FR-PO907

Sparsentan in Patients With Prior or Concurrent Immunosuppressive Treatment (IST) for IgA Nephropathy (IgAN): A Case Series

Luis E. Vélez,[1] Reva Siva,[2] Agness Pelts Block,[3] Christopher Gisler,[3] Pablo Garcia[4]

[1]South Texas Renal Care Group San Antonio, TX, USA; [2]Virginia Nephrology Group, Arlington, VA, USA; [3]Travere Therapeutics, Inc., San Diego, CA, USA; [4]University of New Mexico School of Medicine, Division of Nephrology, Albuquerque, NM, USA

To obtain a PDF of this poster, please scan the Quick Response (QR) code. No personal information is stored.

INTRODUCTION

BACKGROUND

- Sparsentan is a non-immunosuppressive, dual endothelin angiotensin receptor antagonist (DEARA)[1,2] approved in the US and EU for the treatment of adults with IgAN[3,4]
- Systemic IST is being used in the treatment of IgAN; however, concerns remain regarding side effects and patient susceptibility to infection[5]
- The effects of sparsentan in patients receiving prior or ongoing IST for IgAN have not been well characterized

OBJECTIVE

- This case series reports the clinical features and treatment response of 5 cases involving patients with IgAN who received sparsentan after previous or concurrent IST in the real-world setting

CASE SERIES METHODOLOGY

- Patients with biopsy-proven IgAN who received sparsentan for ≥3 months, having received prior or concurrent IST, in routine clinical practice at a tertiary care center between March 2023 and August 2024 were selected by their treating healthcare provider (HCP) for inclusion in this case series (**Figure 1**)
 - According to the prescribing information,[3] sparsentan was dosed at 200 mg/day for 2 weeks before up titration to 400 mg/day
- De-identified patient data, including patient characteristics, treatment history, and clinical assessments, were provided by the patient's HCP

Figure 1. Patient Treatment Summary

Eligibility criteria:

- Biopsy-proven IgAN
- ≥3 months on sparsentan
- Received sparsentan after discontinuing IST or in addition to ongoing IST
- Patient provided consent to HCP

