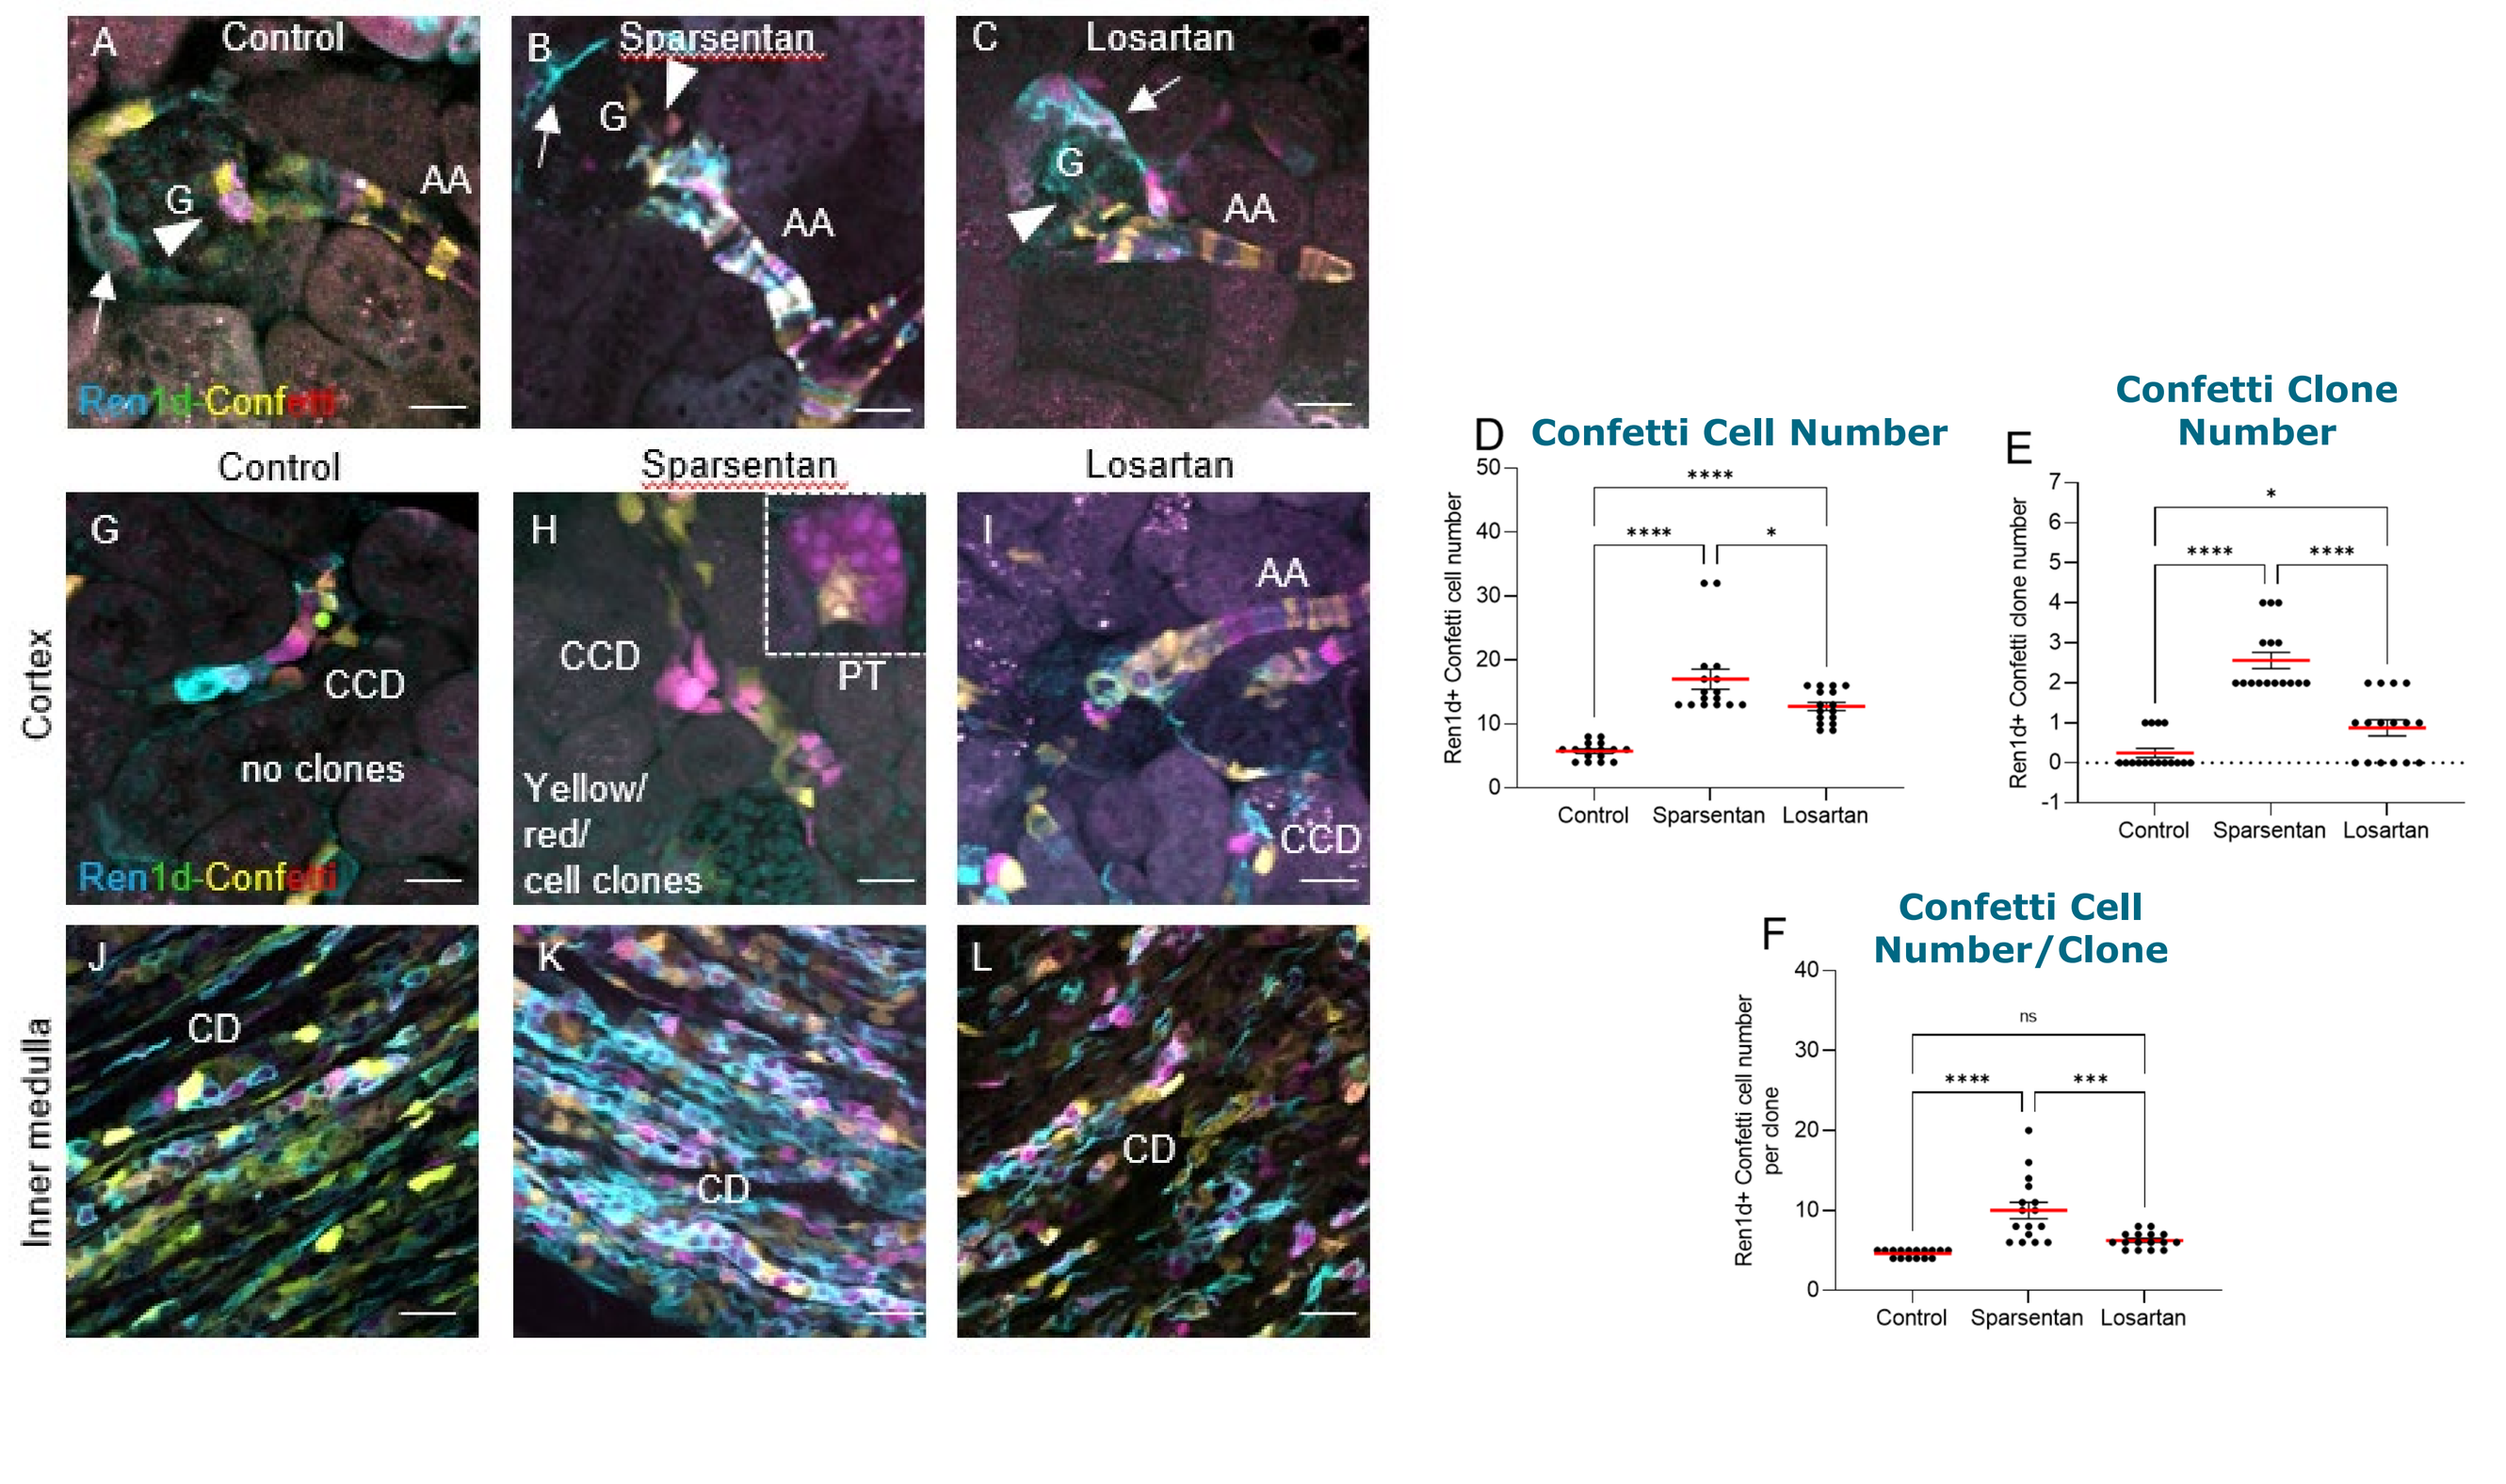
A Control
B Sparsentan
C Losartan
Ren1d-Confetti
Cortex
Inner medulla
CCD
no clones
Yellow/red/cell clones
PT
CD
D Confetti Cell Number
E Confetti Clone Number
F Confetti Cell Number/Clone

