Poster SA-PO901

Sparsentan as First-Line Treatment of Incident Patients With IgA Nephropathy: Preliminary Findings From the SPARTAN Trial

Chee Kay Cheung,[1] Stephanie Moody,[2] Neeraj Dhaun,[3] Siân Griffin,[4] Alexandra Howson,[1] Radko Komers,[2] Alex Mercer,[5] Matthew Sayer,[3] Smeeta Sinha,[6] Lisa Willcocks,[7] Jonathan Barratt[1]

[1]University of Leicester & Leicester General Hospital, Leicester, UK; [2]Travere Therapeutics, Inc., San Diego, CA, USA; [3]Department of Renal Medicine, Royal Infirmary of Edinburgh, Edinburgh, UK; [4]Department of Nephrology and Transplantation, University Hospital of Wales, Cardiff, UK; [5]JAMCO Pharma Consulting, Stockholm, Sweden; [6]Department of Renal Medicine, Salford Royal Hospital Northern Care Alliance NHS Foundation Trust, Salford, UK; [7]Department of Renal Medicine, Addenbrooke's Hospital, Cambridge University Hospitals, Cambridge, UK

INTRODUCTION

- Sparsentan is a novel dual endothelin and angiotensin receptor antagonist (DEARA) that was granted accelerated approval in the US for the treatment of adults with immunoglobulin A nephropathy (IgAN) at risk of disease progression, based on improvements in proteinuria shown in the ongoing phase 3 PROTECT study[1,2]
- Sparsentan has been studied in patients with previous maximized renin-angiotensin system inhibitors,[2] but the effect in newly diagnosed, treatment-naive patients remains unknown
- SPARTAN (NCT04663204) is a phase 2, open-label, single-arm trial investigating the safety, efficacy, and mechanistic actions of sparsentan as first-line therapy in patients newly diagnosed with IgAN[3]

Objective

- Here we report preliminary clinical findings over the first 36 weeks of treatment with sparsentan from SPARTAN

METHODS

- The SPARTAN study is being conducted at 5 participating sites in the UK (**Figure 1**)

Figure 1. SPARTAN Study Design and Patient Assessment Schedule

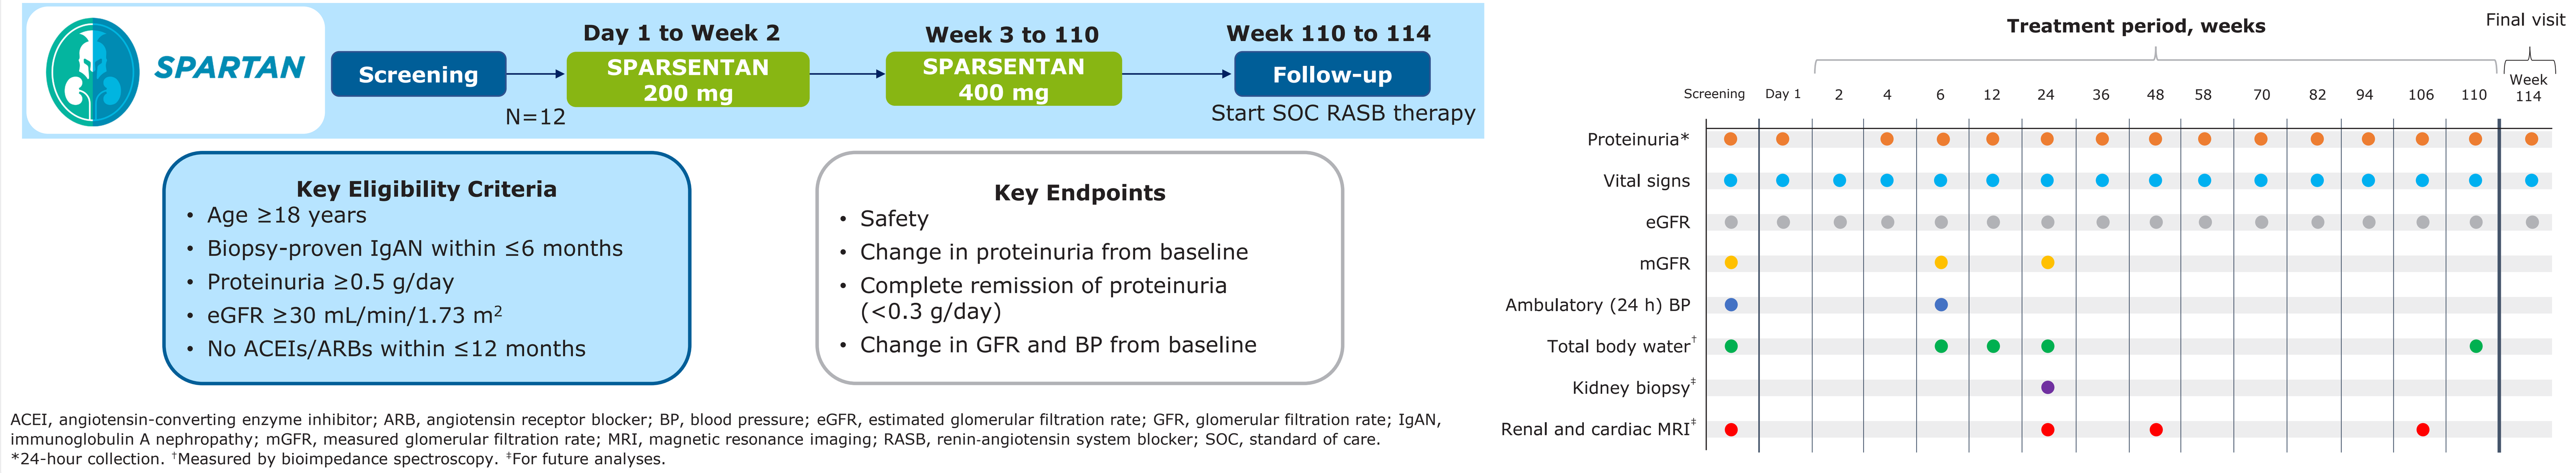

SPARTAN
Screening
N=12
Day 1 to Week 2
SPARSENTAN 200 mg
Week 3 to 110
SPARSENTAN 400 mg
Week 110 to 114
Follow-up
Start SOC RASB therapy
Key Eligibility Criteria
• Age ≥18 years
• Biopsy-proven IgAN within ≤6 months
• Proteinuria ≥0.5 g/day
• eGFR ≥30 mL/min/1.73 m²
• No ACEIs/ARBs within ≤12 months
Key Endpoints
• Safety
• Change in proteinuria from baseline
• Complete remission of proteinuria (<0.3 g/day)
• Change in GFR and BP from baseline
Treatment period, weeks
Final visit
Screening Day 1 2 4 6 12 24 36 48 58 70 82 94 106 110 Week 114
Proteinuria*
Vital signs
eGFR
mGFR
Ambulatory (24 h) BP
Total body water†
Kidney biopsy‡
Renal and cardiac MRI‡

ACEI, angiotensin-converting enzyme inhibitor; ARB, angiotensin receptor blocker; BP, blood pressure; eGFR, estimated glomerular filtration rate; GFR, glomerular filtration rate; IgAN, immunoglobulin A nephropathy; mGFR, measured glomerular filtration rate; MRI, magnetic resonance imaging; RASB, renin-angiotensin system blocker; SOC, standard of care.
*24-hour collection. †Measured by bioimpedance spectroscopy. ‡For future analyses.

RESULTS

Patient Population

- As of the data cutoff (September 26, 2023), 12 patients received sparsentan and participated in the study for ≥6 weeks (**Figures 2** and **3**)