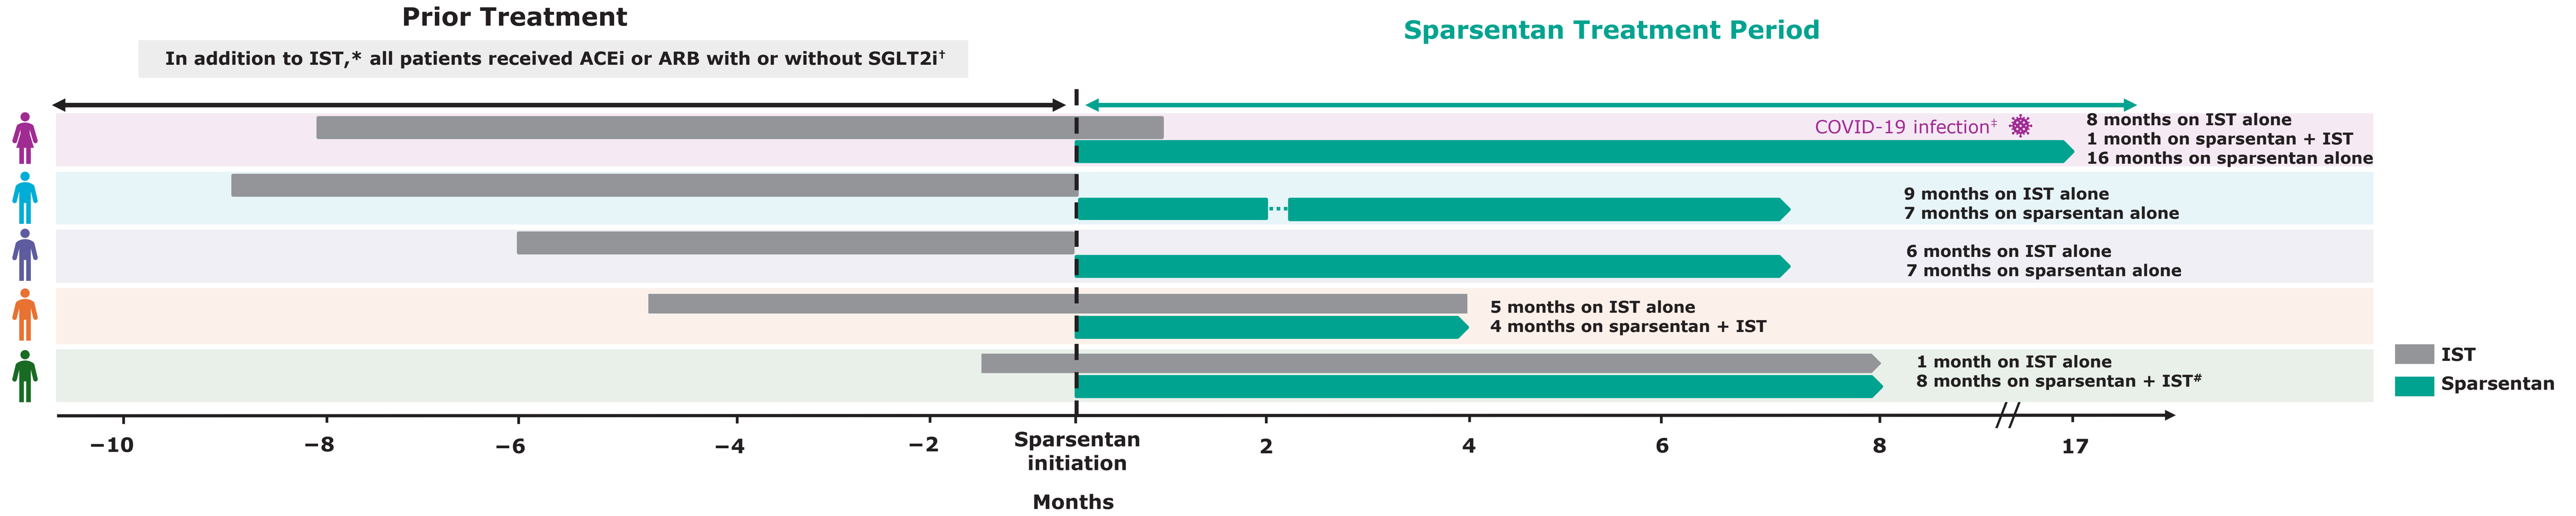

Prior Treatment
In addition to IST,* all patients received ACEi or ARB with or without SGLT2i†
Sparsentan Treatment Period
COVID-19 infection‡
8 months on IST alone
1 month on sparsentan + IST
16 months on sparsentan alone
9 months on IST alone
7 months on sparsentan alone
6 months on IST alone
7 months on sparsentan alone
5 months on IST alone
4 months on sparsentan + IST
1 month on IST alone
8 months on sparsentan + IST#
IST
Sparsentan
−10
−8
−6
−4
−2
Sparsentan initiation
2
4
6
8
17
Months

*IST varied per patient and included: cyclophosphamide, mycophenolate, prednisone, tacrolimus, and TRF-budesonide. †Use of ACEi, ARB, and SGLT2i varied per patient. See **Table 1** for treatment duration and details in each patient.
‡Patient developed COVID-19 3 weeks prior to the last urine collection, and sparsentan was paused for 5 days during nirmatrelvir/ritonavir treatment. #Patient received SGLT2i in addition to ongoing SPAR + IST for <1 month before last follow-up.

CASE DESCRIPTIONS

Patients

- Five patients with biopsy-proven IgAN (ages ≈20-75 years) received sparsentan, after prior or with ongoing IST (**Table 1, Figure 1**)
- Three patients received sparsentan after discontinuing IST
 - Reasons for IST discontinuation included insufficient response (n=1) and completion of treatment course (n=2)
- Two patients received sparsentan in addition to ongoing IST
 - One patient was receiving ongoing IST at last follow-up; the other discontinued IST at the last follow-up visit
 - In 1 patient, who initiated sparsentan after a kidney transplant, an SGLT2i was initiated in addition to ongoing IST and sparsentan (duration of follow-up on SGLT2i, <1 month)
- Duration of follow-up on sparsentan ranged from approximately 4 to 17 months, with all patients receiving ongoing sparsentan treatment at last follow-up

Outcomes

- A decrease in proteinuria (UPCR) was seen in all patients after sparsentan initiation (**Figure 2**)
 - In 1 patient, who developed COVID-19 infection 3 weeks prior to the last urine collection time point, UPCR returned to approximately baseline levels at last follow-up visit
- In 2 patients who achieved complete proteinuria remission (CR; UPCR <0.3 g/g) or UPCR <0.5 g/g at any time after initiating sparsentan, target proteinuria was not previously reached on prior IST (**Figure 3**)
- There were no major fluctuations in eGFR from initiation to follow-up, with a notable improvement in eGFR seen in 1 patient (**Figure 4**)
- Hematuria resolved in 1 out of 3 patients (**Table 1**)
- Overall, there were no major fluctuations in blood pressure (**Table 1**)
- Sparsentan was generally well tolerated by these patients
 - In 1 patient, sparsentan treatment was briefly interrupted due to elevated liver function tests, but treatment was resumed (initially at 200 then 400 mg/day) without further issues

