A Quantitative Systems Pharmacology (QSP) Model for Classical Homocystinuria Predicting Efficacy of Treatment

Tomas Majtan[1], Robert Mines[2], Maryam Khalifa[2], Piet H van der Graaf[2,3], Douglas Chung[2], Ying Chen[4], Steve Rodems[4], Kai Liu[4]

[1]Department of Pharmacology, Faculty of Science and Medicine, University of Fribourg, Chemin du Musee 18, PER17, 1700 Fribourg, Switzerland;
[2]Certara, Sheffield, S1 2BJ, United Kingdom; [3]Leiden University, 2300 RA Leiden, The Netherlands; [4]Travere Therapeutics, Inc., San Diego, California, USA

Poster #79

METHODS

- The QSP model, developed in collaboration with Certara (Princeton, NJ), was based on prior modelling work[7,8] and constructed using published preclinical data, including that from I278T mouse models of HCU[9-12]
- The whole organism QSP model (simplified version in **Figure 2**) is composed of 145 chemical reactions or transport processes
- Together, these reactions change the concentration of all relevant pathway metabolites (including Met, Hcy, cysteine, Bet, SAM, and SAH, along with their oxidized forms) in every organ compartment
- The model can be used to predict the result of any experimental perturbation such as dietary modification, addition of Bet supplements, or treatment with pegtibatinase