Figure 2. Patient Demographics and Baseline Characteristics

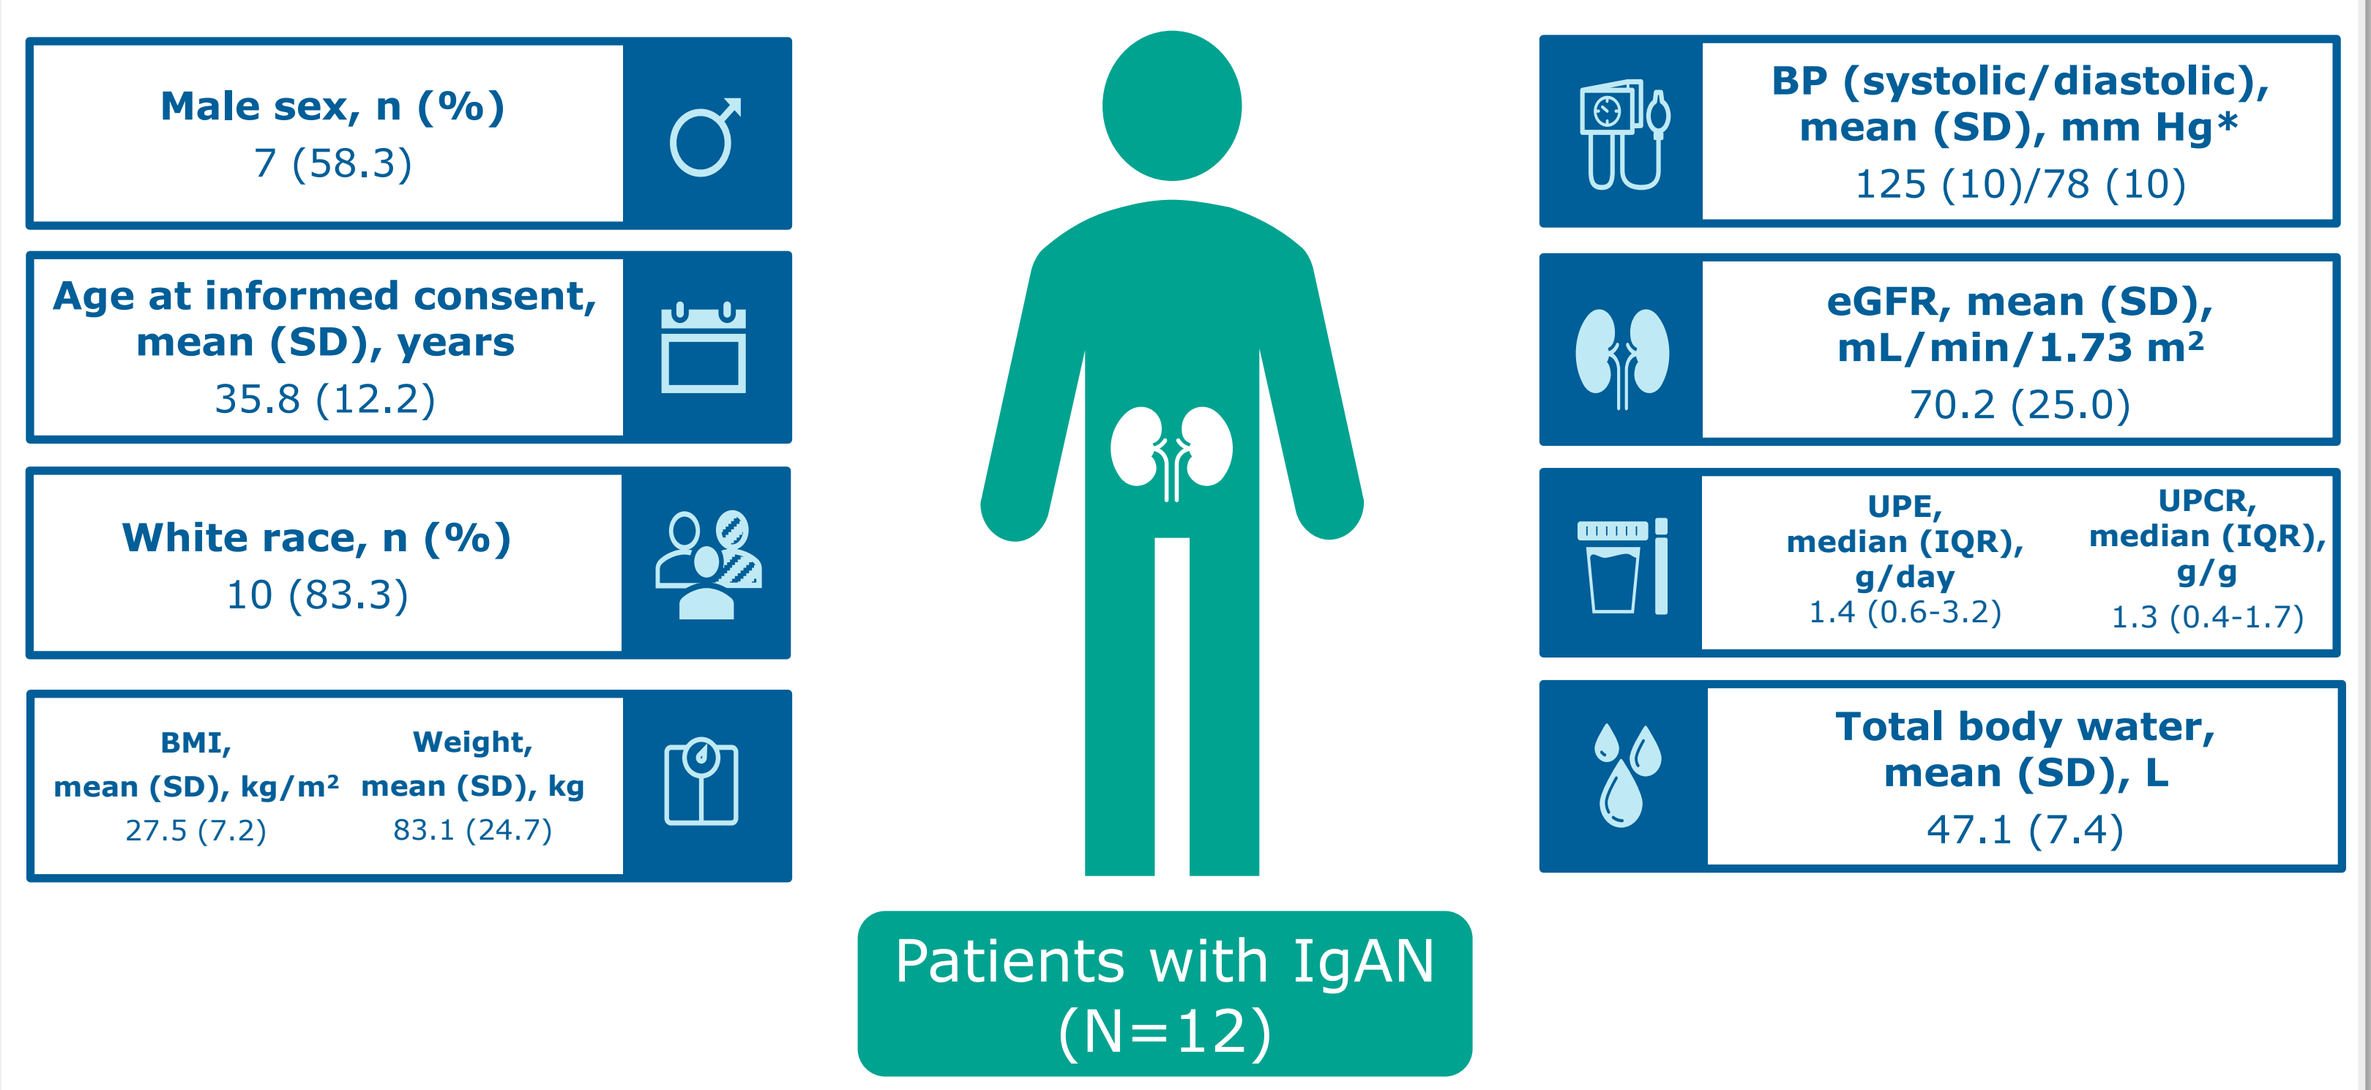

Male sex, n (%)
7 (58.3)
Age at informed consent, mean (SD), years
35.8 (12.2)
White race, n (%)
10 (83.3)
BMI, mean (SD), kg/m²
27.5 (7.2)
Weight, mean (SD), kg
83.1 (24.7)
BP (systolic/diastolic), mean (SD), mm Hg*
125 (10)/78 (10)
eGFR, mean (SD), mL/min/1.73 m²
70.2 (25.0)
UPE, median (IQR), g/day
1.4 (0.6-3.2)
UPCR, median (IQR), g/g
1.3 (0.4-1.7)
Total body water, mean (SD), L
47.1 (7.4)
Patients with IgAN (N=12)

BMI, body mass index; BP, blood pressure; eGFR, estimated glomerular filtration rate; IgAN, immunoglobulin A nephropathy; UPCR, urine protein-to-creatinine ratio; UPE, urine protein excretion.
*Office BP.