Figure 2. Change in UPCR

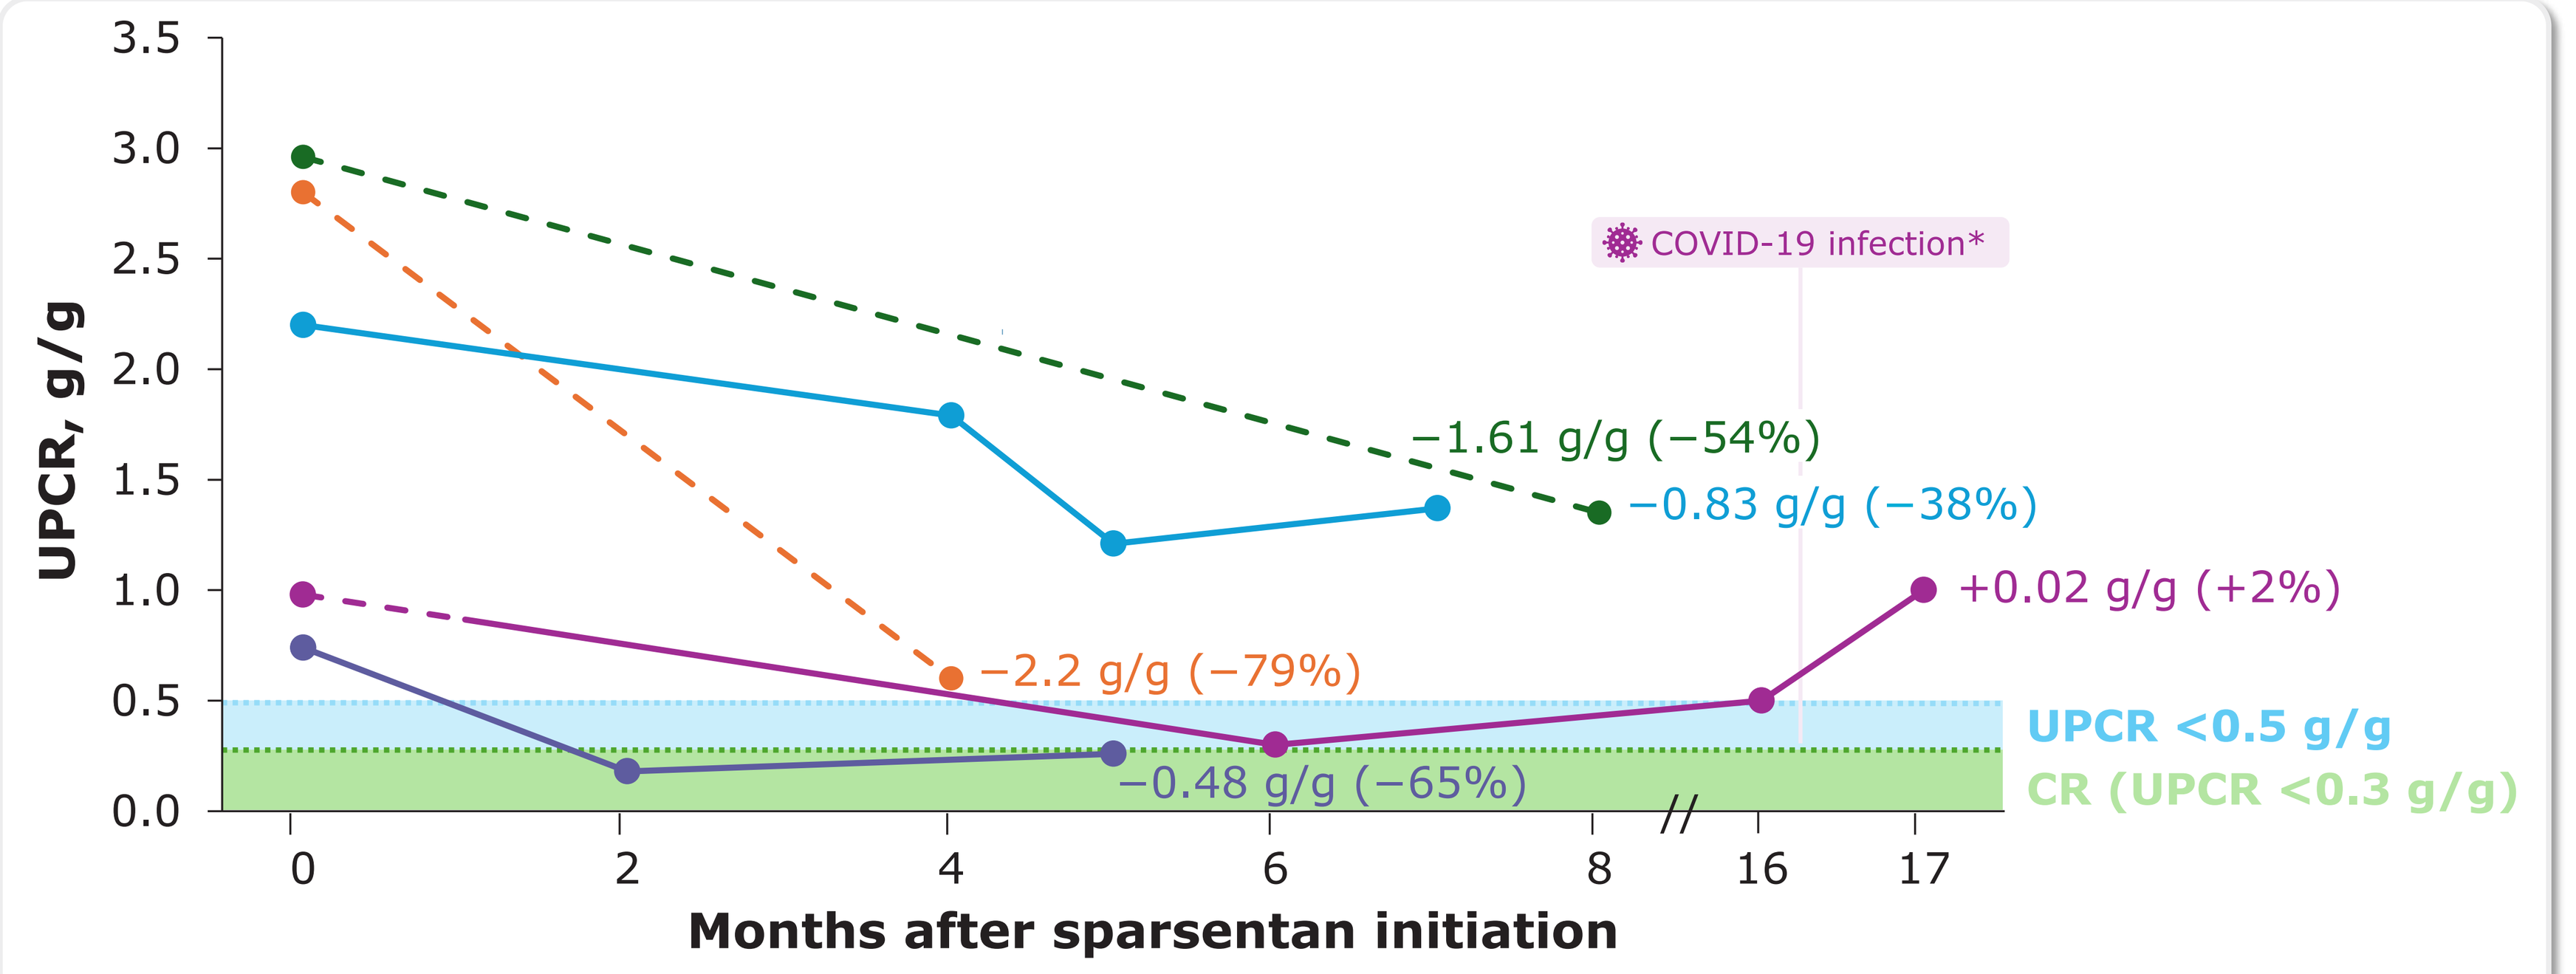

UPCR, g/g
3.5
3.0
2.5
2.0
1.5
1.0
0.5
0.0
COVID-19 infection*
−1.61 g/g (−54%)
−0.83 g/g (−38%)
+0.02 g/g (+2%)
−2.2 g/g (−79%)
−0.48 g/g (−65%)
UPCR <0.5 g/g
CR (UPCR <0.3 g/g)
0
2
4
6
8
16
17
Months after sparsentan initiation

Dashed lines indicate concomitant sparsentan and IST treatment. Solid lines indicate sparsentan treatment alone.
Values reflect change from sparsentan initiation (Month 0) to last follow-up.
*Patient developed COVID-19 3 weeks prior to the last urine collection, and sparsentan was paused for 5 days during nirmatrelvir/ritonavir treatment.

