Proteinuria and kidney survival in focal segmental glomerulosclerosis (FSGS) or immunoglobulin A nephropathy (IgAN): A NEPTUNE analysis

Laura H. Mariani[1], Jonathan Troost[1], Wu Gong[2], Mark E. Bensink[2], Yelena Drexler[3], Ambarish Athavale[4], Kunal Bailoor[1], Pietro Canetta[5], Vimal Derebail[6], Howard Trachtman[1]

[1]Univ. of Michigan; [2]Travere Therapeutics; [3]Univ. of Miami ; [4]Univv of California San Diego; [5]Columbia Univ.; [6]Univ. of North Carolina

Background/Goals

- **Background:** Changes in disease activity markers are important outcomes for clinical trials in rare proteinuric kidney diseases, such as FSGS and IgAN. While novel therapies may be effective in multiple etiologies, the relationship between proteinuria and kidney survival has not been directly described across diseases.
- **Goal:** To describe the relationship between proteinuria reduction and loss of kidney function in FSGS and IgA.

Methods

- We studied children and adults with FSGS or IgAN enrolled in the Nephrotic Syndrome Study Network (NEPTUNE) at the time of a clinically indicated kidney biopsy.
- Participants were followed prospectively with proteinuria and eGFR assessed at each study visit 2-3 times/year.
- We described the proportion of participants with proteinuria at months 4, 12 and 24 post-biopsy.
- We tested the association between lowest proteinuria within 12 months after biopsy and time to the composite of kidney failure or 40% eGFR decline from baseline using unadjusted Kaplan-Meier (K-M) method.

Table 1: Baseline Characteristics

	FSGS			IgAN		
	Overall (n=211)	Children (n=77)	Adults (n=134)	Overall (n=58)	Children (n=9)	Adults (n=49)
Age	32.9 (21.5)	10.7 (4.8)	45.7 (16.4)	36.8 (17.1)	11.9 (4.2)	41.4 (14.4)
UPCR (g/g) at baseline, Mean (SD)	6.3 (7.41)	8.8 (10.30)	4.8 (4.51)	3.6 (4.39)	5.2 (5.53)	3.3 (4.19)
eGFR at baseline, Mean (SD)	75.0 (34.9)	96.4 (32.1)	63.0 (30.5)	69.5 (32.2)	83.9 (29.9)	66.8 (32.2)
Edema	80 (38%)	32 (42%)	48 (36%)	14 (24%)	2 (22%)	12 (24%)
Obese	88 (42%)	29 (38%)	59 (44%)	20 (34%)	4 (44%)	16 (33%)
Stage 1 Hypertension*	55 (26%)	23 (30%)	32 (24%)	13 (22%)	4 (44%)	9 (18%)
Stage 2 Hypertension**	53 (25%)	10 (13%)	43 (32%)	16 (28%)	1 (11%)	15 (31%)

*130 to 139 Sys or 80 to 89 Dias for Adults and Ped >= 13 years old, or SysPct/DiasPct>=95 to <95 + 12mmHg or 130/80 to 139/89, **>=140/90 Sys/Dias for Adults and Ped >= 13 years old, or SysPct/DiasPct >= 95 + 12 mmHg or >= 140/90 Sys/Dias for Ped < 13 years old

Results

Figure 1: Proportion of patients reaching proteinuria thresholds over 24 months of follow-up, by pediatric and adult subgroups

