61st ERA CONGRESS
STOCKHOLM & VIRTUAL
MAY 23-26, 2024

The Humanistic Burden of Rare Kidney Diseases: Understanding the Impact of Immunoglobulin A Nephropathy (IgAN) and Focal Segmental Glomerulosclerosis (FSGS) on Patients and Care-Partners Study (HONUS): Results for IgAN and FSGS in Europe

Justyna Szklarzewicz[1], Ute Floege[2], Daniel Gallego[3], Keisha Gibson[4], Kamyar Kalantar-Zadeh[5], Kelly Helm[6], Dale Robinson[7], Bonnie Schneider[8], Philip Smith[7], Kjell Tullus[9], Mark Bensink[10], Bruce Hendry[10], Nisha C Hazra[11], Chunyi Xu[11], Jingyi Liu[11], Kirsten Gallant[11], Zheng-Yi Zhou[11]

[1]University Hospitals of Leicester NHS Trust, Leicester, UK; [2]University Hospital of the RWTH Aachen University, Aachen, Germany; [3]EKPF European Kidney Patients Federation, FEDERACION ALCER Spanish Kidney Patient Federation, Madrid, Spain; [4]University of North Carolina at Chapel Hill School of Medicine, Chapel Hill, NC, USA; [5]School of Medicine University of California, Irvine, CA, USA; [6]NephCure Kidney International, King of Prussia, PA, USA; [7]Kidney Research UK, Peterborough, UK; [8]The IGA Nephropathy Foundation of America, Wall, NJ, USA; [9]Great Ormond Street Hospital, London, UK; [10]Travere Therapeutics, Inc., San Diego, CA, USA; [11]Analysis Group, Boston, MA, USA.

BACKGROUND

- Rare proteinuric kidney diseases are associated with clinical and economic burden including high health care costs,[1] however, the humanistic burden associated with these conditions has not been directly evaluated.
- HONUS (Humanistic Burden of Rare Nephrotic Diseases: Understanding the impact of FSGS and IgAN on Patients and Caregivers Study) is a multi-national, cross-sectional survey study designed to evaluate the humanistic burden associated with two rare kidney diseases, immunoglobulin A nephropathy (IgAN) and focal segmental glomerulosclerosis (FSGS), from the patient and caregiver perspectives in the United States (US) and Europe.[2]
- Previous US results have been published for the HONUS population[3,4]; the current analysis focuses on results from adults with IgAN or FSGS and their care-partners in Spain, Germany, France, or the United Kingdom who participated in HONUS between January and October 2023.

OBJECTIVE

To quantify the humanistic burden of IgAN and FSGS adult patients in Europe and the burden and impact of IgAN and FSGS from the care-partner perspective.

METHODS

Adult patients with IgAN or FSGS and their care-partners were recruited from one medical center in Germany (Nephrologisches Zentrum Villingen-Schwenningen) and one patient advocacy group in Spain (Federacion Nacional Asociaciones ALCER). Additionally, one patient living in Europe was recruited from a patient advocacy group based in the US (the IGA Nephropathy Foundation of America).

Study Population

Inclusion criteria

- Adult patients (≥ 18 years old) and their paired adult care-partners in Spain, Germany, France, or the United Kingdom who had a self-reported physician-provided diagnosis of IgAN (with renal biopsy confirmation of the diagnosis) or FSGS, and were able to provide informed consent

Exclusion criteria

The following patients (and their care-partners) were excluded from the study:

- Had IgAN or FSGS secondary to another condition
- Had a history of malignancy other than adequately treated basal cell or squamous cell skin cancer
- Had a co-existing glomerular disease (e.g., membranous nephropathy or lupus nephritis)
- Participated in a kidney disease clinical trial, and potentially have received active treatment as part of the trial at the time of recruitment

Study outcomes

- All patients and care-partners completed an online survey, with questions on demographics, clinical characteristics and the following validated questionnaires:

PATIENTS	CARE-PARTNERS
KDQoL-36 (including SF-12)	SF-12
GAD-7	GAD-7
PHQ-9	PHQ-9
Productivity impairment: WPAI:SHP	Productivity impairment: WPAI:SHP

Analysis

All outcomes were summarized descriptively. Continuous variables were summarized as mean, median, and standard deviation (SD); categorical variables were summarized as count and proportion.