Healthy Ren1d-Confetti-Tg: Fixed histological kidney sections (A-C, G-L). Color Key: membrane CFP (blue), nuclear GFP (green), cytosolic YFP (yellow), and RFP (red). Clones appear as identical color groups. (A-C) Labeled CoRL showing extra- and intra-glomerular mesangial cells (arrowheads), parietal epithelial cells, and podocytes of the Bowman's capsule (arrows). (D-F) Measured number of Ren1d Confetti+ clones and cells. (G-L) Confetti+ tubular cells. Presence or absence of identical clonal cell groups in the proximal tubule (PT, inset), cortical (CCD), and medullary collecting duct (CD).

BACKGROUND

- Preliminary preclinical and emerging clinical evidence indicate strong antiproteinuric actions of dual endothelin type A (ET_AR) and angiotensin II type 1 (AT_1R) receptor antagonism with sparsentan
- These nephroprotective effects have been more pronounced in experimental and clinical settings compared to current standard of care using an AT_1R blocker (ARB)
- Dual inhibition of ET_AR and AT_1R using sparsentan is postulated to target multiple renal cell types via a variety of renoprotective mechanisms

AIM

To determine glomerular action of sparsentan compared to the ARB losartan by direct visualization of effects on renal hemodynamics and tissue remodeling in intact living mouse kidney

METHODS

- Intravital multiphoton microscopy (MPM) was performed on transgenic mouse tissue combined with traditional urinalysis and histology-based phenotyping

Transgenic (Tg) mouse models to track tissue remodeling:

- **Healthy-Tg** Model: Ren1d-**GCaMP5**/**tdTomato** transgenic mice specifically express the intensely green and highly calcium-sensitive fluorescent protein GCaMP5G and the calcium-insensitive red fluorescent protein tdTomato in cells of the renin lineage
- **FSGS-Tg** Model: Pod-**GCaMP5**/**tdTomato** **TRPC6** transgenic mice (1.5 years), in which TRPC6 is overexpressed together with the calcium reporter GCaMP5 td tomato in podocytes