Table 1. Case Summaries

	Patient characteristics				Treatment				UPCR, g/g		eGFR, mL/min/1.73 m²		Hematuria		Blood pressure, mm Hg		Safety
	Age range, years	Sex	Race	Time since IgAN diagnosis	Other treatment history (duration)	Steroid/IST (duration)	Other treatment initiated after sparsentan	Duration of follow-up on SPAR*	SPAR initiation†	Follow-up‡	SPAR initiation†	Follow-up‡	SPAR initiation†	Follow-up‡	SPAR initiation	Follow-up‡	
	40-45	Female	White	27 months	• SGLT2i (9 mo; discontinued§) • ACEi (9 mo; discontinued‖)	• TRF-Budesonide§ (9 mo; discontinued¶)	• NA	17 months	0.98	1.0	110	90	Present	Present	108/58	101/59	Well tolerated
	50-54	Male	White	11 years	• ACEi (NS; discontinued‖)	• Prednisone (9 mo; discontinued#)	• NA	7 months**	2.2	1.37	24††	22	Absent	Absent	138/80	123/81	Sparsentan was stopped for LFT elevations; LFTs subsequently stabilized within normal range and sparsentan was restarted
	20-25	Male	Asian	3 years	• ACEi (23 mo; discontinued§)	• Cyclophosphamide (6 mo, discontinued#) • Prednisone (6 mo, discontinued#)	• NA	7 months‡‡	0.74	0.26	127	113	Present	Present	107/67	150/80	Well tolerated
	55-60	Male	White	9 months	• ACEi (NS; discontinued§)	• TRF-Budesonide (9 mo; discontinued)	• NA	4 months	2.8	0.6	35	28	Present	Absent	146/84	132/76	Well tolerated
	70-75	Male	Hispanic	5 years	• ARB (NS; discontinued§)	• TRF-Budesonide (9 mo; ongoing) • Tacrolimus§§ + mycophenolate¶¶ (NS; ongoing)	• Dapagliflozin (<1 mo, ongoing)	8 months	2.96	1.35	68	97	Absent	Absent	150/80	150/80	Well tolerated
									≥0.5	<0.3 (CR)	≤45 / >40 to ≤90	>90	Present	Absent			

*Approximate. †Values at initiation derived from visit at which SPAR was initiated or most recent visit prior to initiation. ‡Values at last follow-up unless otherwise specified. §Discontinued to start sparsentan. ‖Reason for discontinuation not specified. ¶Inadequate response. #Treatment course completed.
**Treatment paused for 2 weeks during this time period due to elevated LFTs (resumed without further issue). ††At 3 months follow-up. ‡‡7 months of follow-up for eGFR, hematuria, and safety data; 5 months of follow-up for UPCR. §§Anti-rejection IST for kidney transplant.