CONTACT INFORMATION
Mark.Bensink@travere.com

RESULTS

Demographics and Patient Disease Characteristics

- Patients (26 IgAN, 9 FSGS) and their care-partners (22 IgAN, 7 FSGS) were recruited from Germany and Spain (**Table 1**).
- IgAN patients had a mean (SD) age of 42.2 (11.6) years and FSGS patients of 51.4 (11.4) years; proportion female was 61.5% and 44.4% for IgAN and FSGS, respectively.
- Average (SD) time since diagnosis (years): 9.6 (8.6) (IgAN), 21.9 (13.9) (FSGS) (**Table 2**).
- Less than half of care-partners (40.9%) for IgAN patients were female, compared to most care partners (85.7%) for FSGS; most paired care-partners were partners of patients (IgAN: 81.8%, FSGS: 71.4%).

Health-Related Quality of Life

- IgAN patients were more impacted on the mental component (Mean [SD] SF-12 PCS: 47.3 [11.6], MCS: 43.2 [10.9]), while FSGS patients were more impacted on the physical (PCS: 40.6 [6.0], MCS: 43.8 [9.1]) (**Table 3**); scores reflected generally worse HRQoL than previously published European general population scores (mean 50.0 for PCS and MCS).[10]
- Care-partners of IgAN patients were more impacted by the mental component and care-partners of FSGS patients by the physical component (**Table 3**).

Anxiety and Depression

- Moderate to severe anxiety (recall period of 2 weeks) was reported by 30.8% of IgAN and 33.3% of FSGS patients (**Figure 1a, 1b**), and moderate to severe depression by 30.8% of IgAN and 44.4% of FSGS patients (**Figure 1c, 1d**).
- Moderate to severe anxiety was reported in 9.2% of IgAN care-partners, and no FSGS care-partners reported moderate to severe anxiety (**Figure 2a, 2b**). Moderate to severe depression was reported in 13.8% (IgAN) and 14.3% (FSGS) of care-partners (**Figure 2c, 2d**).

Work Productivity

- Among IgAN patients who were employed (69.2%), mean percent absenteeism (work time missed) was 13.3%, presenteeism (impairment while working) 17.7%, and overall work productivity loss 17.7% due to IgAN-related reasons (**Figure 2**). Only 3 FSGS patients (33.3%) were employed (outcomes not reported due to small sample size).
- Among employed IgAN care-partners (86.4%), mean percent absenteeism was 14.2%, presenteeism 9.3%, and overall work productivity loss 9.9% due to IgAN-related reasons.
- Work productivity outcomes are not shown for FSGS due to small sample size (3 patients and 6 care-partners employed).

Activity Impairment

- Mean percent activity impairment was reported at 27.3% in IgAN patients and 18.6% in their care-partners.
- Mean percent activity impairment was reported at 43.3% in FSGS patients and 17.1% in their care-partners.

Table 1. Demographics PATIENTS CARE-PARTNERS

	IgAN		FSGS	
	PATIENTS (N = 26)	CARE-PARTNERS (N = 22)	PATIENTS (N = 9)	CARE-PARTNERS (N = 7)
Age				
Mean ± SD	42.2 ± 11.6	46.6 ± 9.5	51.4 ± 11.4	50.1 ± 15.7
Median	39.0	45.0	50.0	49.0
Sex, N (%)				
Male	10 (38.5%)	13 (59.1%)	5 (55.6%)	1 (14.3%)
Female	16 (61.5%)	9 (40.9%)	4 (44.4%)	6 (85.7%)
Country, N (%)				
Spain	16 (57.1%)	14 (63.6%)	8 (88.9%)	6 (85.7%)
Germany	8 (30.8%)	6 (27.3%)	1 (11.1%)	1 (14.3%)
France	1 (3.8%)	1 (4.5%)	0 (0.0%)	0 (0.0%)
UK	1 (3.8%)	1 (4.5%)	0 (0.0%)	0 (0.0%)