Figure 3. Patient Progress in the Study Up to Week 36

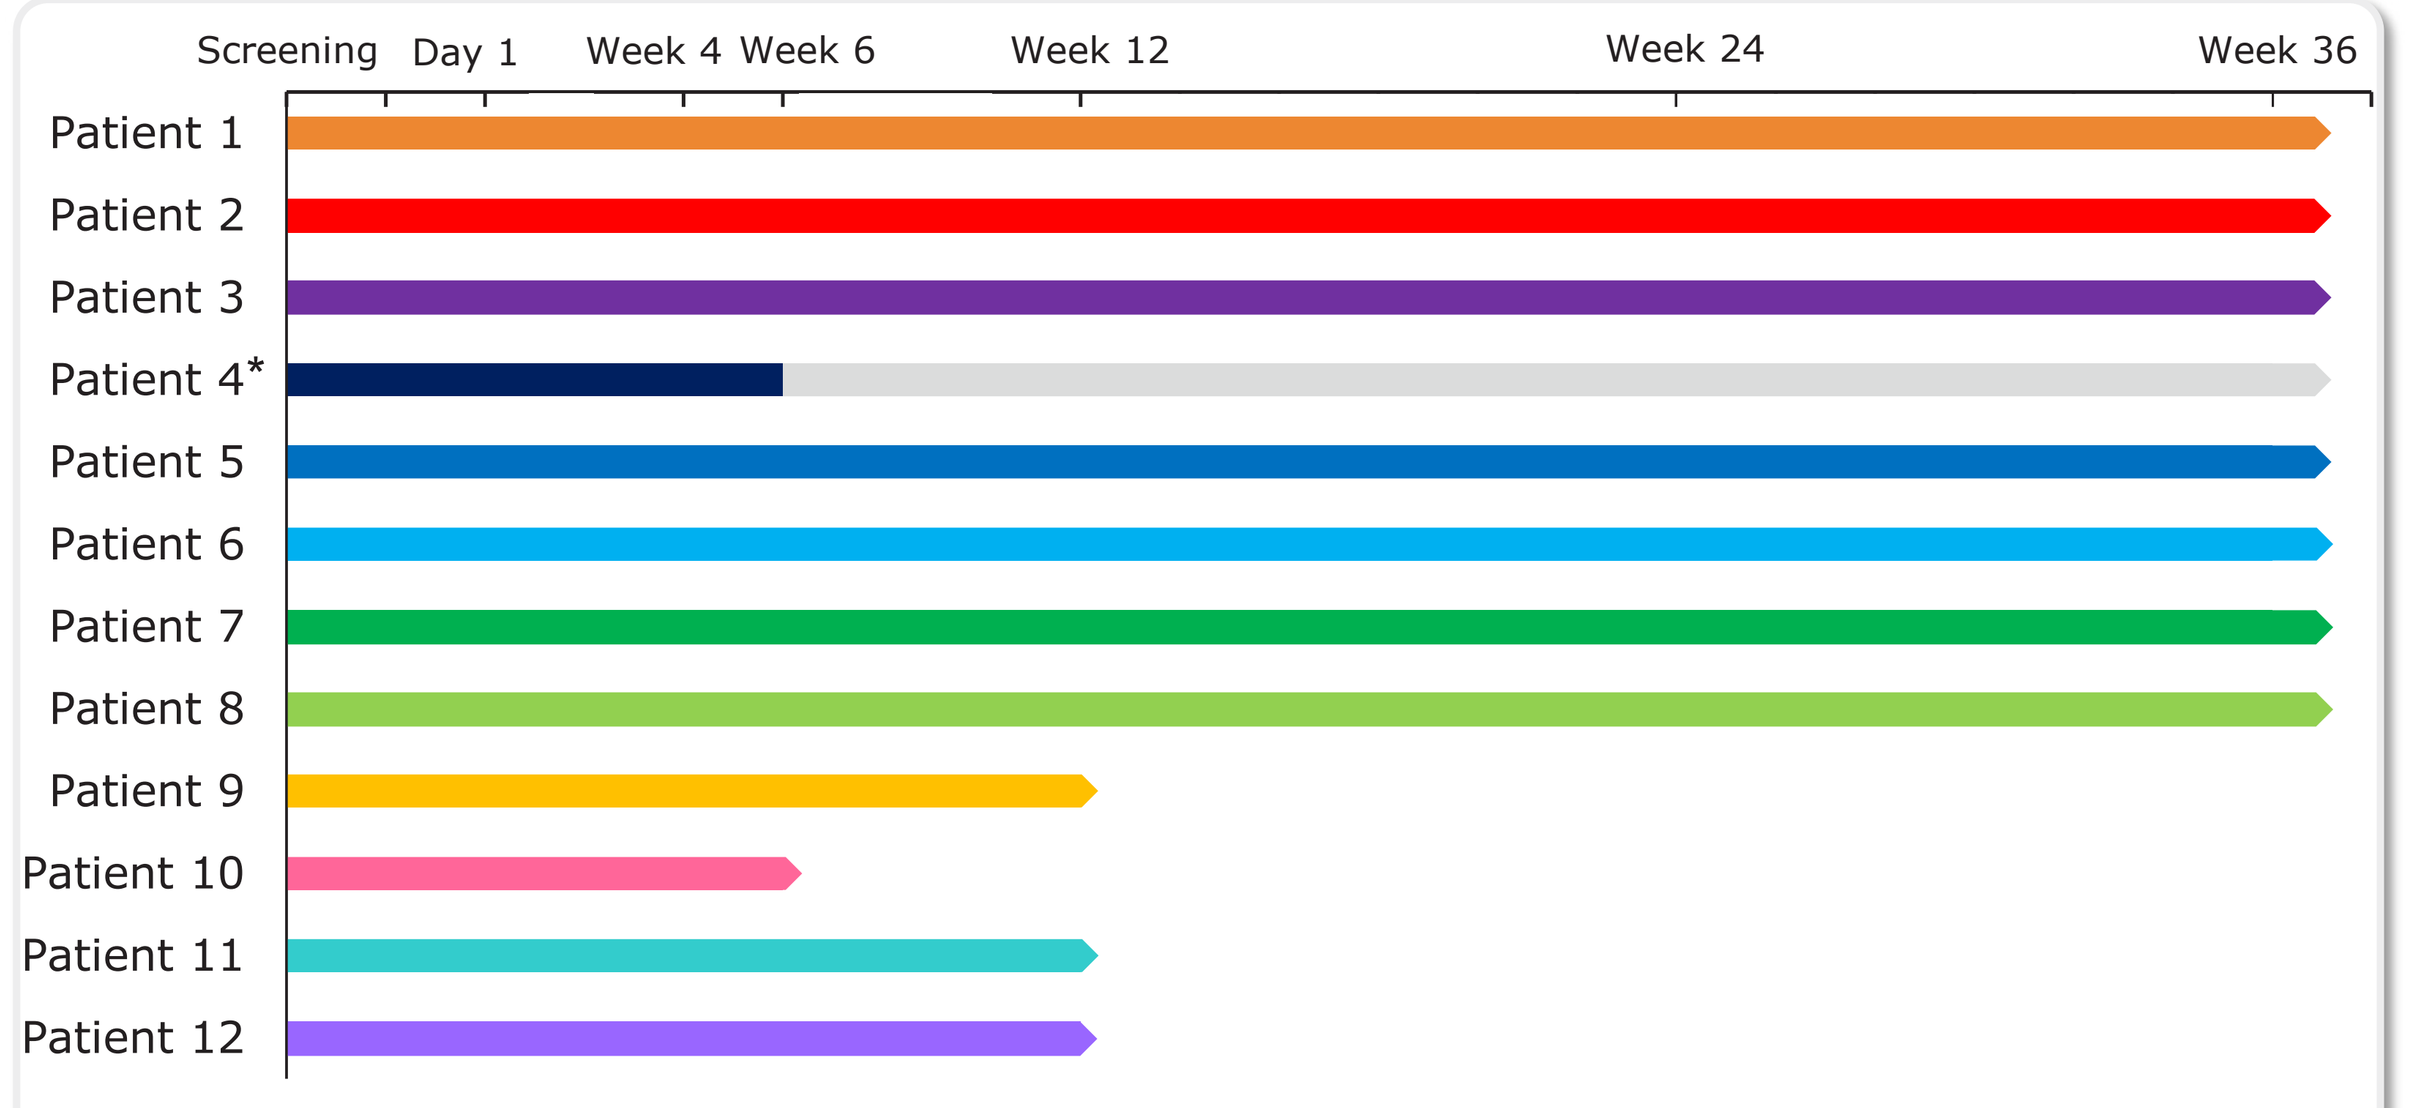

Screening
Day 1
Week 4
Week 6
Week 12
Week 24
Week 36
Patient 1
Patient 2
Patient 3
Patient 4*
Patient 5
Patient 6
Patient 7
Patient 8
Patient 9
Patient 10
Patient 11
Patient 12

*Patient 4 stopped treatment after the week 6 visit but continued in the study off-treatment to the week 24 visit. All analyses in this study are calculated for patients receiving treatment, and data from patient 4 are excluded after week 6.

Proteinuria

- Reduction in proteinuria at week 4 was ≈60% from baseline, which was sustained over 36 weeks of sparsentan treatment (**Figure 4**)
- Among the 4 patients with protein excretion of >2 g/day at baseline, 3 had proteinuria reductions of ≥75% at any time during the first 36 weeks of treatment (**Figure 5**)
- 67% (8/12) of patients achieved complete remission (<0.3 g/day) at any time during the 36-week treatment period