Figure 2. A Whole Organism Model of the Met Metabolic Pathway

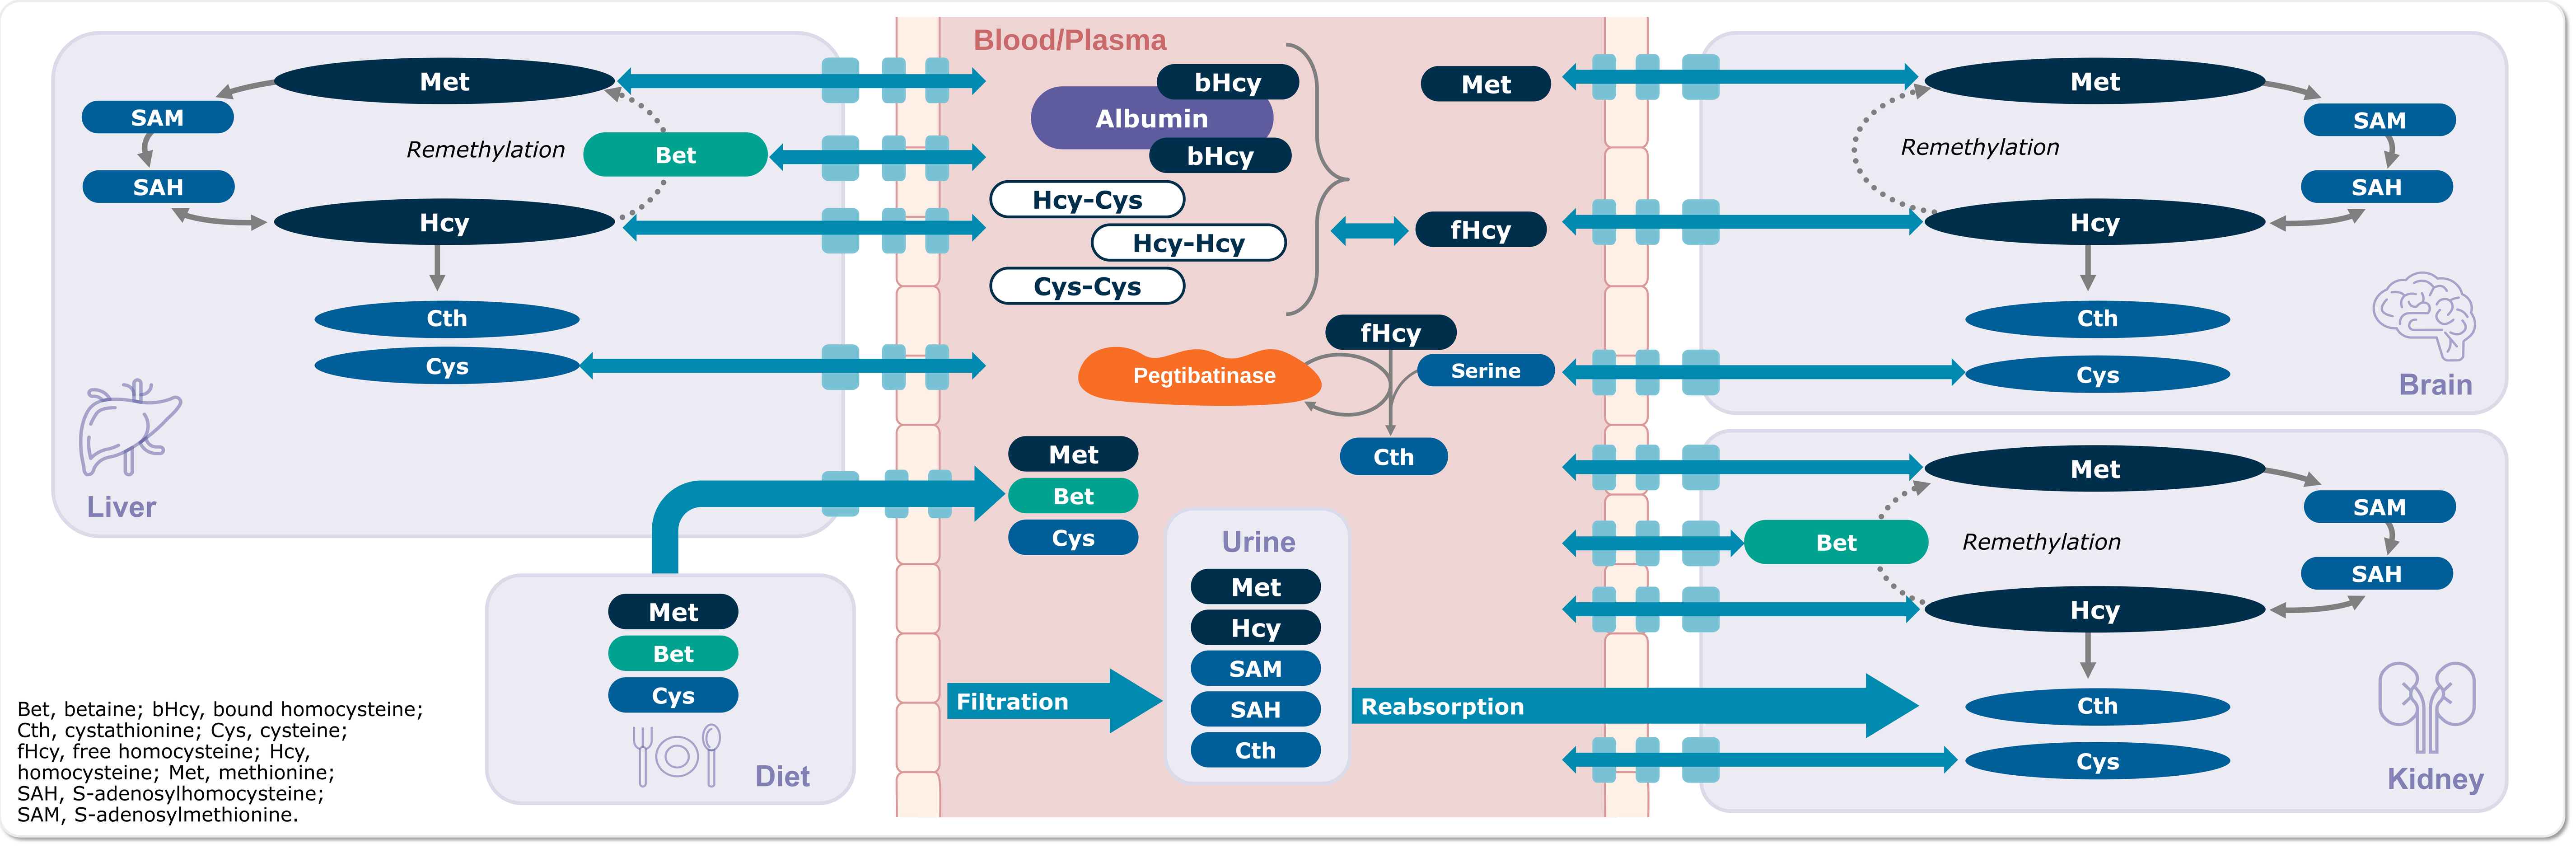

Blood/Plasma
Liver
Met
SAM
SAH
Remethylation
Bet
Hcy
Cth
Cys
bHcy
Albumin
Hcy-Cys
Hcy-Hcy
Cys-Cys
fHcy
Pegtibatinase
Serine
Diet
Urine
Filtration
Reabsorption
Brain
Kidney

Bet, betaine; bHcy, bound homocysteine; Cth, cystathionine; Cys, cysteine; fHcy, free homocysteine; Hcy, homocysteine; Met, methionine; SAH, S-adenosylhomocysteine; SAM, S-adenosylmethionine.

RESULTS

Validation

- The model was validated by its ability to reproduce known pathophysiological metabolic consequences in transgenic I278T mouse models of HCU, where Met intake was unrestricted (ie, standard mouse chow provided *ad libitum*)

Effects of Treatments and Diet

- **Figure 3A-D** shows the close correlation between existing mouse preclinical data (data points shown at Days 1, 7, 12, 16, and 22) and QSP model simulations (solid lines) on plasma levels of tHcy, Met, cysteine, and Bet during dietary acclimation, Bet supplementation, and pegtibatinase administration
- Taking plasma tHcy as an example (**Figure 3A**), tHcy levels were initially high, then were reduced by introduction of the Met 1.0 g/kg Met-restricted diet at Day 0, reduced further by Bet supplementation from Day 6, and even further reduced well below the 100 µM clinically meaningful threshold[1] following pegtibatinase dosing from Day 13; the strong correlation between observed preclinical data and simulated predictions was observed for all metabolites studied
- **Figure 3E-H** follows the same sequence of diet acclimation, Bet supplementation, and pegtibatinase dosing, but illustrates the computational simulations of tissue concentrations of the selected metabolites in the brain (purple), kidney (green), and liver (light blue)
- Similar patterns were observed where pegtibatinase treatment resulted in dramatic changes in the concentrations of tHcy, Met, cysteine, and Bet, over and above what was achieved with Bet supplementation
- Examination of brain, kidney, and liver tissue in the clinical situation is essentially non-existent which makes our QSP model particularly valuable by providing simulations of metabolic changes in the tissues in addition to readily available plasma

