P-205

SSIEM 2019

Skeletal Fragility In A Natural History Study Of Homocystinuria Due To Cystathionine Beta-synthase Deficiency Patients

Sellos-Moura M[6], Ficicioglu C[2], Viau K[1], Bublil E M[6], Crushell E[3], Farkas S[6], Glavin J F[6], Li H[5], Sharma R[4], Wolf J L[7], Levy H L[1]

[1]Boston Childrens Hospital, Boston, United States, [2]The Childrens Hospital of Philadelphia, Philadelphia, United States, [3]Childrens University Hospital, Dublin, Ireland, [4]Salford Royal Foundation NHS Trust, Salford, United Kingdom, [5]Emory University, Atlanta, United States, [6]Orphan Technologies, Rapperswil, Switzerland, [7]The Conjugate Group, Boston, United States

Background

Cystathionine beta-synthase deficient homocystinuria (HCU) is a rare autosomal recessive disorder caused by mutations in the cystathionine beta-synthase (CBS) gene leading to defects in methionine metabolism. The metabolic defect results in increased plasma levels of homocysteine (Hcy) and methionine (Met) and decreased levels of cysteine and cystathionine. Treatment consists of supplementation with B-vitamins (vitamins B6, B12, folate) and/or betaine, and/or restriction of the intake of the essential amino acid Met through a diet that is very low in natural protein to reduce the precursor load on the trans-sulfuration pathway. For patients on dietary treatment, supplementation with a Met-free L-amino acid mixture is also advised (Morris et al. 2017). Clinical manifestations of HCU include involvement of the eye, skeletal system, vascular system (thromboembolism), and neuropsychiatric abnormalities, including learning difficulties, developmental delay, and intellectual disability. Among the skeletal abnormalities seen in HCU are sternal deformities, vertebral changes, scoliosis, genu valgum (Brenton 1977), and reduced bone mineral density (BMD) (Parrot et al. 2000; Weber et al. 2016).

Methods

Study CBS-HCY-NHS-01 is a multicenter (8 sites), international, observational, prospective, natural history study of HCU that enrolled 55 pediatric (5-17 years of age) and adult (>18 years of age) patients to characterize the clinical course of HCU in patients under current clinical management practices over 3 years. It explores the range of plasma concentrations of total Hcy (tHcy) and related sulfur metabolites, and the variability of the clinical sequelae of the disease. Because Study CBS-HCY-NHS-01 is ongoing, this interim analysis was performed using a data cut-off date of 05 July 2019. Patients' characteristics and cognitive impairments were also assessed, and these data are reported in separate posters (0868 and 0853, respectively).
BMD was assessed at baseline and 1 year at three locations (the hip, lumbar spine, and whole body) by dual-energy X-ray absorptiometry (DXA) using either Hologic or General Electric/Lunar densitometers. BMD and Z-scores (number of standard deviations away from the average BMD value of the reference group, normalized for age and gender) were calculated for each location. A Z-score between -1 and -2.5 indicates osteopenia, while a Z-score below -2.5 indicates osteoporosis. tHcy levels were measured in plasma, and median total protein intake (g/day) was calculated from 3-day food records. Total protein intake was defined as the sum of the total natural protein from the diet and the protein from Met-free L-amino acid mixture, if taken by the patient. Correlations between BMD and tHcy or dietary protein, were calculated using the Pearson's correlation coefficient (R). Table 1 shows the percentile ranks associated with different Z-scores.

Table 1. Z-score and percentile bone density

%-ile	5	10	15	20	25	30	35	40	45	50	55	60	65	70	75	80	85	90	95
Z-score	-1.65	-1.29	-1.04	-.84	-.68	-.53	-.39	-.26	-.13	0	.13	.26	.39	.53	.68	.84	1.04	1.29	1.65

Results

Patients Characteristics: 55 subjects were enrolled in the study:
- 45%/55% females/males; most (84%) were white, non-Hispanic.
- Median (range) age at enrollment was 21 (5–53) years.
- Median time on study was 12.2 months.
- Half of the patients (51%) were diagnosed as infants (<1 year of age).
- The median plasma tHcy level was 95 µM (74 µM for pediatric; 104 µM for adult), consistent with underlying undertreated disease.
- 95% of pediatric patients had tHcy greater than the upper limit of normal (ULN), and 82% had tHcy >50 µM. The tHcy levels of almost half (45%) of the pediatric patients were >100 µM.
- All (100%) adult patients had tHcy greater than the ULN, 93% had tHcy levels >50 µM, and 77% had tHcy levels >100 µM.
- Median BMI was in the normal range (21.5 kg/m^2).
- Additional patient characteristics are reported in Poster P-204.