Figure 4. Proteinuria Change (UPCR) From Baseline

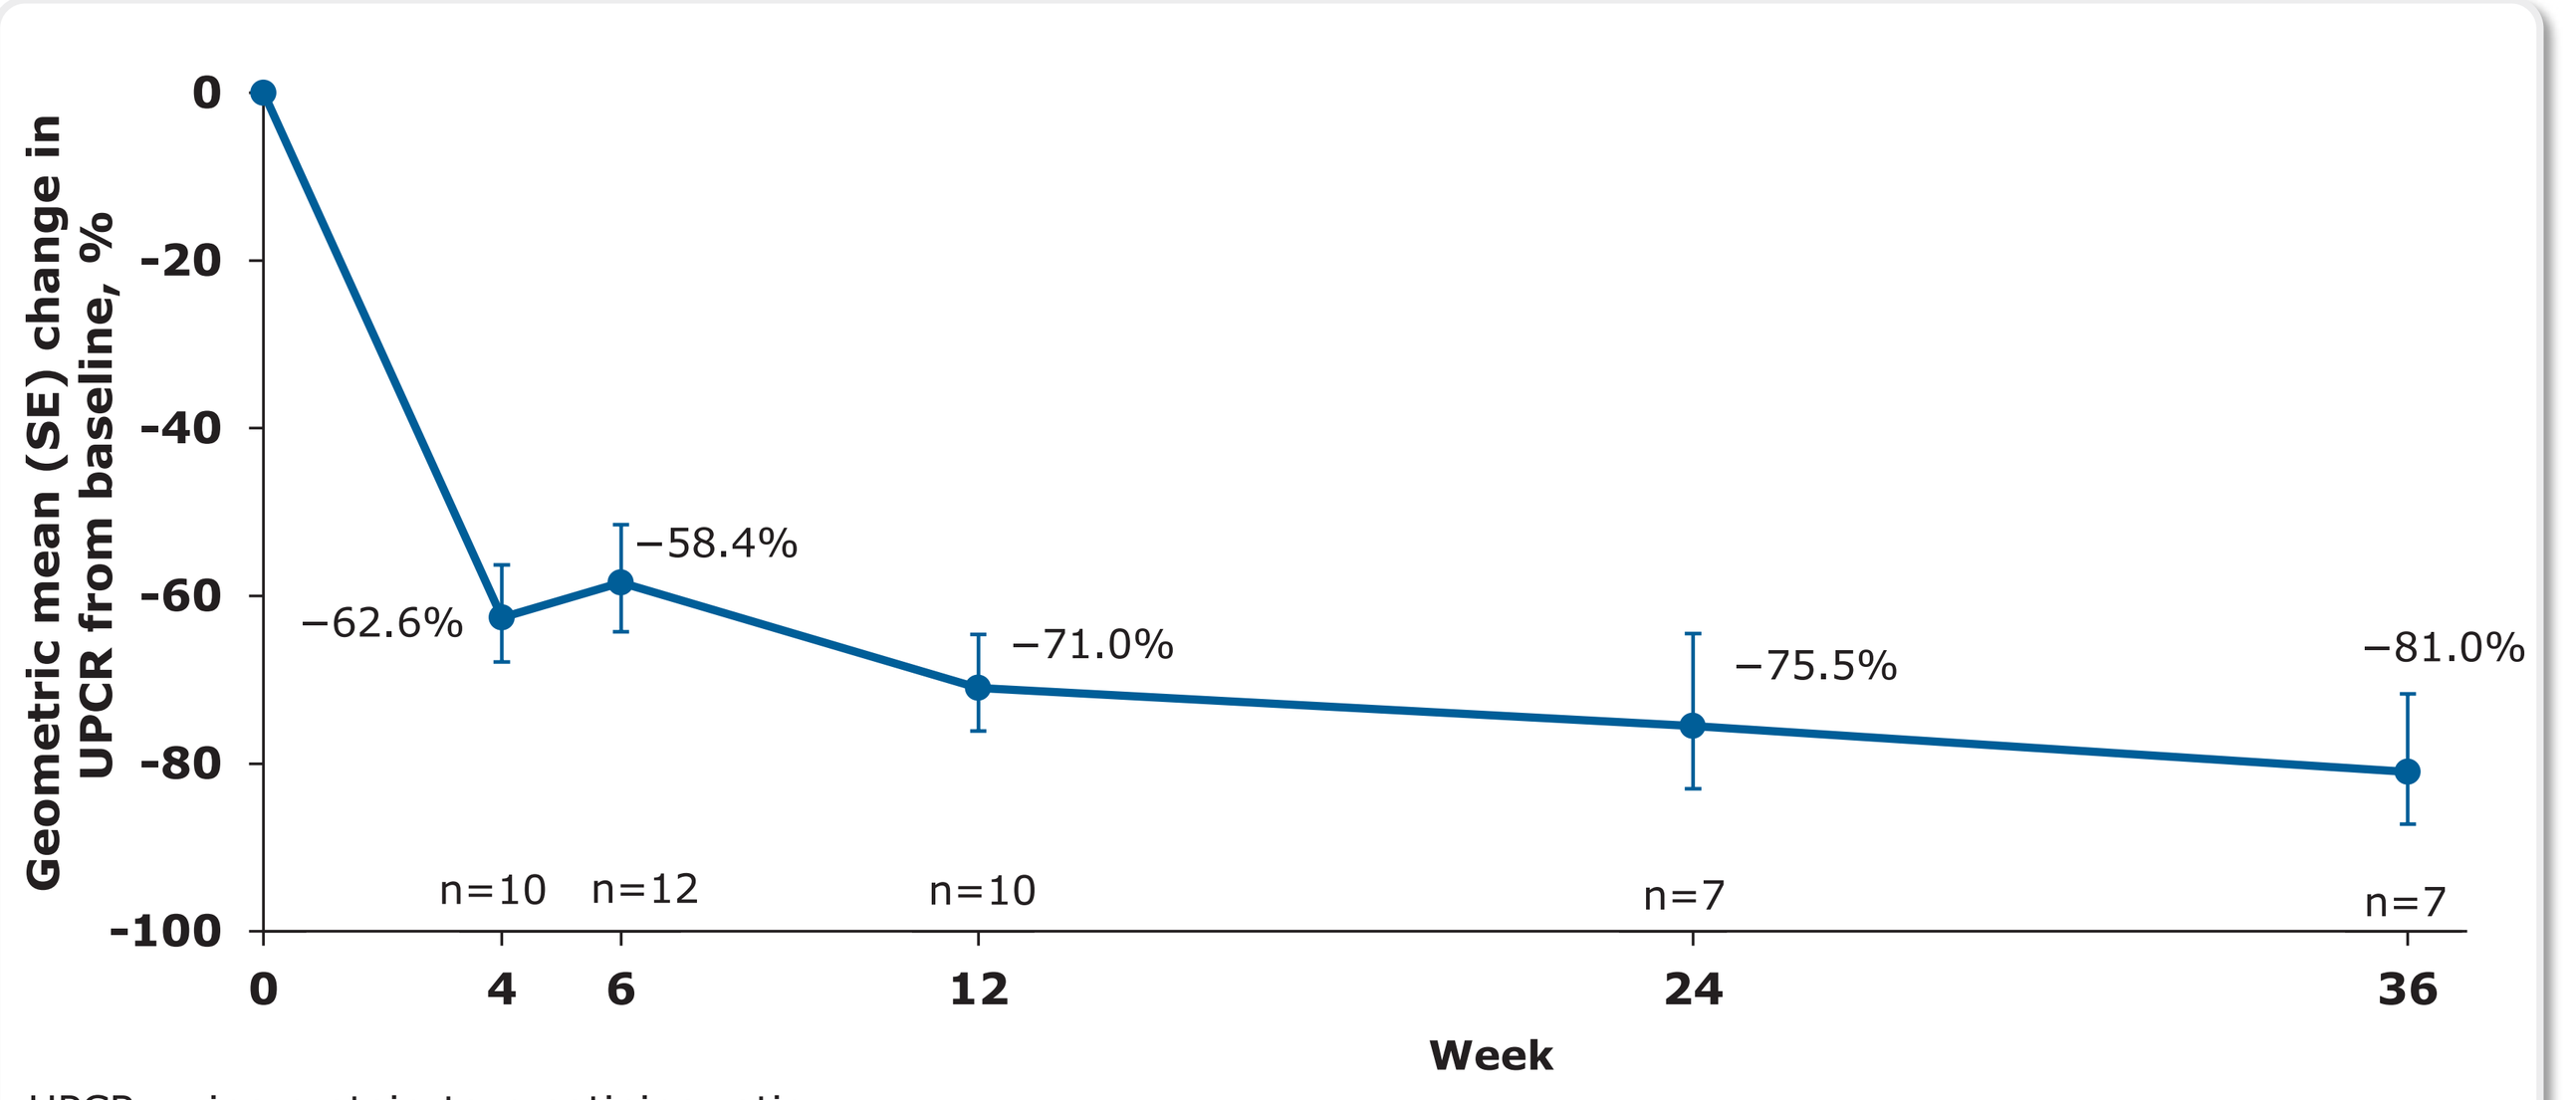

Geometric mean (SE) change in UPCR from baseline, %
0
-20
-40
-60
-80
-100
−62.6%
−58.4%
−71.0%
−75.5%
−81.0%
n=10
n=12
n=10
n=7
n=7
0
4
6
12
24
36
Week

UPCR, urine protein-to-creatinine ratio.