Table 2. Disease Characteristics PATIENTS

	IgAN	FSGS
	PATIENTS (N = 26)	PATIENTS (N = 9)
Time since diagnosis (years)		
Mean ± SD	9.6 ± 8.6	21.9 ± 13.9
Current CKD status, N (%)		
Stage 1	3 (11.5%)	0 (0.0%)
Stage 2	3 (11.5%)	1 (11.1%)
Stage 3	4 (15.4%)	0 (0.0%)
Stage 4	3 (11.5%)	1 (11.1%)
Stage 5, dialysis	4 (15.4%)	3 (33.3%)
Stage 5, no dialysis	0 (0.0%)	0 (0.0%)
Transplant recipient	7 (26.9%)	4 (44.4%)
Unknown	2 (7.7%)	0 (0.0%)

Table 3. HRQOL PATIENTS CARE-PARTNERS

	IgAN		FSGS	
	PATIENTS (N = 26)	CARE-PARTNERS (N = 22)	PATIENTS (N = 9)	CARE-PARTNERS (N = 7)
SF-12 PCS				
Mean ± SD	47.3 ± 11.6	53.3 ± 8.8	40.6 ± 6.0	47.0 ± 10.6
Median	51.6	55.9	40.1	46.5
SF-12 MCS				
Mean ± SD	43.2 ± 10.9	47.9 ± 10.8	43.8 ± 9.1	49.4 ± 9.5
Median	44.6	50.9	42.4	51.9

Abbreviations: HRQOL = Health-Related Quality of Life; MCS = mental component summary; PCS = physical component summary; SD = standard deviation; SF-12 = 12-item short form survey
Note: [1] Lower scores reflect worse HRQOL.