FSGS

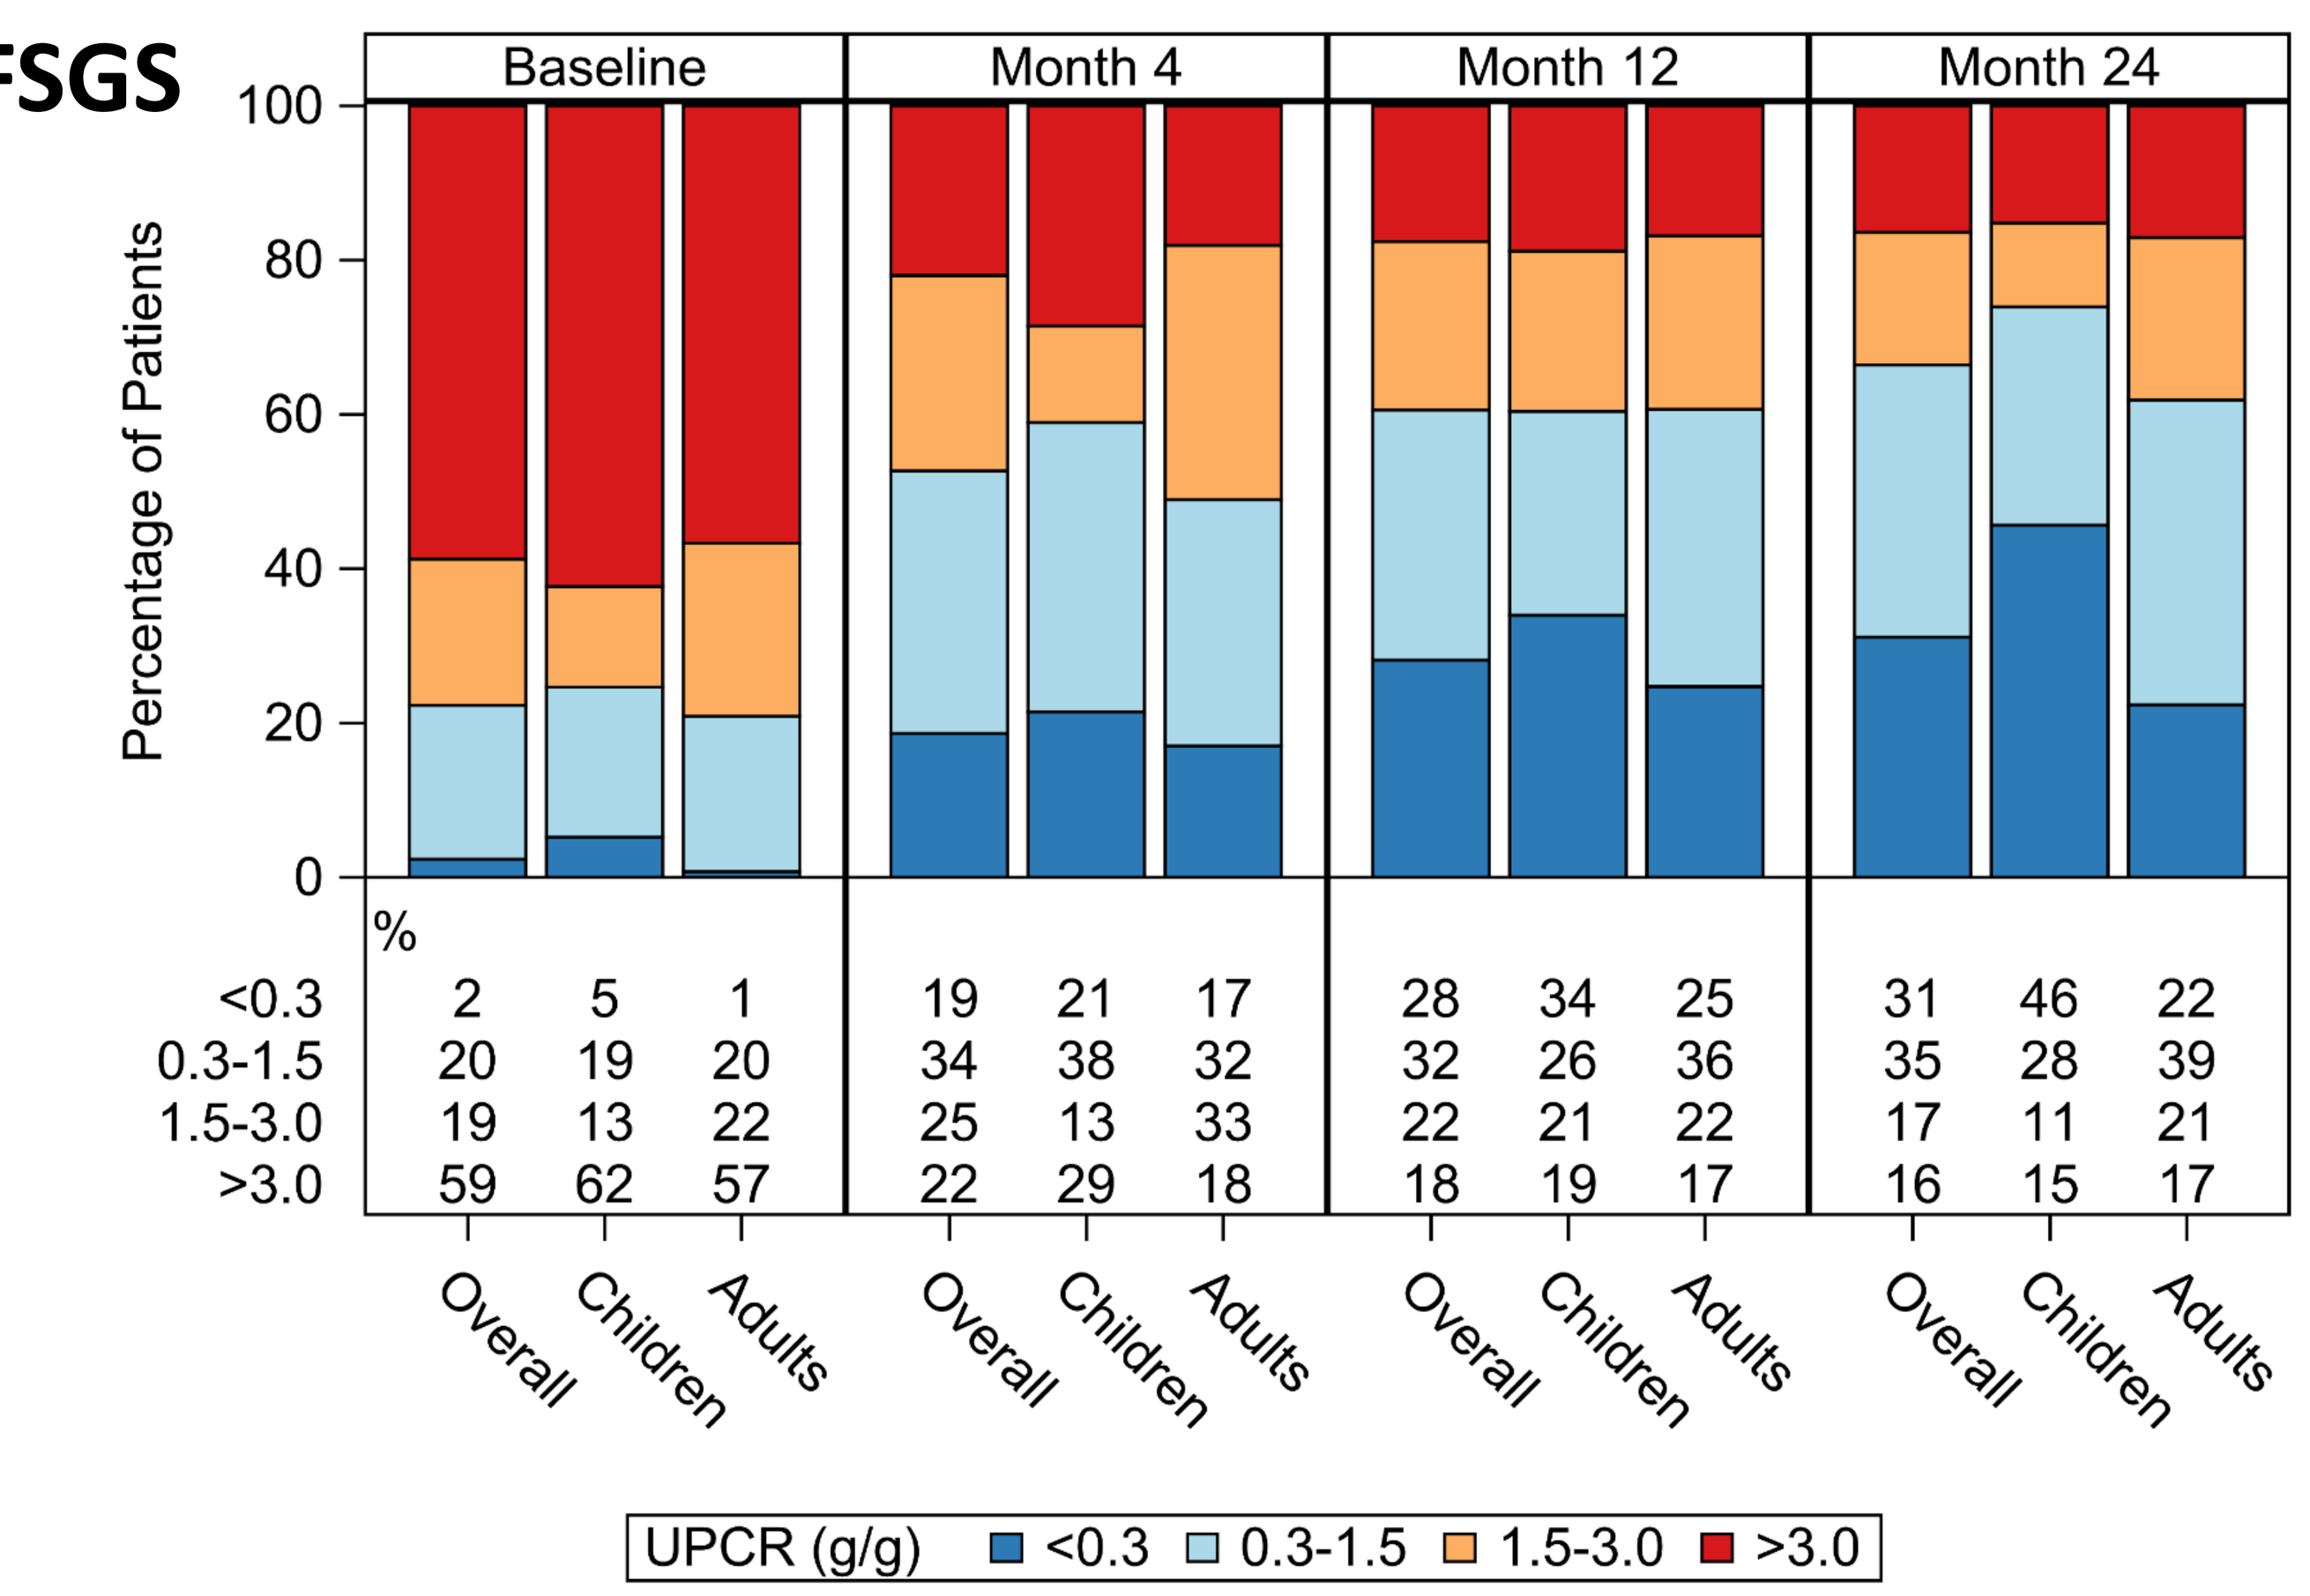
Baseline
Month 4
Month 12
Month 24
Percentage of Patients
100
80
60
40
20
0
%
<0.3 2 5 1 19 21 17 28 34 25 31 46 22
0.3-1.5 20 19 20 34 38 32 32 26 36 35 28 39
1.5-3.0 19 13 22 25 13 33 22 21 22 17 11 21
>3.0 59 62 57 22 29 18 18 19 17 16 15 17
Overall
Children
Adults
UPCR (g/g) <0.3 0.3-1.5 1.5-3.0 >3.0