Figure 5. Proteinuria Per Individual Patient

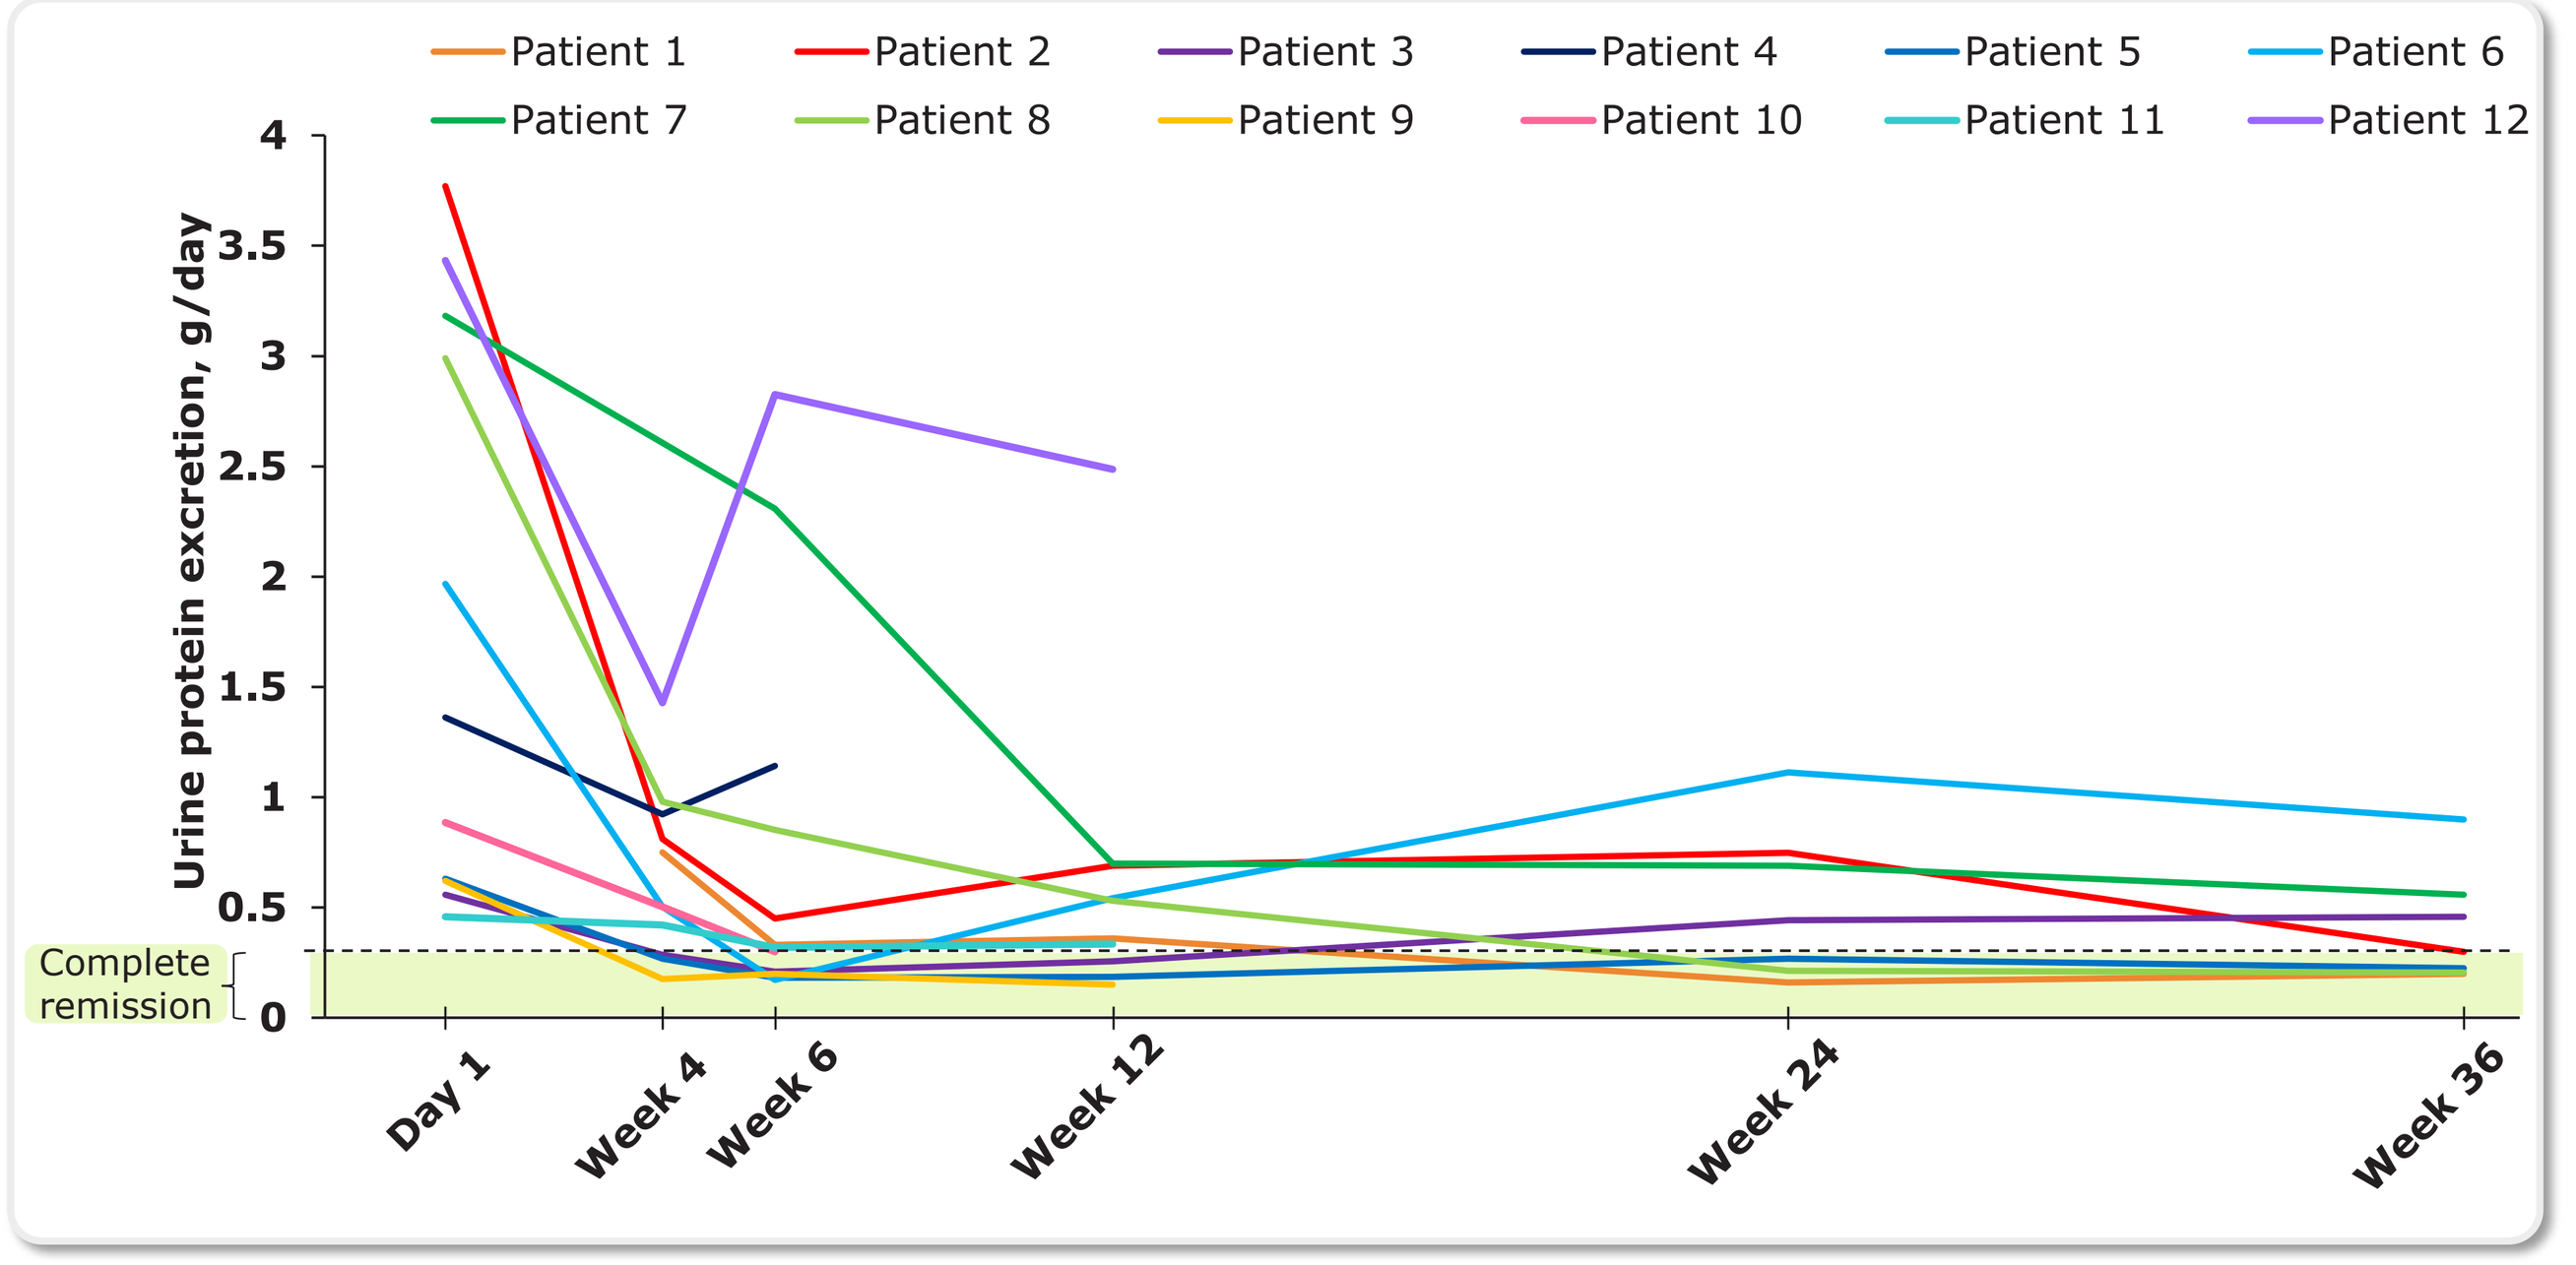

Patient 1
Patient 2
Patient 3
Patient 4
Patient 5
Patient 6
Patient 7
Patient 8
Patient 9
Patient 10
Patient 11
Patient 12
Urine protein excretion, g/day
4
3.5
3
2.5
2
1.5
1
0.5
0
Complete remission
Day 1
Week 4
Week 6
Week 12
Week 24
Week 36

Estimated Glomerular Filtration Rate

- Estimated glomerular filtration rate changes were relatively stable over 36 weeks of treatment with sparsentan (**Figure 6**)