Figure 1: Anxiety (GAD-7) and Depression (PHQ-9) Total Scores PATIENTS

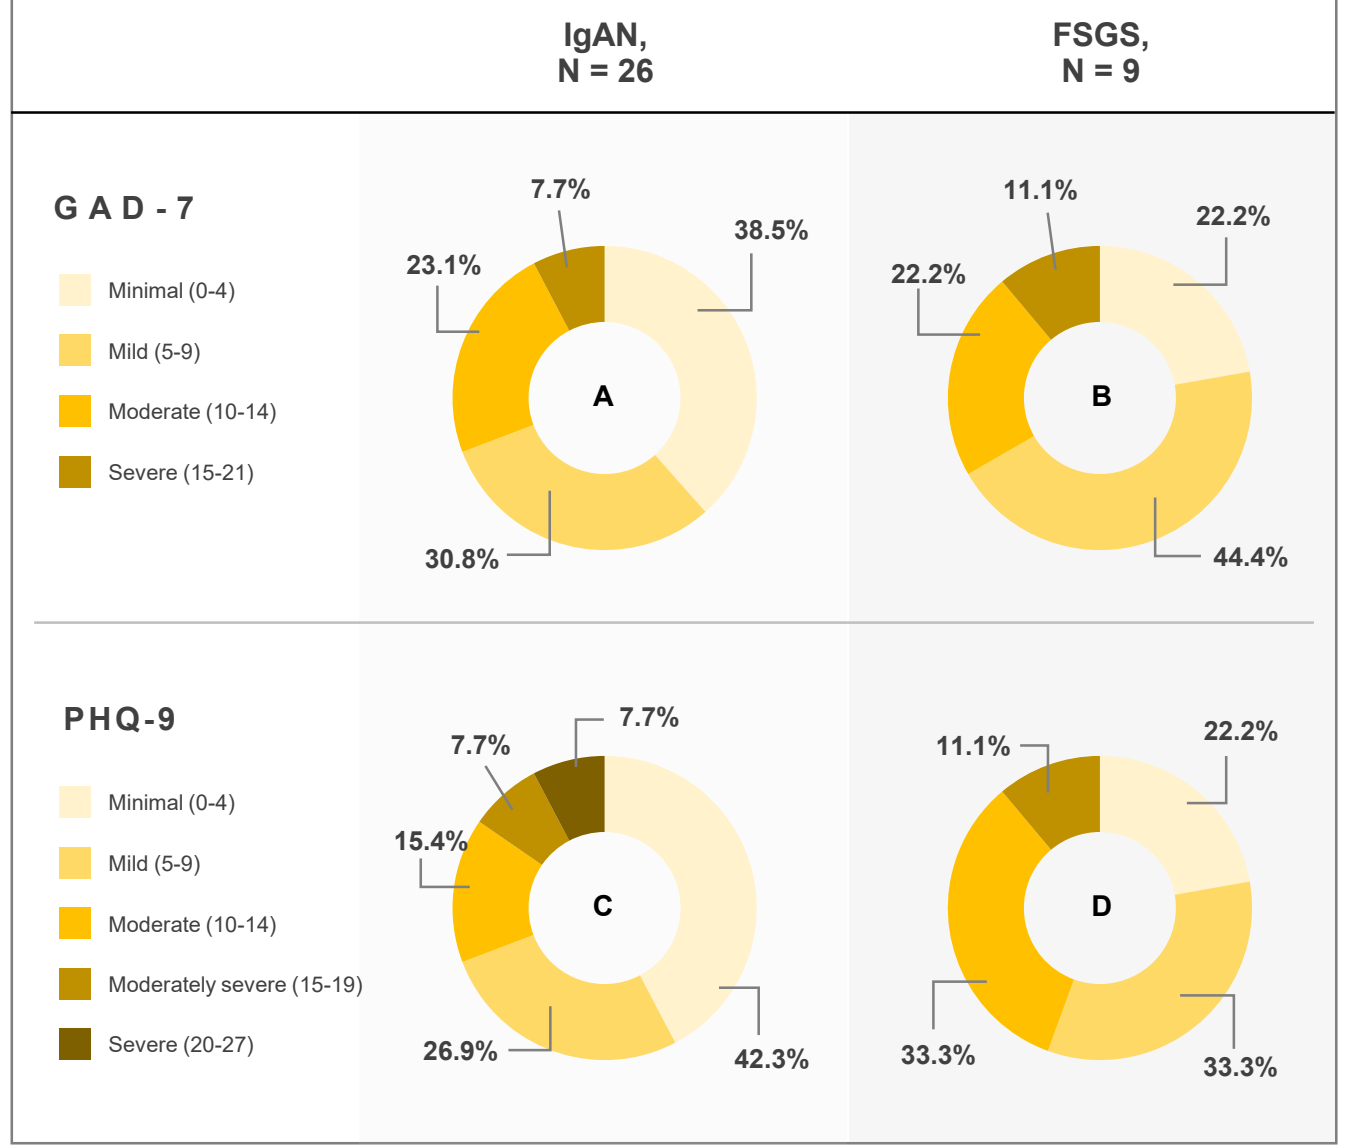

IgAN,
N = 26
FSGS,
N = 9
GAD-7
Minimal (0-4)
Mild (5-9)
Moderate (10-14)
Severe (15-21)
A
7.7%
38.5%
23.1%
30.8%
B
11.1%
22.2%
22.2%
44.4%
PHQ-9
Minimal (0-4)
Mild (5-9)
Moderate (10-14)
Moderately severe (15-19)
Severe (20-27)
C
7.7%
7.7%
15.4%
26.9%
42.3%
D
11.1%
22.2%
33.3%
33.3%