Figure 3. CR (UPCR <0.3 g/g) and UPCR <0.5 g/g at Any Time During Follow-Up

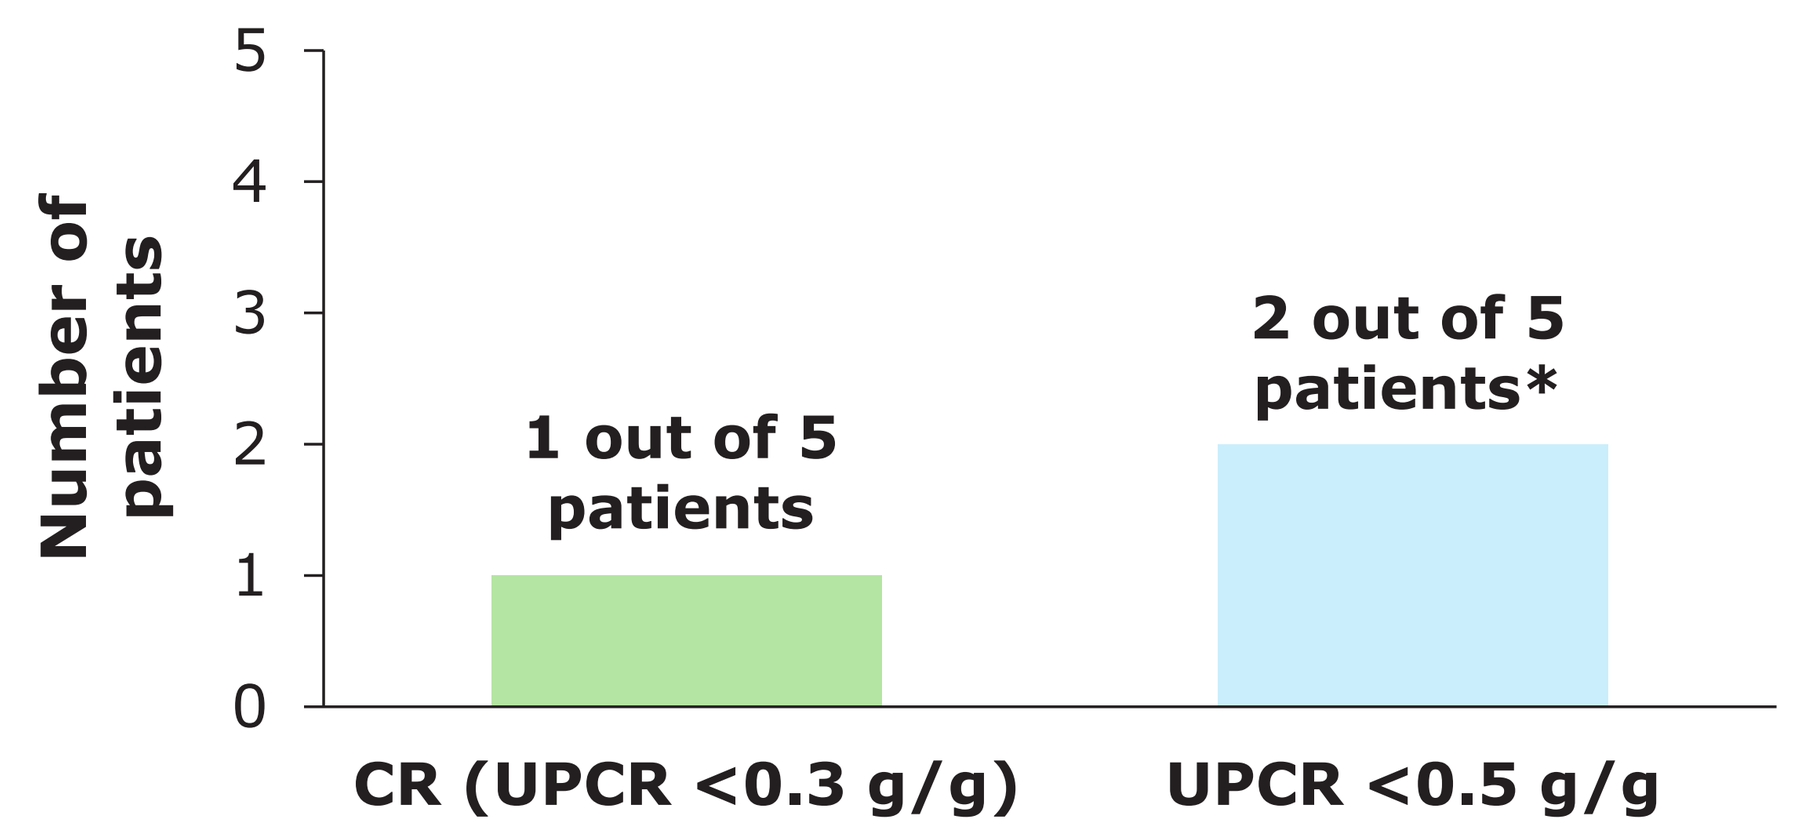

Number of patients
5
4
3
2
1
0
1 out of 5 patients
2 out of 5 patients*
CR (UPCR <0.3 g/g)
UPCR <0.5 g/g

	Achieved CR (UPCR <0.3 g/g) at any time	Achieved UPCR <0.5 g/g at any time	Case details
	✓	✓	• Achieved UPCR <0.3 g/g at 2 months • UPCR maintained at 0.3 g/g at 5 months
	–	✓	• Achieved UPCR <0.5 g/g at 6 months • UPCR increased to 0.5 g/g at 16 months and 1.0 g/g at 17 months in the setting of COVID-19 infection (at ≈16 months)

*The patient who achieved CR (UPCR <0.3 g/g) was also included in the total number achieving UPCR <0.5 g/g.