Healthy Ren1d-Confetti-Tg Model: Ren1d-Confetti mice feature a multicolor **CFP**/**GFP**/**YFP**/**FP** reporter that allows single cell fate tracking of cells of the renin lineage (CoRL)

Measured Parameters

- Glomerular hemodynamic parameters (afferent [AA] and efferent arteriole [EA] diameters, single nephron glomerular filtration rate [SNGFR]), podocyte calcium (Ca^{2+}) entry, glomerular sieving coefficient (GSC), podocyte number, red blood cell velocity (RBCV), and glomerular capillary blood flow (GCBF)
- Urinary albumin/creatinine ratio (ACR) and clonal expansion (confetti model)

Treatment Groups

- No drug control, losartan (10 mg/kg/day), or sparsentan (120 mg/kg/day) administered ad libitum in rodent chow for 6 weeks (FSGS-Tg) or 2 weeks (Healthy-Tg or Healthy Ren1d-Confetti-Tg)

Acute Vasoconstriction induced by:

- Bolus IV injection of ET-1 (50 ng/kg) with or without Ang II (400 ng/kg) into the cannulated carotid artery in vehicle control

Statistics

- Values are expressed as mean ± SEM, *P<0.05, **P<0.01, ***P<0.001, ****P<0.0001 using one-way ANOVA with Tukey's multiple comparison test, n=11-44 measurements from n=8 mice in each group
- Bars in microscopy images are 20 μm

CONCLUSIONS

- Serial MPM imaging directly visualized several mechanisms underlying beneficial antiproteinuric and structural effects of sparsentan in both FSGS-Tg and in normal mouse kidneys (Healthy-Tg and Healthy Ren1d-Confetti-Tg)
- Sparsentan-treatment had a greater impact on reduction in proteinuria (ACR) and increase in podocyte protection in the FSGS-Tg model than losartan, driven by both AA and EA dilation resulting in an increase in capillary blood flow and SNGFR
- The Healthy-Tg models suggest mechanisms involving ET-1 and AngII antagonism and activation of resident progenitor cells and tissue remodeling for sparsentan being more effective in attenuating podocyte injury and renal disease
- These findings suggest multiple layers of renal protective actions by dual ET_AR and AT_1R antagonism

DISCLOSURES

Georgina Gyarmati, Urvi Shroff, Audrey Izuhara, Janos Peti-Peterdi received a research grant from Travere Therapeutics, Inc.

Patricia W. Bedard and Radko Komers are full-time employees of Travere Therapeutics, Inc., and may have an equity or other financial interest in Travere Therapeutics, Inc.

ACKNOWLEDGMENTS

This work was supported by a research collaboration grant by Travere Therapeutics and S10 OD021833 (to the USC Multi-Photon Microscopy Core). Editorial support was provided by Eric Scocchera, PhD, and Courtney Breuel, ELS, of MedVal Scientific Information Services, LLC (Princeton, NJ) and was funded by Travere Therapeutics, Inc.

REFERENCES

1. Saran R, et al. *Am J Kidney Dis*. 2020;75(1 Suppl 1):A6-A7; 2. Campbell KN, Tumlin JA. *Am J Nephrol*. 2018;47 Suppl 1(Suppl 1):14-29; 3. Feng D, et al. *Am J Physiol Renal Physiol*. 2018;314:F921-5; 4. Dryer SE, Reiser J. *Am J Physiol Renal Physiol*. 2010;299:F689-F701; 5. Krall P, et al. *PLoS One*. 2010;5:e12859; 6. Reiser J, et al. *Nat Genet*. 2005;37:739-44; 7. Winn MP, et al. *Science*. 2005;308:1801-4; 8. Eckel J, et al. *J Am Soc Nephrol*. 2011;22:526-35; 9. Komers R, Plotkin H. *Am J Physiol Regul Integr Comp Physiol*. 2016;310:R877-84; 10. Kowala MC, et al. *J Pharmacol Exp Ther*. 2004;309:275-84; 11. Trachtman H, et al. *J Am Soc Nephrol*. 2018;29:2745-54; 12. Burford JL, et al. *Pflugers Archiv*. 2017;469:965-74.

To obtain a PDF of this poster:

Scan the QR code OR visit www.traverepublications.com/DGfN2022/P202/

Charges may apply.

No personal information is stored.

Presented at the German Society of Nephrology 14th Annual Congress 2022 (DGfN2022), 6-9 October 2022, Berlin, Germany

Contact information: Medinfo@Travere.com