IgAN

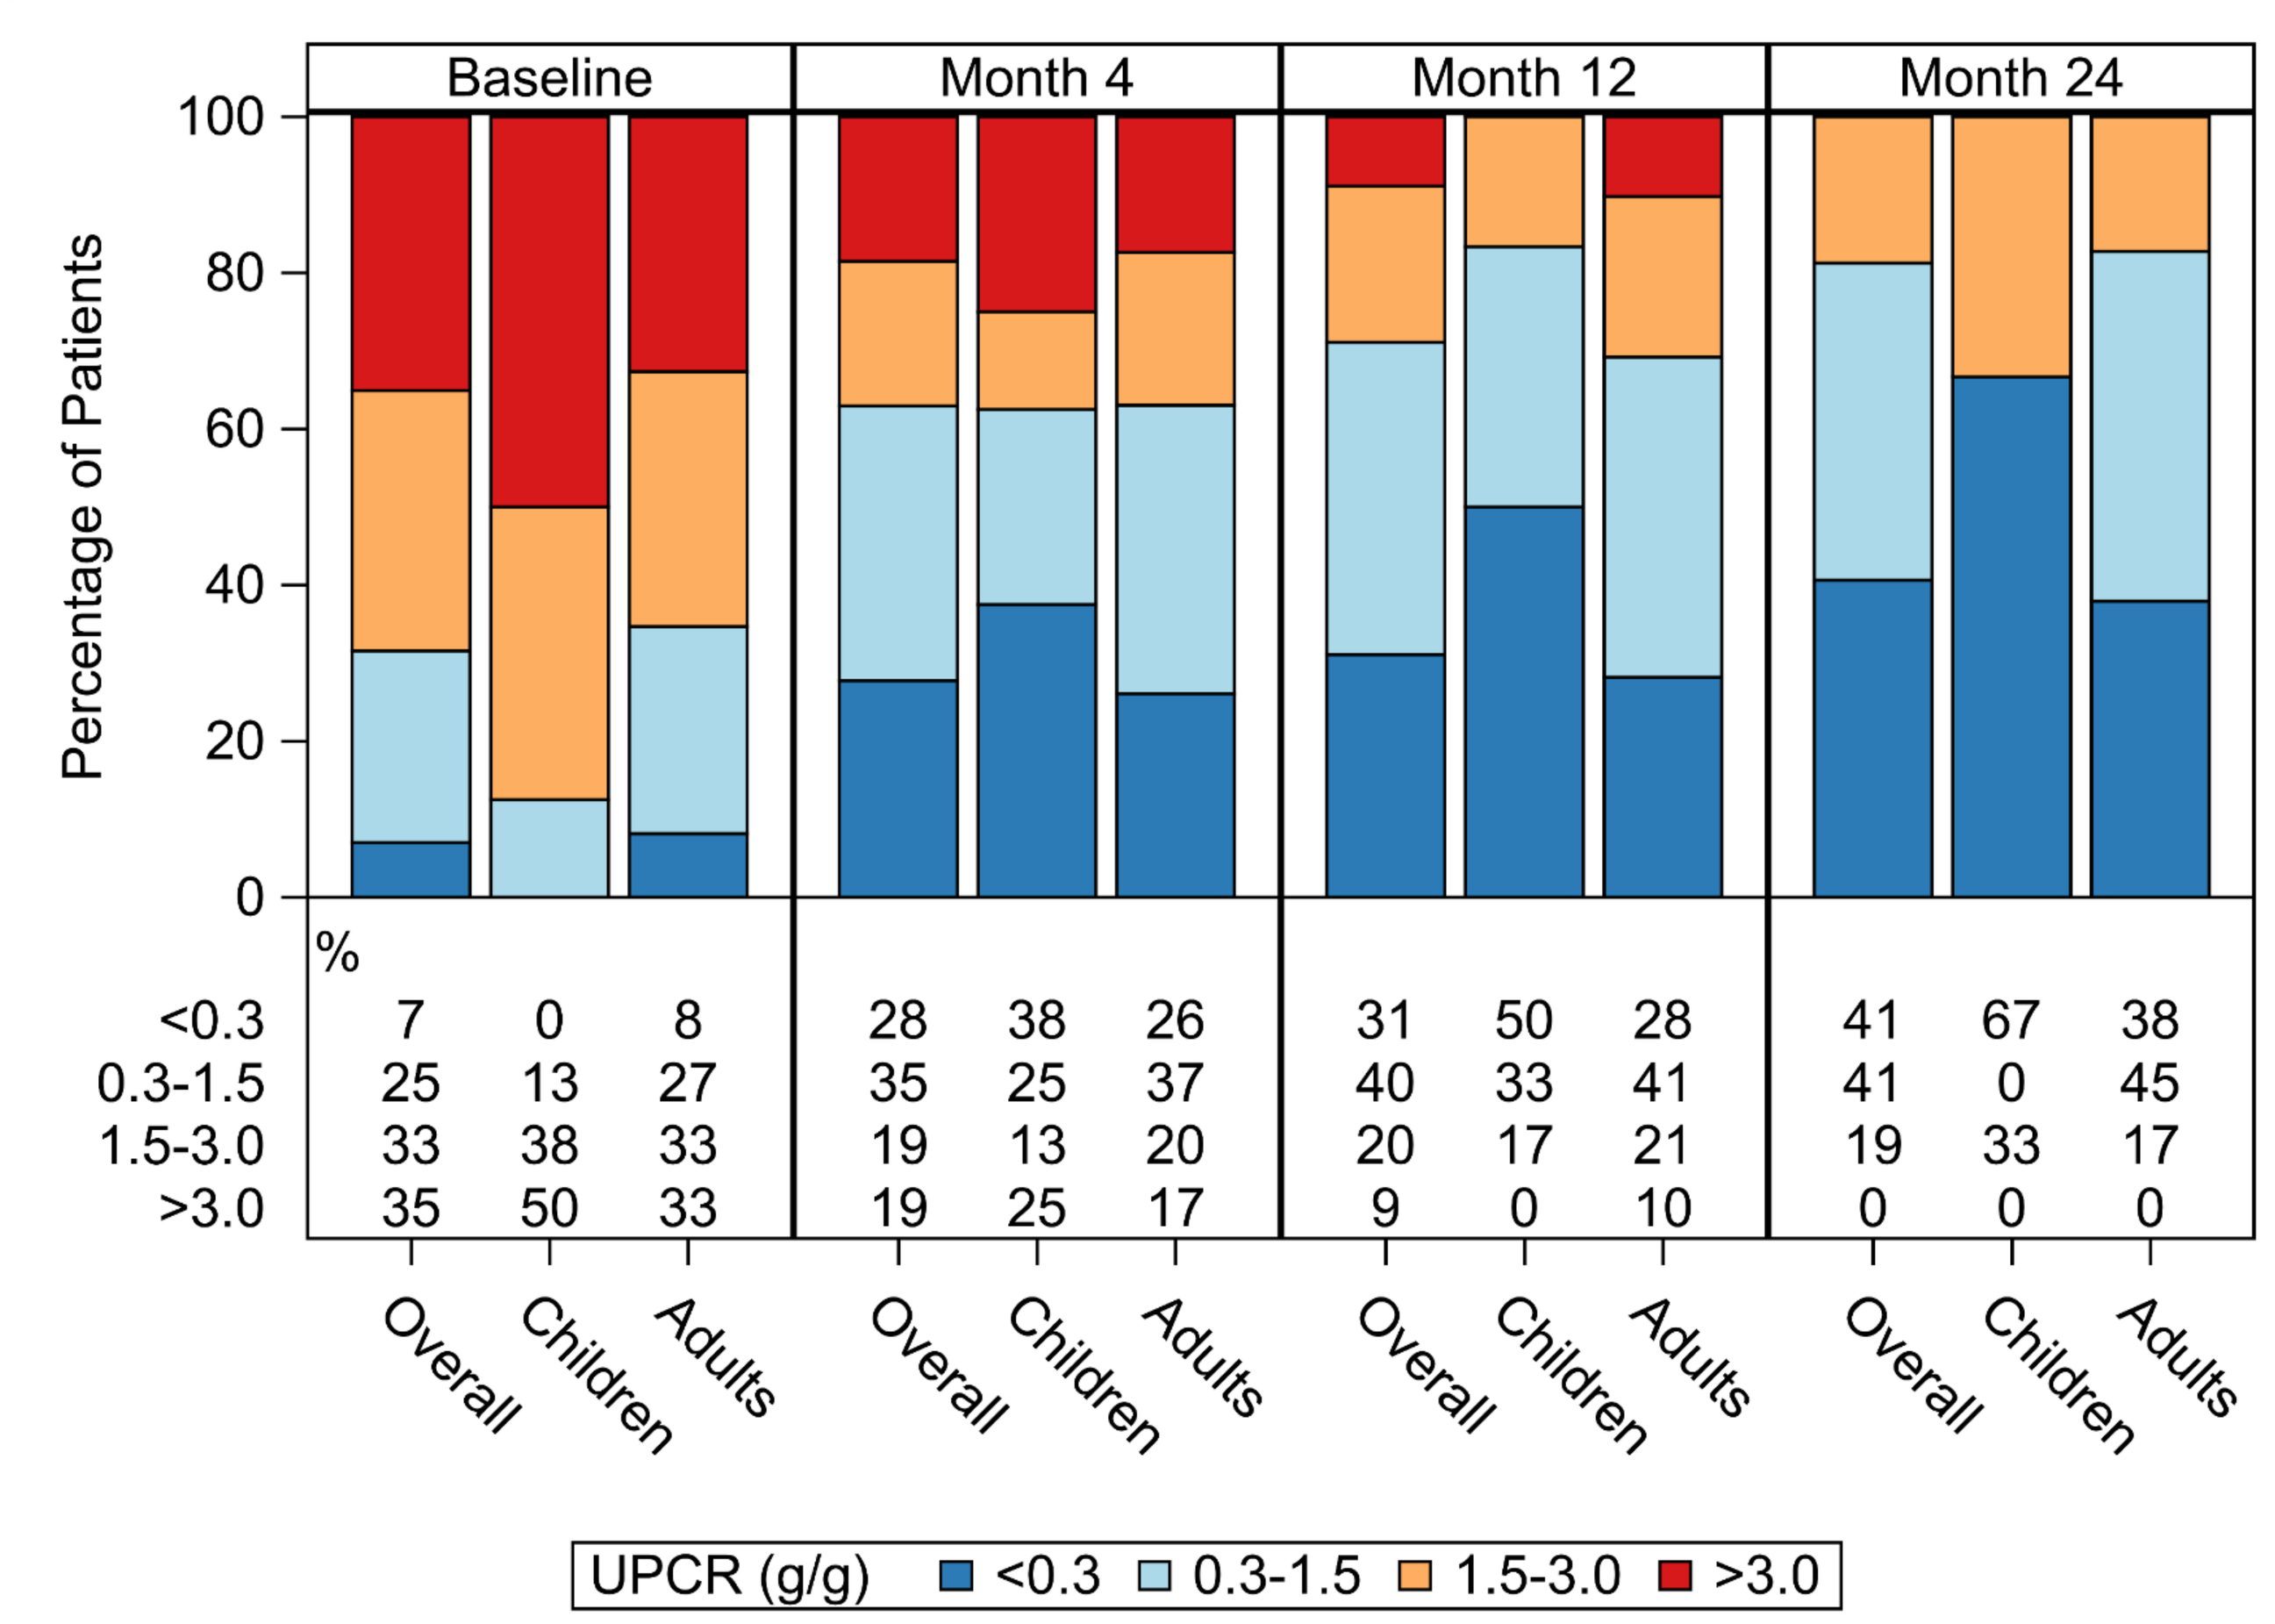
Baseline
Month 4
Month 12
Month 24
Percentage of Patients
100
80
60
40
20
0
%
<0.3 7 0 8 28 38 26 31 50 28 41 67 38
0.3-1.5 25 13 27 35 25 37 40 33 41 41 0 45
1.5-3.0 33 38 33 19 13 20 20 17 21 19 33 17
>3.0 35 50 33 19 25 17 9 0 10 0 0 0
Overall
Children
Adults
UPCR (g/g) <0.3 0.3-1.5 1.5-3.0 >3.0