Figure 6. Mean eGFR Change From Baseline

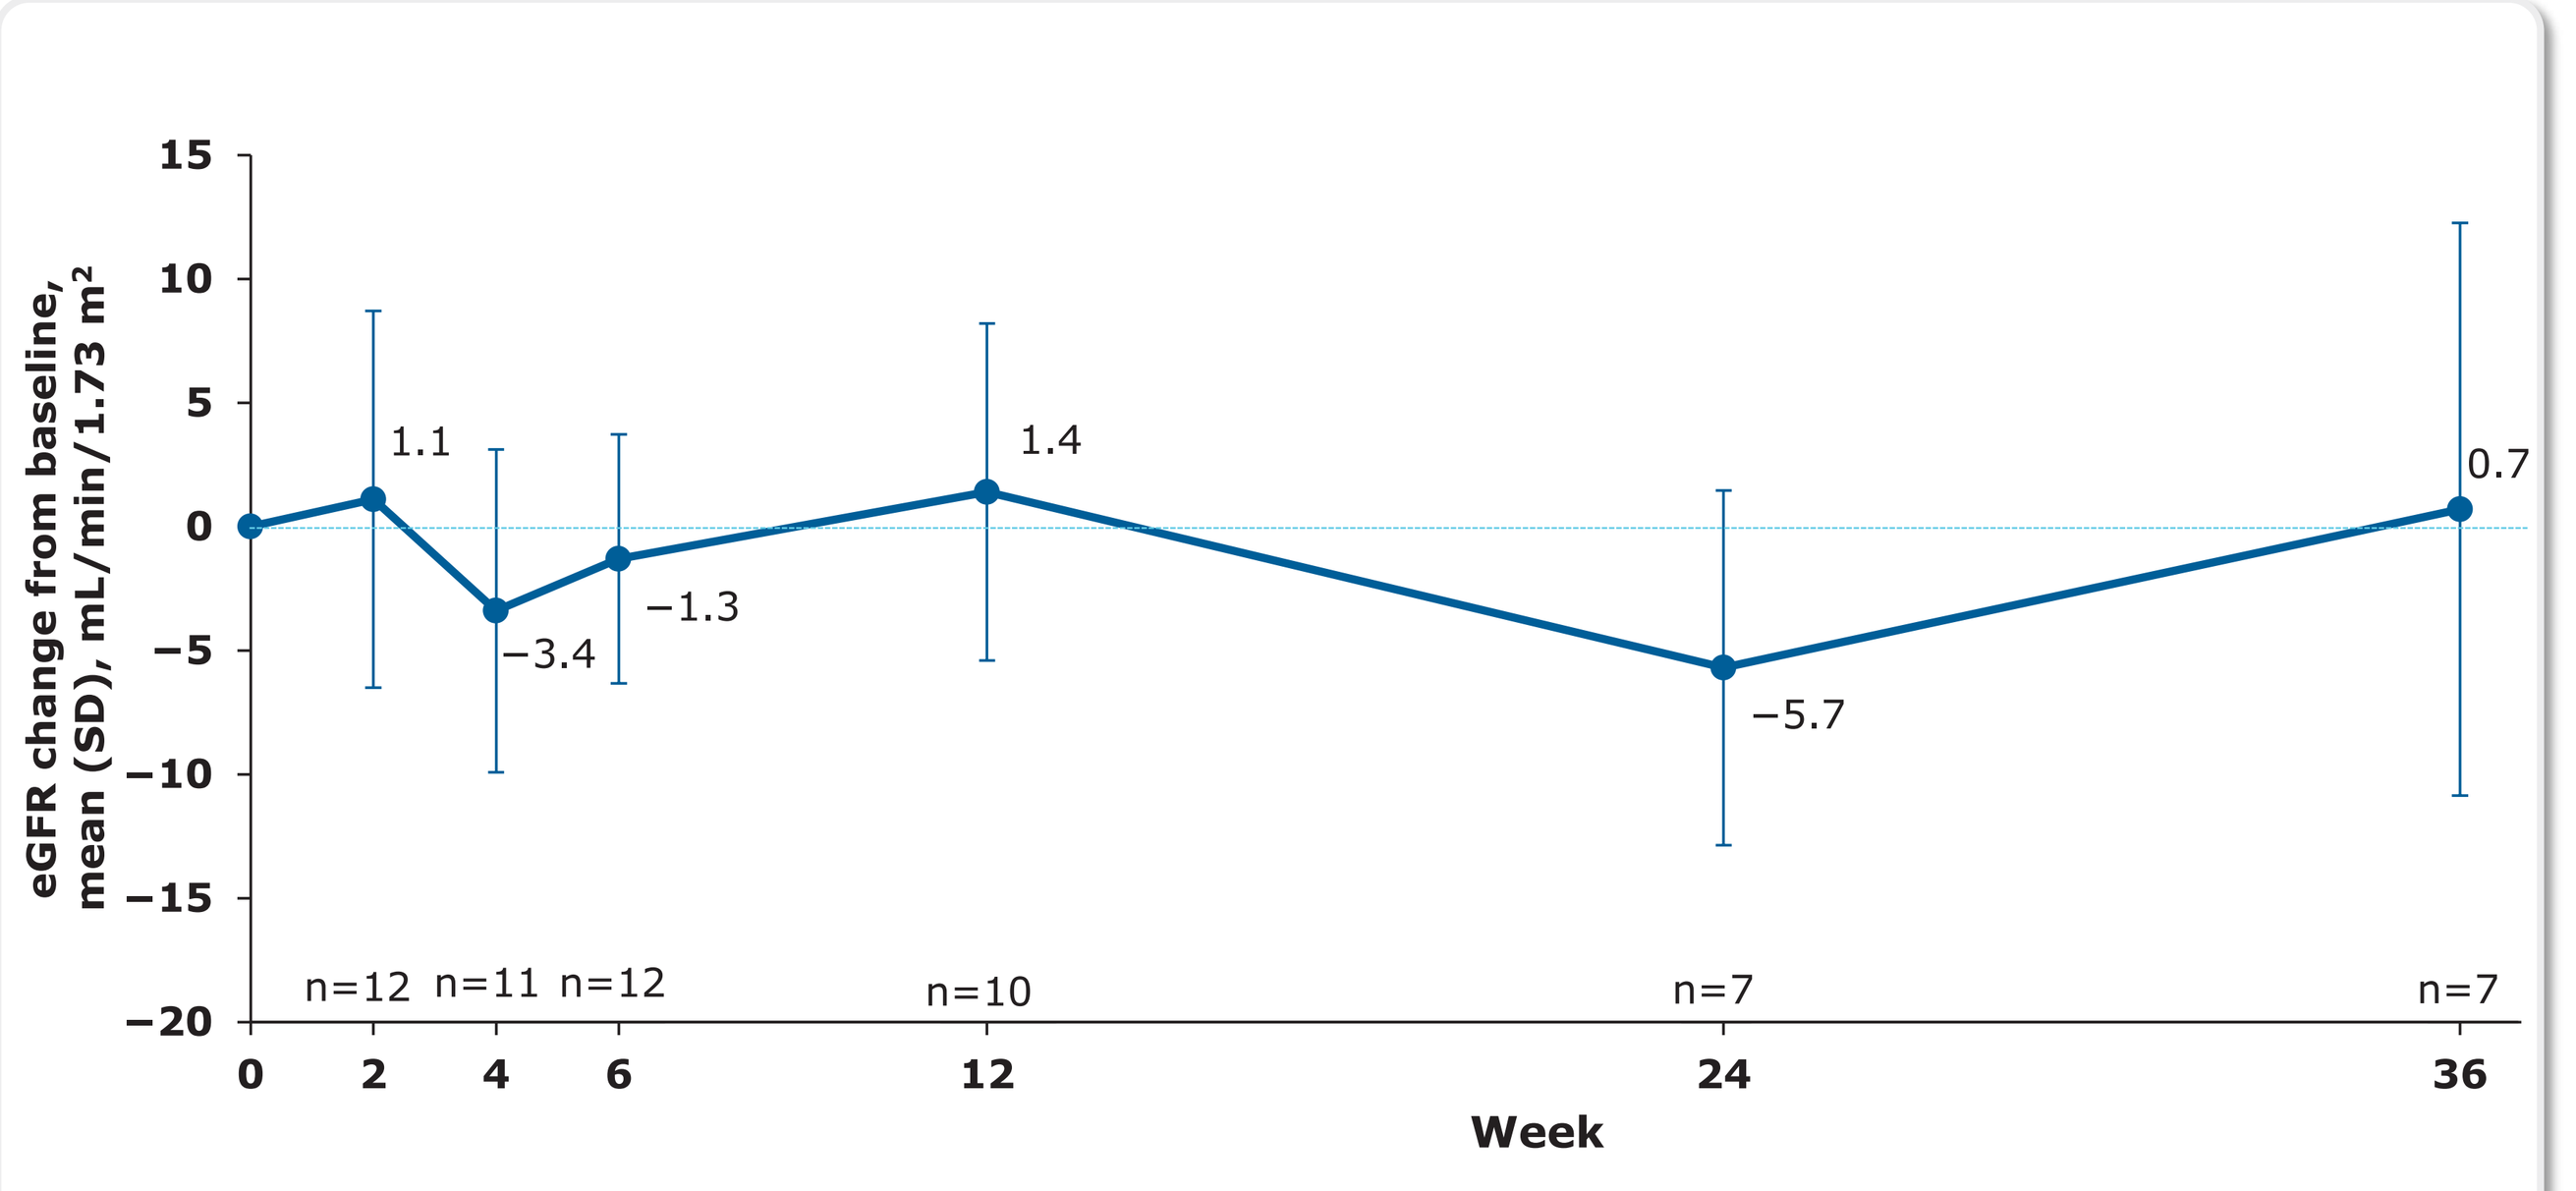

eGFR change from baseline, mean (SD), mL/min/1.73 m²
15
10
5
0
−5
−10
−15
−20
1.1
−3.4
−1.3
1.4
−5.7
0.7
n=12
n=11
n=12
n=10
n=7
n=7
0
2
4
6
12
24
36
Week

eGFR, estimated glomerular filtration rate.

Blood Pressure

- After an initial decrease, blood pressure (BP) remained stable during the rest of the follow-up (**Figure 7**)
- Office and ambulatory BP showed similar measurements of systolic BP and diastolic BP at baseline and week 6, and ambulatory BP showed a slightly greater change from baseline than office BP (**Figure 8**)