Figure 3. Plasma Responses (A-D) and Simulated Tissue Responses (E-H) of Met Cycle Metabolites to Dietary Restriction, Bet Supplementation, and Pegtibatinase

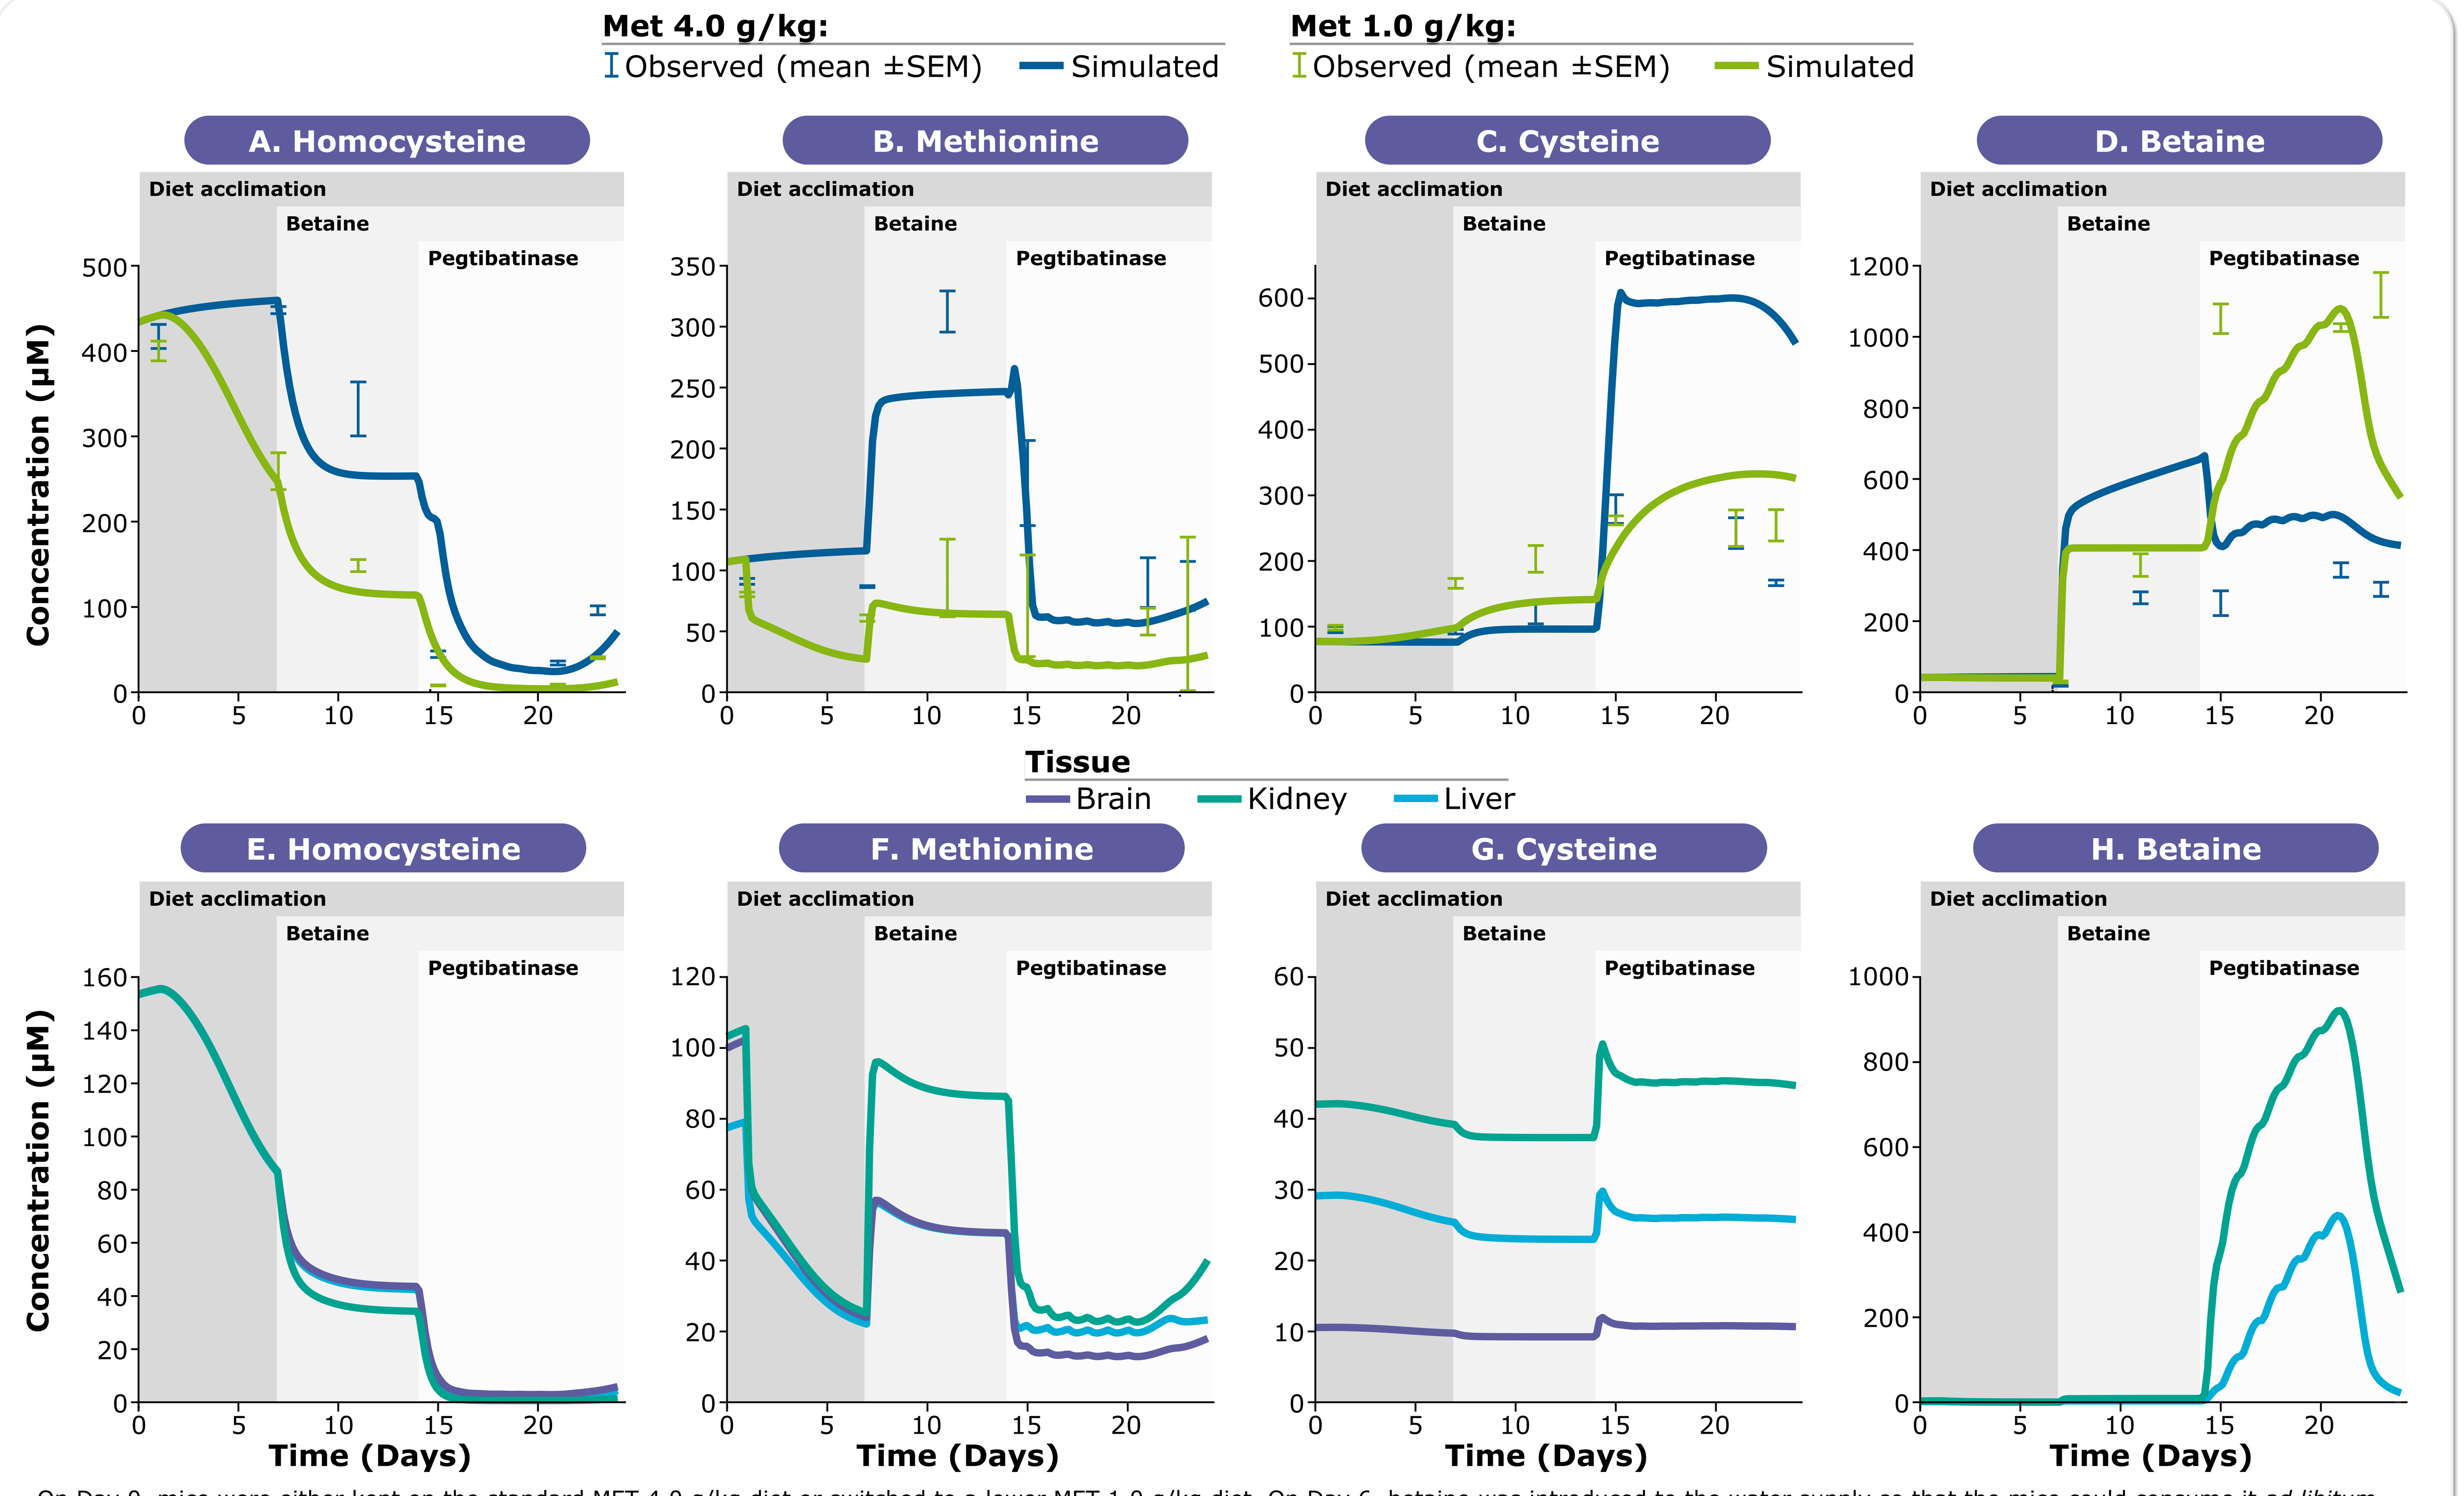

Met 4.0 g/kg:
Observed (mean ±SEM)
Simulated
Met 1.0 g/kg:
Observed (mean ±SEM)
Simulated
A. Homocysteine
B. Methionine
C. Cysteine
D. Betaine
Diet acclimation
Betaine
Pegtibatinase
Concentration (µM)
Tissue
Brain
Kidney
Liver
E. Homocysteine
F. Methionine
G. Cysteine
H. Betaine
Time (Days)

On Day 0, mice were either kept on the standard MET 4.0 g/kg diet or switched to a lower MET 1.0 g/kg diet. On Day 6, betaine was introduced to the water supply so that the mice could consume it *ad libitum*. On Day 13, pegtibatinase was dosed at 8 mg/kg per day for 7 days. Preclinical measurements were taken on Days 1, 7, 12, 16, and 22. SEM, standard error of the mean.

CONCLUSIONS

- Our QSP computational model successfully illustrated the complex interactions that exist between metabolites of the Met cycle and factors such as dietary Met restriction and use of therapies such as Bet and pegtibatinase