Figure 2: Anxiety (GAD-7) and Depression (PHQ-9) Total Scores CARE-PARTNERS

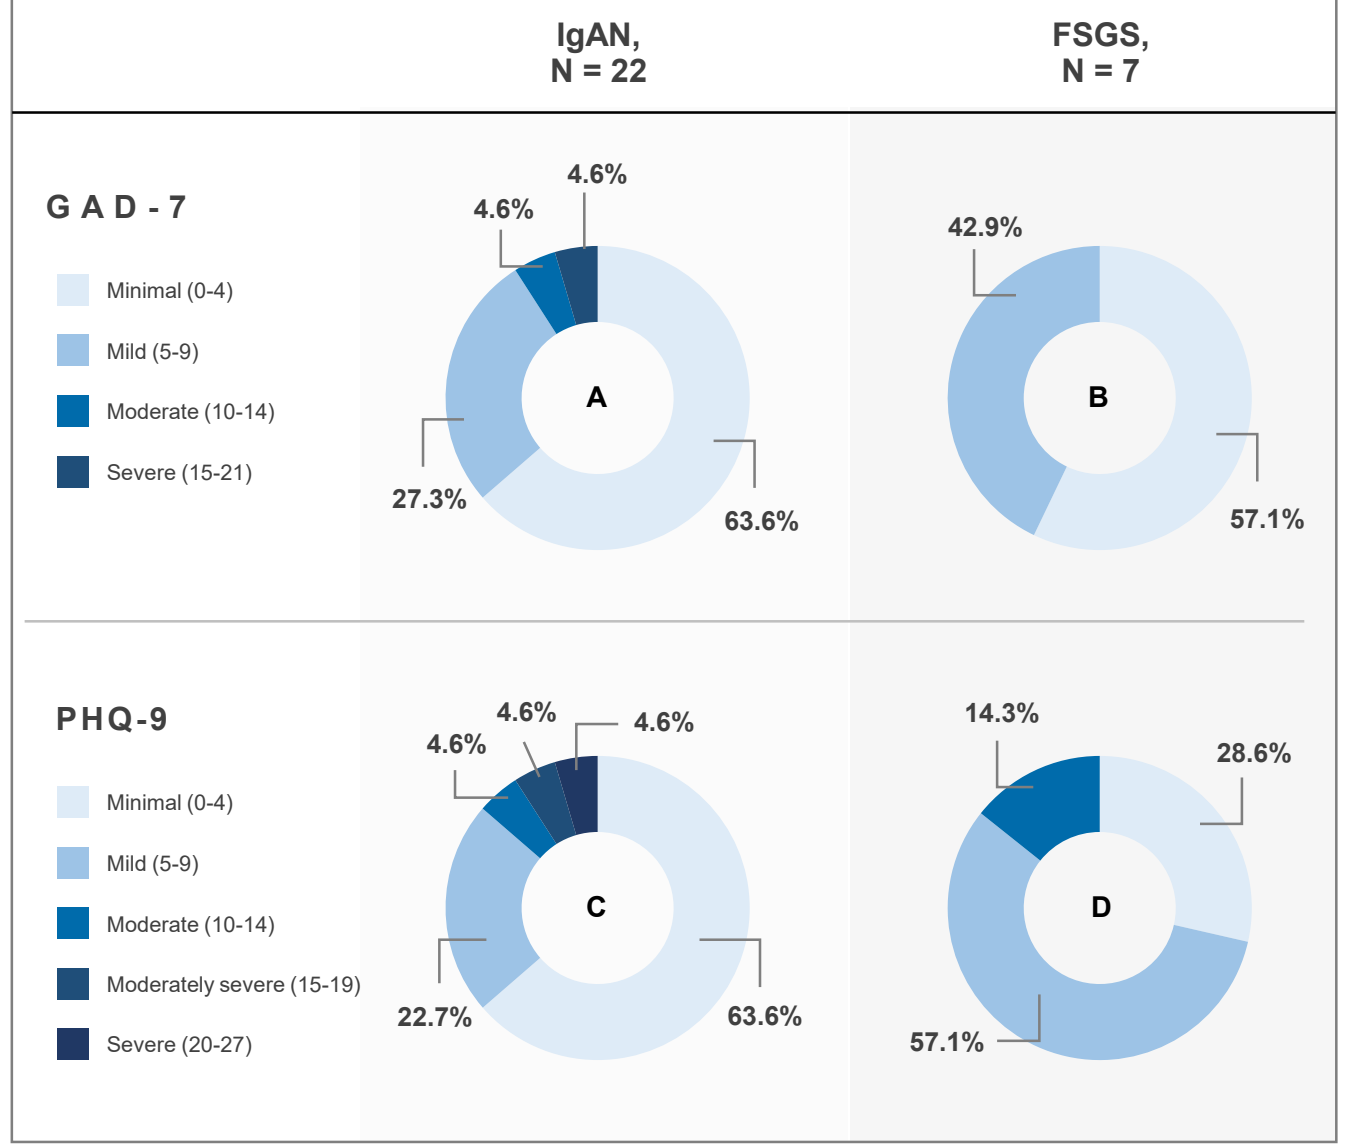

IgAN,
N = 22
FSGS,
N = 7
GAD-7
Minimal (0-4)
Mild (5-9)
Moderate (10-14)
Severe (15-21)
A
4.6%
4.6%
27.3%
63.6%
B
42.9%
57.1%
PHQ-9
Minimal (0-4)
Mild (5-9)
Moderate (10-14)
Moderately severe (15-19)
Severe (20-27)
C
4.6%
4.6%
4.6%
22.7%
63.6%
D
14.3%
28.6%
57.1%