Figure 2: K-M survival for composite of 40% eGFR decline or ESKD, by diagnosis (FSGS and IgAN) and by lowest proteinuria achieved in 12 months follow-up post-biopsy

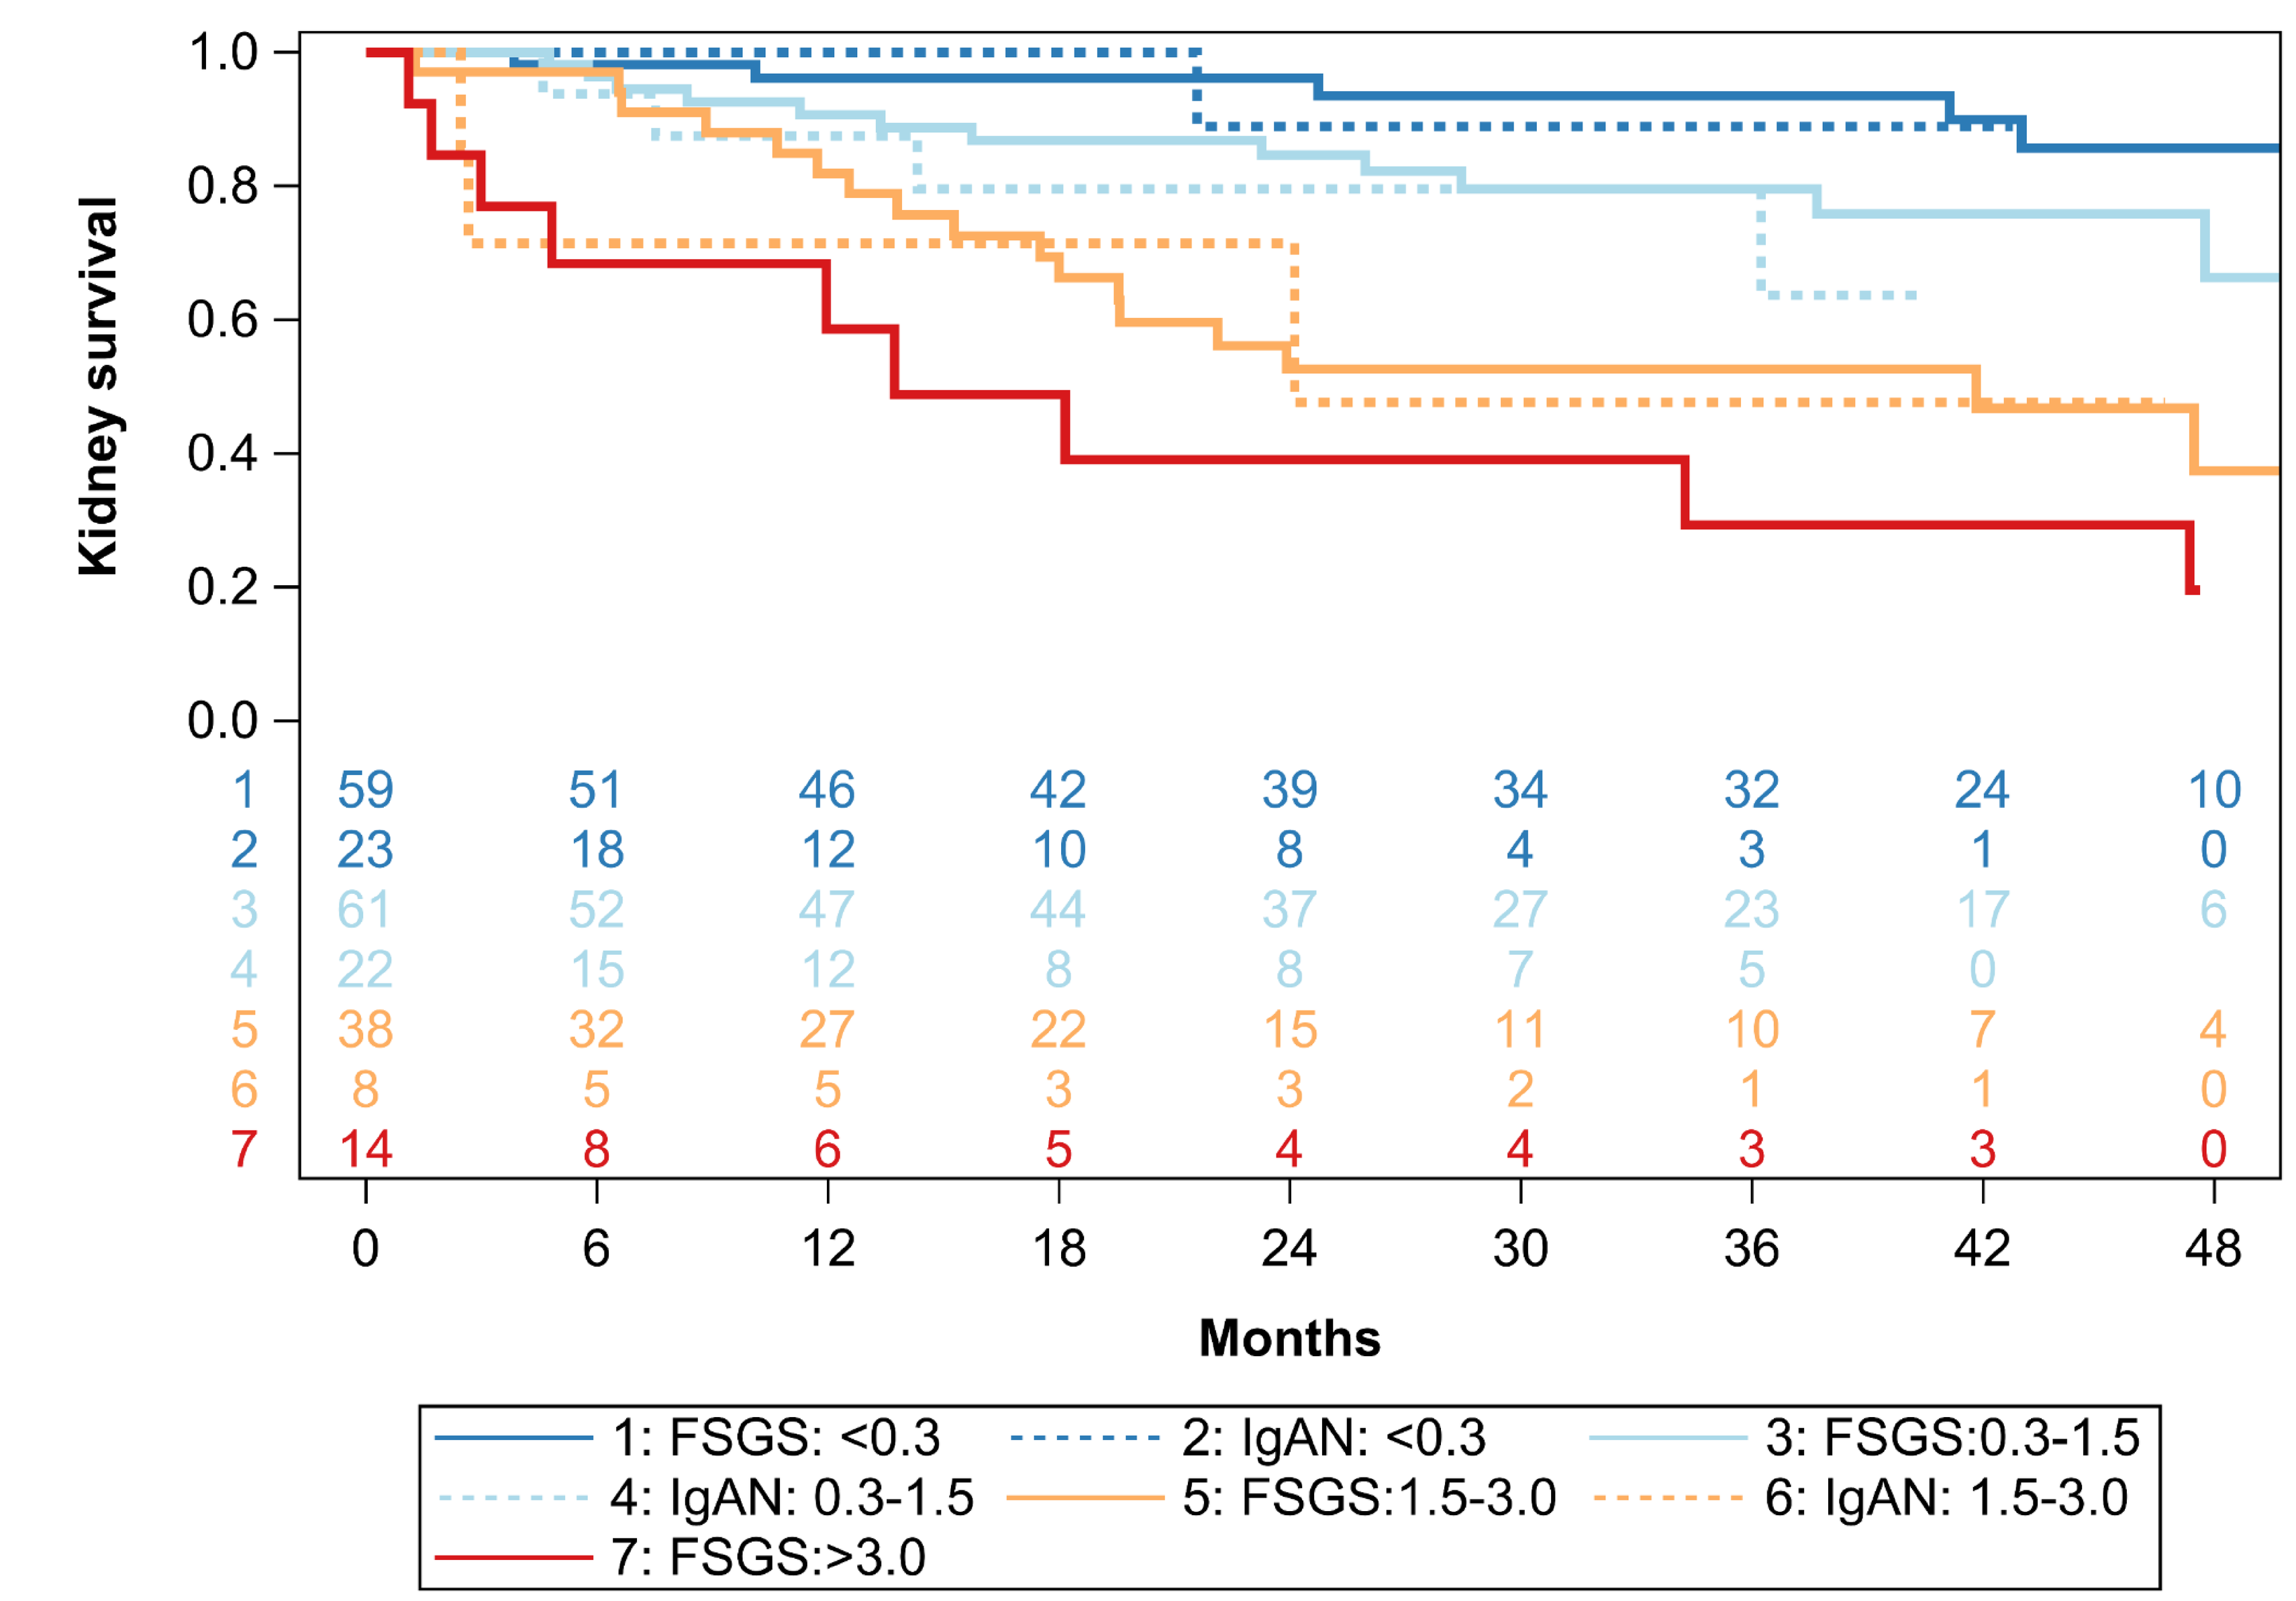
Kidney survival
1.0
0.8
0.6
0.4
0.2
0.0
1 59 51 46 42 39 34 32 24 10
2 23 18 12 10 8 4 3 1 0
3 61 52 47 44 37 27 23 17 6
4 22 15 12 8 8 7 5 0
5 38 32 27 22 15 11 10 7 4
6 8 5 5 3 3 2 1 1 0
7 14 8 6 5 4 4 3 3 0
0 6 12 18 24 30 36 42 48
Months
1: FSGS: <0.3
2: IgAN: <0.3
3: FSGS:0.3-1.5
4: IgAN: 0.3-1.5
5: FSGS:1.5-3.0
6: IgAN: 1.5-3.0
7: FSGS:>3.0

- 211 FSGS and 58 IgAN patients were included (Table 1). FSGS participants had higher baseline eGFR, UPCR and obesity rates, but similar age and blood pressure to IgAN participants.
- Relative change in proteinuria was -42% vs -39% by 12mo and -42% vs -63% by 24mo in FSGS and IgAN, respectively, but a significant proportion with either diagnosis did not achieve complete remission (Fig 1). Only 9% of IgAN participants had proteinuria >3.0 g/g at 12 months.
- Associations between lowest proteinuria achieved within 12mo and longer kidney survival time were similar across diseases (Fig 2).

Summary/Conclusions

SUMMARY:

- Lower proteinuria within 12 months after biopsy was associated with similar improvement in kidney survival in FSGS and IgAN.
- A substantial proportion of patients in this real-world cohort have high proteinuria at 24 months - highlighting unmet need for new treatments.

Abstract #: TH-PO622

Correspondence: lmariani@umich.edu

Acknowledgements: The Nephrotic Syndrome Study Network (NEPTUNE) is funded under U54DK083912 as a collaboration between NCATS and NIDDK. This analysis was funded by Travere Therapeutics

Presented at: American Society of Nephrology (ASN) Kidney Week 2023; November 1-5, 2023; Philadelphia, PA