Figure 7. Mean Office BP

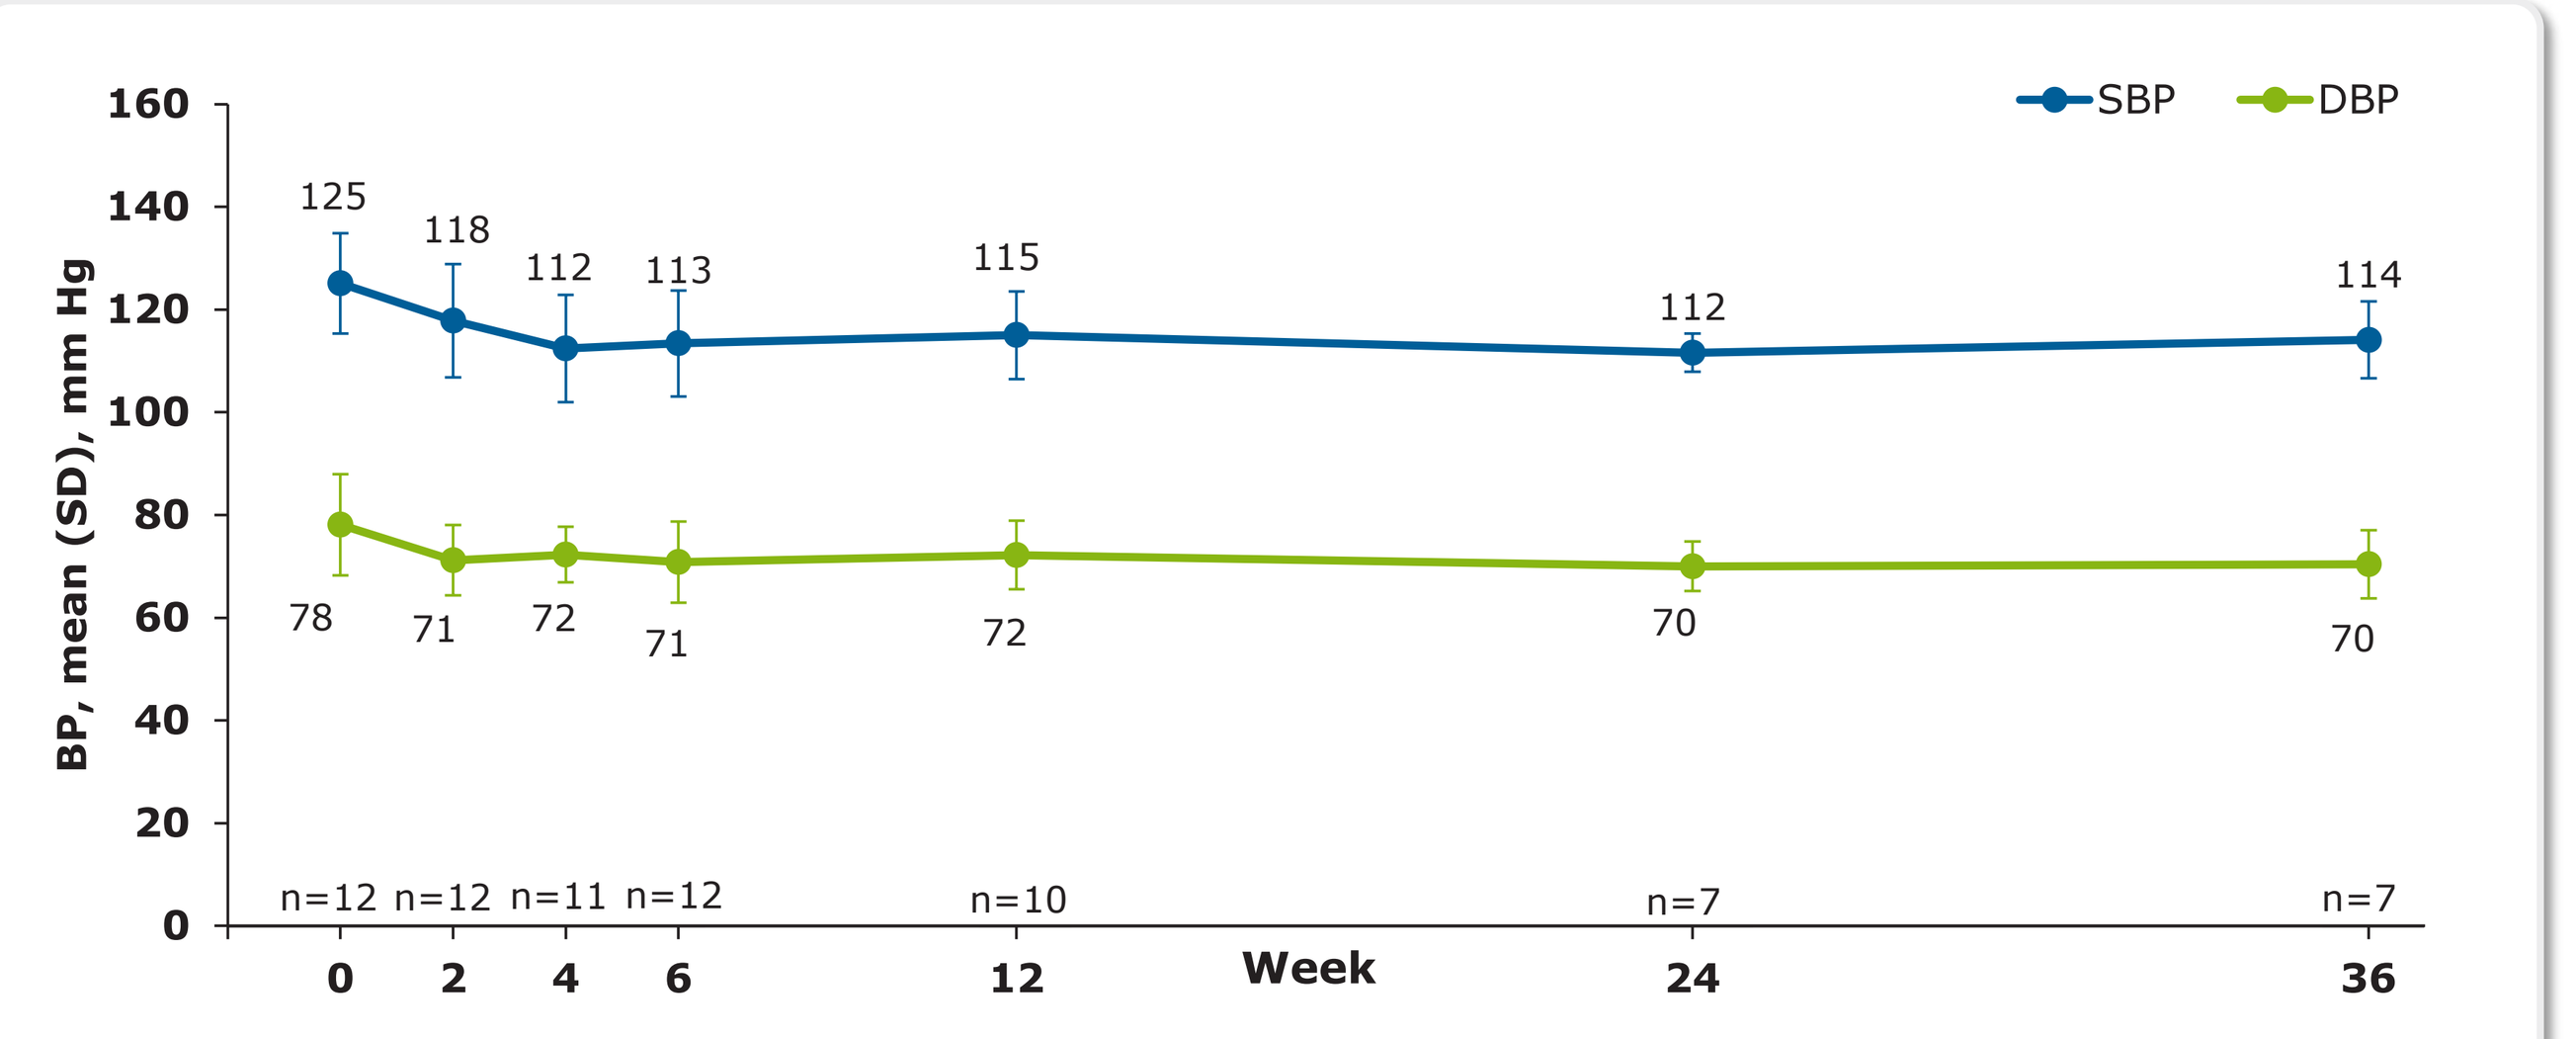

SBP
DBP
BP, mean (SD), mm Hg
160
140
120
100
80
60
40
20
0
125
118
112
113
115
112
114
78
71
72
71
72
70
70
n=12
n=12
n=11
n=12
n=10
n=7
n=7
0
2
4
6
12
Week
24
36

BP, blood pressure; DBP, diastolic blood pressure; SBP, systolic blood pressure.