Figure 3: WPAI: SHP (IgAN) PATIENTS CARE-PARTNERS

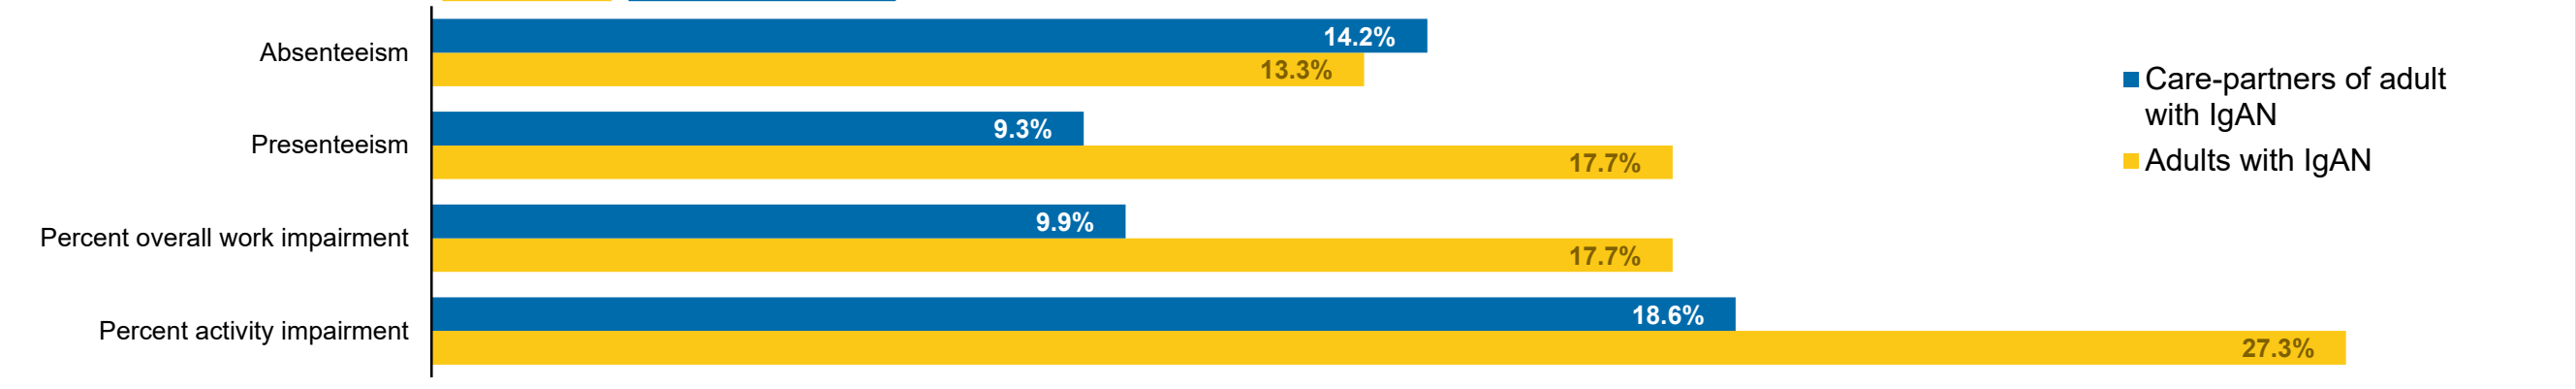

Absenteeism
14.2%
13.3%
Presenteeism
9.3%
17.7%
Percent overall work impairment
9.9%
17.7%
Percent activity impairment
18.6%
27.3%
Care-partners of adult with IgAN
Adults with IgAN

Notes:
1. 18 (69.2%) IgAN patients and 19 (86.4%) IgAN care-partners were employed.
2. Percent work time missed (absenteeism) and overall work impairment were only assessed among respondents who were employed at the time of survey. Impairment while working (presenteeism) was assessed among respondents who were employed and worked >0 hrs in the past 7 days. Activity impairment was assessed amongst all respondents.
3. The recall period is the past 7 days.