- The model recapitulates and extends the same conclusions as numerous preclinical studies, predicting tHcy lowering associated with pegtibatinase use in addition to or independent of dietary protein restriction and Bet supplementation

DISCLOSURES

TM: Ad hoc consultant for Travere Therapeutics, Inc.; Inventor on patents related to pegtibatinase. **RM:** QSP consultant for Travere Therapeutics, Inc. **MK:** QSP consultant for Travere Therapeutics, Inc. **PHvdG:** QSP consultant for Travere Therapeutics, Inc. **DC:** QSP consultant for Travere Therapeutics, Inc. **YC:** Employee and stockholder, Travere Therapeutics, Inc. **SR:** Employee and stockholder, Travere Therapeutics, Inc. **KL:** Employee and stockholder, Travere Therapeutics, Inc.

ACKNOWLEDGMENTS

This study was supported by Travere Therapeutics, Inc. (San Diego, CA). The QSP model was developed in collaboration with Certara (Princeton, NJ). Medical writing and layout assistance was provided by LiNK Health Group (Bridgewater, NJ) and Sephirus Communications (North Hills, CA) and was funded by Travere Therapeutics, Inc.

REFERENCES

1. Morris AA, et al. *J Inherit Metab Dis*. 2017;40:49-74. **2.** Majtan T, et al. *Biomacromolecules*. 2017;18:1747-1761. **3.** Bublil EM, Majtan T. *Biochimie*. 2020;173:48-56. **4.** Levy HL, et al. Poster #55 presented at the Society for Inherited Metabolic Disorders (SIMD) 43rd Annual Meeting; April 10-13, 2022; Orlando, FL. 2022. **5.** Aghamiri SS, et al. *J Pharmacokinet Pharmacodyn*. 2022;49:19-37. **6.** Bai JPF, et al. *CPT Pharmacometrics Syst Pharmacol*. 2021;10:1479-1484. **7.** Reed MC, et al. *J Nutr*. 2006;136:529S-532S. **8.** Martinov MV, et al. *J Theor Biol.* 2000;204:521-532. **9.** Majtan T, et al. *FASEB J.* 2017;31:5495-5506. **10.** Majtan T, et al. *Mol Ther.* 2018;26:834-844. **11.** Park I, et al. *Nutrients.* 2020;12:2895. **12.** Martinov MV, et al. *Biochim Biophys Acta.* 2010;1804:89-96.