Bone Density Results:
- BMD data were available for 43 patients.
- The median Z-score was negative for both adult and pediatric patients at all body locations (Table 2), indicating skeletal fragility.
- 46% of adult and 53% of pediatric patients had bone densities below the 15th percentile (Z-scores ≤-1) in at least one location (Table 2).
- BMD results and Z-scores were stable over 12 months for each patient (N = 15-20; correlation between Visit 1 and Visit 3 for BMD at each location: Pearson's R = 0.94 to 0.97; p<0.0001).

Table 2. Summary of Z-score results by DXA scan location

	Hip			Spine			Body		
	All patients	Adult	Pediatric	All patients	Adult	Pediatric	All patients	Adult	Pediatric
N	43	24	19	42	23	19	37	19	18
Mean	-0.76	-0.60	-0.97	-0.52	-0.63	-0.39	-0.72	-0.81	-0.62
SD	1.32	1.38	1.26	1.30	1.44	1.15	1.37	1.36	1.20
Median	-0.9	-0.75	-1.00	-0.65	-0.80	-0.50	-0.8	-1.00	-0.65
Range	-2.6 to 3.0	-2.4 to 3.0	-2.6 to 1.9	-3.2 to 3.2	-3.2 to 3.2	-2.3 to 1.9	-3.0 to 2.2	-3.0 to 2.2	-2.9 to 1.3
N (%) patients with Z-score ≤ -1.0	21 (48.8%)	11 (45.8%)	10 (52.6%)	14 (33.3%)	10 (43.5%)	4 (21.1%)	17 (45.9%)	10 (52.6%)	7 (38.9%)

Effect of Various Parameters on Bone Density:
- Patients with plasma tHcy levels <100 µM had much greater skeletal fragility than those with levels >100 µM (Fig. 1). There was a positive correlation between plasma tHcy levels and BMD at all three locations (Pearson's R = 0.33 to 0.51; p<0.03; Table 3).
- BMD improves with greater total dietary protein intake, as shown by a positive correlation between BMD at all three locations and total dietary protein intake (Pearson's R = 0.55 to 0.76; p<0.011; Table 3).
- There is a positive correlation between BMD at all three locations and C-reactive protein (hsCRP) Pearson's R = 0.36 to 0.39; p<0.051; Table 3), a marker of inflammation, suggesting that inflammation may be correlated with high tHcy or increased protein intake.

Table 3. Correlations between BMD and other parameters

BMD	N (all, adult, pediatric)	All Subjects	Adult	Pediatric
Pearson Correlation with tHcy				
Hip	45, 26, 19	0.3340 (p=0.0249)	0.1545 (p=0.4510)	0.4963 (p=0.0307)
Spine	41, 24, 17	0.4784 (p=0.0016)	0.5001 (p=0.0128)	0.5005 (p=0.0408)
Body	38, 19, 19	0.5095 (p=0.0011)	0.4772 (p=0.0388)	0.3939 (p=0.0952)
Pearson Correlation with hsCRP				
Hip	33, 26, 7	0.3597 (p=0.0398)	0.3398 (p=0.0894)	0.3334 (p=0.4649)
Spine	30, 24, 6	0.3611 (p=0.0500)	0.2875 (p=0.1732)	0.6717 (p=0.1440)
Body	26, 19, 7	0.3925 (p=0.0473)	0.2999 (p=0.2122)	0.2123 (p=0.6476)
Pearson Correlation with Total Dietary Protein				
Hip	24, 11, 13	0.7566 (p<0.0001)	0.7268 (p=0.0113)	0.6843 (p=0.0099)
Spine	22, 10, 12	0.6870 (p=0.0004)	0.7073 (p=0.0221)	0.5532 (p=0.0621)
Body	21, 8, 13	0.5486 (p=0.0100)	0.1255 (p=0.7672)	0.4748 (p=0.1011

Red: p<0.05.