CONCLUSIONS

Patients with IgAN and FSGS in Europe experience impaired HRQoL compared to previously published European population estimates, as well as anxiety, depression, and impaired productivity.

While data for care-partners of FSGS patients was limited, care-partners of IgAN patients also experience impaired HRQoL, particularly in terms of mental components, and reduced overall productivity.

LIMITATIONS

- Selection bias may exist as participants who voluntarily participated in the survey may differ from those who did not participate; bias from self-reported responses may also be present.
- Due to low numbers, results for FSGS should be interpreted with caution and further research is warranted in this group.
- The study lacks a control group; use of historical European general population estimates is included for reference for SF-12 outcomes.
- Participant responses may be influenced by the COVID-19 pandemic. This broader circumstance may impact socioeconomic status, accessibility of care, health outcomes, and HRQoL.

REFERENCES

1. Bensink ME, Goldschmidt D, et al. Kidney Failure Attributed to Focal Segmental Glomerulosclerosis: A USRDS Retrospective Cohort Study of Epidemiology, Treatment Modalities, and Economic Burden. Kidney Med. 2023 Nov 27;6(2):100760.
2. Szklarzewicz J, Leicester L. Humanistic Burden of Rare Kidney Diseases; Understanding the Impact of FSGS and IgAN on Patients and Caregivers: The HONUS Rationale and Study Design. 2021.
3. Szklarzewicz J, Bensink ME, et al. #5300 HUMANISTIC BURDEN OF RARE KIDNEY DISEASES: UNDERSTANDING THE IMPACT OF IGAN AND FSGS ON PATIENTS AND CARE-PARTNERS STUDY (HONUS): A US IGAN RESULTS UPDATE, Nephrology Dialysis Transplantation, Volume 38, Issue Supplement_1, June 2023, gfad063c_5300
4. Szklarzewicz J, Bensink ME, et al. The Humanistic Burden of Rare Kidney Diseases: Understanding the Impact of Focal Segmental Glomerulosclerosis (FSGS) and Immunoglobulin A Nephropathy (IgAN) on Patients and Caregivers Study (HONUS): Results for FSGS in the United States (US). Poster TH-PO597: Presented at the American Society of Nephrology (ASN) Kidney Week 2023; November 2-5, 2023; Philadelphia, PA.
5. Hays RD. The medical outcomes study (mos) measures of patient adherence. J Behav Med. 1994;17:361-7.
6. Mark Kosinski JEW, Diane M. Turner-Bowker, et al. User's manual for the SF-12v2 health survey : with a supplement documenting the SF-12® health survey. QualityMetric Incorporated, Lincoln, RI; 2007.
7. Spitzer RL, Kroenke K, et al. A brief measure for assessing generalized anxiety disorder: the GAD-7. Archives of internal medicine. 2006;166(10):1092-1097.
8. Kroenke K, Spitzer RL, et al. The PHQ-9: validity of a brief depression severity measure. Journal of general internal medicine. 2001;16(9):606-613.
9. Reilly MC, Zbrozek AS, et al. The validity and reproducibility of a work productivity and activity impairment instrument. Pharmacoeconomics. 1993;4(5):353-365.
10. Gandek B, Ware JE, et al. Cross-validation of item selection and scoring for the SF-12 Health Survey in nine countries: results from the IQOLA Project. International Quality of Life Assessment. J Clin Epidemiol. 1998 Nov;51(11):1171-8.

ACKNOWLEDGEMENTS

We wish to acknowledge all HONUS survey respondents who gave up their time to provide these data. Your contributions are critical to understanding the burden of these rare kidney diseases, thank you.

FUNDING STATEMENT

This study was funded by Travere Therapeutics, Inc.

DISCLOSURES

JS, UF, DG, KG, KK-Z, KH, DR, BS, PS, KT:

Received consultancy fees from Travere Therapeutics, Inc.

MB, BH: Employee, Travere Therapeutics, Inc.

NCH, CX, JL, KG, Z-YZ: Employees of Analysis Group, which received consultancy fees from Travere Therapeutics, Inc.

MA-SP-24-0067 05/2024