To obtain a PDF of this poster:

Scan the QR code OR visit www.traverepublications.com/SIMD2023/79

Charges may apply.

No personal information is stored.

INTRODUCTION

- Classical homocystinuria (HCU) is a rare genetic metabolic disorder involving the methionine (Met) cycle and transsulfuration pathway[1] (**Figure 1**)
- In patients with HCU, deficiency of cystathionine β-synthase (CBS) leads to increased levels of homocysteine (Hcy) and other metabolic abnormalities, causing complications such as stroke, cognitive and ocular impairment, and skeletal abnormalities[1]
- Current treatment for HCU includes a protein restricted diet, Met-free L-amino acid formula, pyridoxine (vitamin B6) therapy for patients who are responsive, and betaine (Bet), but this regimen is burdensome and inadequate, and there remains an unmet medical need[1-2]
- Pegtibatinase is a first-in-class, investigational, PEGylated, truncated human CBS designed as an enzyme replacement therapy for HCU;[3] interim results from a Phase 1/2 study demonstrate clinically meaningful reductions in total Hcy (tHcy) after pegtibatinase administration to patients with HCU[4]
- In mouse models of HCU, the subcutaneous administration of pegtibatinase reduced tHcy levels, improved other metabolic abnormalities, and lessened symptoms associated with the disease[3]
- Quantitative systems pharmacology (QSP) uses computational modelling to explore the dynamic interplays between a drug and the biological system, permitting multiple parameters to be adjusted and explored in parallel in a way that is not possible in clinical trials;[5] QSP is increasingly being used in drug development and regulatory decision making[6]

OBJECTIVE

- The objective of this study was to establish a QSP model of impaired Met metabolism due to a loss of CBS activity based on available HCU animal data to simulate the global metabolic effects of pegtibatinase