Figure 4. Change in eGFR

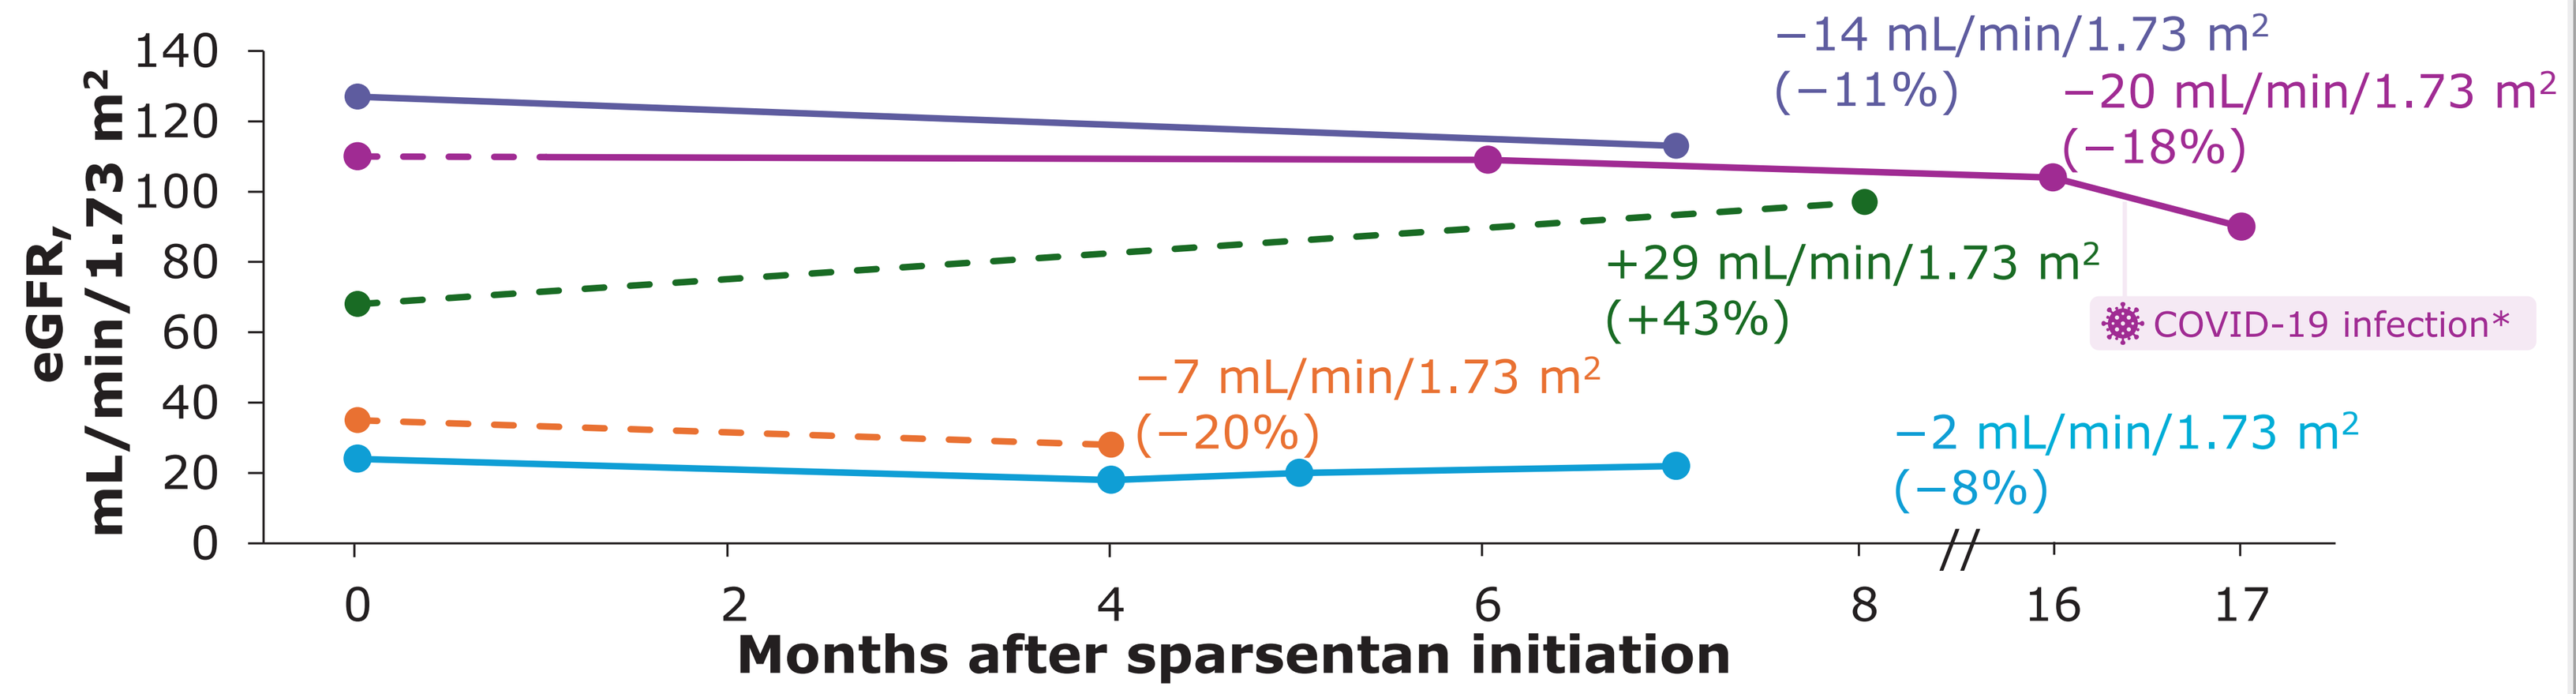

eGFR, mL/min/1.73 m²
140
120
100
80
60
40
20
0
−14 mL/min/1.73 m² (−11%)
−20 mL/min/1.73 m² (−18%)
+29 mL/min/1.73 m² (+43%)
COVID-19 infection*
−7 mL/min/1.73 m² (−20%)
−2 mL/min/1.73 m² (−8%)
0
2
4
6
8
16
17
Months after sparsentan initiation

Dashed lines indicate concomitant sparsentan and IST treatment. Solid lines indicate sparsentan treatment alone.
Values reflect change from sparsentan initiation (Month 0) to last follow-up.
*Patient developed COVID-19 3 weeks prior to the last urine collection, and sparsentan was paused for 5 days during nirmatrelvir/ritonavir treatment.

FINDINGS

- These cases support the safety and efficacy of sparsentan in patients with previous or ongoing IST
- Improvements in proteinuria were observed regardless of eGFR or UPCR at sparsentan initiation, including in patients who previously were unable to achieve target proteinuria with IST alone

ABBREVIATIONS

ACEi, angiotensin-converting enzyme inhibitor; **ARB,** angiotensin receptor blocker; **CR,** complete proteinuria remission; **COVID-19**, coronavirus disease 2019;l **DEARA,** dual endothelin angiotensin receptor antagonist; **eGFR,** estimated glomerular filtration rate; **HCP,** healthcare provider; **IgAN,** immunoglobulin A nephropathy; **IST,** immunosuppressive treatment; **LFT,** liver function test; **NS,** not specified; **SGLT2i,** sodium-glucose cotransporter-2 inhibitor; **SPAR,** sparsentan; **TRF,** targeted release formulation; **UPCR,** urine protein-to-creatinine ratio.