Figure 1. Effect of plasma tHcy levels on Z-scores

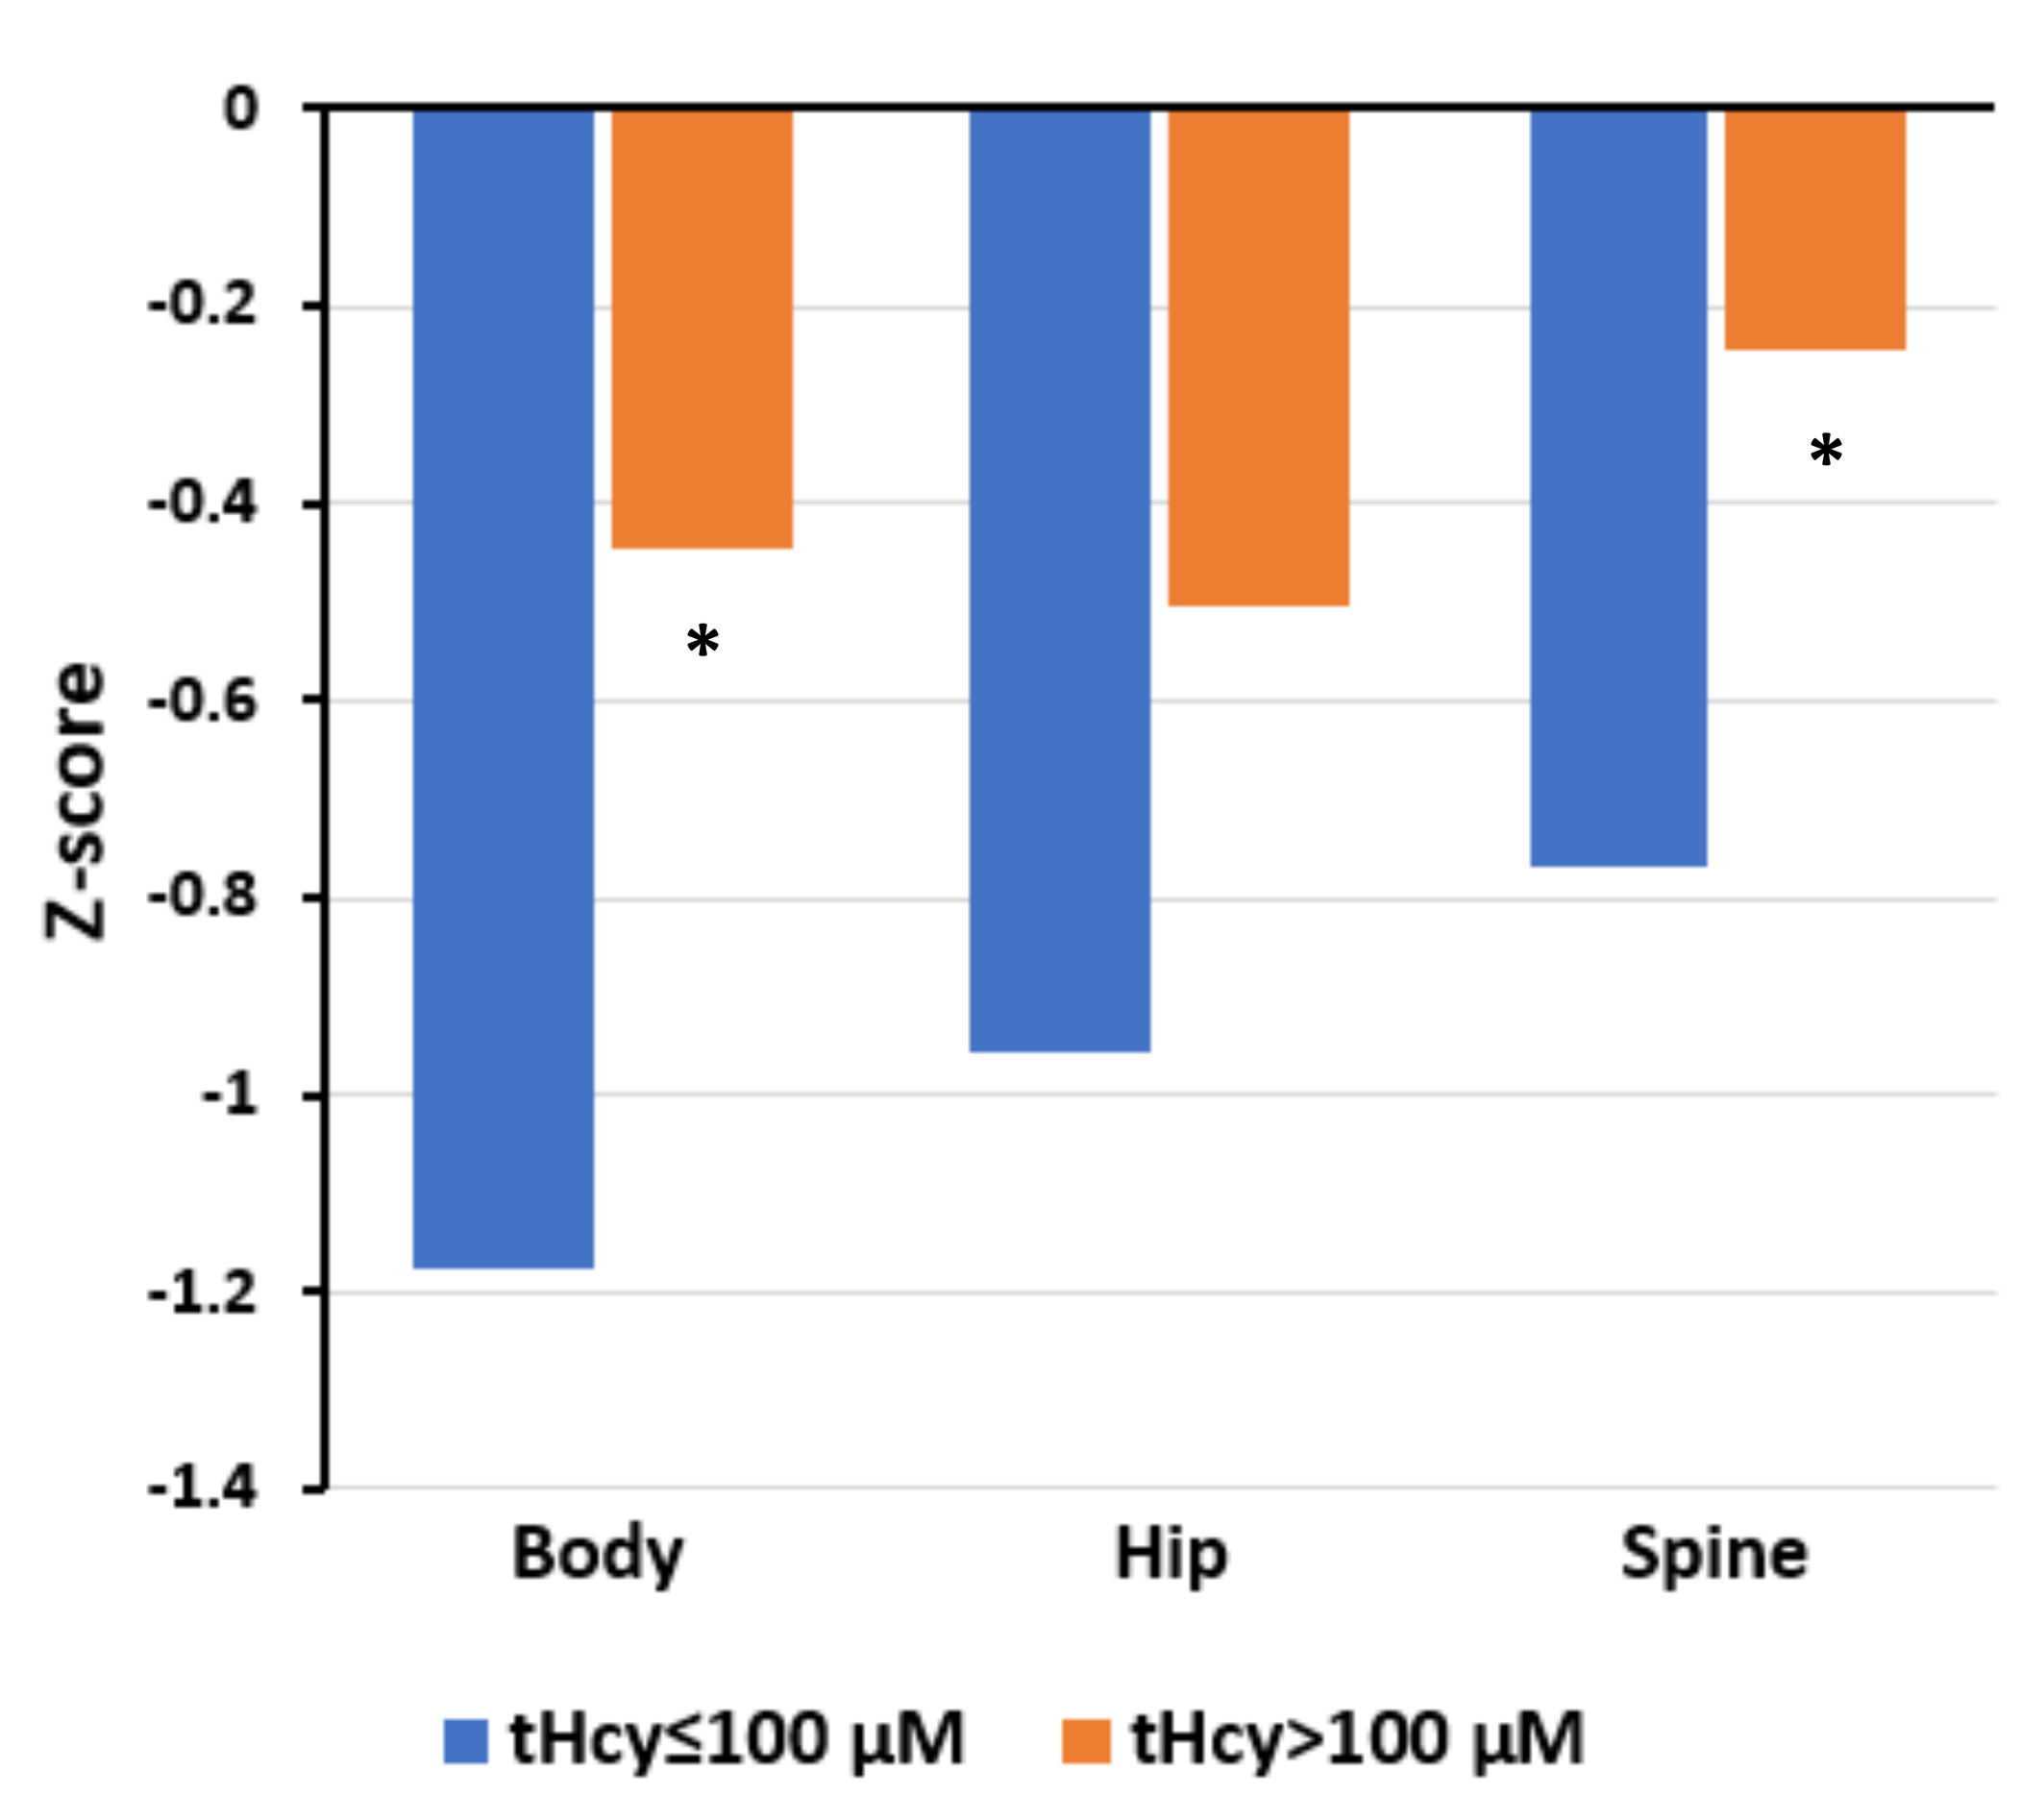

0
-0.2
-0.4
-0.6
-0.8
-1
-1.2
-1.4
Z-score
*
*
Body
Hip
Spine
tHcy≤100 µM
tHcy>100 µM

Significant differences indicated by * (P<0.05; t-test).

Discussion

- This study shows that most HCU patients have skeletal fragility despite a relatively young age, confirming past studies (Parrot et al. 2000; Weber et al. 2016).
- For each patient, BMD results and Z-scores were stable over 12 months. Therefore, BMD and Z-scores assessed by DXA can be considered a reliable endpoint to assess the efficacy of investigational treatments in future clinical trials in HCU patients.
- Bone health in this population is correlated with higher total dietary protein intake and higher tHcy levels, suggesting that, by chronically restricting their total protein intake to control their tHcy levels, patients may be increasing their skeletal fragility.

References

- Brenton DP. Postgrad Med J. Skeletal abnormalities in homocystinuria. 1977;53:488-96.
- Morris AAM, et al. Guidelines for the diagnosis and management of cystathionine beta-synthase deficiency. J Inherit Metab Dis. 2017;40(1):49-74.
- Parrot F, et al. J Inherit Metab Dis. Osteoporosis in late-diagnosed adult homocystinuric patients. 2000;23:338-40.
- Weber DR, et al. Low bone mineral density is a common finding in patients with homocystinuria. Med Genet Metab. 2016;117:351-4.

Disclosure: CBS-HCY-NHS-01 is sponsored by Orphan Technologies. Four authors (Sellos-Moura, Bublil, Farkas, and Glavin) are employees of the company and the remaining authors are Investigators in this clinical trial.

orphan technologies