Figure 8. Mean Office and Ambulatory BP at Baseline and Week 6

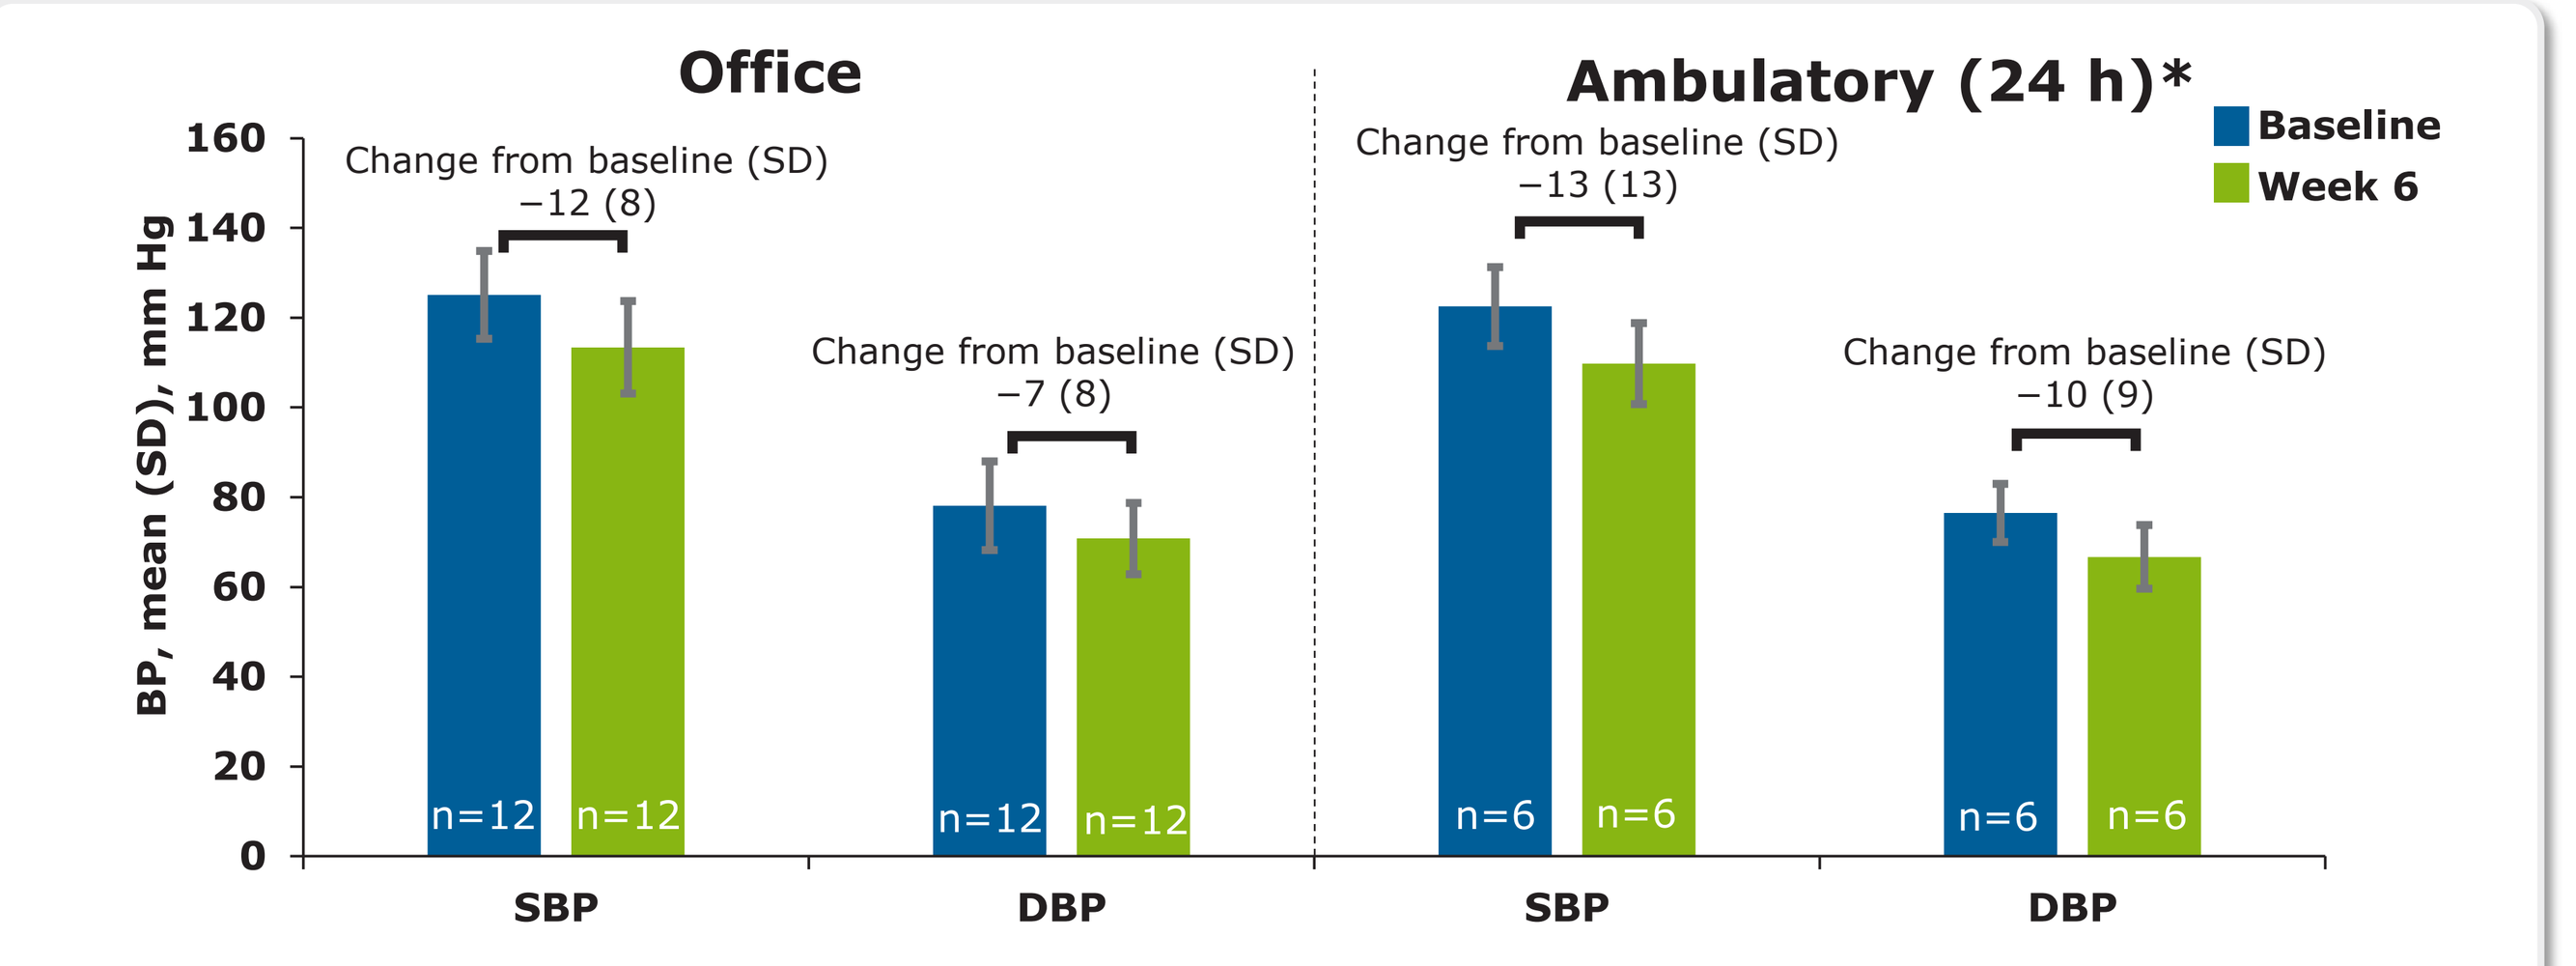

Office
Ambulatory (24 h)*
Baseline
Week 6
BP, mean (SD), mm Hg
160
140
120
100
80
60
40
20
0
Change from baseline (SD)
−12 (8)
Change from baseline (SD)
−7 (8)
Change from baseline (SD)
−13 (13)
Change from baseline (SD)
−10 (9)
n=12
n=12
n=12
n=12
n=6
n=6
n=6
n=6
SBP
DBP
SBP
DBP

BP, blood pressure; DBP, diastolic blood pressure; SBP, systolic blood pressure.
*Patients who had ambulatory BP data available at baseline and week 6.

Body Weight and Total Body Water

- Mean body weight changes showed minor fluctuations over 36 weeks (**Table 1**)
- Mean total body water change from baseline showed modest reductions during follow-up

Table 1. Mean Weight and Total Body Water Change From Baseline

Mean (SD) change from baseline	Week 2	Week 4	Week 6	Week 12	Week 24	Week 36
n	12	11	12	10	7	7
Weight, kg	−0.3 (0.7)	−0.2 (1.4)	0.3 (1.4)	0.1 (2.8)	−1.2 (3.2)	0.8 (3.0)
Total body water, L	–	–	−2.0 (7.2)	−1.9 (7.9)	−3.6 (9.1)	–

Safety

- Sparsentan was generally well tolerated over 36 weeks of treatment
- One patient discontinued treatment due to hypotension after 6 weeks
- There have been 3 serious adverse events, none related to treatment

CONCLUSIONS

- In these preliminary results, sparsentan as a first-line treatment in patients newly diagnosed with IgAN was effective at reducing proteinuria and controlling BP
- The rapid and sustained reductions in proteinuria, the achievement of complete remission and safety profile of sparsentan in this study are comparable with those from the phase 3 PROTECT study[2]
- There was no substantial effect on body weight, which was generally maintained over 36 weeks
- Total body water showed modest reduction over the treatment period with sparsentan, with no evidence of fluid retention observed in the study participants
- During the reported study period sparsentan treatment was generally well tolerated
- Planned analyses of cardiac and renal MRIs, repeat kidney biopsies, and other serum, plasma, and urinary biomarkers will investigate the mechanistic actions of sparsentan in this patient population and its potential renoprotective effects

DISCLOSURES

CKC: consulting fees from George Clinical, and Vera Therapeutics; honoraria from Stada; research funding from GSK, and Travere Therapeutics, Inc.; advisory boards for Calliditas, CSL Vifor, and Novartis; steering committee/DSMC for CSL Vifor, Alpine Immune Sciences, and Roche; and travel support from Otsuka, and Chinook Therapeutics. **SM, RK:** employees of Travere Therapeutics, Inc., and may have equity or other financial interest in Travere Therapeutics, Inc. **ND, JB**: consulting fees and research funding from Travere Therapeutics, Inc. **SG:** consulting fees from CSL Vifor, and Alexion; honoraria from Bayer, and Travere Therapeutics, Inc.; advisory board for Emmes, and ICON plc; and travel support from Alexion. **AM**: consulting fees from Travere Therapeutics, Inc., Vera Therapeutics, and HI-Bio. **SS**: research funding from Johnson and Johnson (JnJ), AstraZeneca, CSL Vifor, and Sanofi Genzyme; consulting fees from Novartis, Bayer, Sanofi-Genzyme, Vifor Pharma, Boehringer-Ingelheim, AstraZeneca, GSK, Sanifit, and Inozyme Pharma Inc.; honoraria from AstraZeneca, Menarini, Napp, CSL Vifor, GSK, Novartis, Bayer, Sanofi Genzyme, and Medscape; National Clinical Director of Renal Medicine for NHS England; travel support from AstraZeneca, Novartis, and CSL Vifor. **LW**: consulting fees from Travere Therapeutics, Inc., Novartis, Chinook, and Goldfinch Bio; honoraria from Travere Therapeutics, Inc., Novartis, and Otsuka; and advisory boards for Eledon. **AH, MS**: have no conflicts of interest.

ACKNOWLEDGMENTS

This study was funded by Travere Therapeutics, Inc. Medical writing support was provided by Taryn Ralph, PhD, of Medical Expressions, Inc., a part of Nucleus Global, an Inizio Company, and was funded by Travere Therapeutics, Inc.

REFERENCES

1. Filspari (sparsentan). Prescribing information. Travere Therapeutics, Inc.; 2023. **2.** Heerspink HJL, et al. *Lancet*. 2023;401(10388):1584-1594. **3.** ClinicalTrials.gov. Accessed September 11, 2023. https://clinicaltrials.gov/study/NCT04663204

To obtain a PDF of this poster:

Please scan the Quick Response (QR) code.

No personal information is stored.

UNIVERSITY OF LEICESTER

Leicester IgA Nephropathy Research Group

TRAVERE THERAPEUTICS

Contact information: Chee Kay Cheung, Cheekay.Cheung@uhl-tr.nhs.uk

Presented at the American Society of Nephrology (ASN) Kidney Week 2023; November 2-5, 2023; Philadelphia, PA, USA