Figure 1. Met Cycle and Transsulfuration Pathway

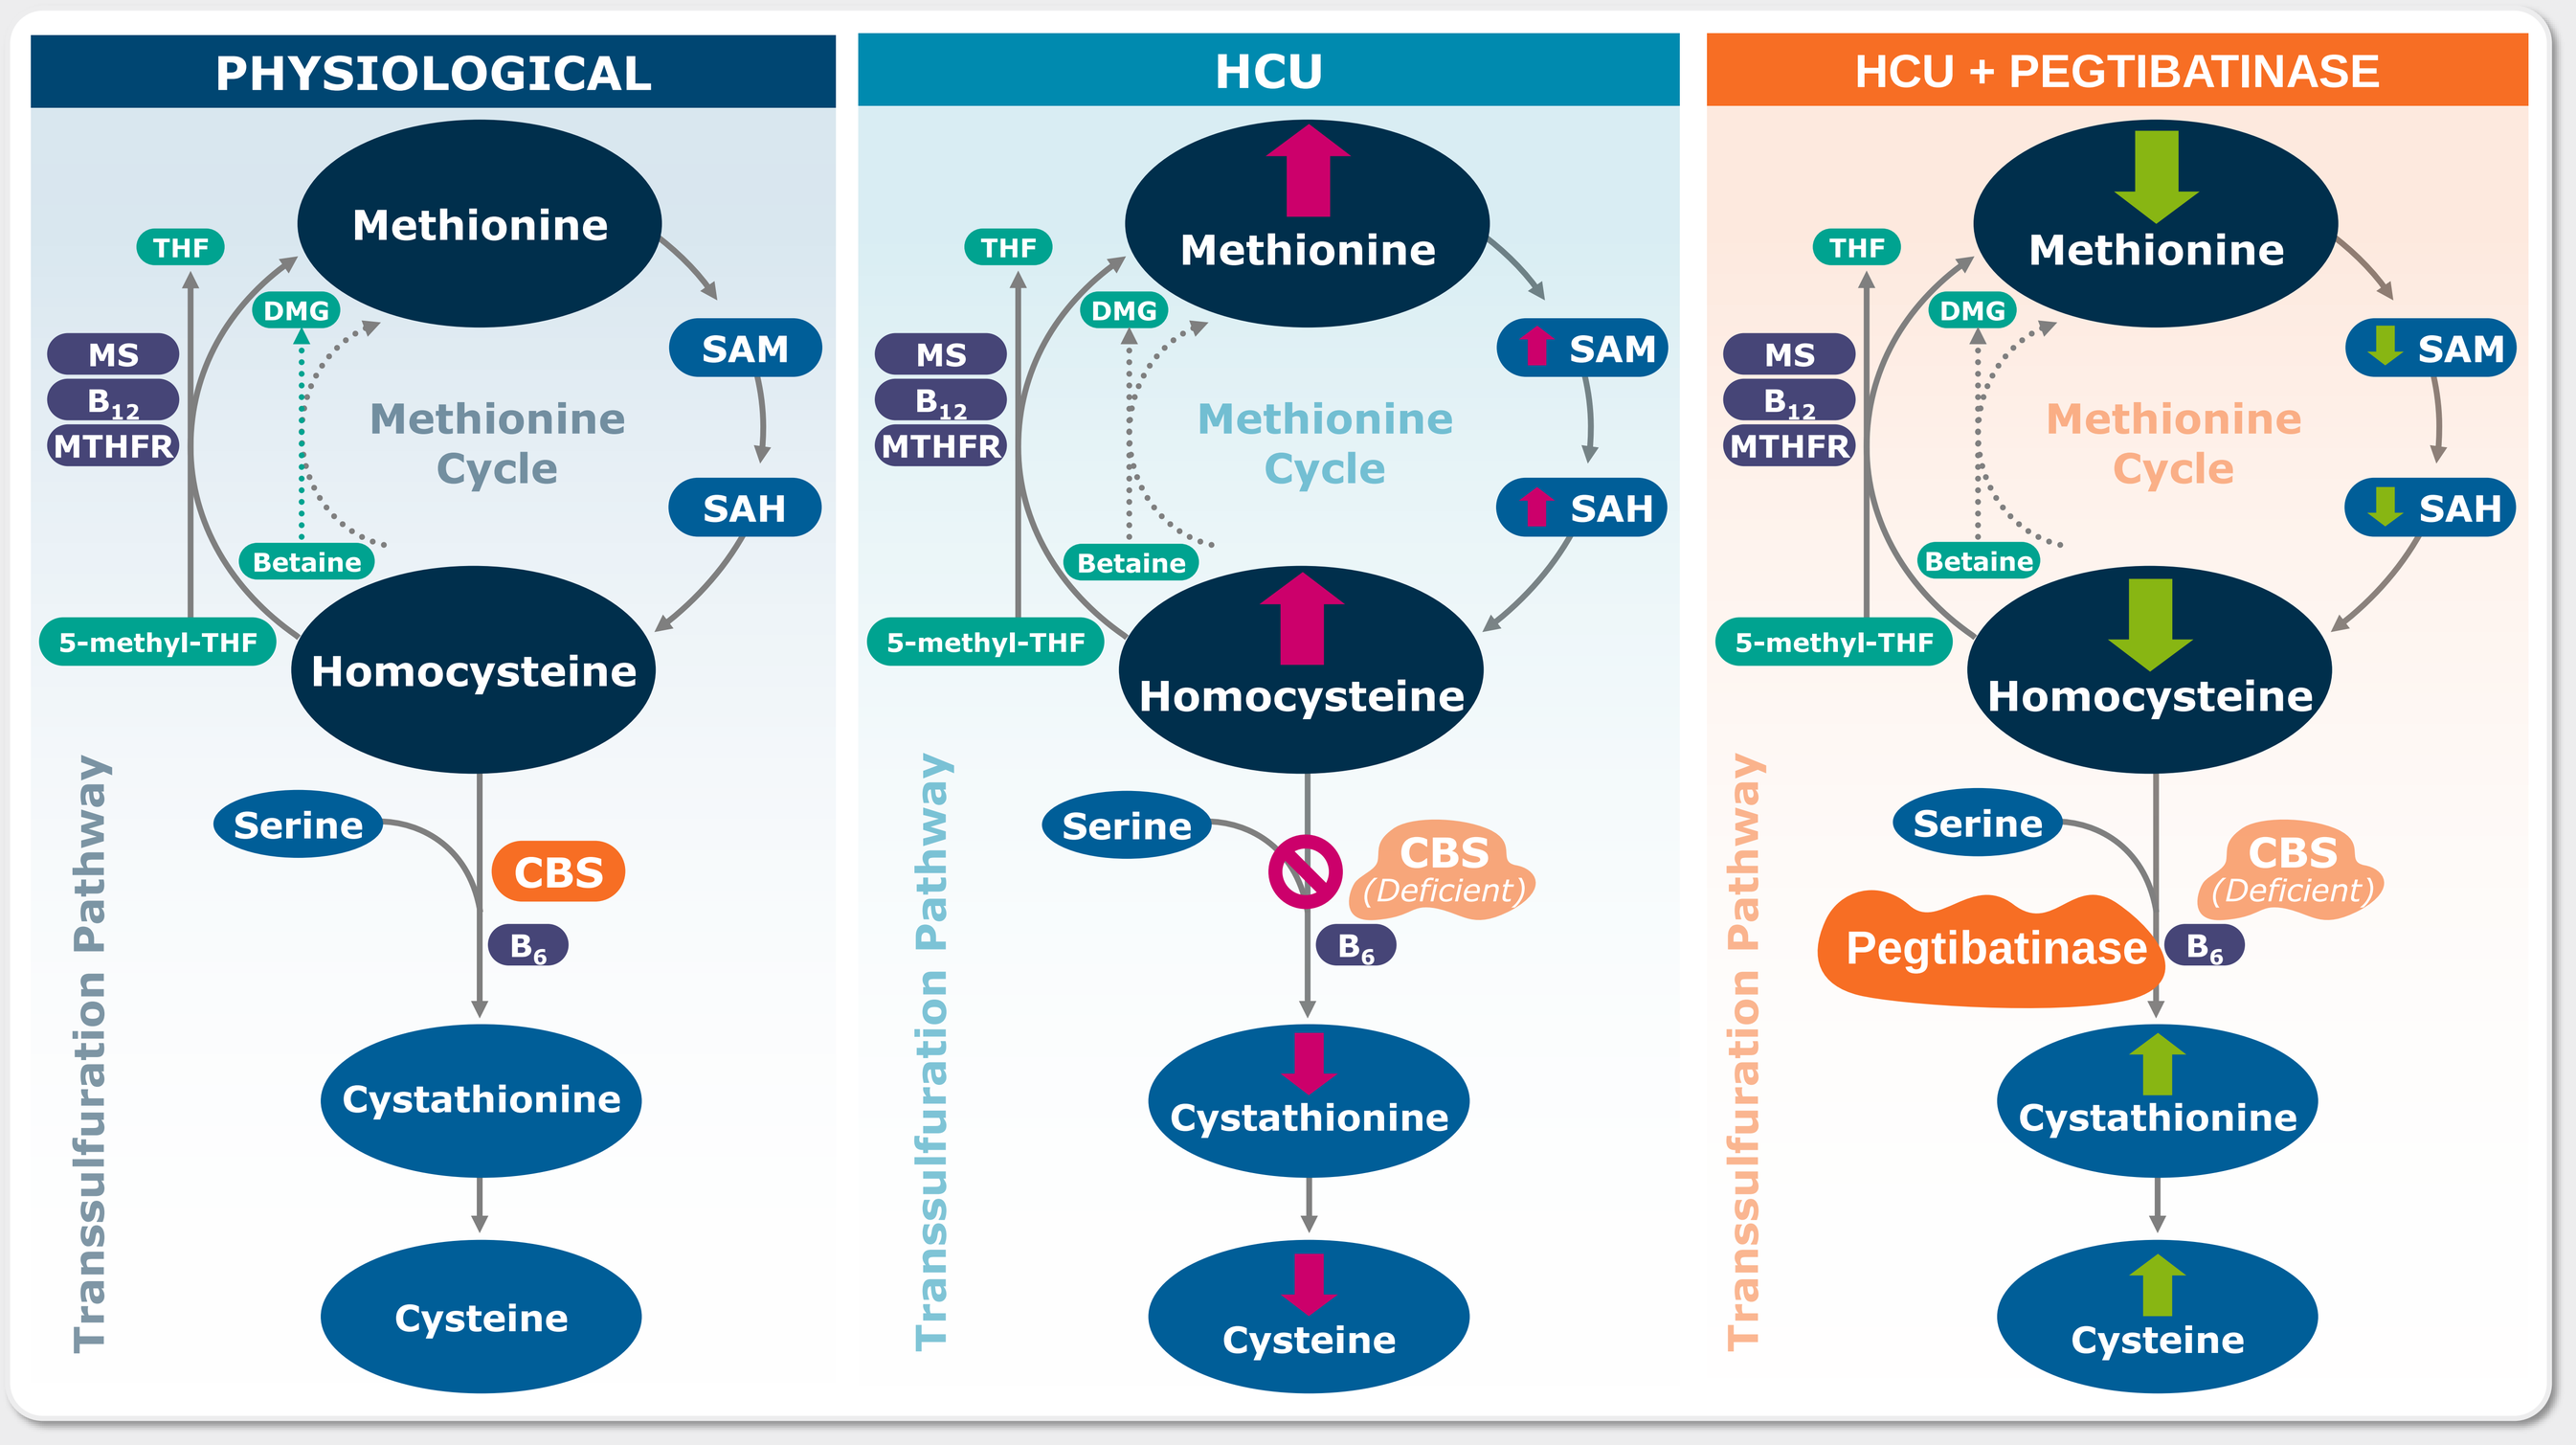

PHYSIOLOGICAL
HCU
HCU + PEGTIBATINASE
Methionine
SAM
SAH
Methionine Cycle
THF
DMG
MS
B_{12}
MTHFR
Betaine
5-methyl-THF
Homocysteine
Serine
CBS
CBS (Deficient)
B_6
Pegtibatinase
Cystathionine
Cysteine
Transsulfuration Pathway

CBS, cystathionine β-synthase; DMG, dimethylglycine; MS, methionine synthase; MTHFR, methylenetetrahydrofolate reductase; SAH, S-adenosylhomocysteine; SAM, S-adenosylmethionine; THF, tetrahydrofolate.

DISCUSSION

- Next steps include extending the model into data derived from forthcoming clinical studies, such as the ongoing HCU Natural History Study (ACAPPELLA), the Phase 1/2 COMPOSE Study, and the upcoming Phase 3 Study (HARMONY)
- When combined with clinical data, this model will assist in predicting optimal pegtibatinase dosing

LIMITATIONS

- These data are derived from a computational model based on preclinical data inputs and should be used to support, but not replace, clinical trial observations
- The findings reported here, based on mouse model data, will be re-examined when the model is populated with human data derived from clinical trials

Contact information: Travere Therapeutics, Inc. Medical Information, MedInfo@Travere.com

Presented at the Society for Inherited Metabolic Disorders (SIMD) 44th Annual Meeting; March 18 - 21, 2023; Salt Lake City, UT