DISCLOSURES

LEV reports honoraria from Travere Therapeutics, Inc. and Otsuka America Pharmaceutical speaker bureaus. **RS** and **PG** report no financial disclosures related to this case series. **APB** and **GC** are employees and stockholders of Travere Therapeutics, Inc.

ACKNOWLEDGMENTS

This case series was supported by Travere Therapeutics, Inc.
Medical writing assistance and editorial support were provided under the direction of the authors by Marina Dragovic, MRes, and Ari Simenauer, of Nucleus Global, an Inizio company, in accordance with Good Publication Practice 2022 guidelines, and were funded by Travere Therapeutics, Inc.

REFERENCES

1. Kohan DE, et al. *Clin Sci (Lond).* 2024;138(11):645-662. **2.** Trachtman H, et al. *Expert Rev Clin Immunol*. 2024;20(6):571-576. **3.** FILSPARI (sparsentan) Prescribing Information. September 2024. Travere Therapeutics, Inc. San Diego, CA, USA. **4.** FILSPARI (sparsentan) SmPC. April 2024. CSL Vifor. Paris, France. **5.** Kidney Disease: Improving Global Outcomes (KDIGO) Glomerular Diseases Work Group. *Kidney Int.* 2021;100(4S):S1-S276.

Contact information: Luis E. Vélez, levelez@texaskidneycare.com

Presented at the American Society of Nephrology (ASN) Kidney Week 2024: October 23-27, 2024; San Diego, CA, USA